695

Higher Baseline Comorbidities and Liver Disease Severity Among Patients With Hepatitis Delta Infection vs Hepatitis B Monoinfection: a National US Claims Database Analysis

Ira Jacobson,[1] Robert Gish,[2] Joseph Lim,[3] Ankita Modi Kaushik,[4] Nandita Kachru,[4] Xiaoxi Sun,[5] Robert Wong[6]

[1]NYU Grossman School of Medicine, New York, NY; [2]University of Nevada, Reno School of Medicine, Kirk Kerkorian School of Medicine at UNLV, Las Vegas, NV, and UC San Diego Skaggs School of Pharmacy and Pharmaceutical Sciences, Hepatitis B Foundation, La Jolla, CA; [3]Yale School of Medicine, New Haven, CT; [4]Global Value and Access–HEOR, Gilead Sciences, Inc., Foster City, CA; [5]STATinMED, Plano, TX; [6]Stanford University School of Medicine, Stanford, and VA Palo Alto Healthcare System, Palo Alto, CA

Gilead Sciences, Inc.
333 Lakeside Drive
Foster City, CA 94404
800-445-3235

Introduction

- Hepatitis delta virus (HDV) leads to the most severe form of viral hepatitis[1]
- HDV can infect individuals with hepatitis B virus (HBV) infection simultaneously (HBV/HDV coinfection) or can occur as a superinfection in patients with known chronic HBV[1]
- People who inject drugs and public safety officials working with blood or blood-contaminated bodily fluids are at a high risk of getting HDV infection; US foreign-born individuals also have a higher burden of disease[2,3]
- Compared to patients with HBV monoinfection, HDV infection is associated with accelerated disease progression to liver cirrhosis and decompensation[4]; a retrospective cohort study found that HDV infection was associated with a higher risk of liver decompensation and was the only independent predictor of mortality when compared with patients with cirrhosis due to HBV monoinfection[5]
- Limited information is currently available on patients with HDV infection and their characteristics compared to patients with HBV monoinfection in the USA

Objective

- To compare baseline characteristics of adult patients with HDV infection versus HBV monoinfection in a national US claims database analysis

Methods

- The study population included adult patients (aged ≥18 years) with ≥1 claim based on International Classification of Diseases 9th/10th Clinical Revision (ICD-9/10-CM) diagnosis codes for HDV infection or HBV infection in all-payer claims databases (APCD) from 1/1/2014 to 12/31/2020 (study period)
- HDV infection and HBV monoinfection cohorts were further identified from 1/1/2015 to 12/31/2020 (identification period), with their respective first diagnoses claim defined as the index date
- Patients were categorized under HDV infection or HBV monoinfection cohorts based on presence or absence of HDV diagnosis during the study period
- All patients were required to have ≥12 months of continuous capture prior to the index date
- Baseline characteristics including age, gender, payer type, Charlson Comorbidity Index (CCI) score, liver disease, and comorbidities were assessed prior to the index date
- Descriptive statistics were summarized and comparisons were made via chi-square tests

