Antifibrotic treatment prevalence, clinical outcomes, and economic burden among veterans with idiopathic pulmonary fibrosis in the United States

Poster 6969

Ying Qiu,[1] Julia Zhu,[1] Brandon Elpers,[1] Christopher Dieyi,[2] Clare Byrne,[2] Risho Singh,[2] Aryeh Fischer[1]

[1]Bristol Myers Squibb, Princeton, NJ; [2]STATinMED Research, Plano, TX

Introduction

- Idiopathic pulmonary fibrosis (IPF) is a progressive and fatal disease characterized by lung fibrosis, leading to loss of lung function and a poor clinical prognosis; the median estimated survival time after definitive IPF diagnosis is 2 to 5 years[1]
- In 2014, the antifibrotic therapies nintedanib and pirfenidone were approved for the treatment of IPF after they were shown to slow the decline in lung function in clinical trials[2,3]
 - Although these therapies have changed the treatment landscape, both are limited by a high side effect burden
- Few studies have examined the effect of antifibrotic therapies on real-world outcomes, particularly in the US Veterans Health Administration (VHA) population
 - Patients in the VHA system are predominantly male, elderly, and White; these characteristics align with some of the demographic risk factors for IPF[4]
- The overall aims of this study were:
 - To determine the percentage of patients with newly diagnosed IPF who received antifibrotic treatment over the study period
 - To compare clinical outcomes, healthcare resource utilization (HCRU), and costs between patients with IPF who received antifibrotic treatment (treated) and those who did not (untreated)

Methods

Study design

- This retrospective cohort study used deidentified claims data from the VHA, which is the largest integrated health system in the US, serving over 9 million veterans per year at 1,293 healthcare facilities[5]
- Eligible patients were adults with an initial IPF diagnosis claim (International Classification of Disease, Ninth Revision, Clinical Modification [ICD-9-CM]: 516.31; ICD-10-CM: J84.112) during the study period of October 1, 2014, to September 30, 2019; exclusion criteria included a medical claim for any other interstitial lung disease on or after the last IPF diagnosis date and any medical or prescription claims for IPF or antifibrotic therapies prior to the first IPF diagnosis date
- All included patients were further required to have continuous enrollment for ≥12 months prior to the index date (baseline period) and ≥12 months after the index date (follow-up period), unless the patient died within 12 months after the index date
- Patients who had a pharmacy claim for nintedanib or pirfenidone on or after their first IPF diagnosis date were assigned to the treated cohort, while those with no claim for either drug during the study period were assigned to the untreated cohort (**Figure 1**)
- For treated patients, the index date was the date of the first prescription for antifibrotic treatment; for untreated patients, the index date was randomly selected within the same identification window to reflect the randomness of index date assignment in the treated cohort and minimize selection bias

