Impact of Patient Race on Insurance Payor Switch Rate, Healthcare Resource Utilization, and Cost Among Adult Patients with Schizophrenia in the United States: An Analysis of RWD Insights All-Payer Claims Database

Keshia Maughn[1], Mosadoluwa Afolabi[1], Abimbola Michael-Asalu[1], Ishveen Chopra[1]
[1]STATinMED, Dallas, TX, USA

STATinMED
The Authority in RWE

Objectives

This study aimed to describe insurance payor switch rates by race, differences in household income, healthcare resource utilization (HCRU), and cost for patients who switched insurance, stratified by race.

Background

Behavioral health issues affect a considerable number of people in the United States and are notably common among people with low incomes. Patients access to care and treatment may change, transitioning from commercial insurance to Medicaid.

Medicaid plays a major role in financing behavioral health care and providing access to services for people who need them. In 2015, 21% of adults with mental illness, and 26% of adults with serious mental illness had Medicaid insurance. Compared to other insurance types, 14% of the overall adult population, and in total, approximately 9.1 million adults with Medicaid had a mental illness.[1]

In addition to psychiatric hospital visits, Medicaid is more likely than other insurance types to cover additional comprehensive services, including medication management, individual and group therapy, psychosocial rehabilitation, and case management.[2]

Methods

This is a retrospective assessment of the STATinMED RWD Insights all-payer medical and pharmacy claims data population with insight into approximately 80% of the US health care system.

Patients were included if they met the following criteria: ≥18 years of age; ≥1 inpatient or ≥2 outpatient diagnosis claims for schizophrenia (ICD-9-CM: 295.0x-295.9x excluding 295.7x [schizoaffective disorder]; ICD-10-CM: F20.XX) during the identification period (01JAN2015 - 30JUN2020); the earliest such claim was defined as the initial diagnosis date (IDD); claim(s) for any antipsychotic after the IDD; enrollment in a commercial plan on the IDD; had 12 months continuous enrollment pre- and post-IDD; and had race information available.

Patients were defined as switchers if they changed insurance payor to Medicaid after the IDD (first Medicaid claim date = index date). Switchers were further required to have 12 months of continuous enrollment pre- and post-switch date.

Patients with evidence of Medicare Fee-For-Service or Advantage coverage during the study period and those with <12 months pre- and post-switch date were excluded.

Results

Black patients were significantly more likely than their Asian and White counterparts to switch from commercial to Medicaid.

Mean household income among switchers was $27,655 (SD=$35,688) for Black, $35,912 (SD=$41,146) for White, and $43,538 (SD=$37,853) for Asian patients. Black patients had the lowest mean income among switchers and non-switchers, vs other races (Figure 2).

Figure 1. Medicaid Switch Distribution by Race

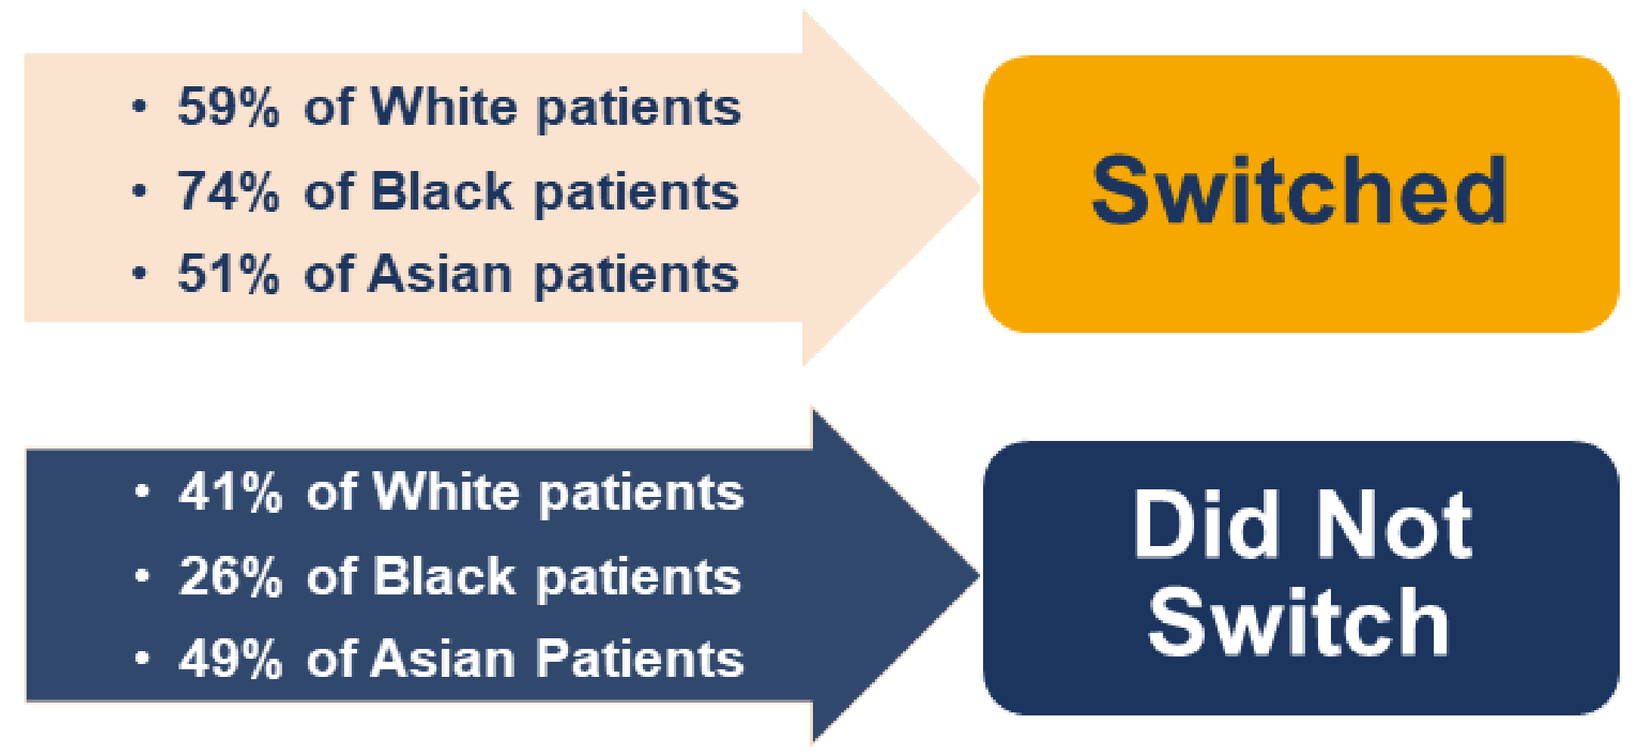
• 59% of White patients
• 74% of Black patients
• 51% of Asian patients
Switched
• 41% of White patients
• 26% of Black patients
• 49% of Asian Patients
Did Not Switch