Results

Attrition Flowchart: Patient Selection for HDV and HBV-Monoinfected Patients

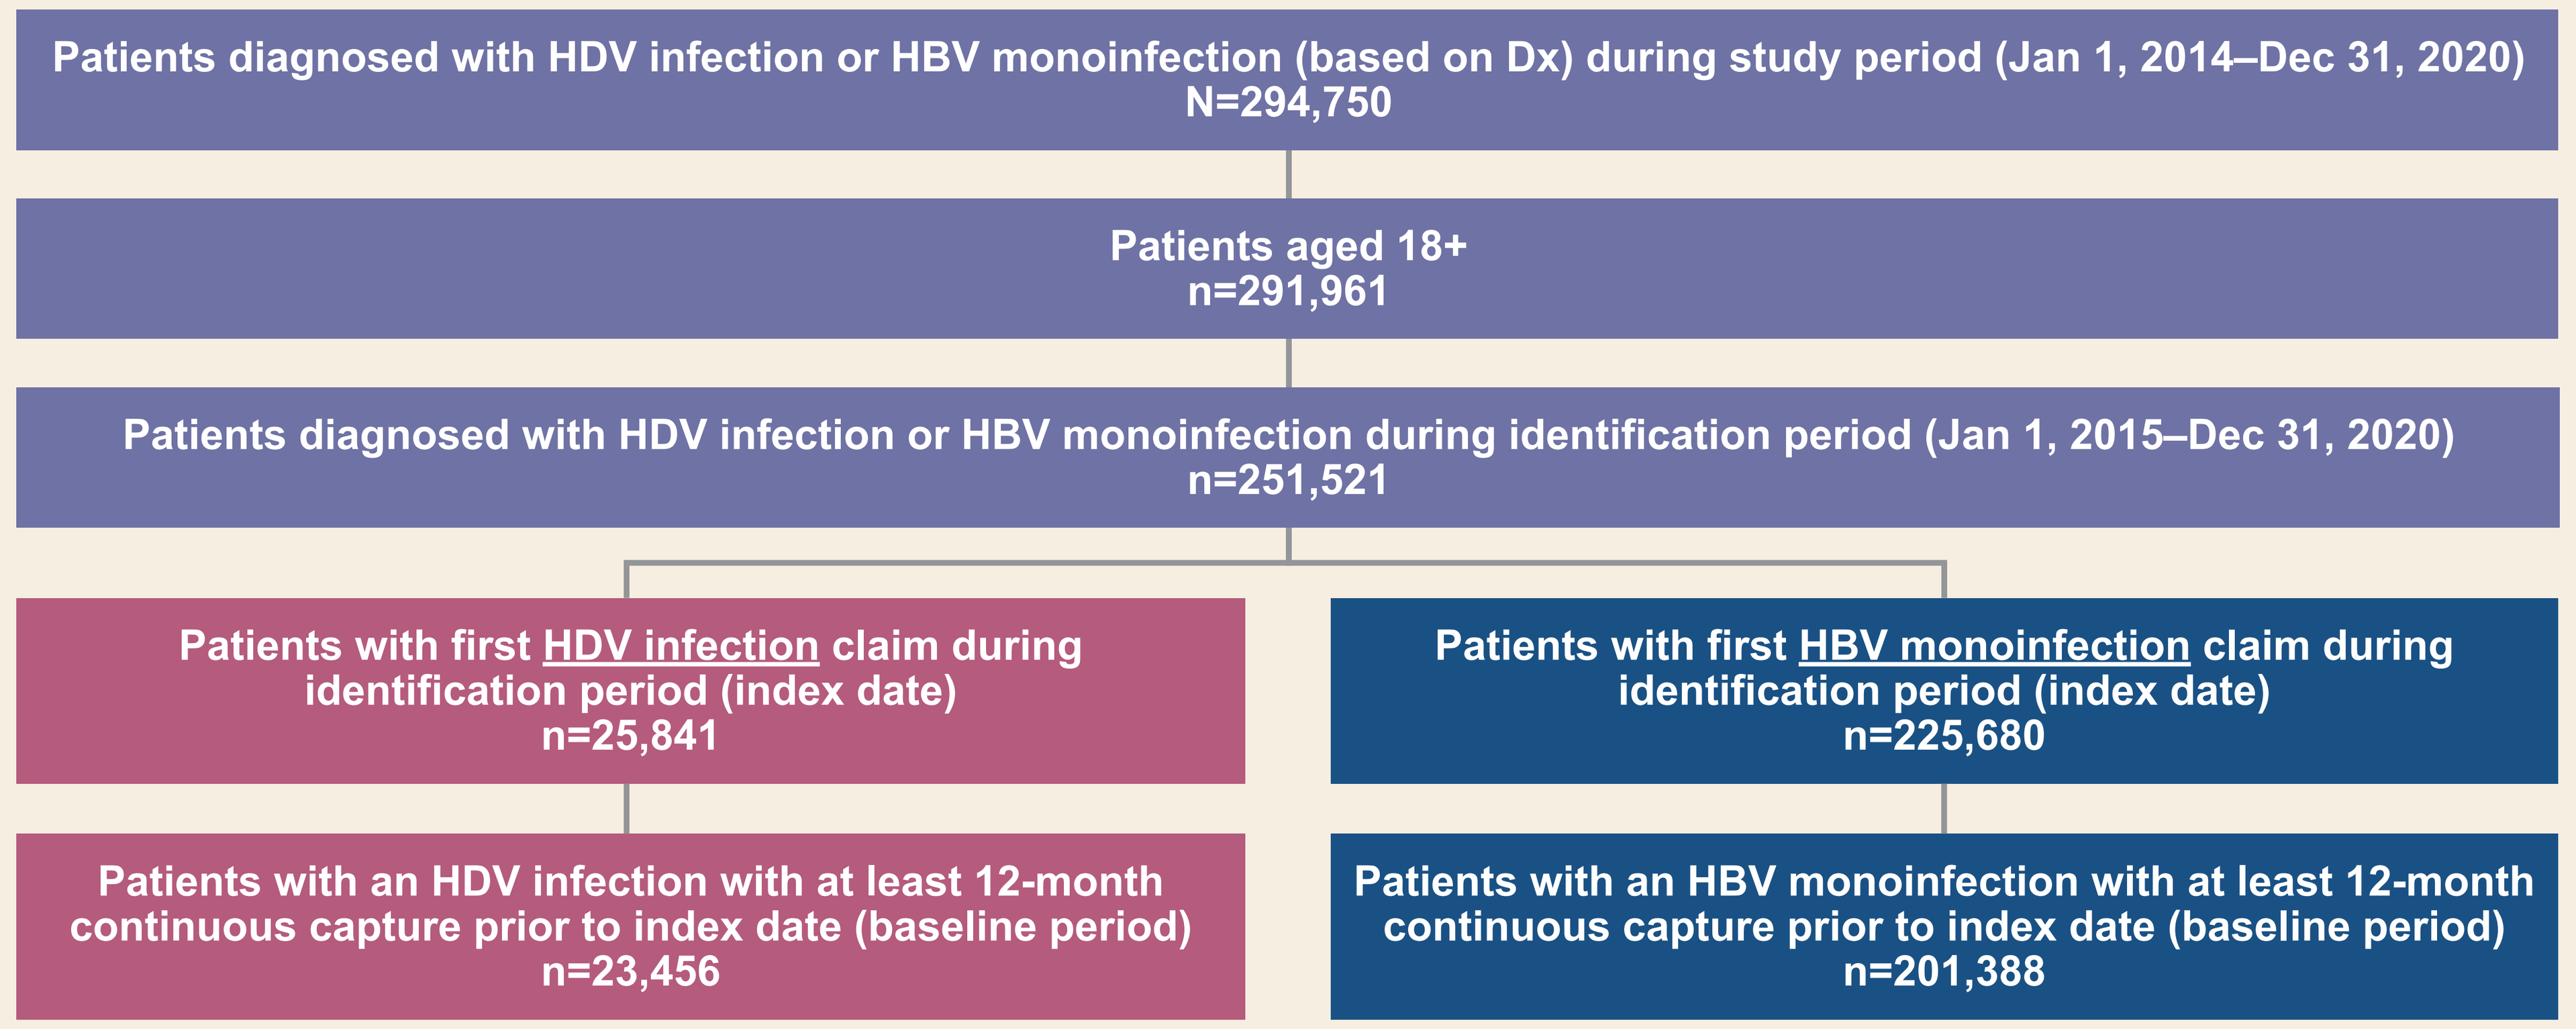
Patients diagnosed with HDV infection or HBV monoinfection (based on Dx) during study period (Jan 1, 2014–Dec 31, 2020)
N=294,750
Patients aged 18+
n=291,961
Patients diagnosed with HDV infection or HBV monoinfection during identification period (Jan 1, 2015–Dec 31, 2020)
n=251,521
Patients with first HDV infection claim during identification period (index date)
n=25,841
Patients with first HBV monoinfection claim during identification period (index date)
n=225,680
Patients with an HDV infection with at least 12-month continuous capture prior to index date (baseline period)
n=23,456
Patients with an HBV monoinfection with at least 12-month continuous capture prior to index date (baseline period)
n=201,388