Figure 1. Study design

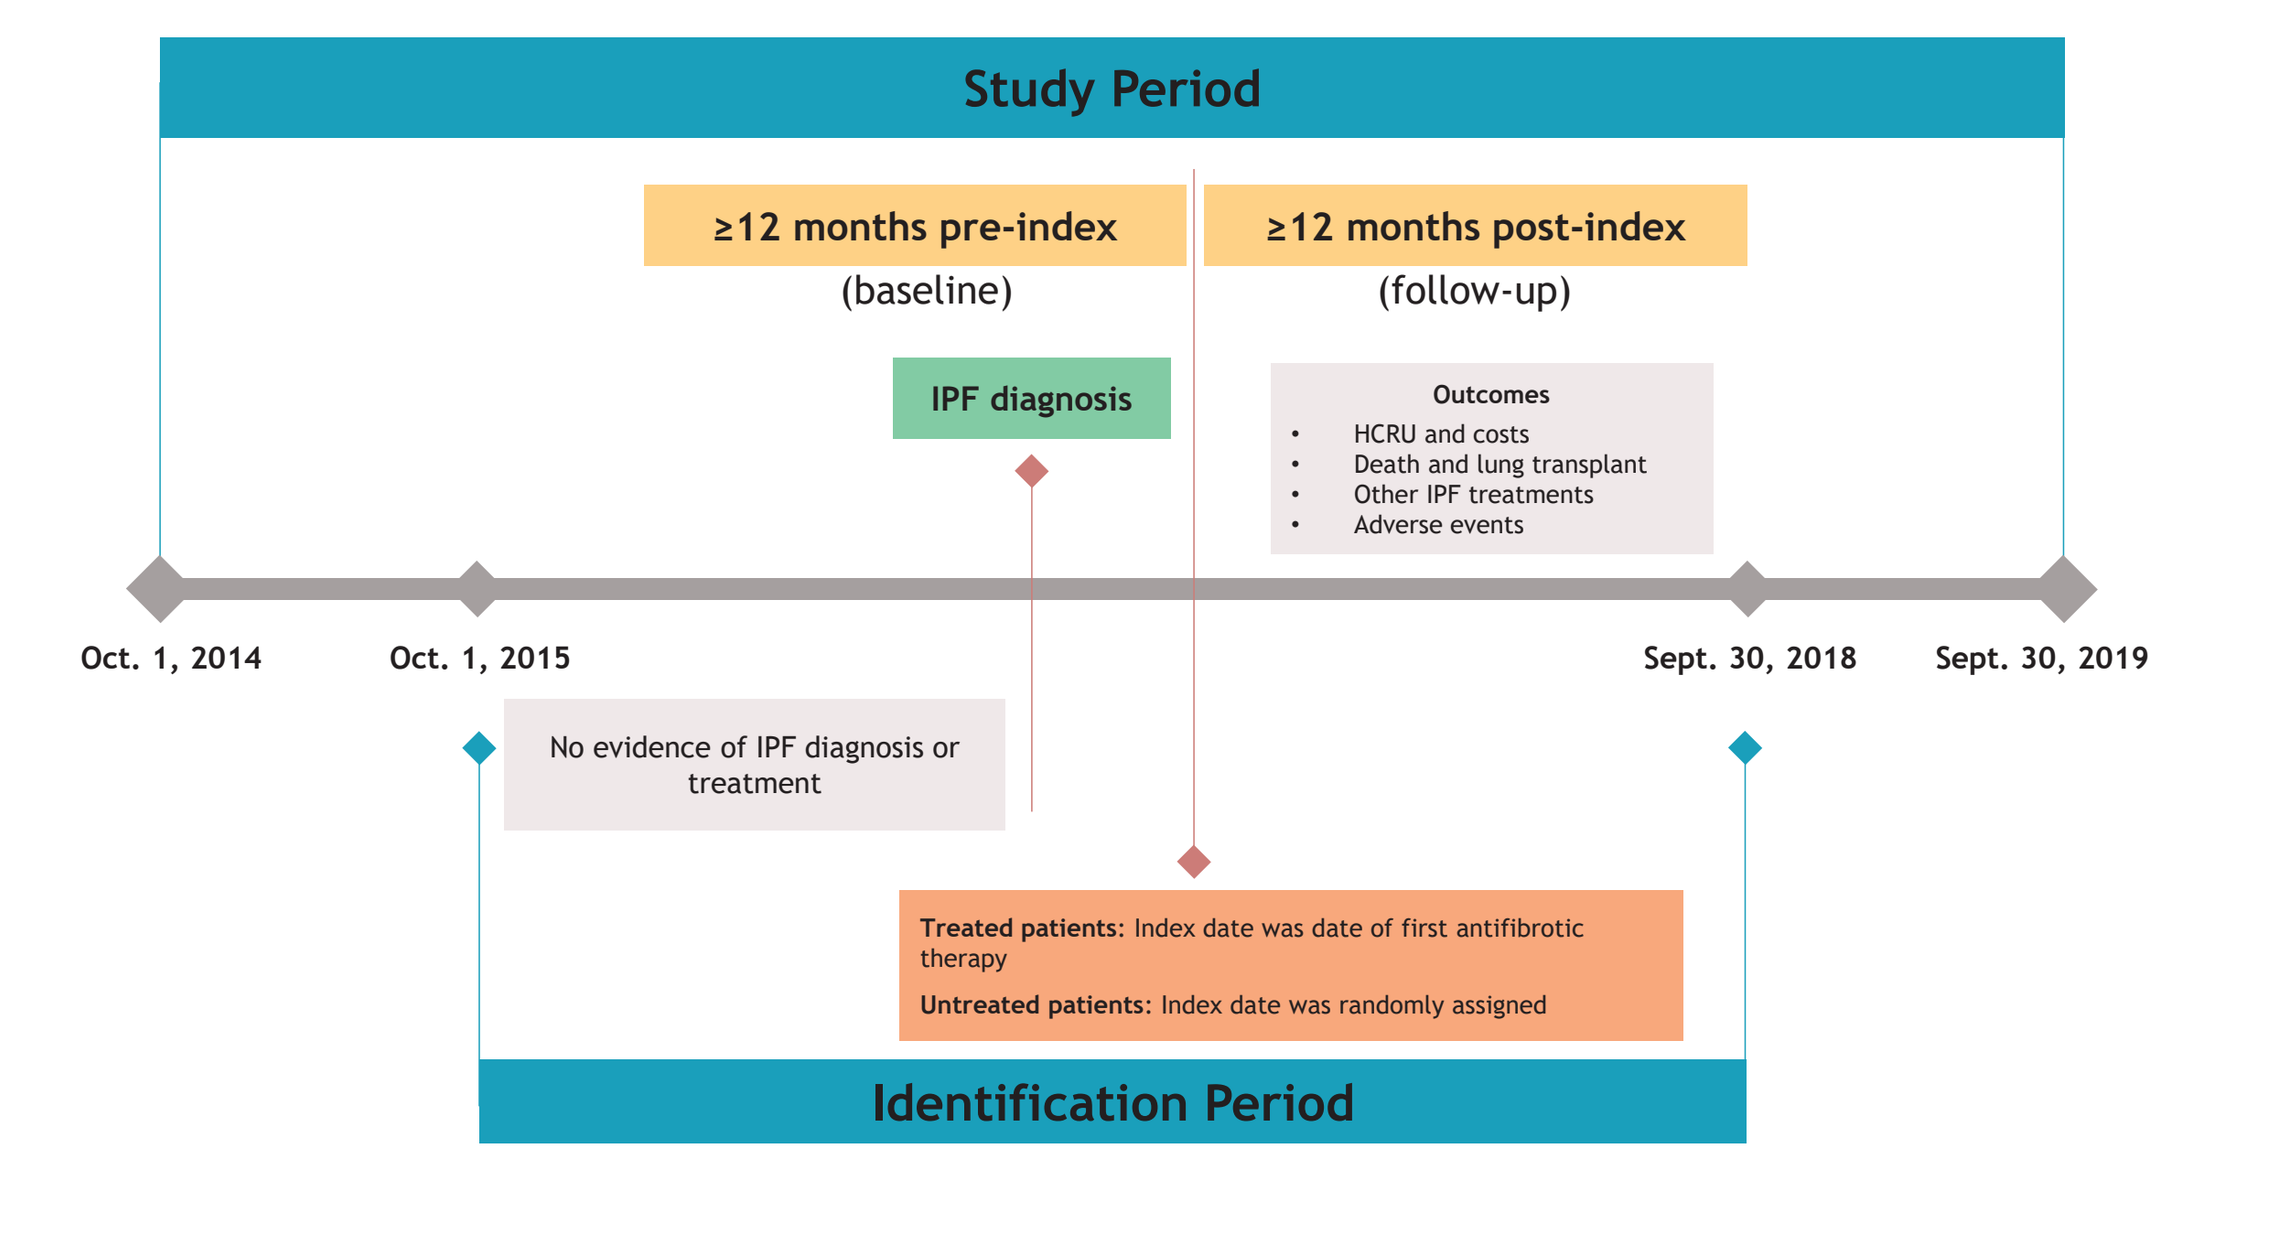
Study Period
≥12 months pre-index
(baseline)
≥12 months post-index
(follow-up)
IPF diagnosis
Outcomes
HCRU and costs
Death and lung transplant
Other IPF treatments
Adverse events
Oct. 1, 2014
Oct. 1, 2015
Sept. 30, 2018
Sept. 30, 2019
No evidence of IPF diagnosis or treatment
Treated patients: Index date was date of first antifibrotic therapy
Untreated patients: Index date was randomly assigned
Identification Period

HCRU, healthcare resource utilization; IPF, idiopathic pulmonary fibrosis.

Study variables

- Prevalence of antifibrotic therapy was calculated as the proportion of patients with IPF who had pharmacy claims for nintedanib or pirfenidone in each quarter of the study period
- Patient demographics (age, sex, race) and clinical characteristics (Charlson Comorbidity Index [CCI]) score, individual comorbidities, prior IPF treatments, smoking status, diagnostic testing, time from diagnosis to treatment, and HCRU and costs) were examined on the index date and during the baseline period, respectively
- HCRU and costs were also examined during the follow-up period on a per-patient-per-month basis to account for variable follow-up
- Mortality and time to death were examined over the follow-up period

Statistical analysis

Descriptive analysis

- Percentages and counts were provided for dichotomous and polychotomous variables
- Mean, SD, median, and interquartile range were provided for continuous variables
- Bivariate comparisons were performed using x^2 tests for dichotomous and polychotomous variables and Student *t* tests for continuous variables
- Kaplan-Meier curves were used to compare mortality rates in the 2 cohorts