Figure 2. Mean Income by Race

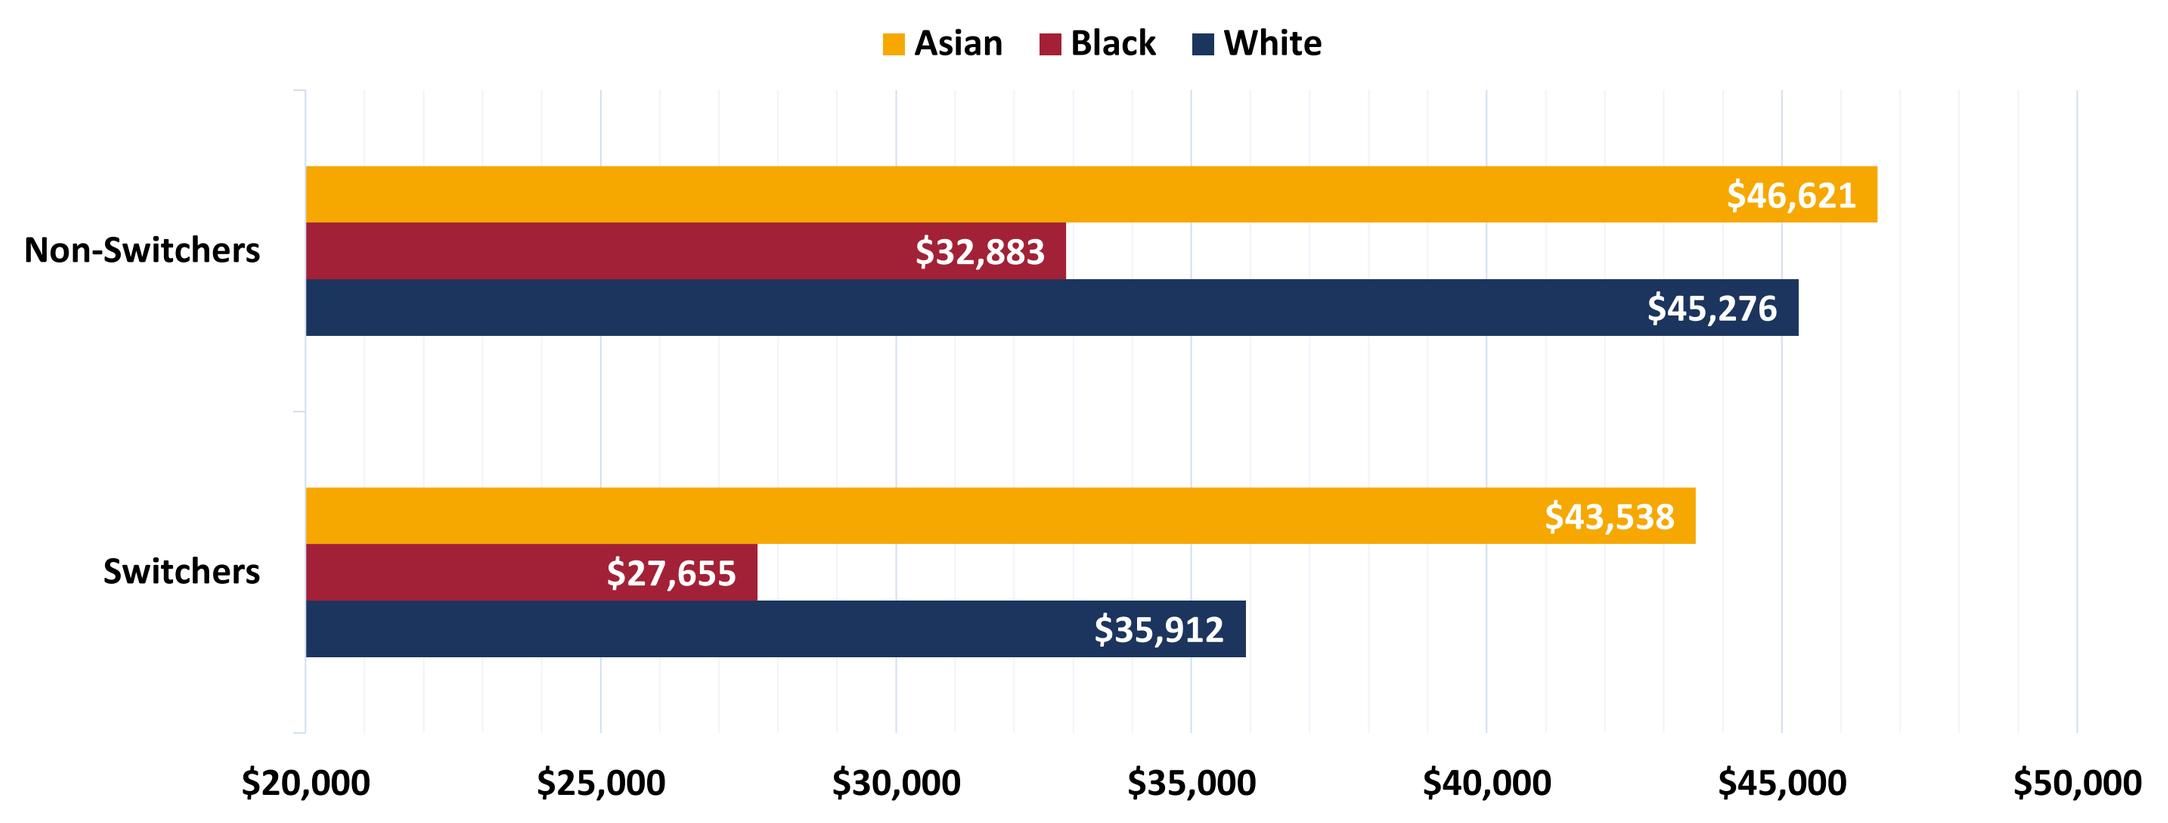
Asian Black White
Non-Switchers
$46,621
$32,883
$45,276
Switchers
$43,538
$27,655
$35,912
$20,000 $25,000 $30,000 $35,000 $40,000 $45,000 $50,000

Results - cont'd

Total mean costs decreased for White ($17,848 vs $17,596) and Black patients ($15,754 vs $15,511) and increased for Asian patients ($12,578 vs $14,164) pre- and post-switch. Pharmacy costs accounted for the majority of the total health care costs across all races. Inpatient stay costs decreased and pharmacy costs increased across all races (Figure 3).

Black patients who switched had a higher rate of all-cause inpatient stays pre-and-post switch. After commercial-to-Medicaid switch, all-cause inpatient stays increased for all groups; highest changes observed: Black (pre-switch=57% vs post-switch=66%; %Diff=9%), White (55% vs 61%; %Diff=6%), and Asian patients (46% vs 51%; %Diff=5%).

Length of hospital stay and number of inpatient stays increased post switch for all race categories and especially for Asian patients (Figures 4a-4c).

