Comparative Effectiveness and Safety Between Non-VKA Oral Anticoagulants in Nonvalvular Atrial Fibrillation Patients with Differential Duration of Treatment: An Analysis of the ARISTOPHANES Study

S. Deitelzweig[1], A. Keshishian[2,3], A. Kang[4], A. Dhamane[4], X. Luo[5], X. Li[4], N. Balachander[4], L. Rosenblatt[4], J. Mardekian[6], J. Jiang[4], M. Di Fusco[6], A. B. Garcia Reeves[4,7], H. Yuce[3], G. Y. H. Lip[8]

[1]Ochsner Clinic Foundation, Department of Hospital Medicine, New Orleans, LA, USA and The University of Queensland School of Medicine, Ochsner Clinical School, New Orleans, LA, USA; [2]STATinMED Research, Ann Arbor, MI, USA; [3]New York City College of Technology, City University of New York, New York, NY, USA; [4]Bristol-Myers Squibb Company, Lawrenceville, NJ, USA; [5]Pfizer, Inc., Groton, CT, USA; [6]Pfizer, Inc., New York, NY, USA; [7]University of North Carolina, Chapel Hill, NC, USA; [8]Liverpool Centre for Cardiovascular Science, University of Liverpool and Liverpool Heart & Chest Hospital, Liverpool, United Kingdom; and Aalborg Thrombosis Research Unit, Department of Clinical Medicine, Aalborg University, Aalborg, Denmark

INTRODUCTION

- Four non-VKA oral anticoagulants (NOACs) - apixaban, dabigatran, edoxaban, and rivaroxaban - have been approved in recent years for stroke prevention in atrial fibrillation (AF) patients, based on this non-inferior efficacy and safety compared to warfarin in randomized controlled trials (RTCs).[1]
- The ARISTOPHANES (Anticoagulants for Reduction In STroke: Observational Pooled analysis on Health outcomes ANd Experience of patientS; ClinicalTrials.gov ID: NCT03087487) study showed that apixaban was associated with a lower risk of stroke/systemic embolism (SE) and major bleeding (MB) versus dabigatran and rivaroxaban; dabigatran was associated with a lower risk of MB and similar risk of stroke/SE compared to rivaroxaban.[2]
- There is evidence from the RCTs for the differential clinical effects of short-term (1-3 months) versus long-term (>1 year) OAC treatment.[3] However, limited real-world evidence exists on the comparative effectiveness and safety of NOAC versus NOAC by treatment duration, especially after a long-term treatment.[4]

OBJECTIVES

This subgroup analysis of the ARISTOPHANES study aimed to assess long-term use of NOAC versus NOAC by evaluating the risk of stroke/SE and MB among nonvalvular AF (NVAF) patients by treatment duration (<1 and ≥1 year).

METHODS

Data Source

This was a retrospective observational study using US Centers for Medicare and Medicaid Services, Truven MarketScan® Commercial Claims and Encounter and Medicare Supplemental and Coordination of Benefits Database ("MarketScan"), IMS PharMetrics Plus™ Database ("PharMetrics"), Optum Clinformatics™ Data Mart ("Optum"), and the Humana Research Database ("Humana"), covering >180 million beneficiaries annually (~56% of the US population).

Study Sample

- AF patients (International Classification of Diseases, 9th Revision, Clinical Modification [ICD-9-CM] code 427.31) aged ≥18 years with ≥1 pharmacy claim for apixaban, dabigatran, edoxaban, or rivaroxaban from January 1, 2013 to September 30, 2015 were included in the study.
- The first NOAC prescription date was designated as the index date.
- Patients were required to have continuous medical and pharmacy health plan enrollment for ≥12 months prior to the index date (baseline period).
- Inclusion and exclusion criteria are shown in Figure 1.
- The follow-up period was from the day after the index date to 30 days post discontinuation date, switch date, death (only inpatient death for the commercial databases and all-cause death for Medicare database), end of continuous health plan enrollment, or the end of the study period, whichever occurred first.