- The final study population included 291,961 adults diagnosed with HDV infection or HBV monoinfection, of whom 23,456 HDV-infected (mean age ± standard deviation [SD] 51.5 ± 15.9 years) and 201,388 HBV-monoinfected (53.5 ± 14.7 years) patients met the inclusion criteria

Baseline Characteristics by Cohort

Baseline Characteristics	HDV Infection n=23,456	HBV Monoinfection n=201,388	p-Value
Mean age, years (SD)	51.5 (15.9)	53.5 (14.7)	<0.001
Mean CCI score (SD)*	1.5 (2.26)	1.3 (2.02)	<0.001
Women, %	53	47	<0.001
Insurance type, %			
Commercial	49	53	<0.001
Medicare	23	26	
Medicaid	23	16	

*CCI: Charlson Comorbidity Index weights 15 comorbid conditions to determine patient's 1-year mortality risk; higher score is associated with higher risk of death within 1 year; CCI was estimated during a 12 month period prior to index date.

- The proportion of patients who were women was significantly greater in the HDV infection vs HBV monoinfection cohort
- Although both cohorts were largely covered by commercial insurance, patients with HDV infection were significantly more likely to be covered by Medicaid and less likely by Medicare versus the HBV monoinfection cohort
- Mean baseline Charlson Comorbidity Index score was significantly higher among patients with HDV infection versus HBV monoinfection