Figure 3. Mean Cost: Total, Pharmacy, and Inpatient by Race

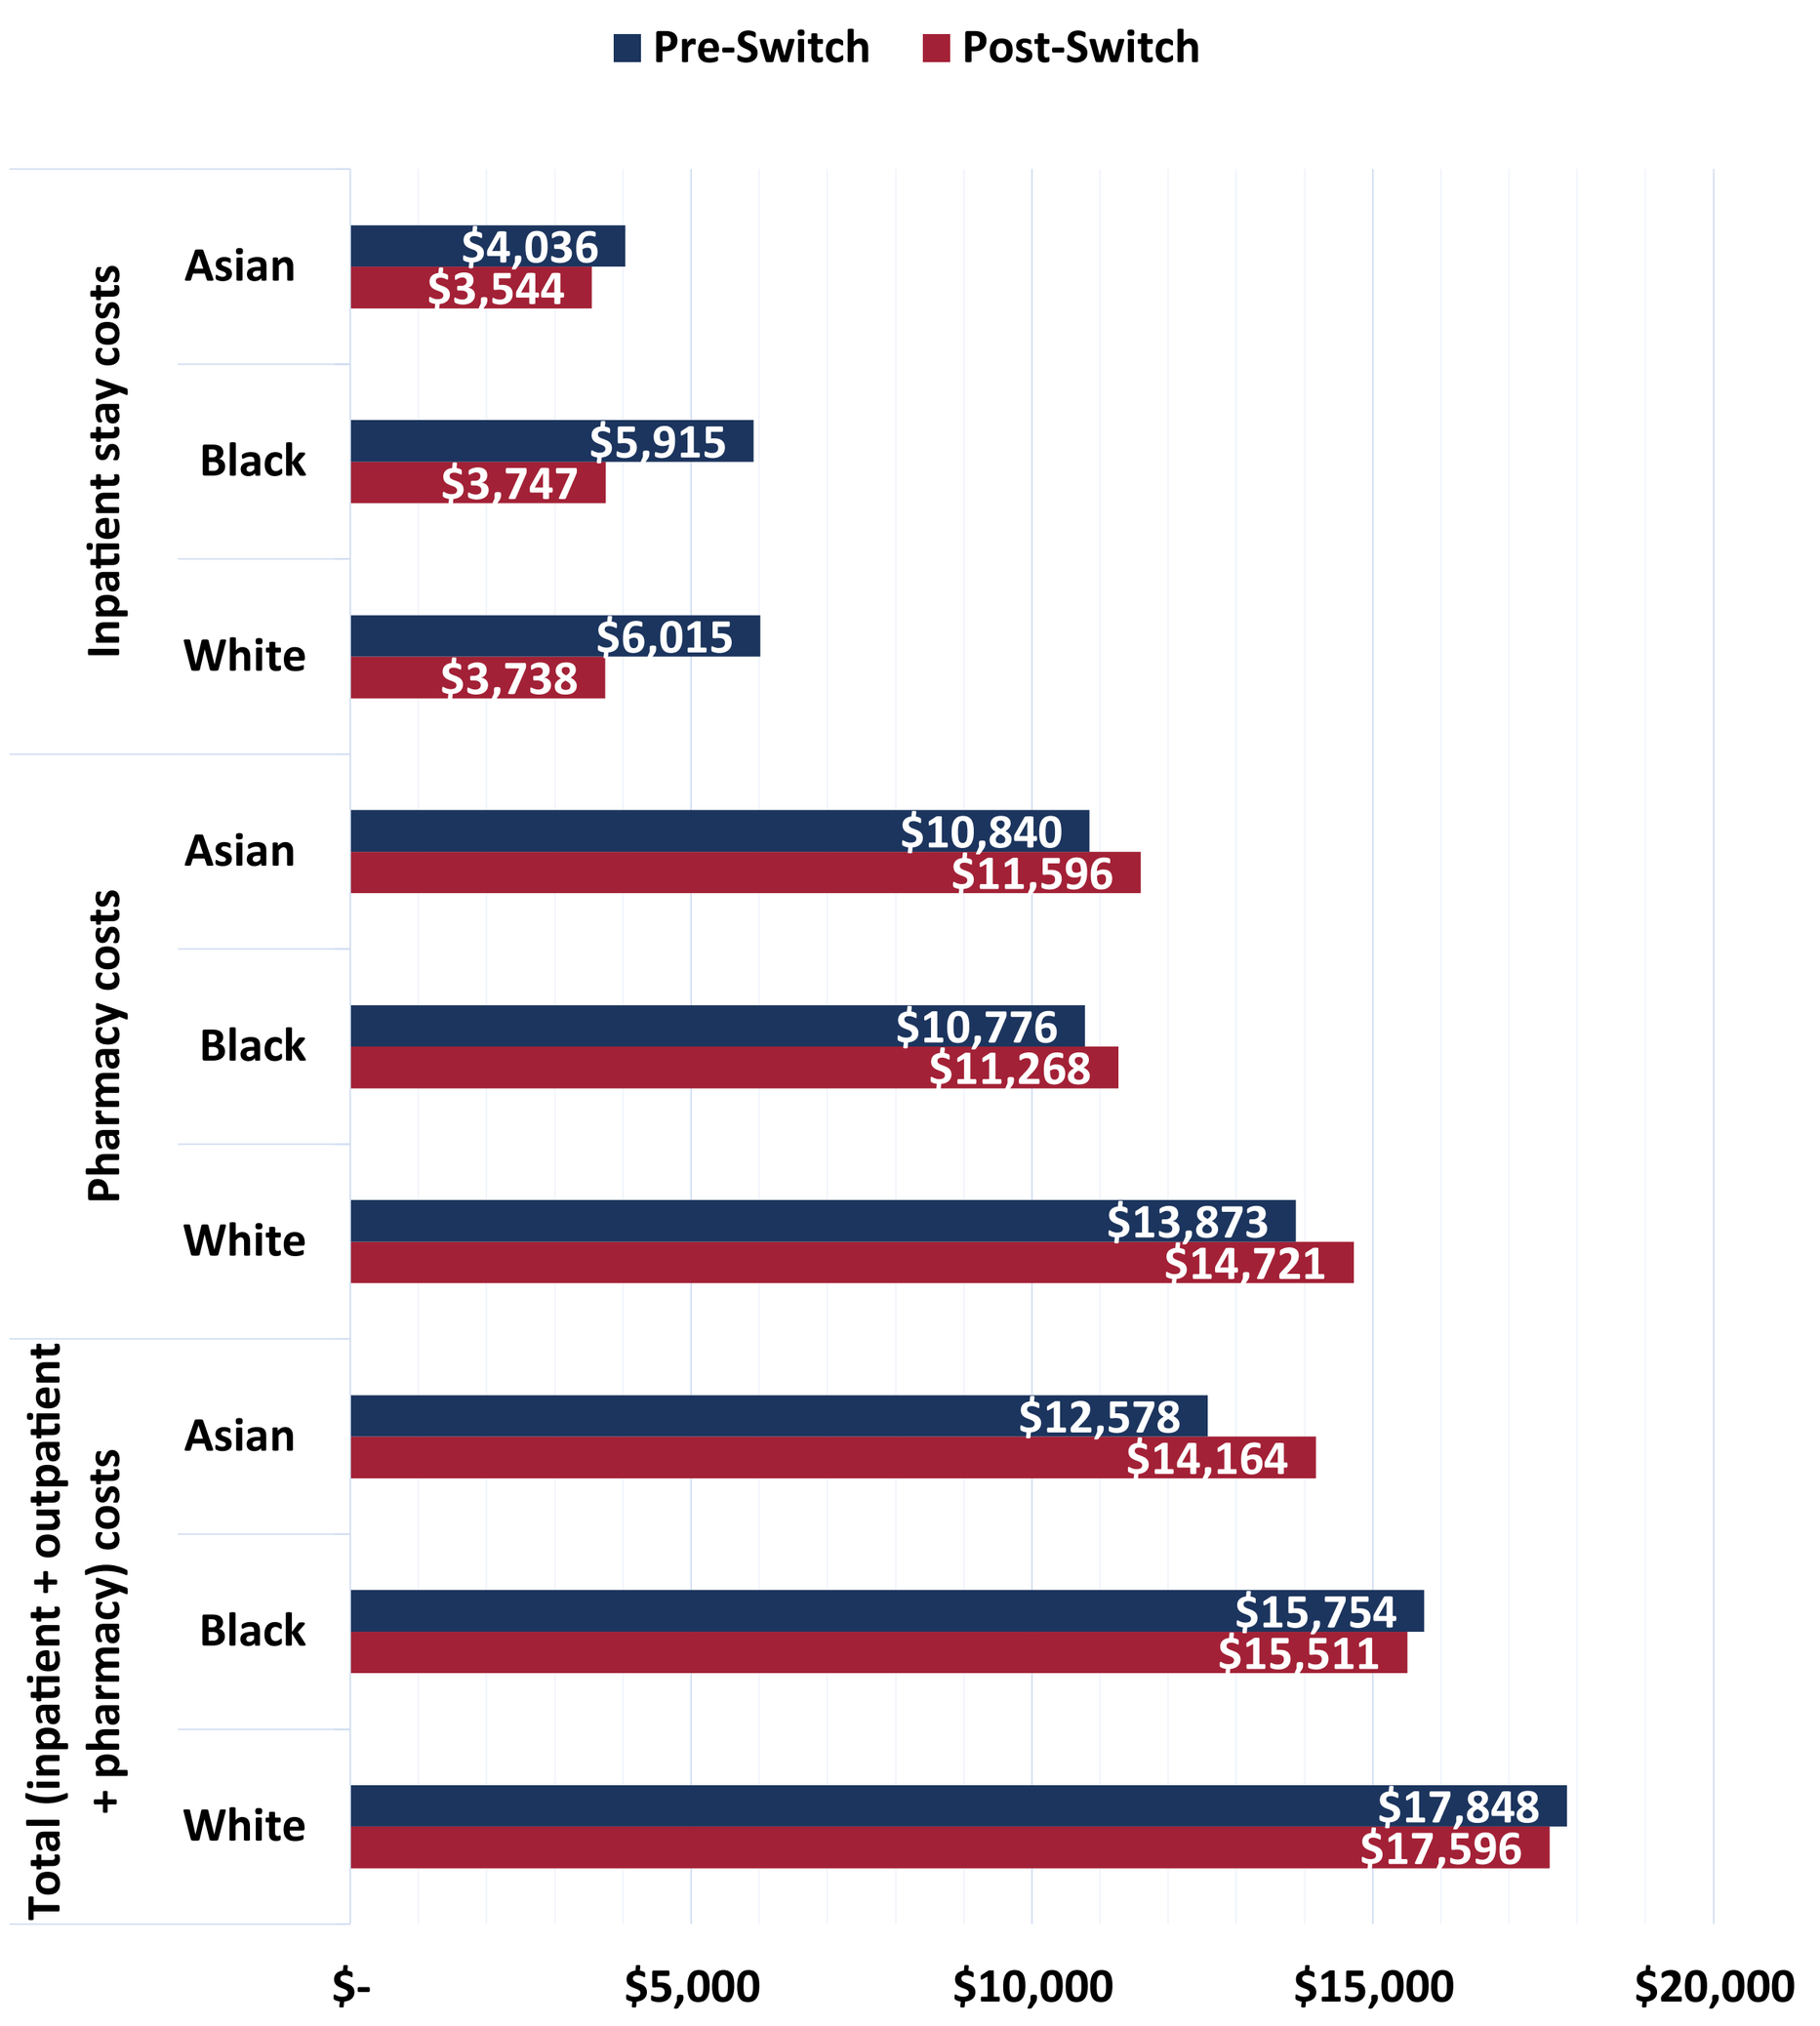
Pre-Switch Post-Switch
Inpatient stay costs
Asian $4,036 $3,544
Black $5,915 $3,747
White $6,015 $3,738
Pharmacy costs
Asian $10,840 $11,596
Black $10,776 $11,268
White $13,873 $14,721
Total (inpatient + outpatient + pharmacy) costs
Asian $12,578 $14,164
Black $15,754 $15,511
White $17,848 $17,596
$- $5,000 $10,000 $15,000 $20,000