Figure 1. Patient Selection Criteria

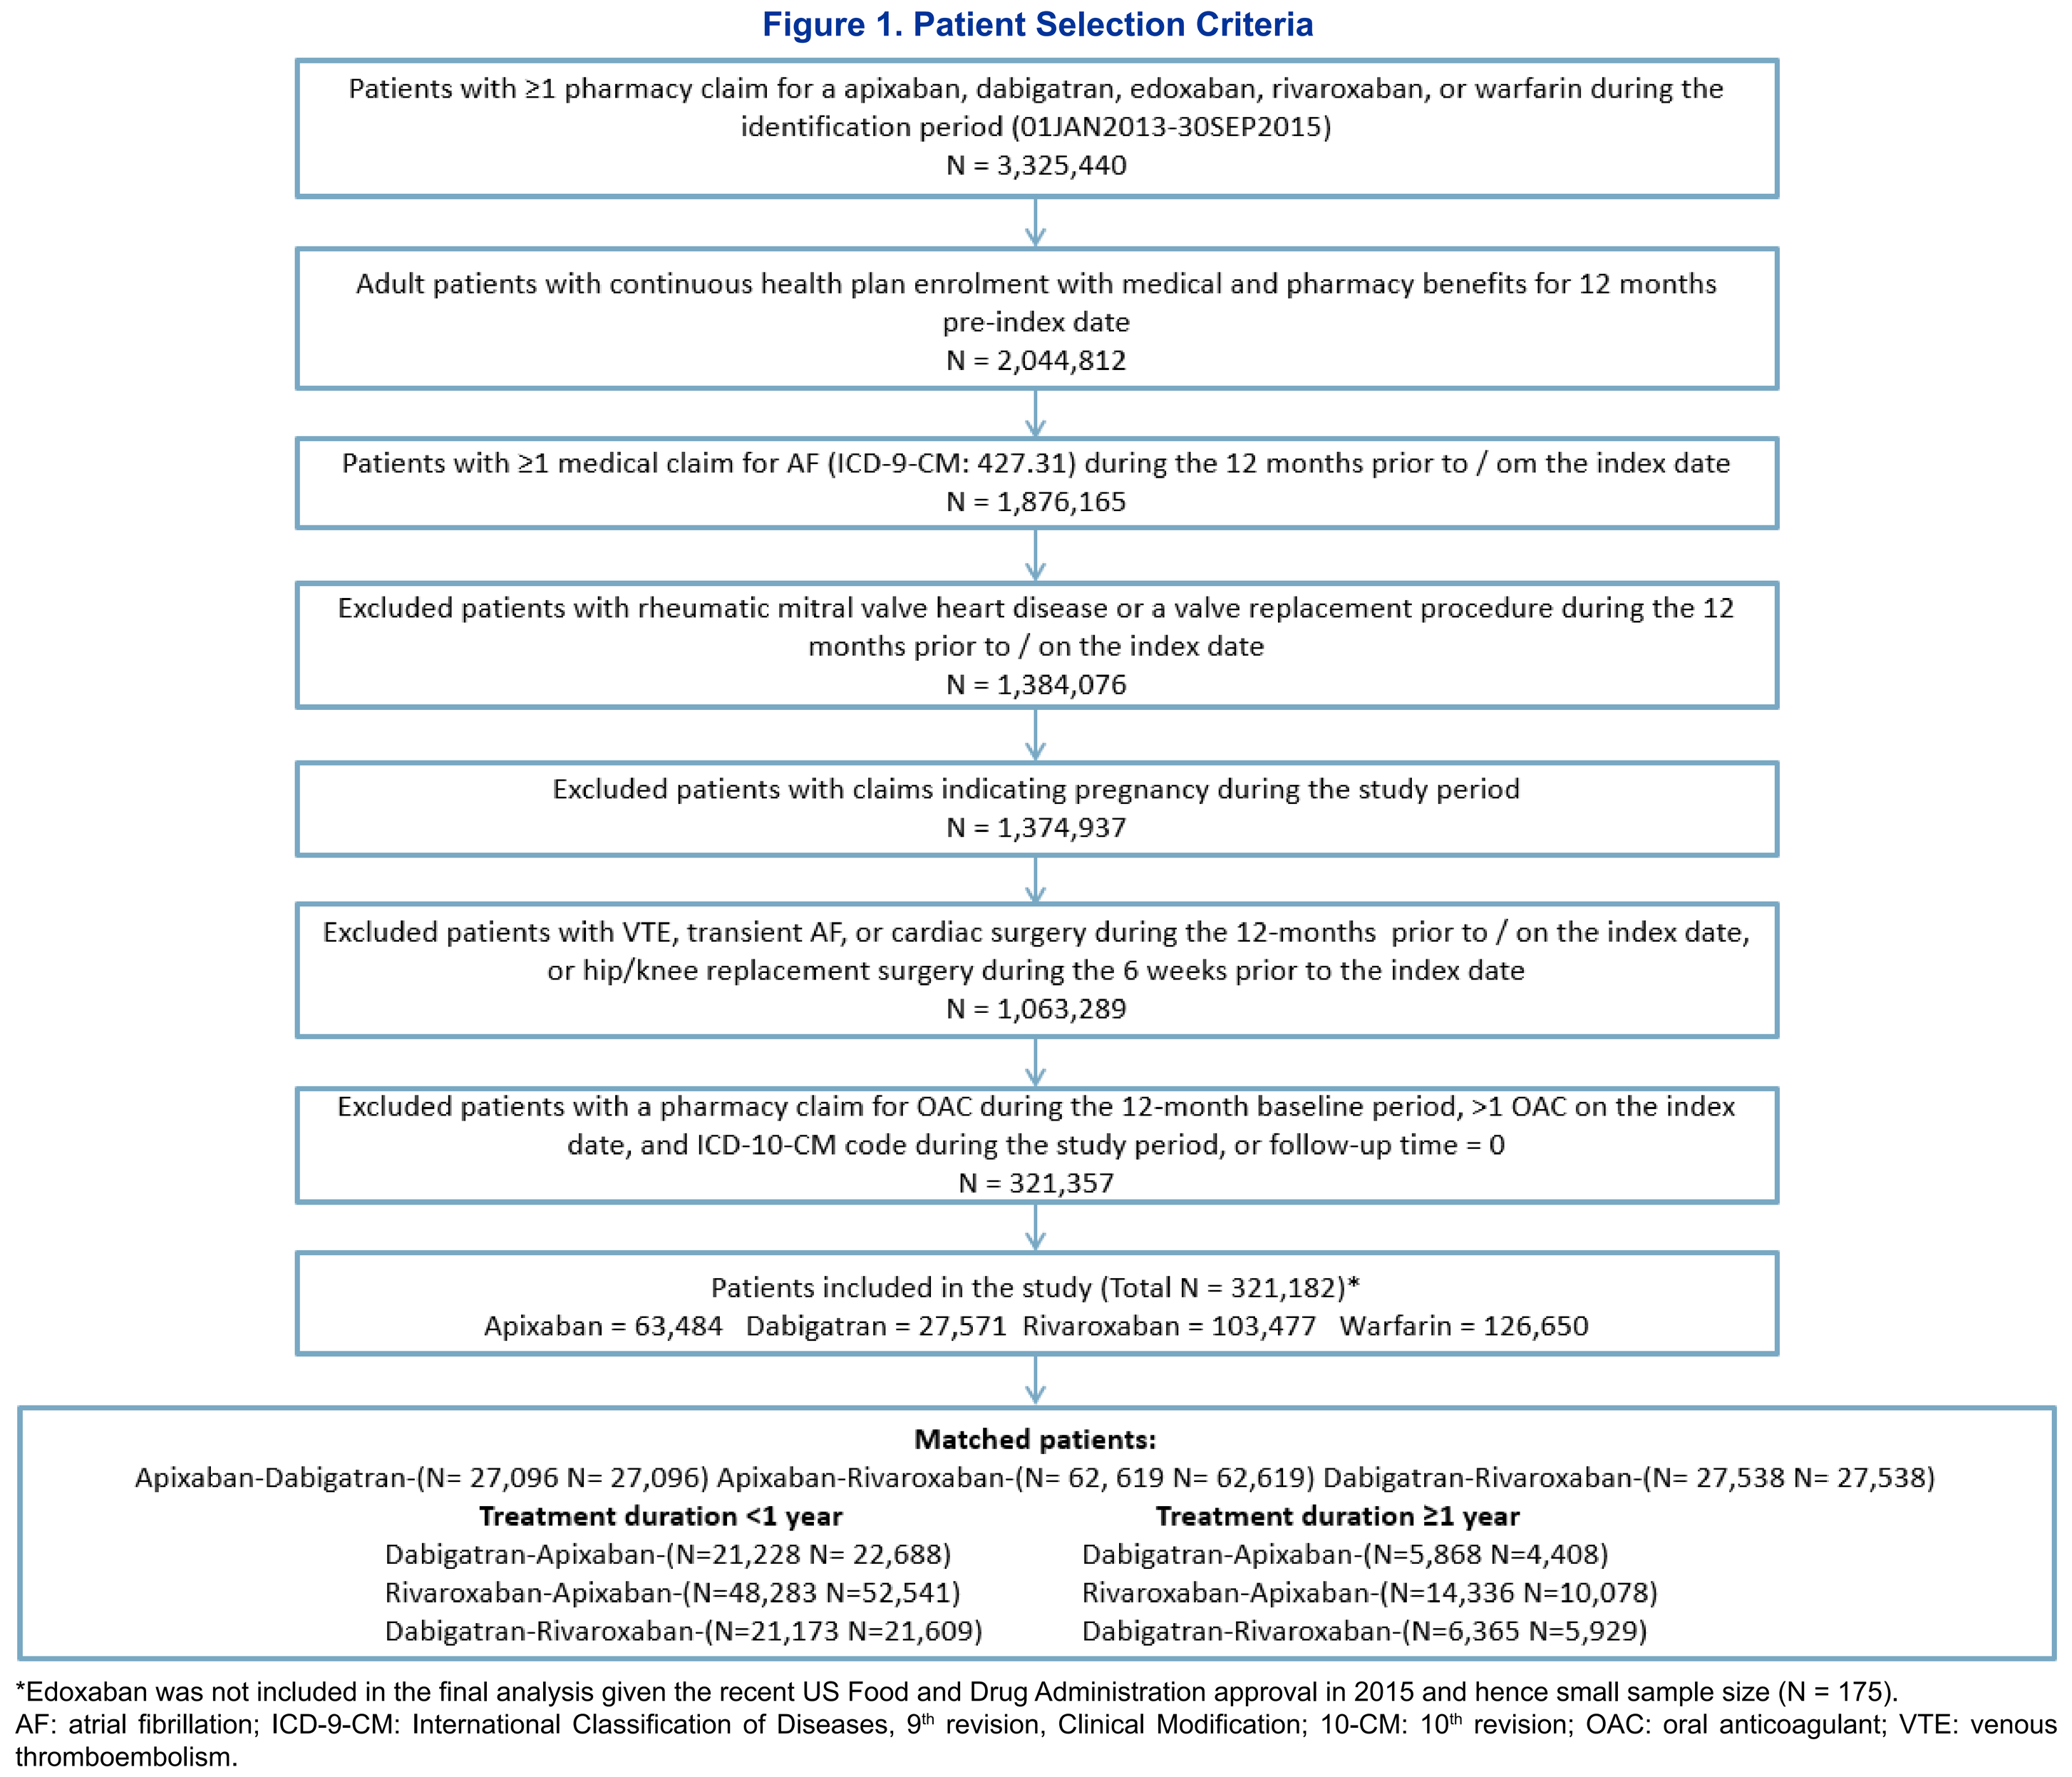
Patients with ≥1 pharmacy claim for a apixaban, dabigatran, edoxaban, rivaroxaban, or warfarin during the identification period (01JAN2013-30SEP2015)
N = 3,325,440
Adult patients with continuous health plan enrolment with medical and pharmacy benefits for 12 months pre-index date
N = 2,044,812
Patients with ≥1 medical claim for AF (ICD-9-CM: 427.31) during the 12 months prior to / om the index date
N = 1,876,165
Excluded patients with rheumatic mitral valve heart disease or a valve replacement procedure during the 12 months prior to / on the index date
