American Society of Clinical Oncology - Genitourinary Cancers Symposium (ASCO-GU) 2019 | February 14-16, 2019 | San Francisco, CA

Impact of Prostate Specific Antigen Doubling Time on Time to Metastasis and Overall Survival in Non-Metastatic Castration-resistant Prostate Cancer Patients

Stephen J. Freedland[1,2], Krishnan Ramaswamy[3], Stanislav Lechpammer[4], Jack Mardekian[3], Neil M. Schultz[5], Ahong Huang[6], Li Wang[6], Onur Baser[7], Daniel J. George[8]

[1]Division of Urology, Department of Surgery, Samuel Oschin Comprehensive Cancer Institute, Cedars-Sinai Medical Center, Los Angeles, CA, USA; [2]Section of Urology, Durham VA Medical Center, Durham, NC, USA; [3]Pfizer Inc., New York, NY, USA; [4]Pfizer Inc., San Francisco, CA, USA; [5]Astellas Pharma Inc., Northbrook, IL, USA; [6]STATinMED Research, Plano, TX, USA; [7]The University of Michigan, Ann Arbor, MI, USA; [8]Duke Cancer Institute, University School of Medicine, Durham, NC, USA

BACKGROUND

- Castration-resistant prostate cancer (CRPC) is an advanced form of prostate cancer (PC) that occurs despite surgical or pharmacological (androgen deprivation therapy [ADT]) castration.[1]
- Progression to CRPC typically presents as any of several factors including—increases in serum levels of prostate-specific antigen (PSA) levels, emergence of new metastases, or increased PSA levels in the absence of detectable metastases—a form of CRPC classified as non-metastatic (nmCRPC).[2,3]
- Important endpoints in CRPC management include PSA response, metastasis, and overall survival.[4,5]
- PSA doubling time (PSADT), defined as the time it would take for PSA levels to double, has been reported as a powerful marker for clinical endpoints.[6]
- Time to metastases has been shown to predict for overall survival in recurrent prostate cancer patient populations,[7] and metastasis-free survival has been recognized as a regulatory endpoint for new drug approvals in prostate cancer.[8]
- Several studies have examined the impact of post-surgical PSADT on patient outcomes, such as risk of metastasis and all-cause mortality.[6,9,10,11,12]
- To date, only one study has assessed the impact of PSADT on metastasis and all-cause mortality in veterans with nmCRPC.[13]

OBJECTIVES

- To validate the prognostic impact of PSADT on time to metastasis and overall survival among nmCRPC patients in the Veterans' Health Administration (VHA) database.

METHODS

Data Source

- Veterans' Health Administration (VHA) database from 01JAN2012-31DEC2016 was used to conduct the analysis.

Sample Selection

- See Figure 1 for patient selection criteria.

Study Measures

- Demographics and baseline clinical characteristics were measured for nmCRPC patients
- Post-index PSA values were used to calculate PSADT, and patients were grouped into 2-month PSADT cohorts as ≤2, >2 to ≤4, >4 to ≤6, >6 to ≤8, >8 to ≤10, >10 to ≤12 and >12 months.
- Metastasis was identified based on the presence of diagnosis codes for secondary malignant neoplasms (ICD-9-CM: 197-199.1) or a prescription claim for abiraterone acetate, enzalutamide, or docetaxel following the CRPC date.
- Overall survival was defined as time from the index date to the date of death.

Statistical Analysis

- Sociodemographic and clinical characteristics were described for all patients. Continuous variables were described as mean with standard deviation (SD), median with interquartile range (IQR) while categorical variables were described as counts (n) and percentages (%).
- Multivariable Cox proportional hazards regression was used to determine the hazards of metastasis and overall survival across PSADT groups, using the >12-month cohort as the reference. Age, race, and clinical comorbidities were included as covariates in these models.
- Adjusted hazard ratios (HR) and 95% confidence intervals (CI) were used to show the strength and significance of these findings, respectively. All statistical analyses were performed using SAS version 9.4 (SAS Institute Inc, Cary, NC).