Figure 4a. Inpatient Number of Stays by Race Pre/Post Switch

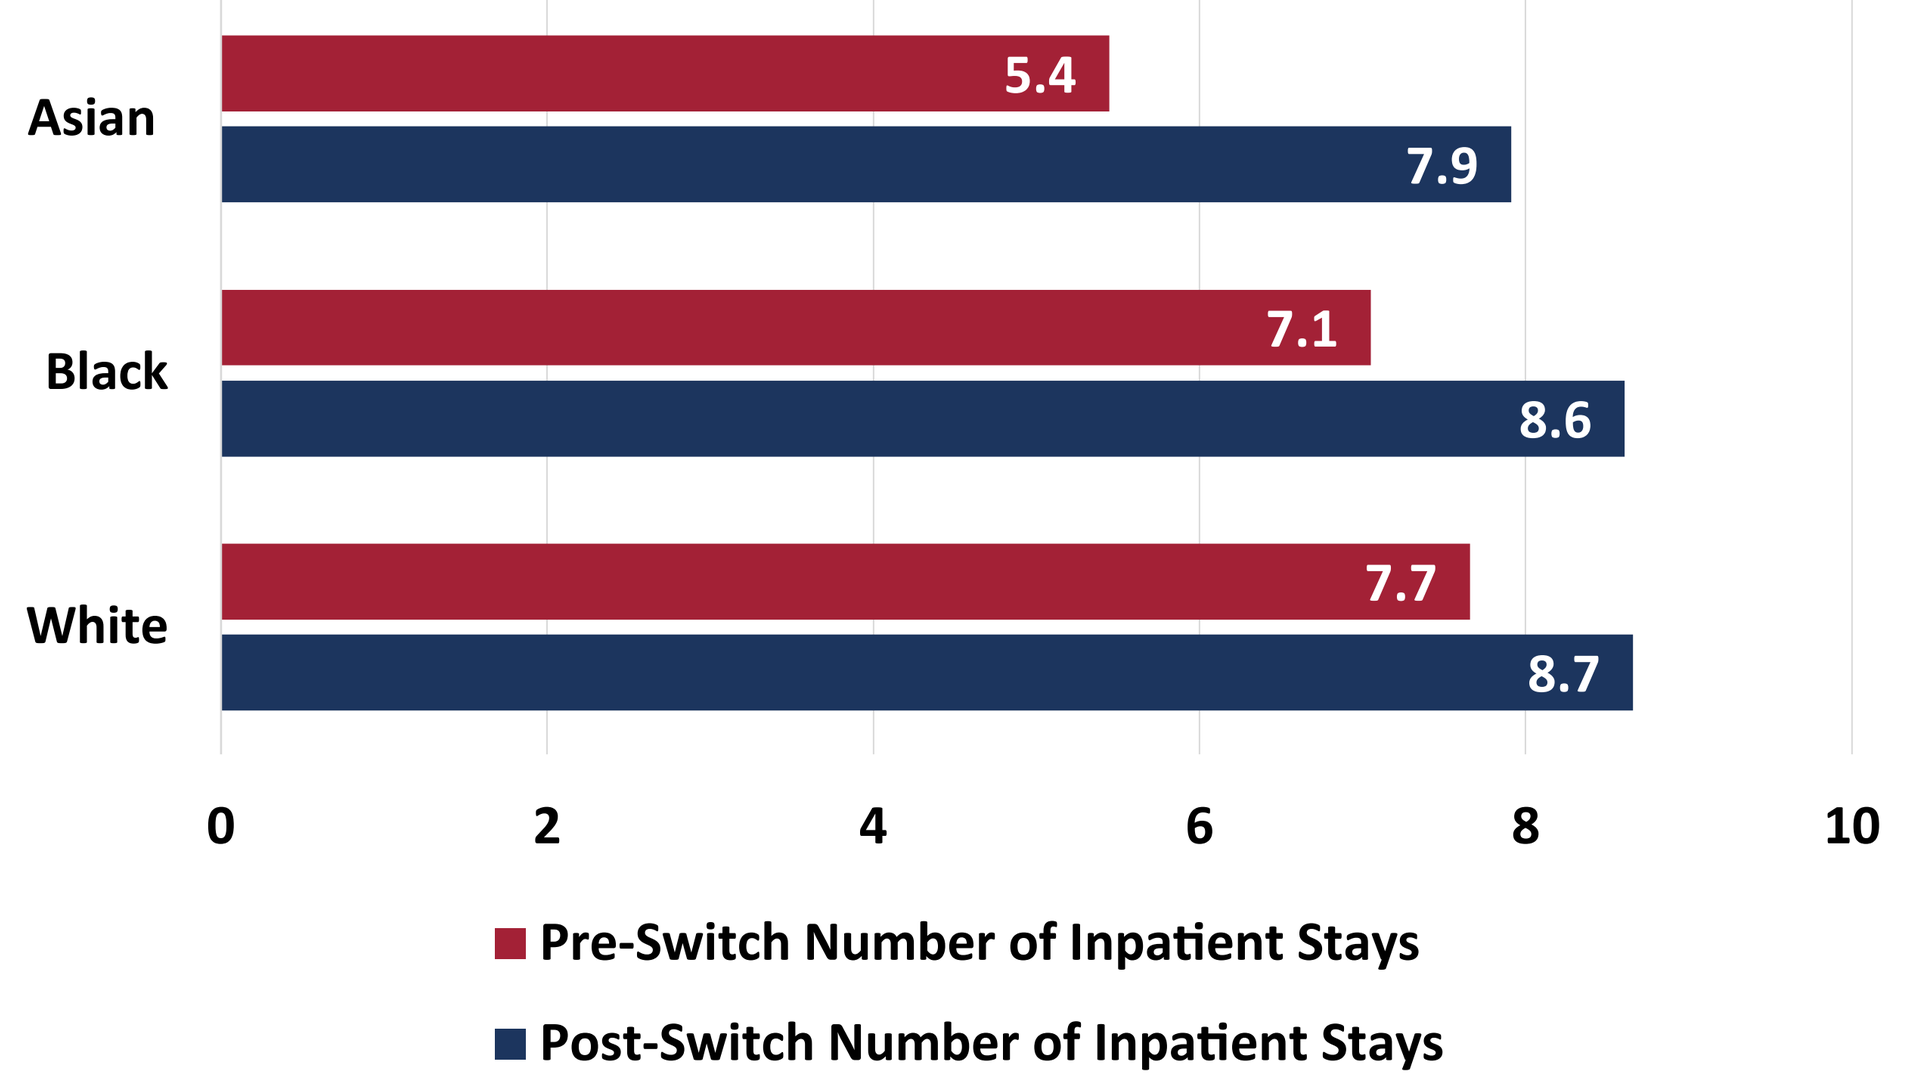
Asian 5.4 7.9
Black 7.1 8.6
White 7.7 8.7
0 2 4 6 8 10
Pre-Switch Number of Inpatient Stays
Post-Switch Number of Inpatient Stays