N = 1,384,076
Excluded patients with claims indicating pregnancy during the study period
N = 1,374,937
Excluded patients with VTE, transient AF, or cardiac surgery during the 12-months prior to / on the index date, or hip/knee replacement surgery during the 6 weeks prior to the index date
N = 1,063,289
Excluded patients with a pharmacy claim for OAC during the 12-month baseline period, >1 OAC on the index date, and ICD-10-CM code during the study period, or follow-up time = 0
N = 321,357
Patients included in the study (Total N = 321,182)*
Apixaban = 63,484 Dabigatran = 27,571 Rivaroxaban = 103,477 Warfarin = 126,650
Matched patients:
Apixaban-Dabigatran-(N= 27,096 N= 27,096) Apixaban-Rivaroxaban-(N= 62, 619 N= 62,619) Dabigatran-Rivaroxaban-(N= 27,538 N= 27,538)
Treatment duration <1 year
Dabigatran-Apixaban-(N=21,228 N= 22,688)
Rivaroxaban-Apixaban-(N=48,283 N=52,541)
Dabigatran-Rivaroxaban-(N=21,173 N=21,609)
Treatment duration ≥1 year
Dabigatran-Apixaban-(N=5,868 N=4,408)
Rivaroxaban-Apixaban-(N=14,336 N=10,078)
Dabigatran-Rivaroxaban-(N=6,365 N=5,929)