Figure 1. Selection Criteria for nmCRPC Patients

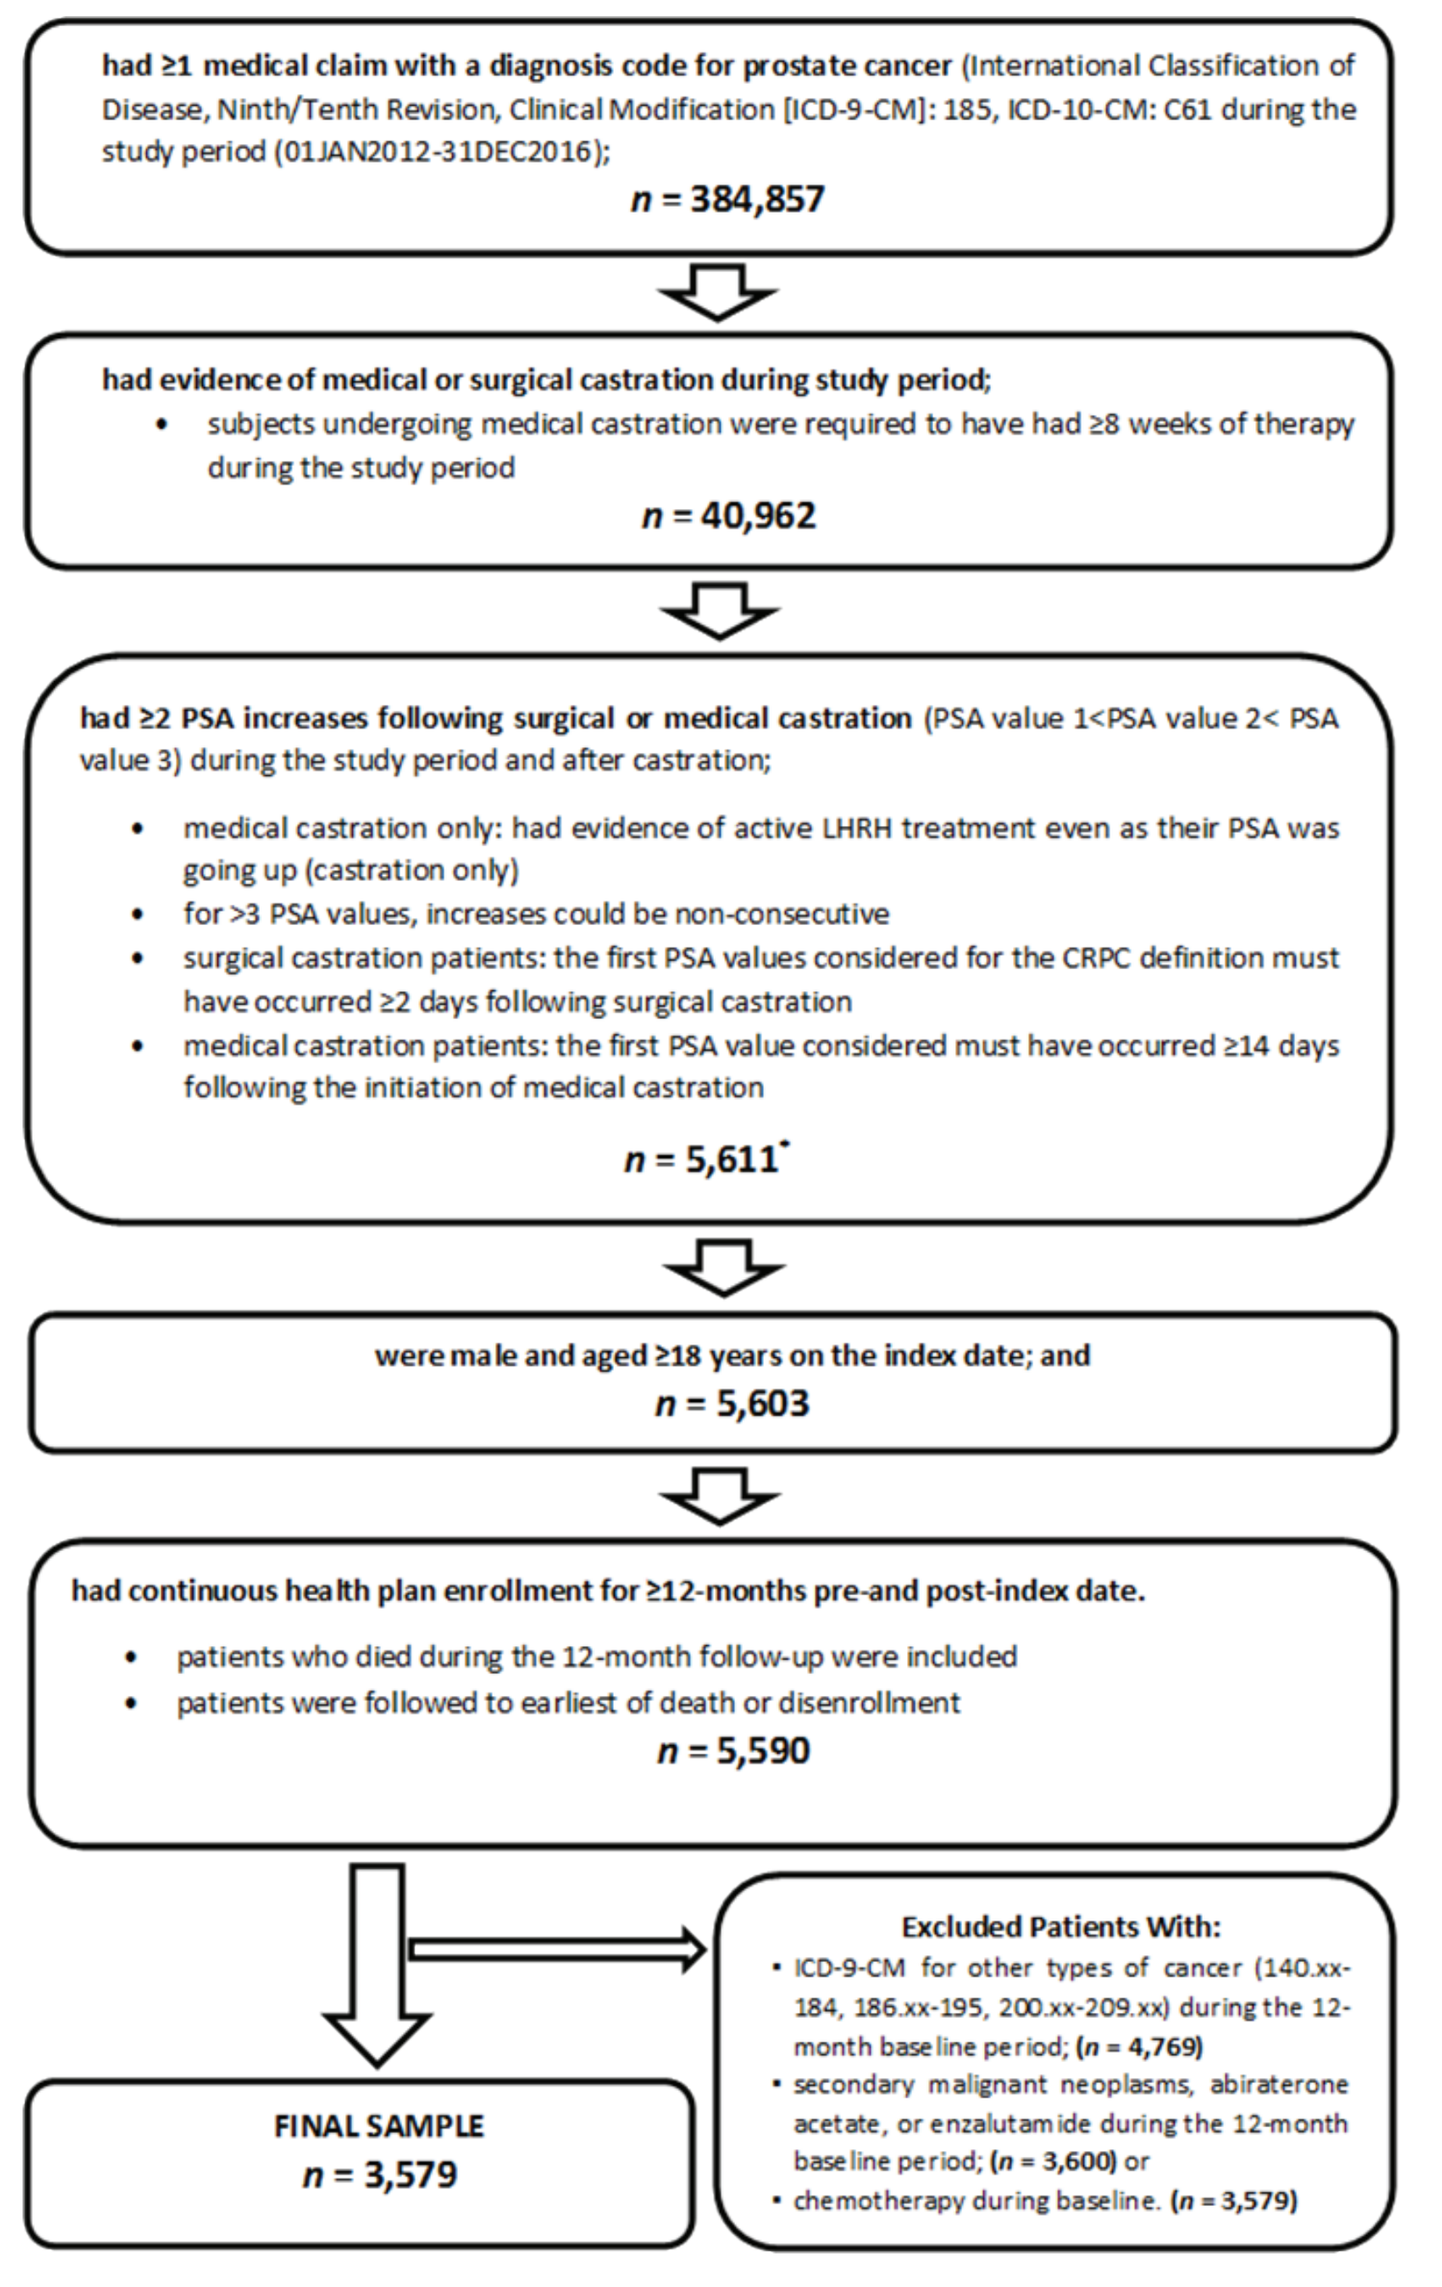
had ≥1 medical claim with a diagnosis code for prostate cancer (International Classification of Disease, Ninth/Tenth Revision, Clinical Modification [ICD-9-CM]: 185, ICD-10-CM: C61 during the study period (01JAN2012-31DEC2016);
n = 384,857
had evidence of medical or surgical castration during study period;
• subjects undergoing medical castration were required to have had ≥8 weeks of therapy during the study period
n = 40,962
had ≥2 PSA increases following surgical or medical castration (PSA value 1<PSA value 2< PSA value 3) during the study period and after castration;
• medical castration only: had evidence of active LHRH treatment even as their PSA was going up (castration only)
• for >3 PSA values, increases could be non-consecutive
• surgical castration patients: the first PSA values considered for the CRPC definition must have occurred ≥2 days following surgical castration
• medical castration patients: the first PSA value considered must have occurred ≥14 days following the initiation of medical castration
n = 5,611*
were male and aged ≥18 years on the index date; and
n = 5,603
had continuous health plan enrollment for ≥12-months pre-and post-index date.
• patients who died during the 12-month follow-up were included
• patients were followed to earliest of death or disenrollment
n = 5,590
Excluded Patients With:
• ICD-9-CM for other types of cancer (140.xx-184, 186.xx-195, 200.xx-209.xx) during the 12-month baseline period; (n = 4,769)
• secondary malignant neoplasms, abiraterone acetate, or enzalutamide during the 12-month baseline period; (n = 3,600) or
• chemotherapy during baseline. (n = 3,579)
FINAL SAMPLE
n = 3,579