Figure 4b. Inpatient Length of Stay by Race Pre/Post Switch

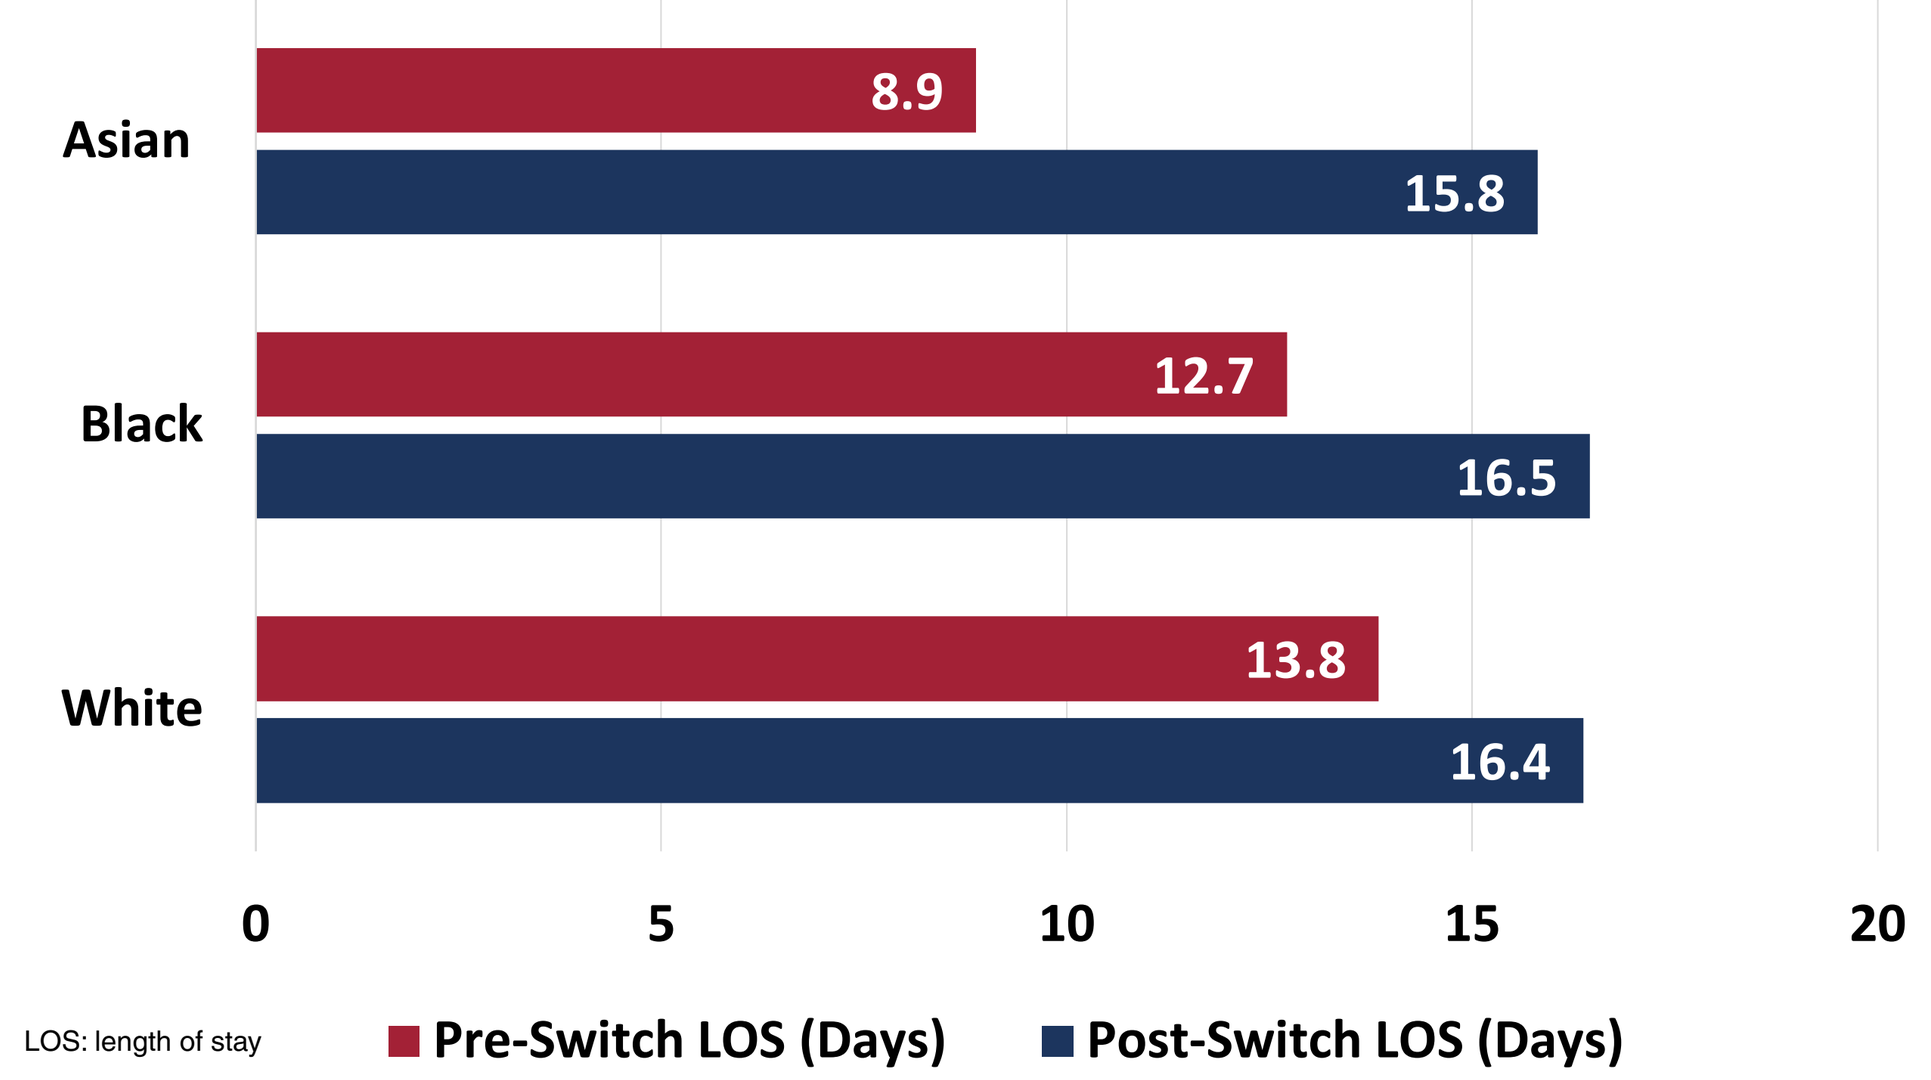
Asian 8.9 15.8
Black 12.7 16.5
White 13.8 16.4
0 5 10 15 20
LOS: length of stay
Pre-Switch LOS (Days)
Post-Switch LOS (Days)