Baseline Comorbidities and Liver Disease Severity by Cohort

Comorbidity Rates

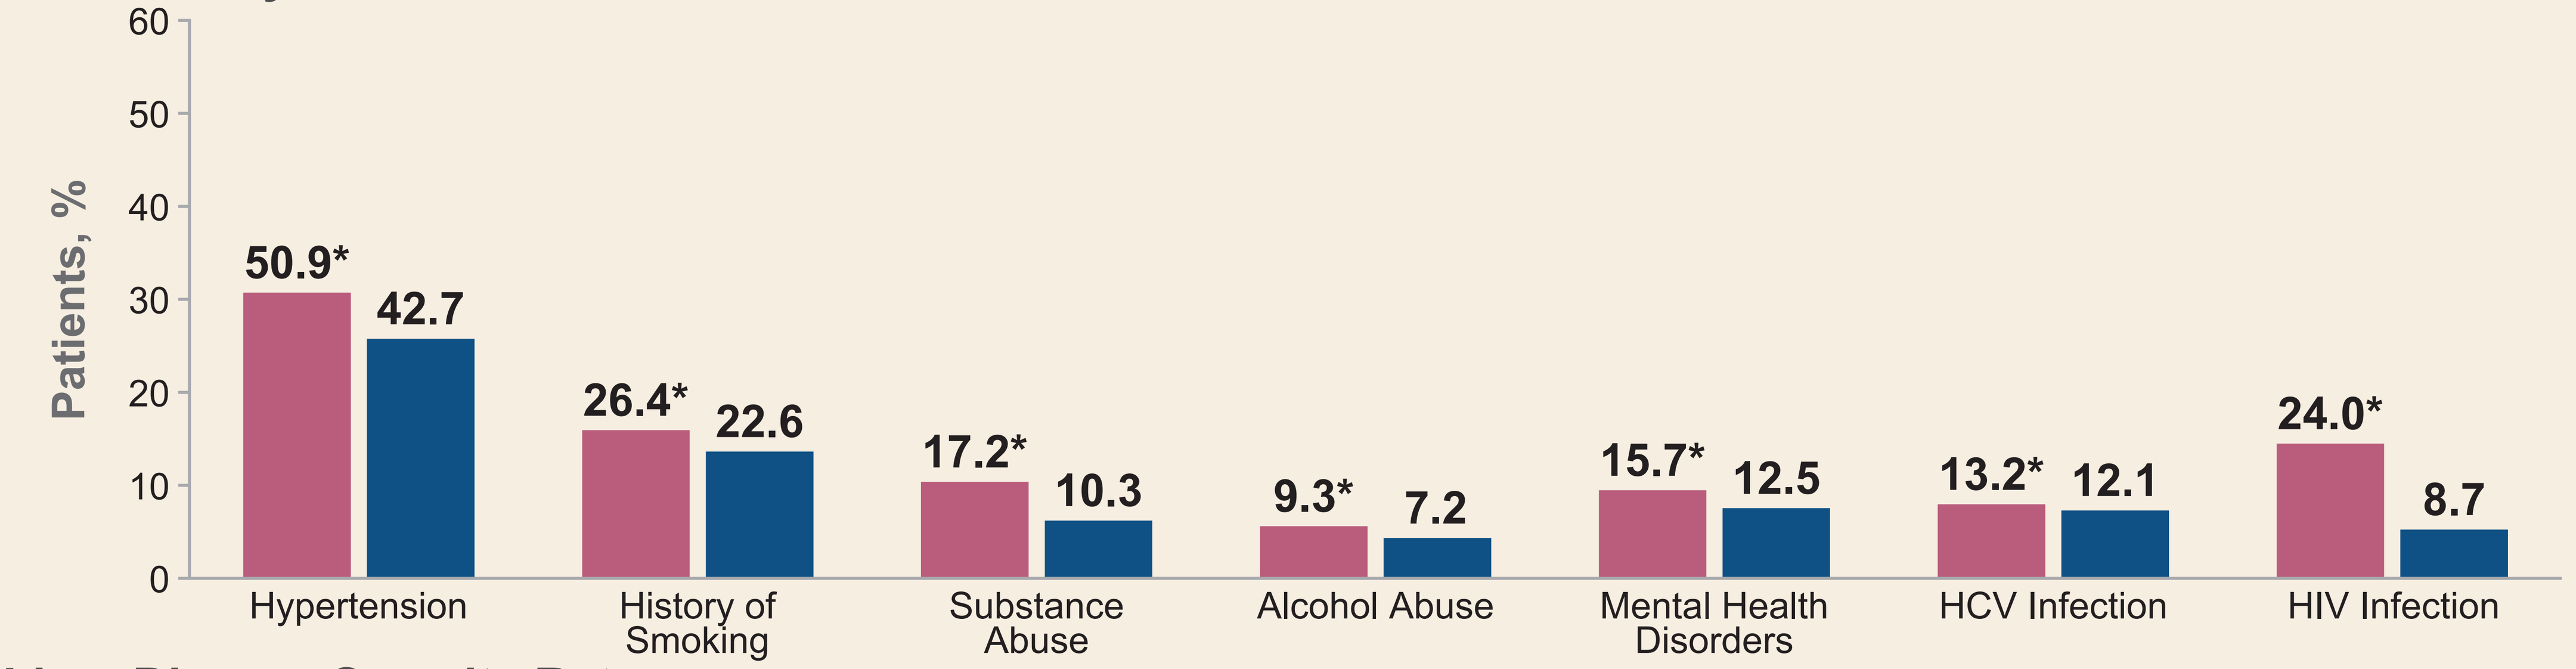
HDV infection
HBV monoinfection
Patients, %
0
10
20
30
40
50
60
50.9*
42.7
Hypertension
26.4*
22.6
History of Smoking
17.2*
10.3
Substance Abuse
9.3*
7.2
Alcohol Abuse
15.7*
12.5
Mental Health Disorders
13.2*
12.1
HCV Infection
24.0*
8.7
HIV Infection