Multivariate analysis

- HCRU and cost estimates were obtained using a generalized linear model; costs were inflated to 2019 US dollars using the annual Consumer Price Index medical expenditure category
- Risk of death was obtained using a Cox proportional hazards model; hazard ratios (HRs) and 95% CIs were acquired
- Both models were adjusted to account for differences in baseline patient characteristics; covariates included in the models were age, sex, race, CCI score, select comorbidities (ie, chronic obstructive pulmonary disease, lung cancer, gastroesophageal reflux disease [GERD], obstructive sleep apnea, and obesity), antacid and/or corticosteroid use, time from diagnosis to index date, and baseline use of inpatient, outpatient, and pharmacy services

Results

- In VHA enrollees, antifibrotic treatment prevalence increased sharply around the fourth quarter of 2014, corresponding to the time of US Food and Drug Administration approval (**Figure 2**)
- As of the third quarter of 2019, 71% of VHA enrollees with an IPF diagnosis were untreated

Figure 2. Prevalence of IPF antifibrotic therapy use in VHA patients

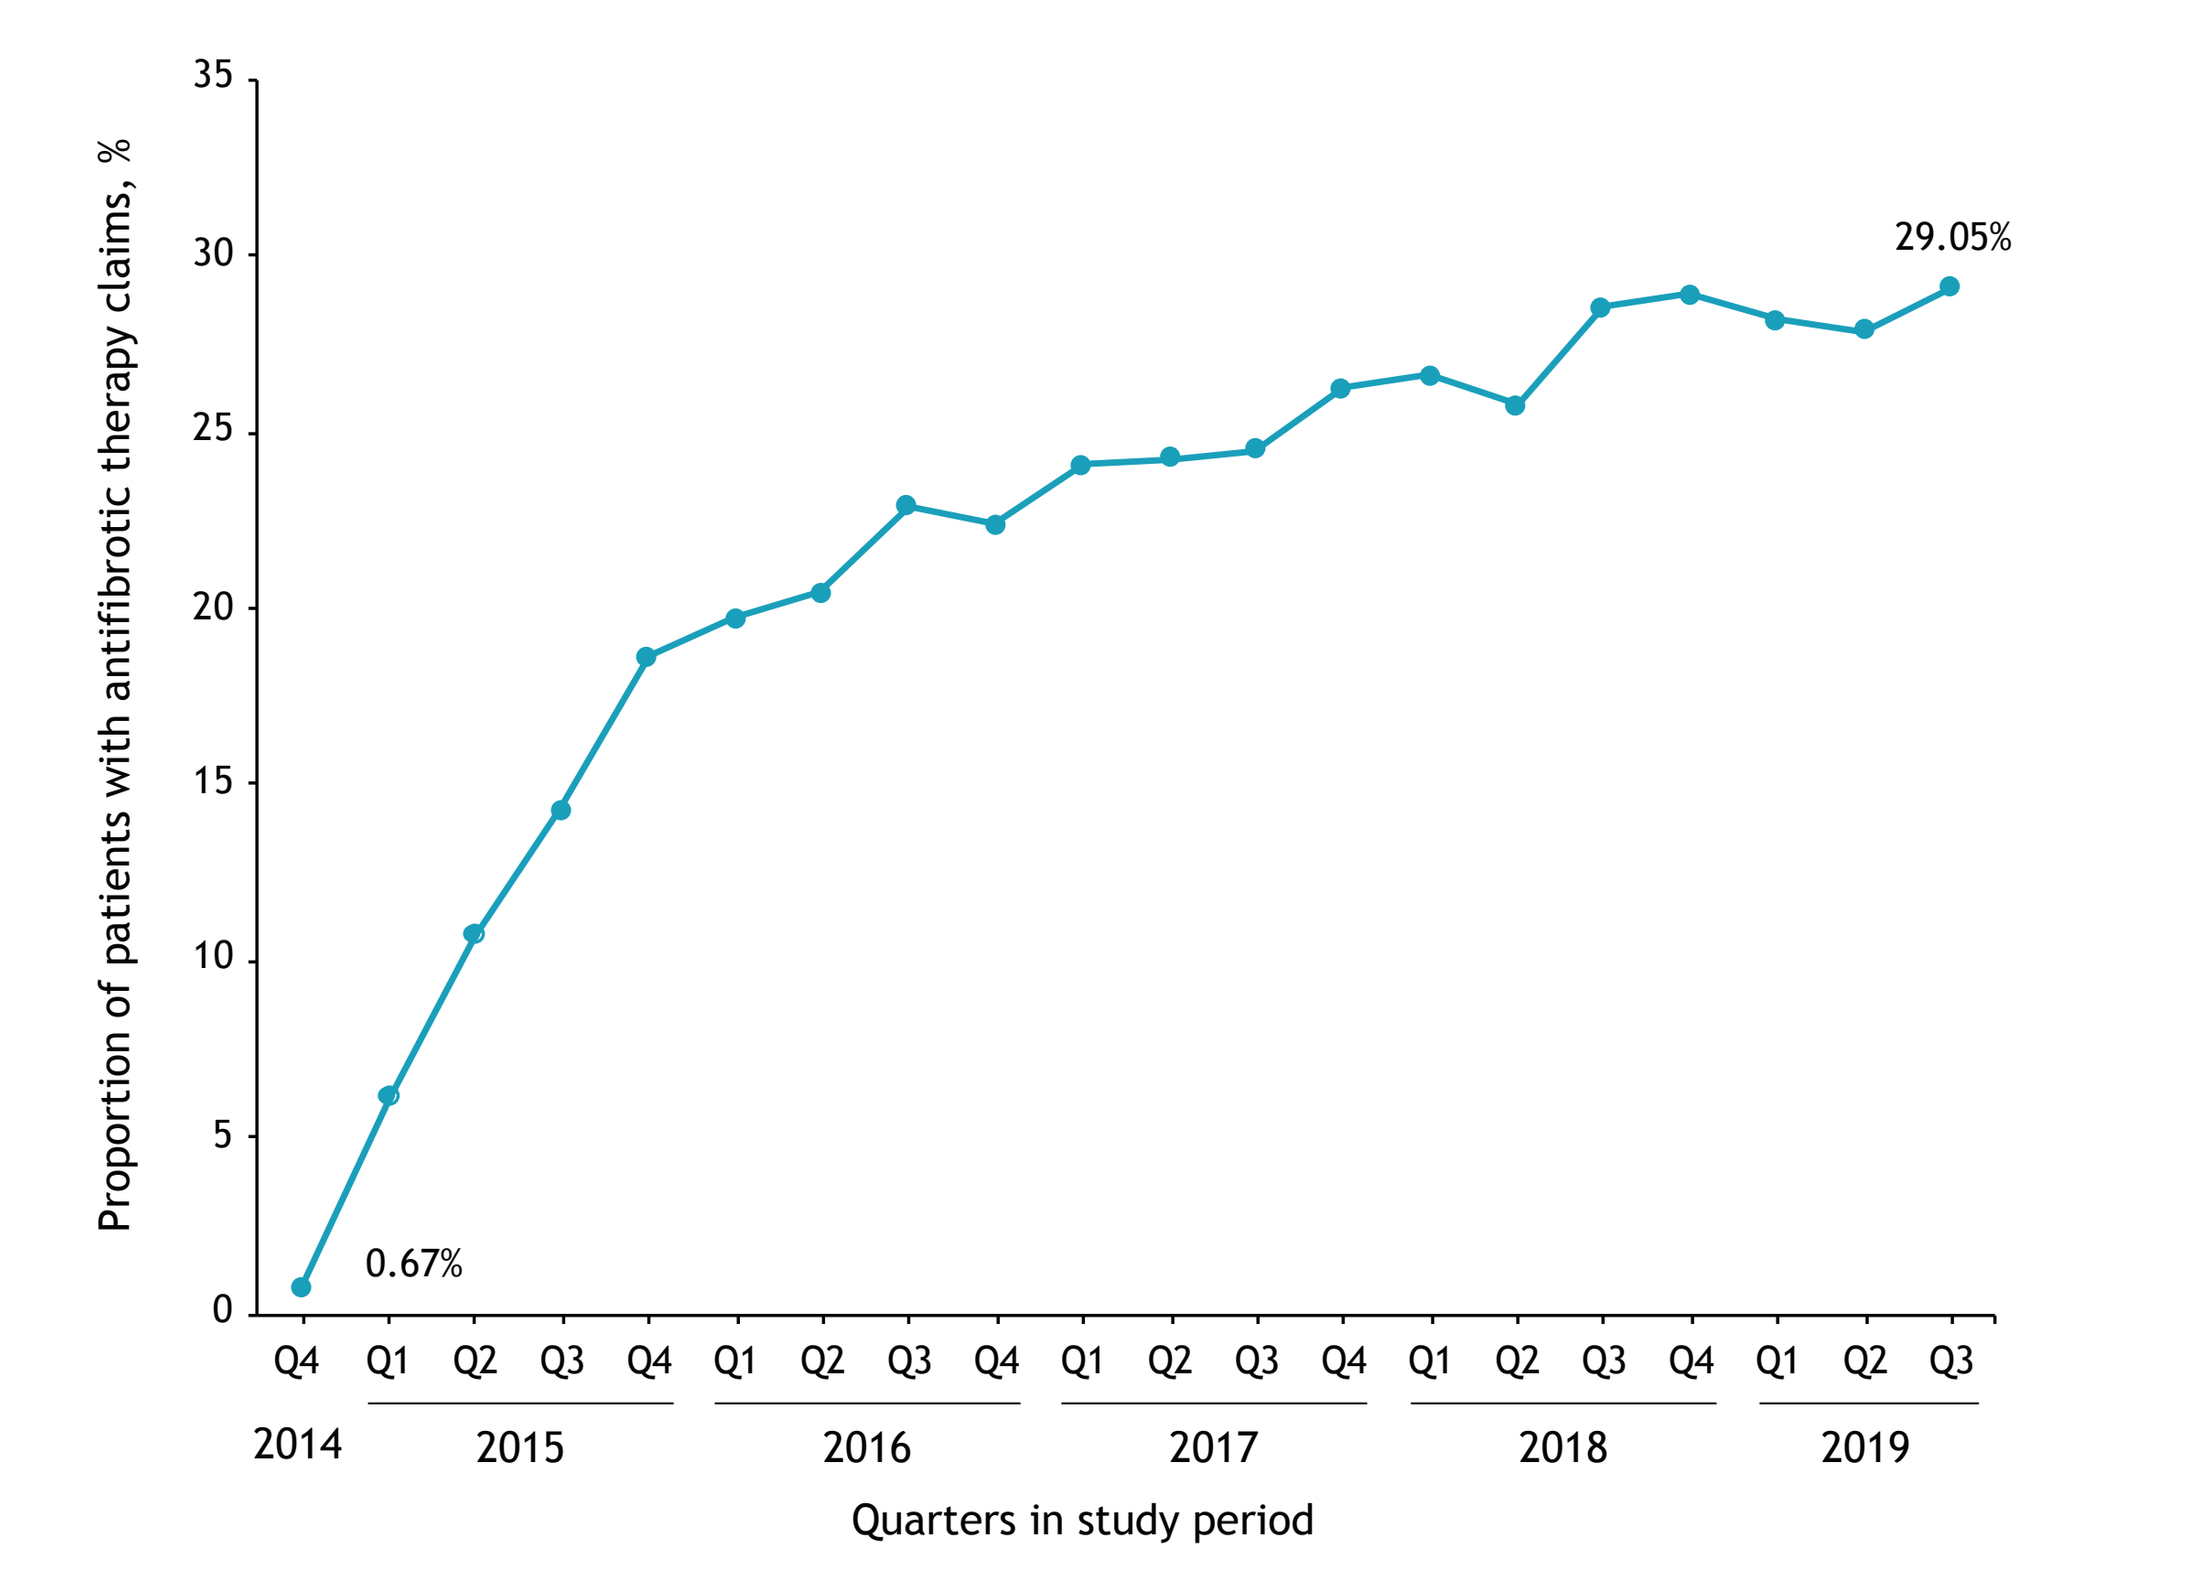
Proportion of patients with antifibrotic therapy claims, %
35
30
25
20
15
10
5
0
0.67%
29.05%
Q4 Q1 Q2 Q3 Q4 Q1 Q2 Q3 Q4 Q1 Q2 Q3 Q4 Q1 Q2 Q3 Q4 Q1 Q2 Q3
2014 2015 2016 2017 2018 2019
Quarters in study period