Figure 4c. Patients with ≥1 Inpatient Stay Pre/Post Switch

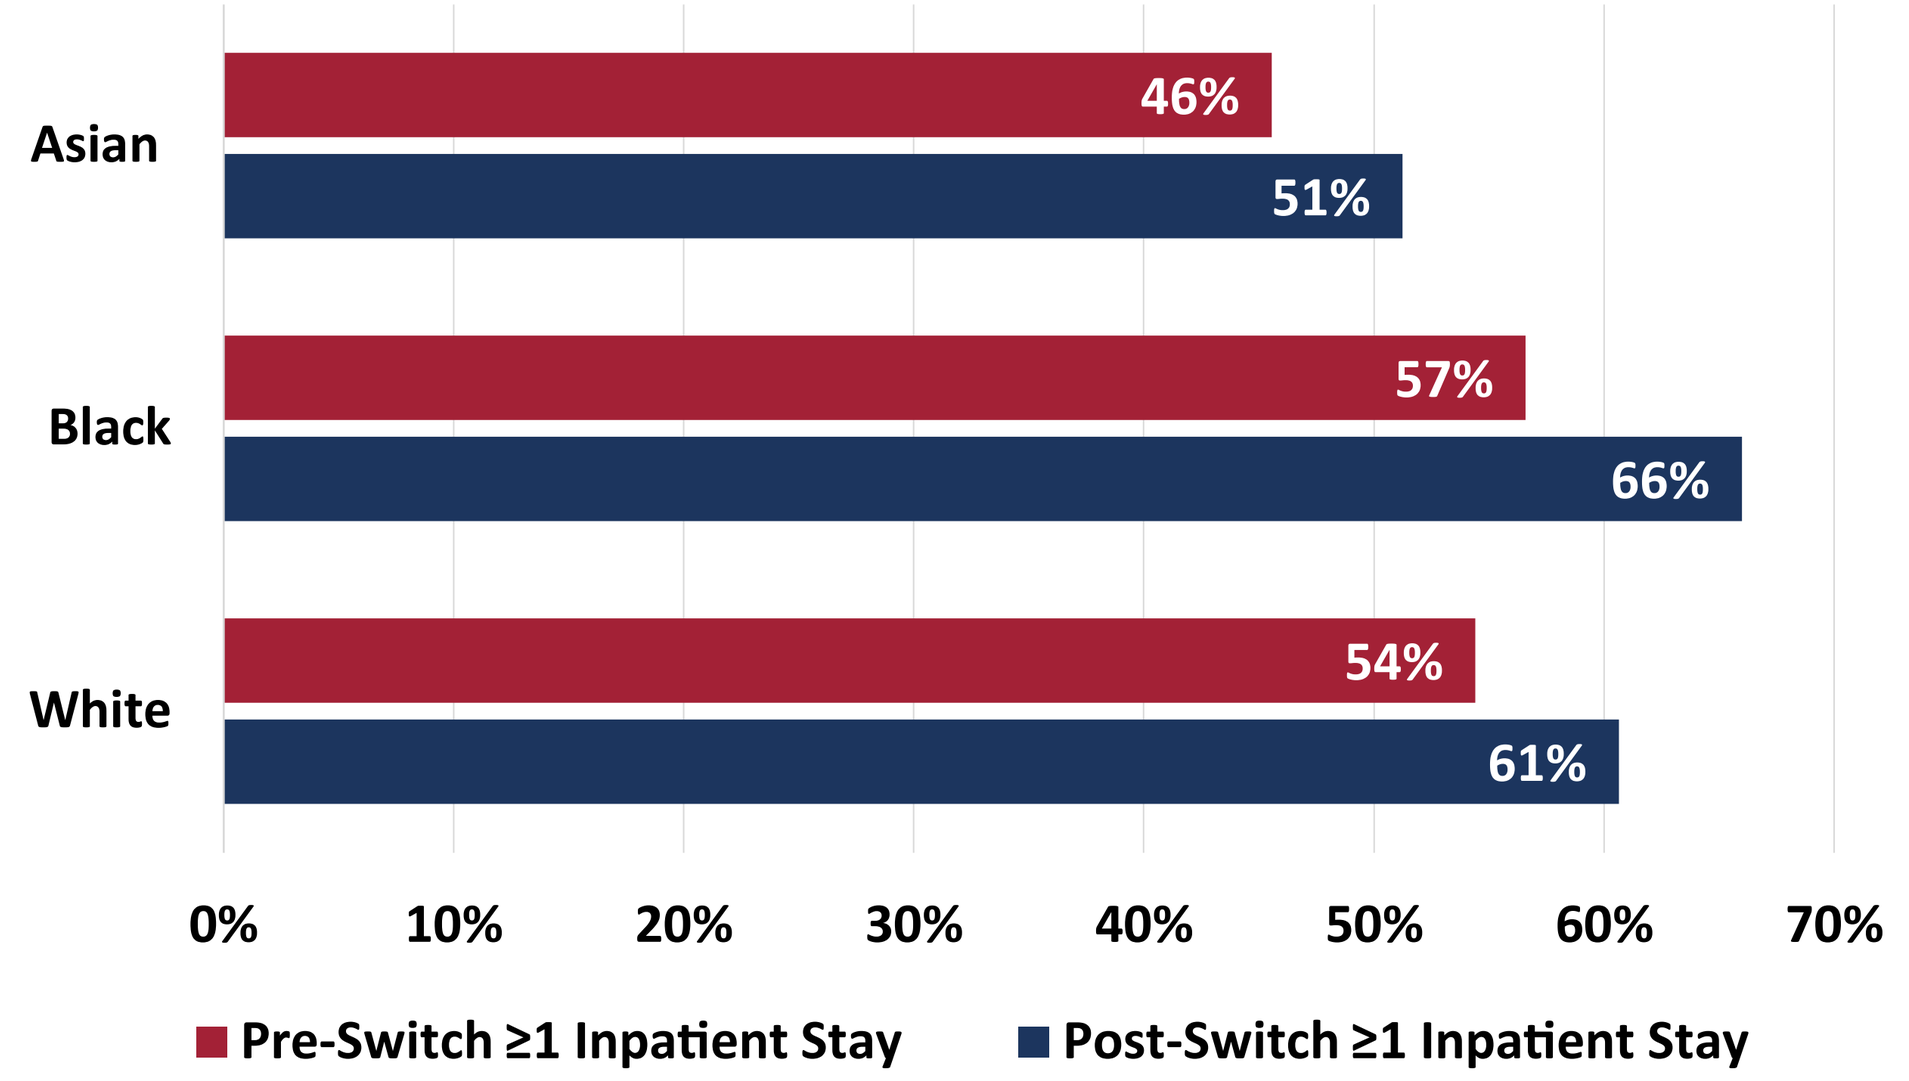
Asian 46% 51%
Black 57% 66%
White 54% 61%
0% 10% 20% 30% 40% 50% 60% 70%
Pre-Switch ≥1 Inpatient Stay
Post-Switch ≥1 Inpatient Stay

After switching, pharmacy visits increased the most for Asian (17.7 vs 19.4; Diff=1.7) vs White (22.5 vs 24.0; Diff=1.5) and Black patients (17.7 vs 18.1; Diff=0.4) (Figures 5a and 5b).