*NOTE: For all patients, the third PSA value must have been ≥25% and 2 ng/ml higher than the first PSA value, and each of the three PSA values must have been ≥14 days apart. The third PSA value (PSA3) date needed to be during the identification period (01 January 2013-31 December 2015) and was designated as the CRPC date. The index date was defined as the CRPC date.

RESULTS

Patient Characteristics (Table 1)

- A total of 3,579 nmCRPC patients were identified after the application of the inclusion and exclusion criteria.
- Among these patients, the mean age was ~73 years, and 2,100 patients (59%) were white.
- The majority of patients had ≥ 1 comorbid condition; the most prevalent comorbidities were hypertension (73%), hyperlipidemia (58%), and type II diabetes (33%) and the median Quan-CCI score was 3.00.

PSA-related Measures (Table 2)

- Of the 3,579 patients identified, 2,800 patients (78%) had ≥3 measurable PSA readings and had a median PSA value of 3.30ng/ml (IQR: 12.76ng/ml).
- Among these patients, the median PSADT was 17 months (IQR: 113 months) and the following PSADT cohorts were identified: ≤2 months (n = 94, 3%), >2 to ≤4 months (n = 274, 10%), >4 to ≤ 6 months (n = 264, 9%), >6 to ≤8 months (n = 186, 7%), >8 to ≤10 months (n = 172, 6%), >10 to ≤12 months (n = 140, 5%), and >12 months (n = 1670, 60%).

Table 1. Characteristics of nmCRPC Patients

Demographics and Clinical Characteristics	nmCRPC Patients (n=3,579)	
	N/Mean	%/SD
Mean Age (years)	72.95	8.02
18-64	405	11%
65-74	1,245	35%
75-87	1,286	36%
>88	643	18%
Race		
White	2,100	59%
Black	1,120	31%
Other	75	2%
Unknown	284	8%
Baseline Clinical Characteristics		
Quan-Charlson Comorbidity Index Score, Median (IQR)	3.00	2.00
Individual Comorbidities		
Hypertension	2,612	73%
Stroke	272	8%
Angina pectoris perforation	44	1%
Arrhythmia	206	6%
Congestive heart failure	234	7%
Hyperlipidemia	2,076	58%
Type II diabetes	1,191	33%
Liver damage/abnormality	163	5%
Acute coronary syndrome/myocardial infarction	90	3%

n: number; SD: standard deviation; IQR: interquartile range; nmCRPC: non-metastatic castration-resistant prostate cancer

Table 2. PSA-related Measures

	nmCRPC Patients (n=3,579)	
	N/Mean	%/SD
Length of follow-up, months, Median (IQR)	27	18
Patients with ≥3 PSA values in the follow-up, including PSA3	2,800	78%
PSA values, including PSA3, ng/ml, Median (IQR)	3.30	12.76
PSADT, months, Median (IQR)	17	113
PSADT Groups		
≤2 months	94	3%
2-4 months	274	10%
4-6 months	264	9%
6-8 months	186	7%
8-10 months	172	6%
10-12 months	140	5%
>12 months	1,670	60%

n: number; SD: standard deviation; IQR: interquartile range; ng/ml: nanograms per milliliter; nmCRPC: non-metastatic castration-resistant prostate cancer; PSA: prostate-specific antigen; PSADT: prostate-specific antigen doubling time

PSADT and Metastasis Risk (Figure 2)

- Compared to the reference cohort, the HR for metastasis increased dramatically from 1.82 for the PSADT >10 to ≤12 months cohort to 33.77 for the PSADT ≤2 months cohort.