Liver Disease Severity Rates

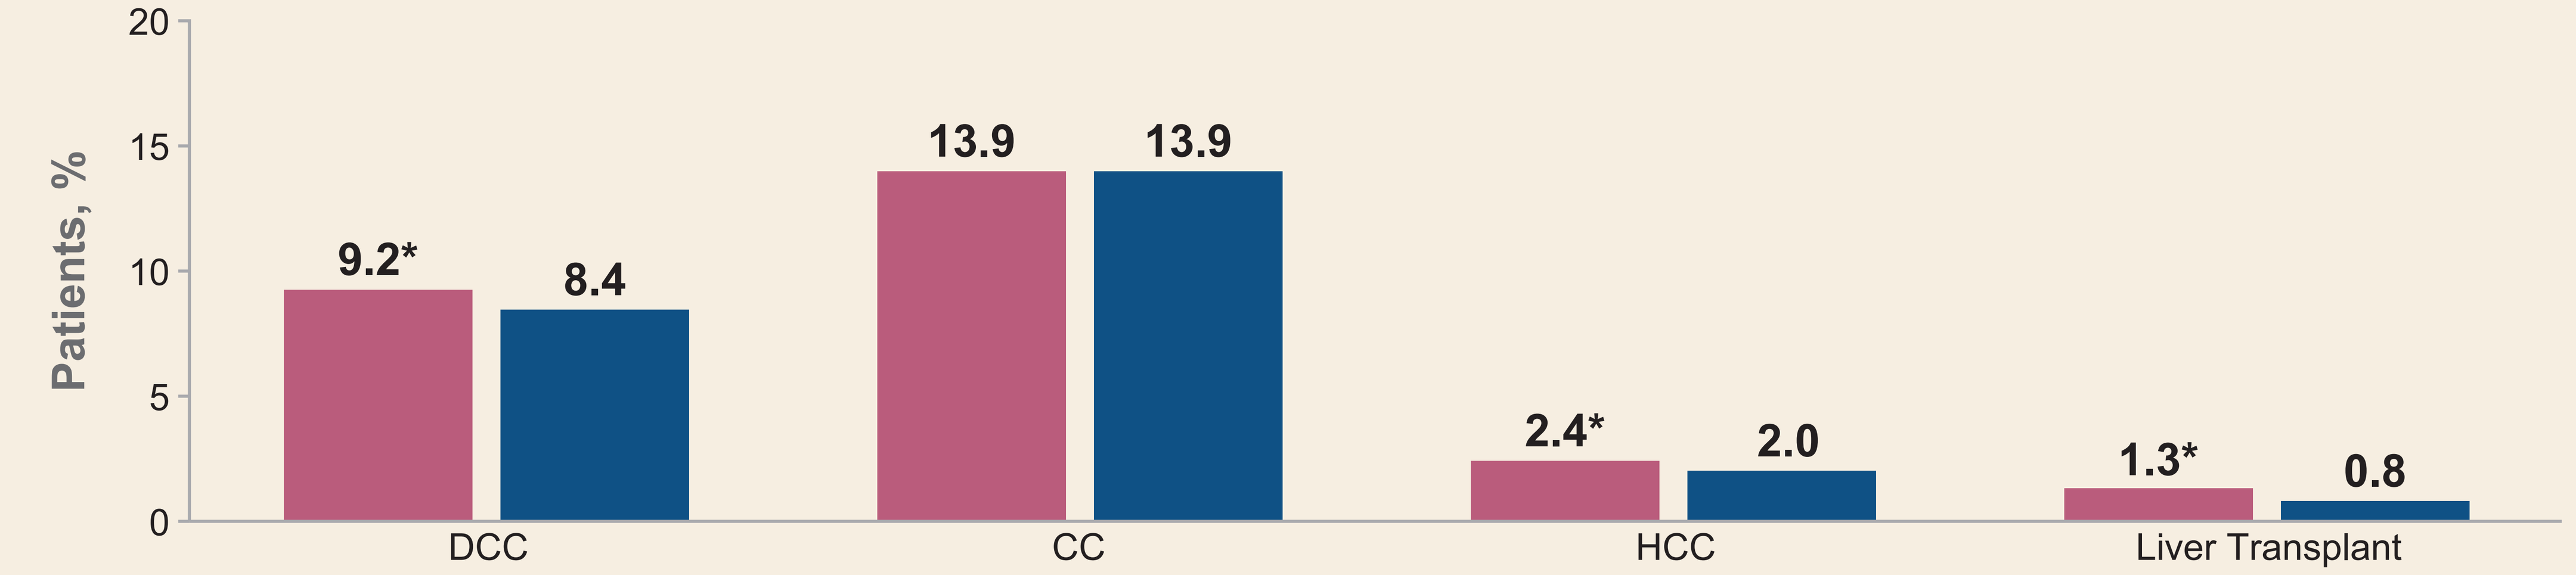
Patients, %
0
5
10
15
20
9.2*
8.4
DCC
13.9
13.9
CC
2.4*
2.0
HCC
1.3*
0.8
Liver Transplant

*p<0.05 for HDV infection vs HBV monoinfection. CC, compensated cirrhosis; DCC, decompensated cirrhosis; HCC, hepatocellular carcinoma; HCV, hepatitis C virus. All comorbidities and liver disease severity were assessed prior to index date (1st diagnosis claim during study identification period).