Figure 5a. Patients with Pharmacy Visits by Race Pre/Post Switch

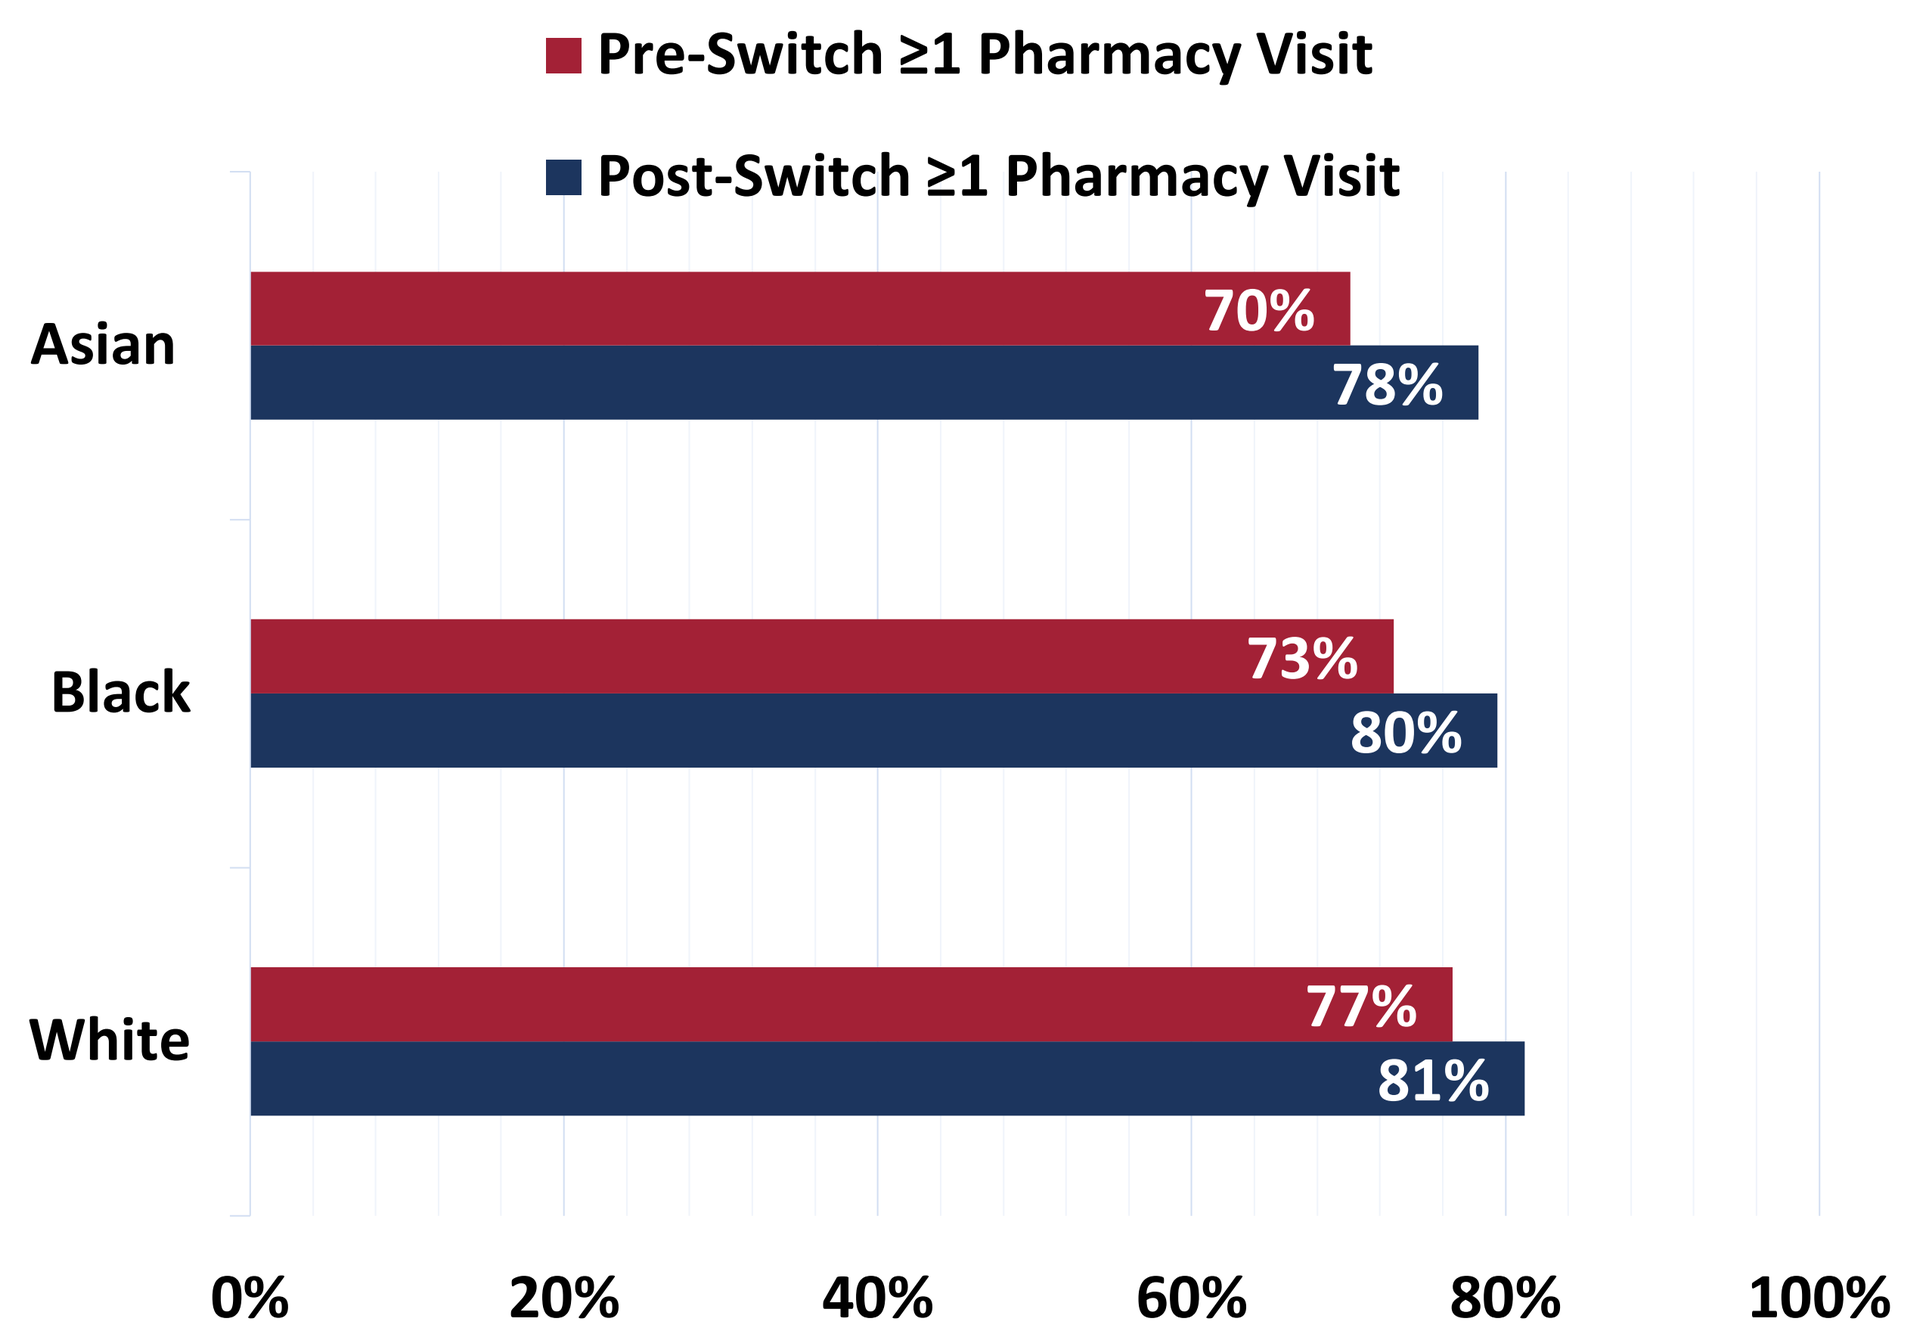
Pre-Switch ≥1 Pharmacy Visit
Post-Switch ≥1 Pharmacy Visit
Asian 70% 78%
Black 73% 80%
White 77% 81%
0% 20% 40% 60% 80% 100%