PSADT and Overall Survival (Figure 3)

- Compared to the reference cohort, the HR for death increased from 1.64 for PSADT >8 to ≤10 months cohort to 12.27 for the PSADT ≤2 months cohort.
- Although not statistically significant, the PSADT >10 to ≤12 months cohort was also associated with increased risk of death.

Figure 2. Adjusted Hazard Ratios of PSADT and Metastasis

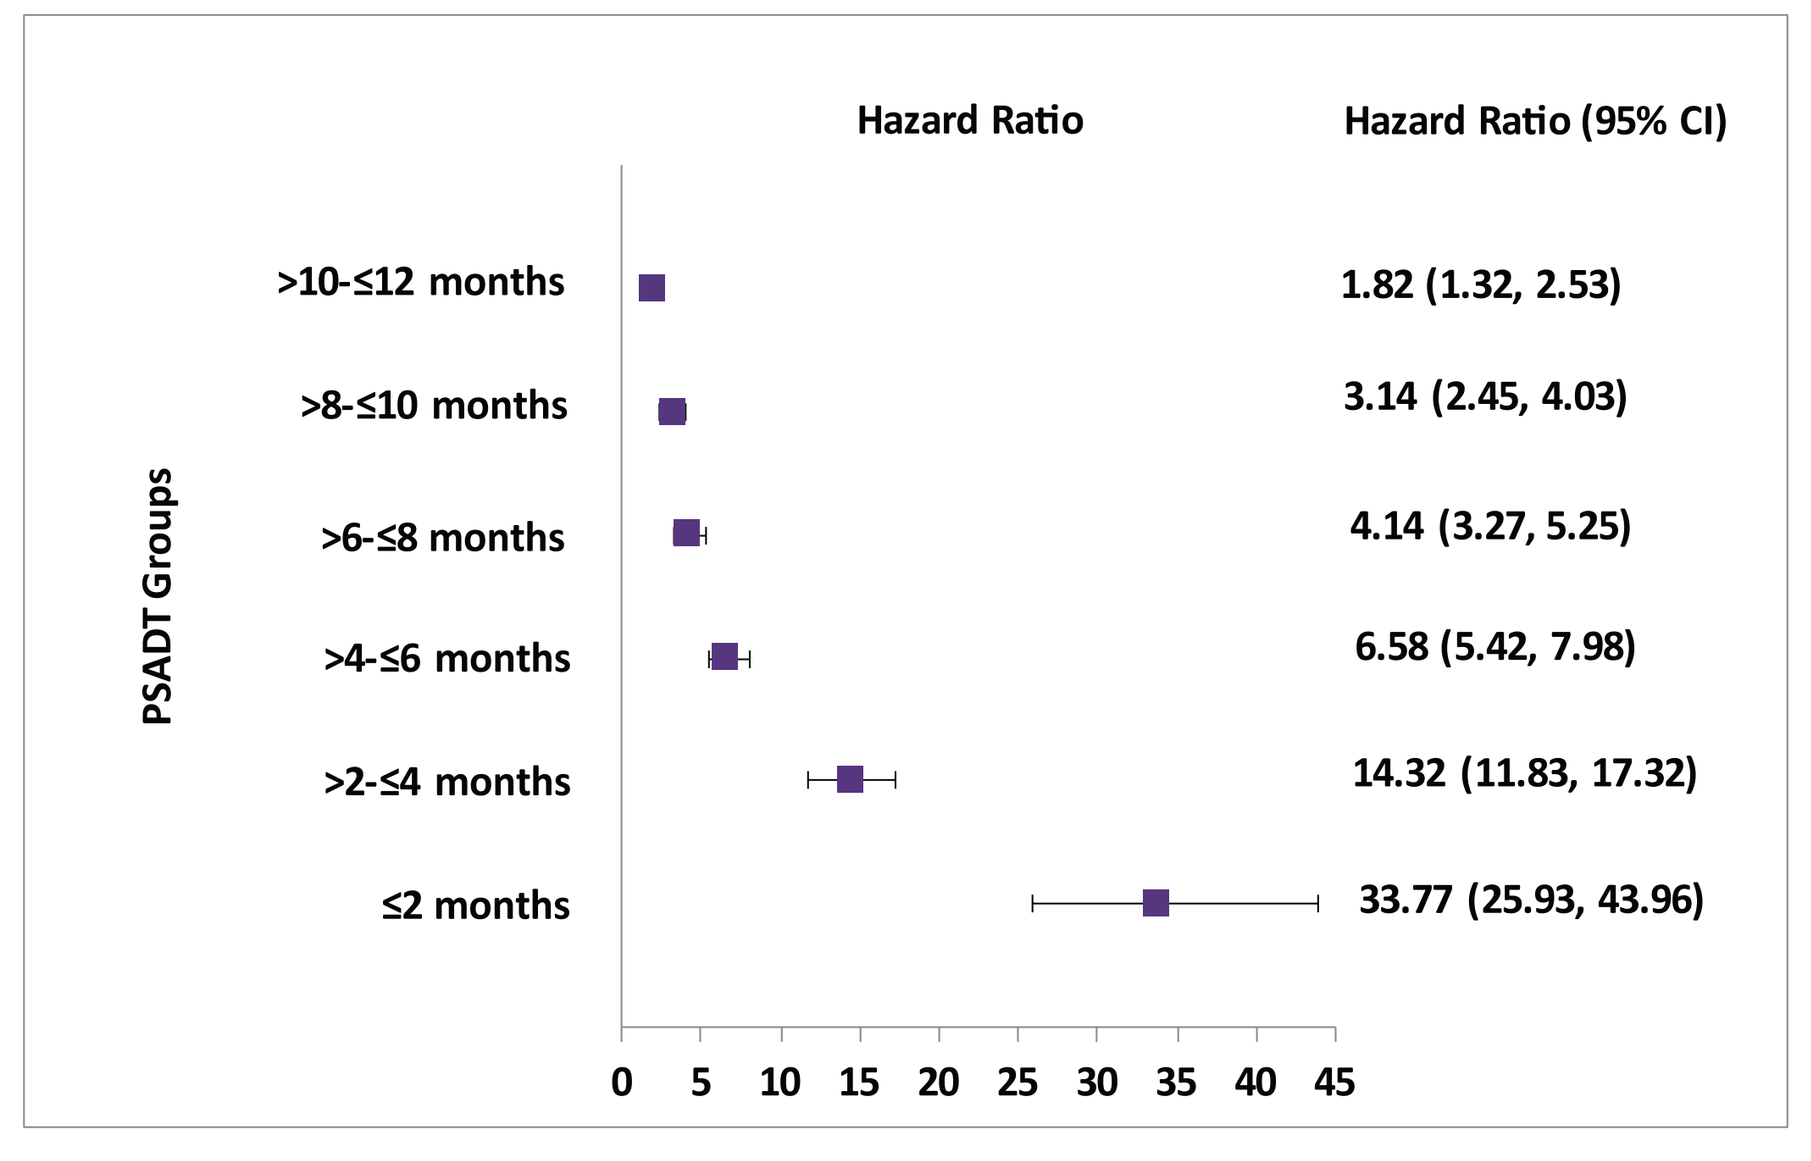
Hazard Ratio
Hazard Ratio (95% CI)
PSADT Groups
>10-≤12 months
1.82 (1.32, 2.53)
>8-≤10 months
3.14 (2.45, 4.03)
>6-≤8 months
4.14 (3.27, 5.25)
>4-≤6 months
6.58 (5.42, 7.98)
>2-≤4 months
14.32 (11.83, 17.32)
≤2 months
33.77 (25.93, 43.96)
0 5 10 15 20 25 30 35 40 45