IPF, idiopathic pulmonary fibrosis; Q, quarter; VHA, Veterans Health Administration.

- Consistent with IPF demographic risk factors, the overall study population was predominantly elderly (>65 y), White, and male; compared with untreated patients, treated patients were significantly younger (mean age: 79.5 vs 73.4 y; *P*<.0001), and more were male (97.6% vs 99.1%; *P*=.0076) and White (83.1% vs 86.7%; *P*=.0102; **Table 1**)
- Treated patients had a significantly lower mean CCI score compared with untreated patients (2.1 vs 2.3, respectively; *P*=.0167) but had a significantly higher burden of important IPF-related comorbidities compared with untreated patients, including GERD (37.2% vs 25.8%; *P*<.0001), obstructive sleep apnea (25.7% vs 16.3%; *P*<.0001), obesity (18.2% vs 11.9%; *P*<.0001), and higher use of antacids (60.0% vs 44.4%; *P*<.0001) and corticosteroids (48.6% vs 41.5%; *P*=.0002)

Table 1. Baseline patient characteristics

Patient characteristics	Treated patients n=850	Untreated patients n=3,609	*P* value
Demographic variables			
Age, mean (SD), y	73.40 (9.61)	79.52 (17.11)	**<.0001**
Age group, n (%)			
18-34 y	0	<12[a]	.3315
35-54 y	<12[a]	63 (1.75)	**.0271**
55-64 y	74 (8.71)	311 (8.62)	.9341
≥65 y	770 (90.59)	3,231 (89.53)	.3588
Sex, n (%)			
Male, n (%)	842 (99.06)	3,522 (97.59)	**.0076**
Race, n (%)			
White	737 (86.71)	2,999 (83.10)	**.0102**
Black	53 (6.24)	264 (7.32)	.2704
Other	15 (1.76)	95 (2.63)	.1424
Unknown	45 (5.29)	251 (6.95)	.0802
CCI score, mean (SD)	2.07 (1.89)	2.25 (2.29)	**.0167**
Individual comorbidities, n (%)			
COPD	311 (36.59)	1,274 (35.30)	.4805
Bacterial pneumonia	27 (3.18)	131 (3.63)	.5201
Lung cancer	<12[a]	110 (3.05)	**.0006**
Cardiovascular conditions	371 (43.65)	1,520 (42.12)	.4167
GERD	316 (37.18)	931 (25.80)	**<.0001**
Obstructive sleep apnea	218 (25.65)	589 (16.32)	**<.0001**
Obesity	155 (18.24)	431 (11.94)	**<.0001**
Depression	17 (2.00)	45 (1.25)	.0916
Diabetes	333 (39.18)	1,236 (34.25)	**.0068**
Anxiety	90 (10.59)	322 (8.92)	.1313
Pulmonary hypertension	73 (8.59)	250 (6.93)	.0928
Prior adjunctive therapies, n (%)			
Antacids	510 (60.00)	1,603 (44.42)	**<.0001**
H2 receptor blockers	68 (8.00)	285 (7.90)	.9202
PPIs	482 (56.71)	1,452 (40.23)	**<.0001**
Corticosteroids	413 (48.59)	1,497 (41.48)	**.0002**
Budesonide	249 (29.29)	858 (23.77)	**.0008**
Dexamethasone	14 (1.65)	73 (2.02)	.4762
Methylprednisolone	24 (2.82)	89 (2.47)	.5508
Prednisolone	33 (3.88)	117 (3.24)	.3515
Prednisone	194 (22.82)	677 (18.76)	**.0072**
Triamcinolone	73 (8.59)	233 (6.46)	**.0270**
Supplemental oxygen	19 (2.24)	40 (1.11)	**.0097**
Diagnostic tests			
HRCT, n (%)	599 (70.47)	1357 (37.60)	**<.0001**
Smoking status: yes, n (%)	185 (21.76)	542 (15.02)	**<.0001**
Time from diagnostic testing to diagnosis, mean (SD), days	104 (90)	141 (101)	**.0019**
Time from diagnosis to index date, mean (SD), days	168 (231)	283 (278)	**<.0001**

[a]VHA patient privacy specifications prohibit the disclosure of exact patient numbers when the n is <12. CCI, Charlson Comorbidity Index; COPD, chronic obstructive pulmonary disease; GERD, gastroesophageal reflux disease; HRCT, high-resolution computed tomography; PPI, proton pump inhibitor.

- Overall, treated patients incurred higher total costs during the baseline period than untreated patients ($2,683 vs $2,119; *P*=.0003; **Table 2**)

Table 2. Baseline all-cause HCRU and costs in treated vs untreated VHA patients with IPF

Patient characteristics	Treated patients n=850	Untreated patients n=3,609	*P* value
Baseline all-cause HCRU			
Patients with ≥1 inpatient visit, n (%)	178 (21)	675 (19)	.1356
Patients with ≥1 outpatient visit, n (%)	850 (100)	3,576 (99)	**.0051**
Patients with ≥1 pharmacy visit, n (%)	823 (97)	3,241 (90)	**<.0001**
Inpatient visits, PPPM, mean (SD)	0.03 (0.06)	0.03 (0.08)	.1234
Length of stay, PPPM, mean (SD), days	0.18 (0.87)	0.28 (1.76)	**.0112**
Outpatient visits, PPPM, mean (SD)	4.06 (2.24)	3.66 (2.77)	**<.0001**
Pharmacy visits, PPPM, mean (SD)	2.26 (1.70)	2.12 (1.75)	.4935
Baseline all-cause healthcare costs, mean (SD), USD			
Inpatient costs	484 (1,745)	645 (2,371)	**.0249**
Outpatient costs	1,663 (1,850)	1,223 (1,806)	**<.0001**
Pharmacy costs	536 (1,530)	251 (1,178)	**<.0001**
Total medical costs (inpatient + outpatient)	2,147 (2,982)	1,868 (3,354)	**.0168**
Total costs (inpatient + outpatient + pharmacy)	2,683 (4,241)	2,119 (4,108)	**.0003**

HCRU, healthcare resource utilization; IPF, idiopathic pulmonary fibrosis; PPPM, per-patient-per-month; USD, US dollars; VHA, Veterans Health Administration.

- Over the follow-up period, the cohort treated with antifibrotics had a significantly lower adjusted risk of death than the untreated cohort (adjusted HR, 0.69; 95% CI, 0.61-0.78); in addition, the treated cohort experienced significantly longer median overall survival (1,198 days) than the untreated cohort (923 days; *P*<.0001; **Figure 3**)