- Patients with HDV infection vs HBV monoinfection had significantly higher (p<0.001) baseline rates of comorbidities and severe liver disease (except for CC only; p=0.82)

Limitations

- The APCD data is representative of individuals that are insured by commercial plans, Medicare, or Medicaid; patients under federal programs (Veterans Administration and Department of Defense) are captured here; subjects in closed plans such as Kaiser and those who are uninsured are not captured
- The usual limitations of any retrospective claims analyses apply; any diagnoses done via ICD-9/10-CM codes are subjected to miscoding and can lead to misclassification bias; no standard coding algorithm exists for liver cirrhosis (compensated or otherwise); baseline frequency of cirrhosis should be interpreted with caution and may or may not be linked with HDV infection
- Comorbidity designation is based on identifying a code of interest >12 months look-back until 2014; it is possible that patients may be missing diagnosis codes for comorbidities due to study period limitations
- There is a lack of FDA-cleared assays, as well as suboptimal screening practices to determine HDV and HBV status, which could cause this study to underestimate the actual number of patients with an HDV and HBV infection

Conclusions

- In a national payer database representing 80% of US claims, patients with an HDV infection were associated with significantly higher rates of decompensated cirrhosis, hepatocellular carcinoma, and liver transplant than patients with an HBV monoinfection
- HDV-infected patients had a significantly higher baseline Charlson Comorbidity Index score, as well as significantly higher baseline rates of comorbidities such as hypertension, smoking history, substance abuse, alcohol abuse, mental health disorders, and HCV and HIV infection, versus the HBV-monoinfected patients
- The study findings underscore the need for early screening and diagnosis, and eventual treatment of HDV infection to mitigate future disease progression
- In order to increase awareness and diagnosis of HDV infection, future studies should be conducted to focus on better understanding disease progression and cost burden related to HDV infection

References: **1.** World Health Organization. Hepatitis D; 7/8/21; **2.** Centers for Disease Control and Prevention. Hepatitis D Questions and Answers for Health Professionals; 3/9/20; **3.** Weinbaum CM, et al. Hepatology 2009;49(Suppl):S35-44; **4.** Roulot D, et al. J Hepatol 2020;73:1046-62; **5.** Romeo R, et al. Gastroenterology 2009;136:1629-38.
Acknowledgments: This study was funded by Gilead Sciences, Inc. STATinMED is a paid consulting institute to Gilead. **Disclosures:** **I. Jacobson:** Gilead, AbbVie, Aligos, ANA Therapeutics, Arbutus, Arrowhead, Assembly Biosciences, Atea, BMS, Durect, Eli Lilly, Enanta, Galmed, Genfit, GSK, Intercept, Janssen, Myr, Poptest, Redhill; **R. Gish:** Gilead, Abbott, AbbVie, Access Biologicals, Altimmune, Antios, Arena, Arrowhead, Bayer, Biocollections, BMS, CoCrystal, Cymabay, Dova, Durect, Dynavax, Eiger, Eisai, Enyo, eStudySite, Exelixis, Fibronostics, Forty-Seven Inc, Fujifilm/Wako, Genlantis, Gerson Lehrmann Group, Hatch Biofund, Helios, HepaTX, HepQuant, Intercept, Ionis, Janssen, Laboratory for Advanced Medicine, Lilly, Medimmune, MSD, Perspectum, Pfizer, Prodigy, Quest, RiboSciences, Salix, Shiongi, Spring Bank, Sonic Incytes, Topography Health, Venatorx, Viking; **J. Lim:** Gilead, Allergan, Conatus, Genfit, Intercept; A. **Kaushik, N. Kachru, Y. Liu,** and **R. Wong:** Gilead; **X. Sun:** nothing to disclose.

Presented at The Liver Meeting Digital Experience™ 2021 (TLMdX®) of the American Association for the Study of Liver Diseases (AASLD), Nov 12–15, 2021

©2021 Gilead Sciences, Inc. All rights reserved.