CI: confidence interval; PSADT: prostate-specific antigen doubling time.

Figure 3. Adjusted Hazard Ratios of PSADT and Overall Survival

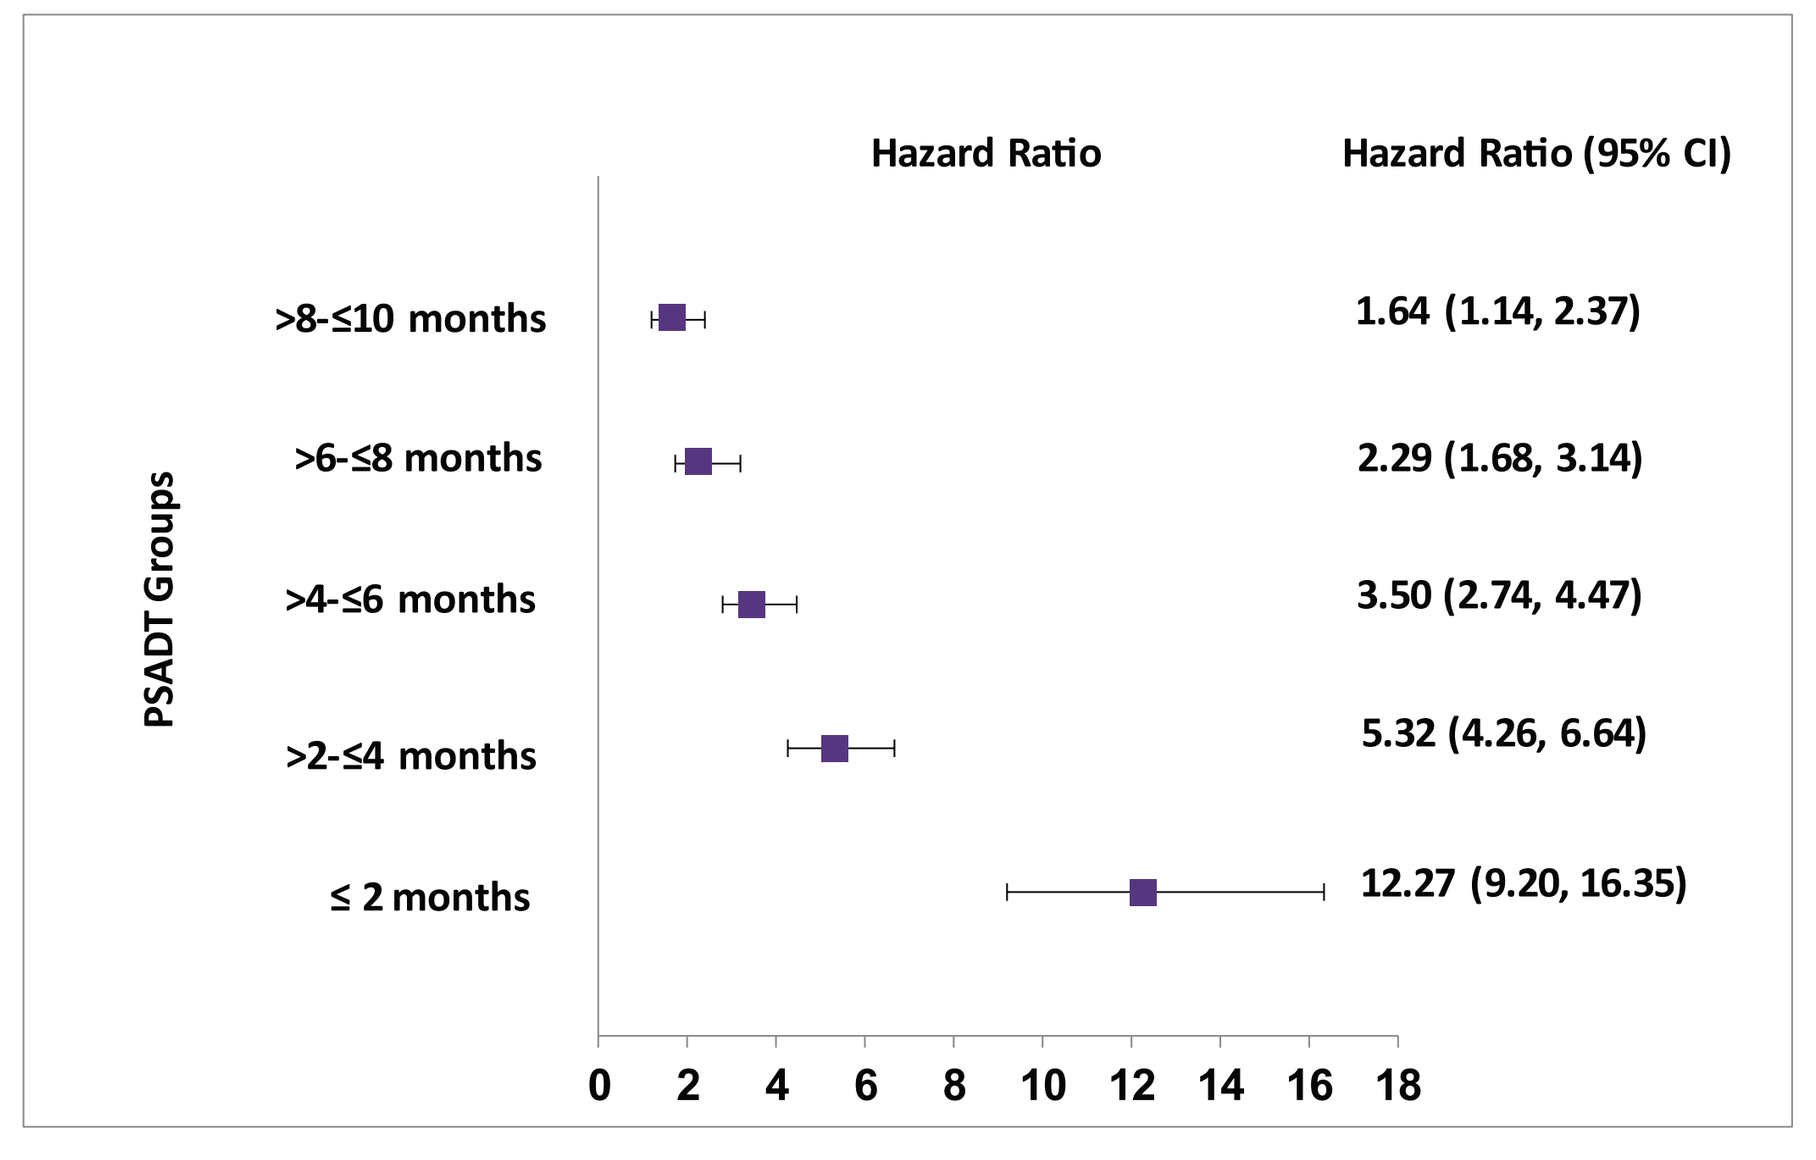
Hazard Ratio
Hazard Ratio (95% CI)
PSADT Groups
>8-≤10 months
1.64 (1.14, 2.37)
>6-≤8 months
2.29 (1.68, 3.14)
>4-≤6 months
3.50 (2.74, 4.47)
>2-≤4 months
5.32 (4.26, 6.64)
≤ 2 months
12.27 (9.20, 16.35)
0 2 4 6 8 10 12 14 16 18

CI: confidence interval; PSADT: prostate-specific antigen doubling time.