Figure 3. Mortality outcomes in treated vs untreated VHA patients with IPF

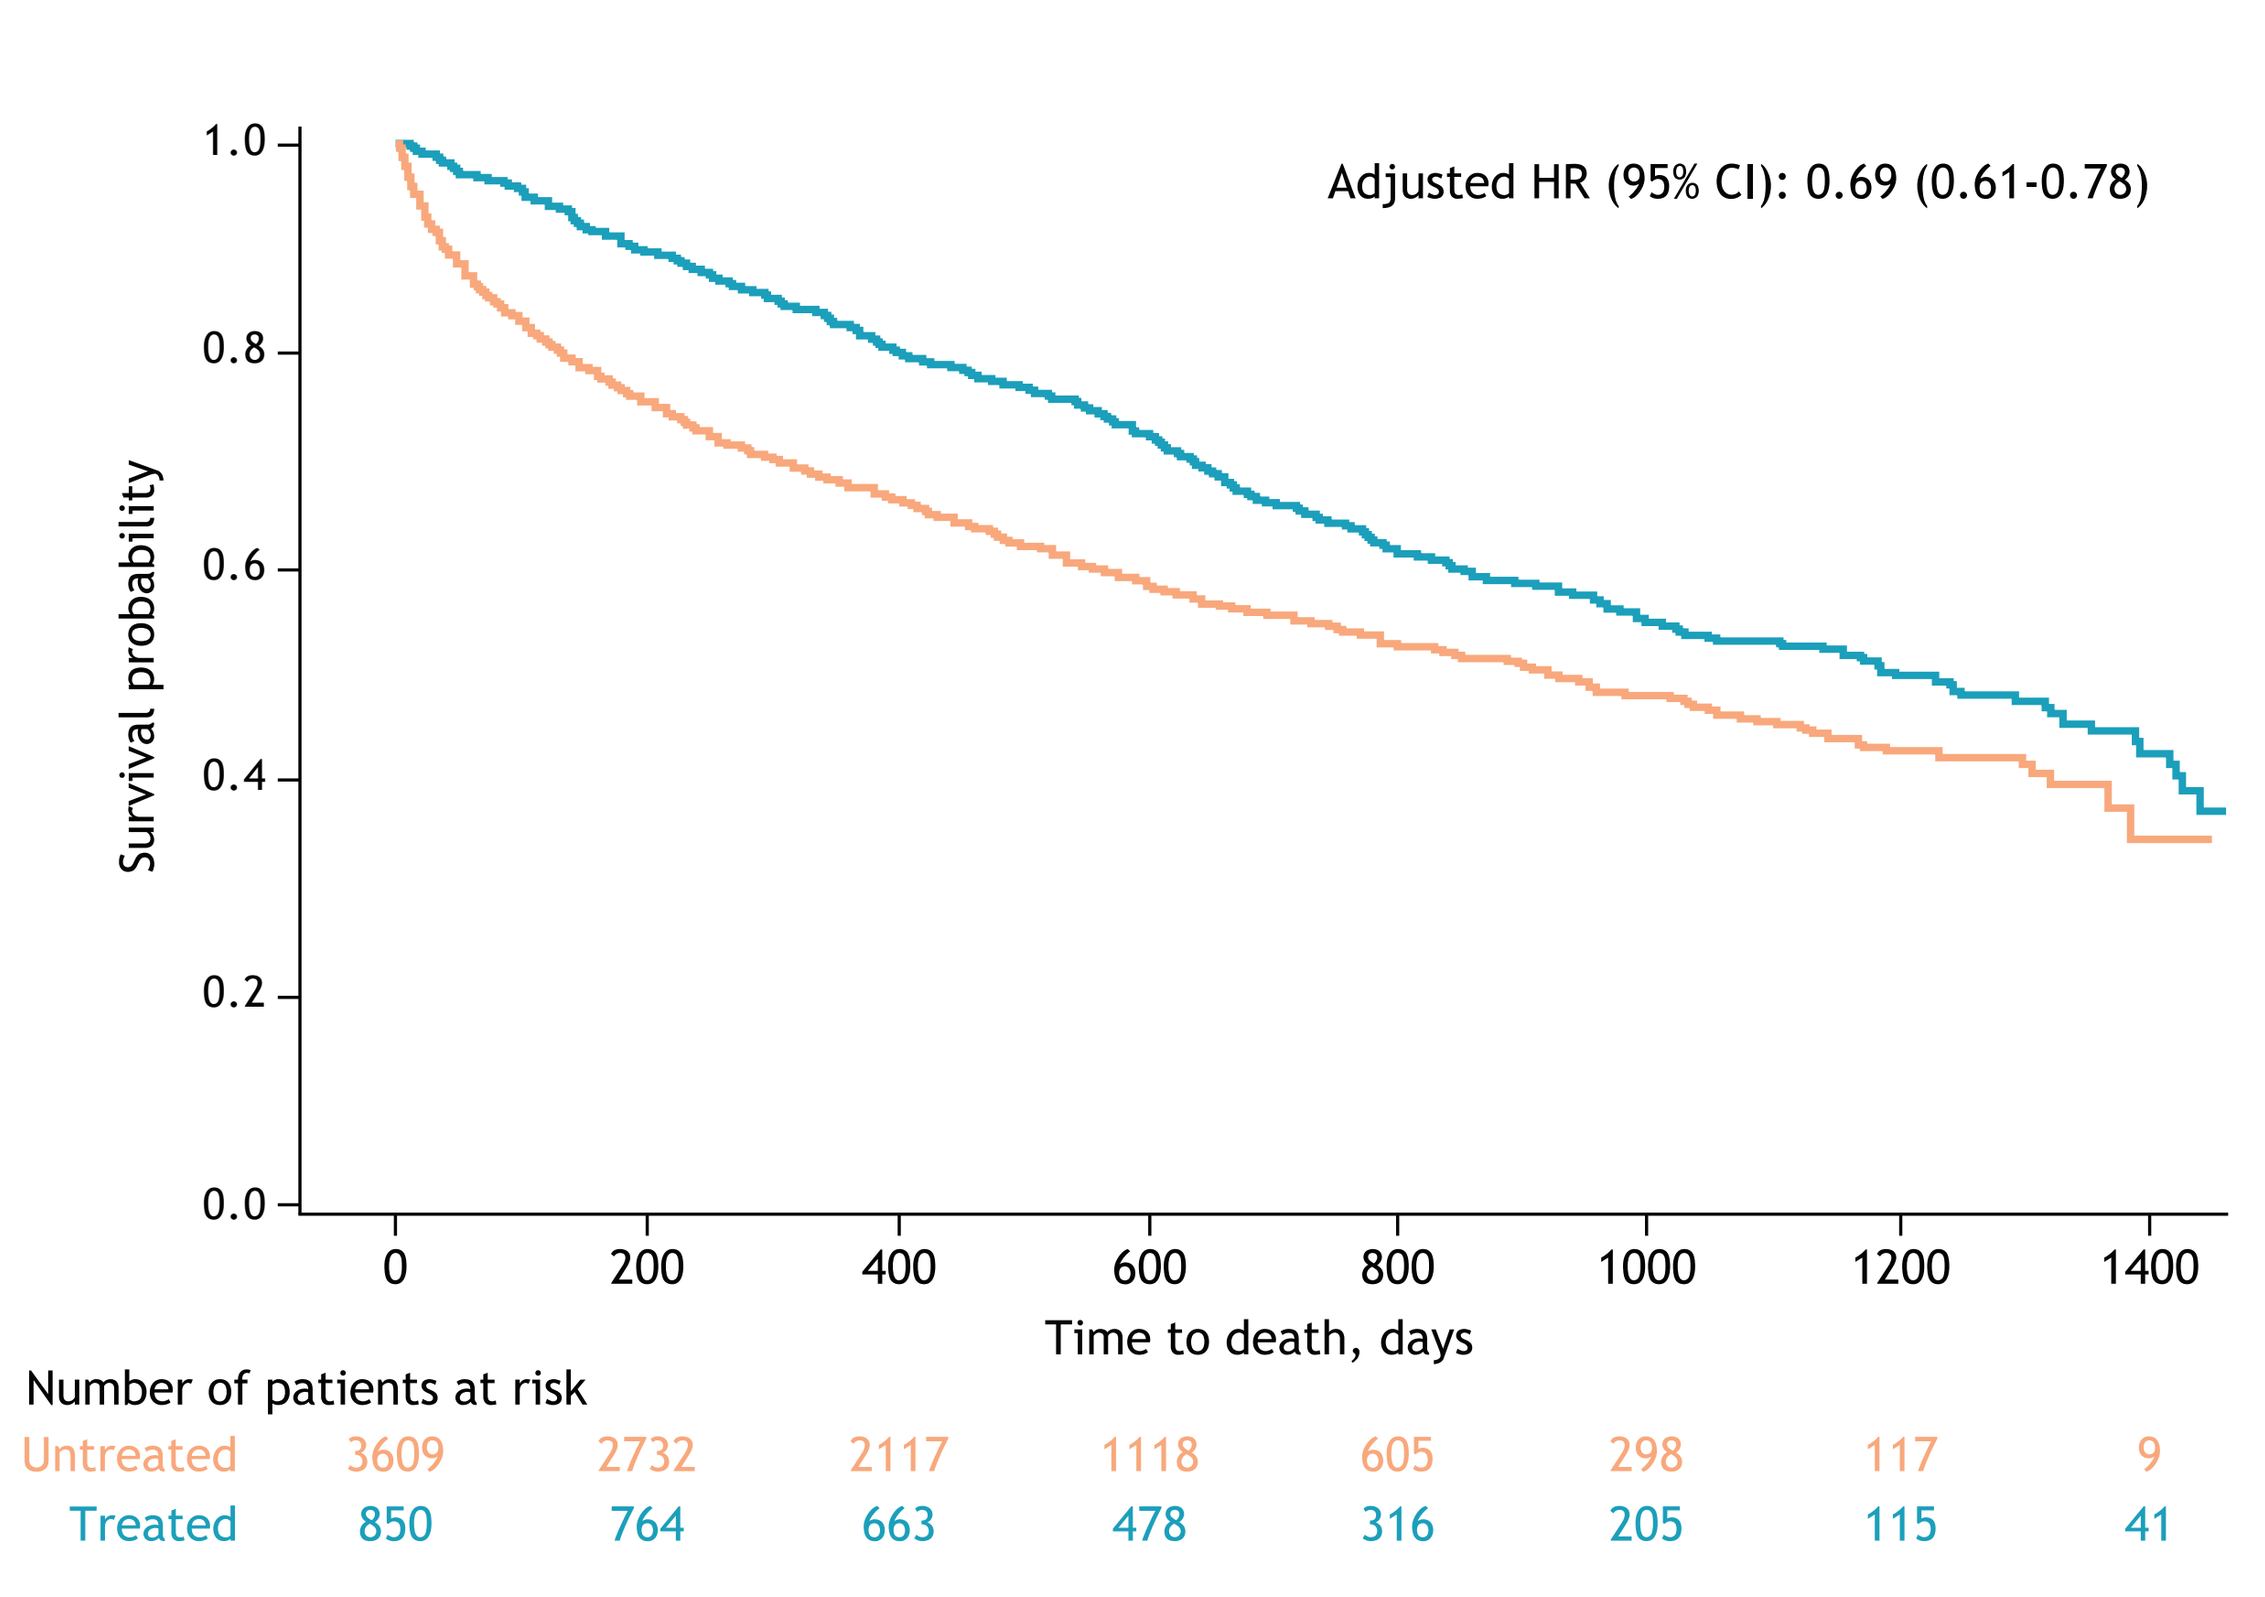
Adjusted HR (95% CI): 0.69 (0.61-0.78)
Survival probability
1.0
0.8
0.6
0.4
0.2
0.0
0 200 400 600 800 1000 1200 1400
Time to death, days
Number of patients at risk
Untreated 3609 2732 2117 1118 605 298 117 9
Treated 850 764 663 478 316 205 115 41