Figure 5b. Number of Pharmacy Visits by Race Pre/Post Switch

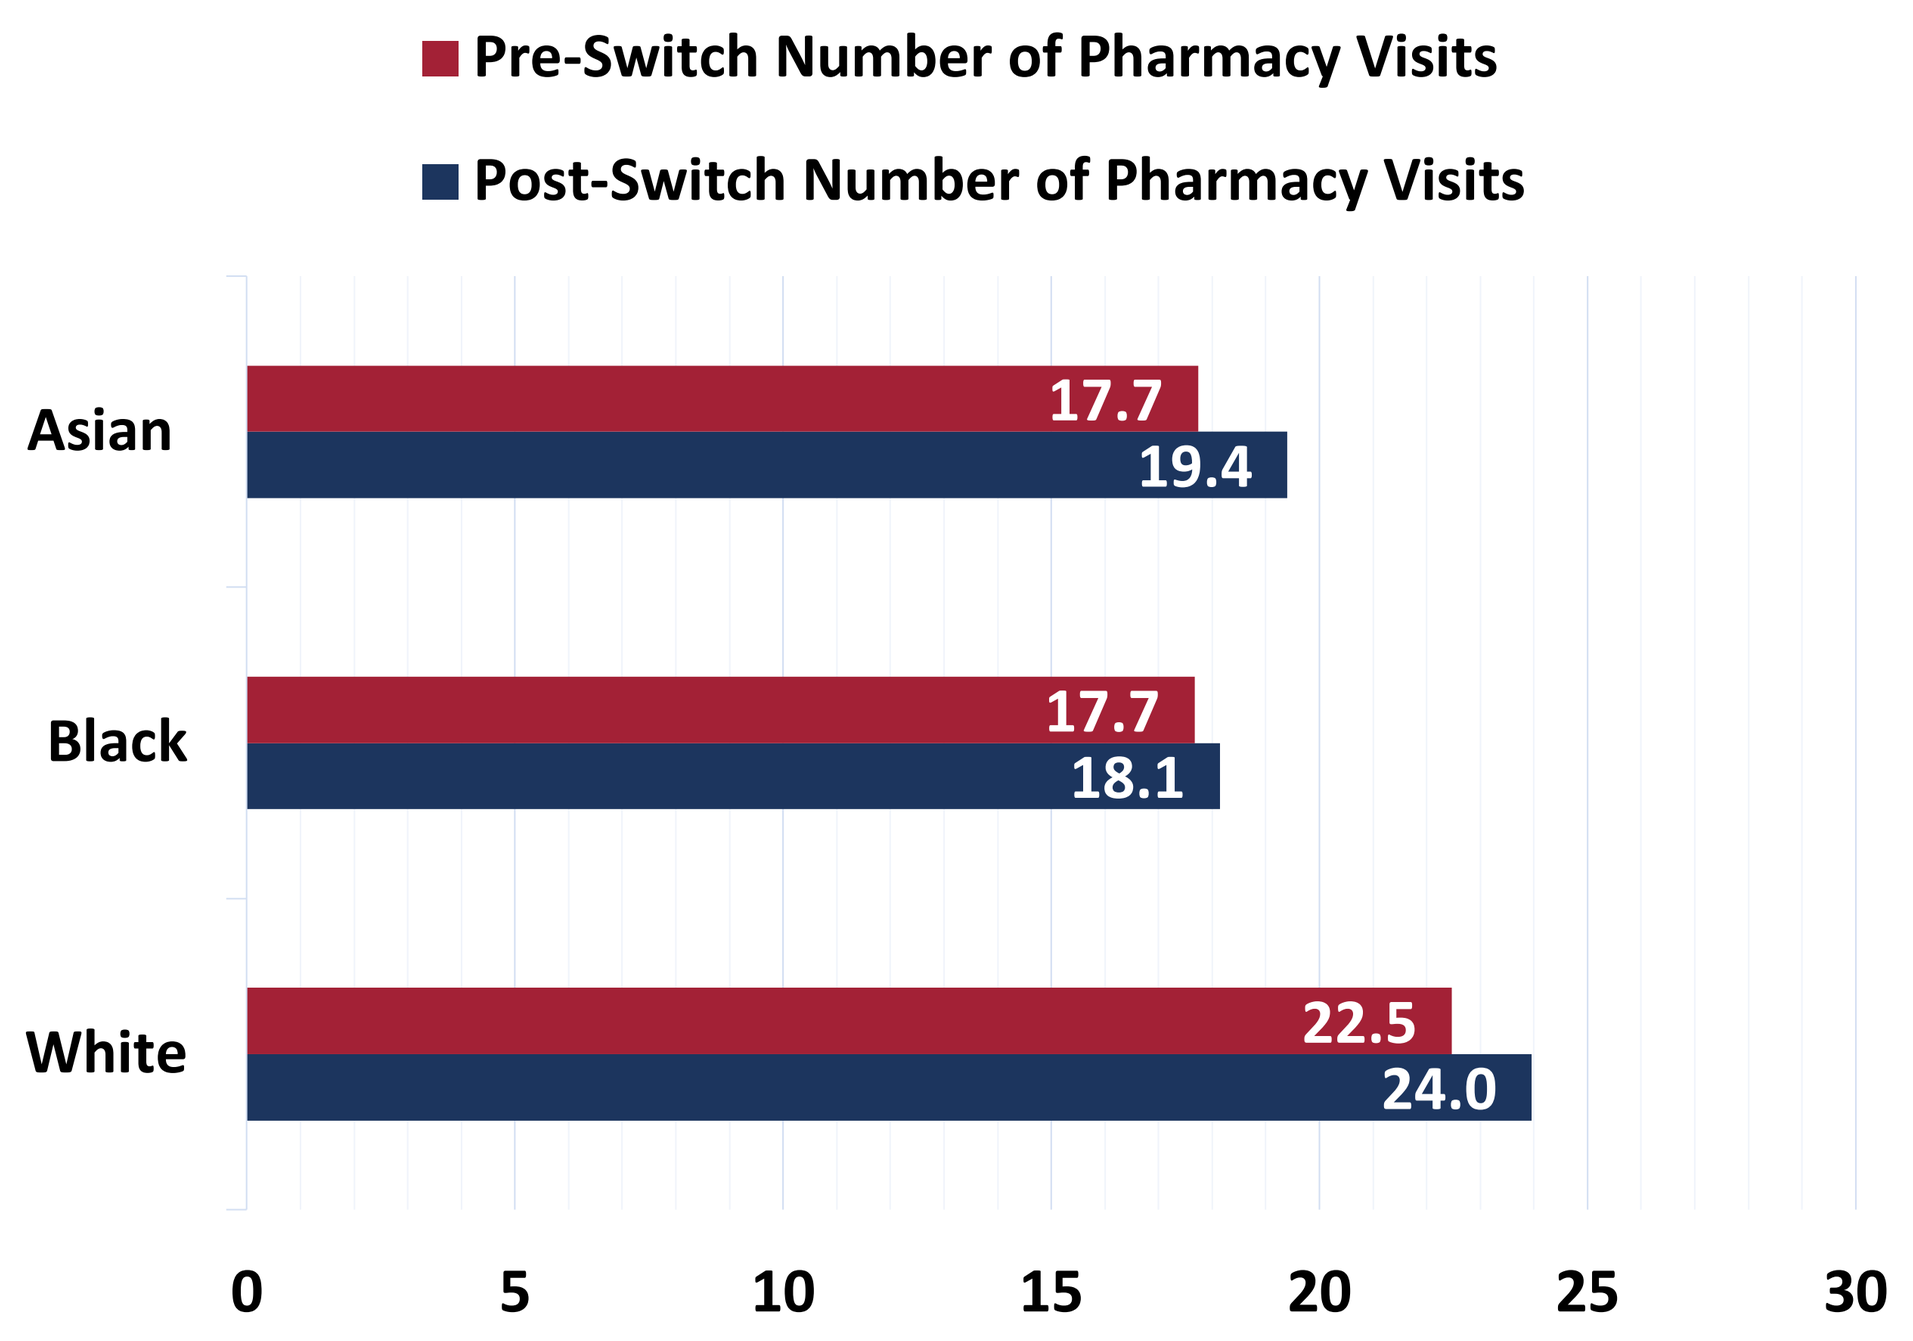
Pre-Switch Number of Pharmacy Visits
Post-Switch Number of Pharmacy Visits
Asian 17.7 19.4
Black 17.7 18.1
White 22.5 24.0
0 5 10 15 20 25 30

Limitations

In this study, we linked all-payer claims data to other data sources; as race information is not readily available in the all-payer claims data, the information was collected from an aggregation of 5 sources of national consumer marketing data. The study sample size was low due to the characteristics of schizophrenia, including social and occupational dysfunction, which impact consuming habits of patients.

Conclusions

Black patients with schizophrenia were significantly more likely to switch to Medicaid compared to other patient groups. Total costs were flat post-switch for Black and White patients and increased for Asian patients, even though Asian patients had lower reimbursement rates. Despite coverage switches with favorable cost sharing, medication use among Black patients declined and hospitalization increased.

References

[1] Zur J, Musumeci M, Garfield R. Medicaid's role in financing behavioral health for low-income individuals. Kaiser Family Foundation Website. 2017. https://files.kff.org/attachment/Issue-Brief-Medicaids-Role-in-Financing-Behavioral-Health-Services-for-Low-Income-Individuals. Accessed September 13, 2022.

[2] Cannon K, Burton J, Musumeci M. Adult behavioral health benefits in Medicaid and the marketplace. 2015. Kaiser Family Foundation Website. https://www.kff.org/medicaid/report/adult-behavioral-health-benefits-in-medicaid-and-the-marketplace/. Accessed September 13, 2022.

Poster presented at *Academy of Managed Care Pharmacy (AMCP) Nexus 2022*; October 11-14, 2022; National Harbor, MD

publications@STATinMED.com
www.STATinMED.com

STATinMED
13101 Preston Rd, Suite 110 #3395
Dallas, TX 75240