LIMITATIONS

- Pharmacy claims are not an indication that a medication was consumed as directed and do not account for non-prescription medication or over-the-counter samples.
- Diagnosis codes, used as evidence for the presence of disease may have been incorrectly coded or used as rule-out criteria rather than the indication of actual disease.
- Based on algorithms used in prior studies, PSA levels were used as a proxy for identifying CRPC. This may have resulted in misclassification of patients since the true CRPC status of these patients is unknown.

CONCLUSIONS

- **In this sample of male veterans with nmCRPC, majority had long PSADT (>12 months).**
- **PSADT is iassociated with metastasis and overall survival. Specifically, those with shorter PSADT (<10 months) have a heightened risk of metastasis and death.**
- **This retrospective study's findings align with the existing literature to confirm that PSADT is a strong predictor of the risk of metastasis and all-cause death among patients with nmCRPC.**
- **This information can be important for clinical trial managers and clinicians to better determine high-risk patients who are eligible for clinical trials or more aggressive treatments and identify low-risk patients who require conservative therapeutic strategies.**

REFERENCES

1 Kirby M, Hirst C, Crawford ED. Characterising the castration-resistant prostate cancer population: a systematic review. *Int J Clin Pract.* 2011;65(11):1180-92.
2 Engel-Nitz NM, Alemayehu B, Parry D, Nathan F. Differences in treatment patterns among patients with castration-resistant prostate cancer treated by oncologists versus urologists in a US managed care population. *Cancer Manag Res.* 2011;3:233
3 Luo J, Beer TM, Graff JN. Treatment of nonmetastatic castration-resistant prostate cancer. *Oncology.* 2016;30(4):336-44.
4 Scher HI, Morris MJ, Basch E, Heller G. End points and outcomes in castration-resistant prostate cancer: from clinical trials to clinical practice. *J Clin Oncol.* 2011;29(27):3695.
5 Maluf FC, Smaletz O, Herchenhorn D. Castration-resistant prostate cancer: systemic therapy in 2012. *Clinics.* 2012;67(4):389-94.
6 Teeter AE, Presti Jr JC, Aronson WJ, et al. Does PSADT after radical prostatectomy correlate with overall survival?—a report from the SEARCH database group. *Urology.* 2011;77(1):149-53.
7 Sweeney CJ, Chen YH, Carducci M, et al. Chemohormonal therapy in metastatic hormone-sensitive prostate cancer. *N Engl J Med.* 2015;373(8):737-46.
8 Schweizer MT, Zhou XC, Wang H, et al. Metastasis-free survival is associated with overall survival in men with PSA-recurrent prostate cancer treated with deferred androgen deprivation therapy. *Ann Oncol.* 2013;24(11):2881-6.
9 D'amico AV, Moul JW, Carroll PR, Sun L, Lubeck D, Chen MH. Surrogate end point for prostate cancer–specific mortality after radical prostatectomy or radiation therapy. *J Natl Cancer Inst.* 2003;95(18):1376-83.
10 Freedland SJ, Humphreys EB, Mangold LA, et al. Risk of prostate cancer–specific mortality following biochemical recurrence after radical prostatectomy. *JAMA.* 2005;294(4):433-9.
11 Arlen PM, Bianco F, Dahut WL, et al. Prostate Specific Antigen Working Group guidelines on prostate specific antigen doubling time. *J Urol.* 2008;179(6):2181-6.
12 Klayton TL, Ruth K, Buyyounouski MK, et al. PSA doubling time predicts for the development of distant metastases for patients who fail 3DCRT OR IMRT using the Phoenix definition. *Pract Radiat Oncol.* 2011;1(4):235.
13 Howard LE, Moreira DM, De Hoedt A, et al. Thresholds for PSA doubling time in men with non-metastatic castration-resistant prostate cancer. *BJU Int.* 2017;120(5B):E80-6.

ABSTRACT PLAIN LANGUAGE SUMMARY

Please scan this quick response (QR) code with your smartphone app to view a plain language summary of the accepted scientific abstract

Please scan this QR code with your smartphone app to view an electronic version of this poster. If you don't have a smartphone, access the poster via the internet at: xxxxxxxxxxxxxxxxxxx xxxxxxxxxxxxxxxxxxxxxx
Copies of this poster obtained through QR code are for personal use only and may not be reproduced without permission from ASCO® and the authors of this poster.

This research was sponsored by Pfizer, Inc.