HR, hazard ratio; IPF, idiopathic pulmonary fibrosis; VHA, Veterans Health Administration.

- As shown in **Figure 4**, treated patients had significantly higher usage and costs of outpatient and pharmacy services compared with untreated patients
- For care received in inpatient settings, the numbers of visits and costs were similar between the cohorts (treated vs untreated: inpatient visits PPPM, 0.07 vs 0.07; inpatient costs PPPM, $1,051 vs $790)

Figure 4. Economic burden in treated vs untreated VHA patients with IPF

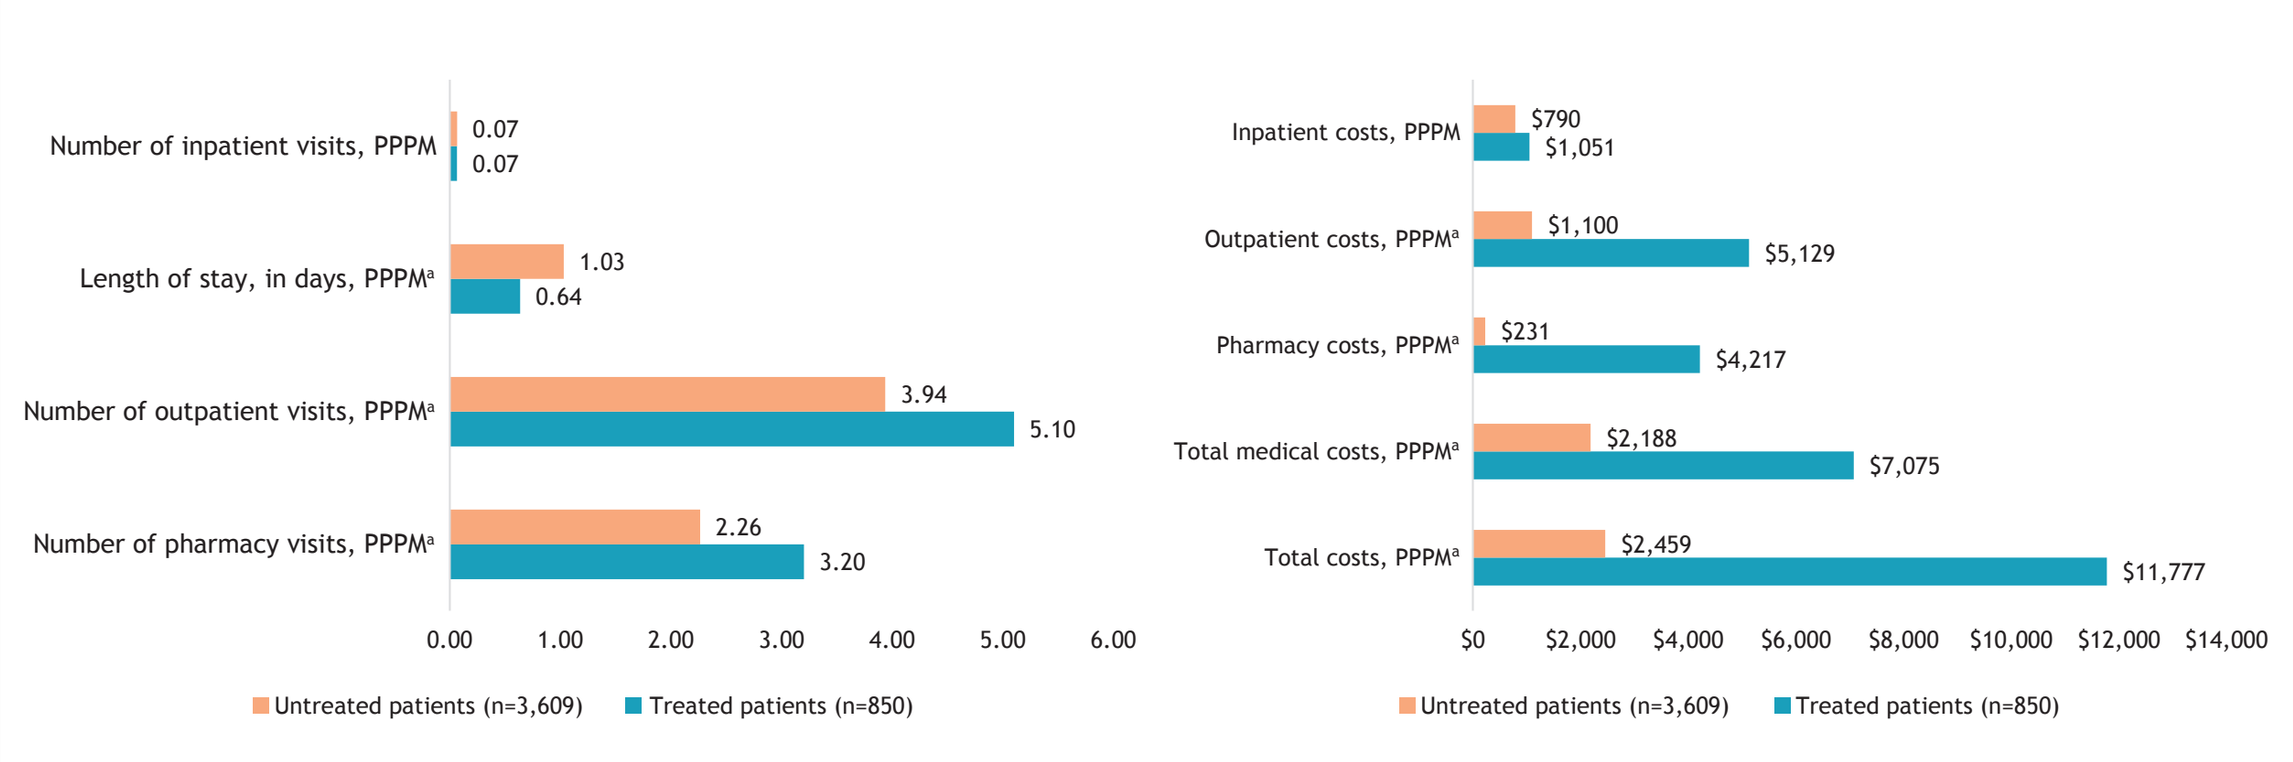
Number of inpatient visits, PPPM 0.07 0.07
Length of stay, in days, PPPM[a] 1.03 0.64
Number of outpatient visits, PPPM[a] 3.94 5.10
Number of pharmacy visits, PPPM[a] 2.26 3.20
0.00 1.00 2.00 3.00 4.00 5.00 6.00
Untreated patients (n=3,609) Treated patients (n=850)
Inpatient costs, PPPM $790 $1,051
Outpatient costs, PPPM[a] $1,100 $5,129
Pharmacy costs, PPPM[a] $231 $4,217
Total medical costs, PPPM[a] $2,188 $7,075
Total costs, PPPM[a] $2,459 $11,777
$0 $2,000 $4,000 $6,000 $8,000 $10,000 $12,000 $14,000
Untreated patients (n=3,609) Treated patients (n=850)

[a]*P*<.05. IPF, idiopathic pulmonary fibrosis; PPPM, per-patient-per-month; VHA, Veterans Health Administration.

Limitations

- As a claims study, it is possible that the dataset could have been affected by the following:
 - Missing or incorrectly inputted data
 - A diagnosis code could indicate probable, rather than confirmed, disease
 - Important clinical information, eg, forced vital capacity scores, was unavailable within VHA data
 - No clear indications of whether filled medications were taken as prescribed were available
- Despite multivariable analysis, residual confounding in the mortality comparison may be present
- Due to data user agreements with the VHA, granular information on patient samples <12 could not be explored

Conclusions

- Nintedanib and pirfenidone are antifibrotic therapies approved for IPF; however, both are associated with side effects that can lead to treatment discontinuation
- In this real-world analysis of VHA patients with IPF, the rate of IPF antifibrotic therapy use was very low (29%) compared with those reported in other US and European IPF patient populations (~60%)[6]; this may be explained, in part, by differences in medication access in the VHA compared with other health systems
- After FDA approval in late 2014, the rate of IPF antifibrotic therapy use in VHA patients increased sharply, but has since leveled off
- Antifibrotic treatment was associated with longer overall survival; however, patients treated with antifibrotic therapies had higher HCRU and direct care costs than untreated patients, which may be related to treatment-associated side effects or other relevant comorbidities in treated patients
- Overall, these results suggest that ongoing unmet needs in IPF management exist among VHA enrollees, despite availability of approved antifibrotic treatments
- Greater utilization of existing antifibrotic treatments as well as more tolerable treatment options may help to address this need

References

1. du Bois RM. *Eur Respir Rev.* 2012;21(124):141-146.
2. Richeldi L, et al. *N Eng J Med.* 2014;370(22):2071-2082.
3. King TE Jr, et al. *N Eng J Med.* 2014;370(22):2083-2092.
4. Raghu G, et al. *Am J Respir Crit Care Med.* 2006;174(7):810-816.
5. Veterans Health Administration website: https://www.va.gov/health. Accessed February 2022.
6. Maher TM, ME Strek. *Respir Res.* 2019;20(1):205.

Acknowledgments & disclosures

- Y Qiu, J Zhu, B Elpers, and A Fischer are employees of Bristol Myers Squibb and may own company stock/stock options
- C Dieyi, C Byrne, and R Singh are employees of STATinMED Research, which received consultancy fees from Bristol Myers Squibb
- Medical writing support was provided by Hayley White, PhD, and Amanda Martin, PhD, of Medical Expressions (Chicago, IL) and was funded by Bristol Myers Squibb

Presented at: ATS 2022; May 13–18, 2022; San Francisco, CA

Email: Ning.Zhu@bms.com