*Edoxaban was not included in the final analysis given the recent US Food and Drug Administration approval in 2015 and hence small sample size (N = 175).
AF: atrial fibrillation; ICD-9-CM: International Classification of Diseases, 9th revision, Clinical Modification; 10-CM: 10th revision; OAC: oral anticoagulant; VTE: venous thromboembolism.

Outcomes

- Outcome measures were time to first stroke/SE and MB events based on the principal or first-listed ICD-9-CM diagnosis on an inpatient claim. The diagnosis codes used for stroke/SE and MB were based on validated administrative claims-based algorithms and the International Society on Thrombosis and Haemostasis' definition of MB.[5,6]
- Stroke/SE was further categorized into ischemic stroke, hemorrhagic stroke, and SE; MB was further categorized into gastrointestinal (GI) bleeding, intracranial hemorrhage, and other MB.

METHODS - cont'd

Statistical Analysis

- Patients were 1:1 matched based on the propensity scores generated by logistic regression using the nearest neighbor matching method without replacement with a caliper of 0.01.[7] Demographics, Charlson Comorbidity Index score, baseline bleeding and stroke/SE history, comorbidities, and baseline co-medications were included in the models.
- After 1:1 propensity score matching (PSM) in each database between NOACs (apixaban-dabigatran, dabigatran-rivaroxaban, and dabigatran-rivaroxaban), the resulting patient records were pooled.
- Within the matched NOAC-NOAC cohorts, patients were analyzed separately by the length of treatment duration (<1 and ≥1 year). The balance of matched covariates was checked (Tables 1 and 2).
- The incidence of stroke/SE and MB in the PSM-adjusted cohorts was calculated as the number of stroke/SE and MB events, respectively, per 100 person-years.
- Cox proportional hazards models with robust sandwich estimates were used to estimate hazard ratios of stroke/SE and MB during observed treatment duration.

RESULTS

- After applying the selection criteria, a total of 194,532 NVAF patients were identified, including 63,484 apixaban, 27,571 dabigatran, and 103,477 rivaroxaban patients. After PSM, 27,096 apixaban-dabigatran, 62,619 apixaban-rivaroxaban, and 27,538 dabigatran-rivaroxaban PSM pairs were included. Edoxaban was not included in the final patient population given the small sample size (N = 175).
- Given the variety of time for market entry, around 16%, 22%, and 23% of apixaban, dabigatran, and rivaroxaban patients, respectively, had treatment duration ≥1 year.
- After separating the matched drug cohorts by treatment duration, baseline characteristics remained balanced. The mean treatment duration for patients with shorter (<1 year) versus longer (≥1 year) duration was ~4 months versus 18-21 months, respectively, across the three matched cohorts.
- The mean ages for the <1-year treatment duration cohorts were 71-73 years with mean CHA_2DS_2-VASc scores of 3.2-3.5 and HAS-BLED scores ranging from 2.6-2.9 (Table 1).
- The mean ages for the ≥1-year follow-up cohorts were 73-75 years with mean CHA_2DS_2-VASc scores of 3.5-3.7 and HAS-BLED scores of 2.8-2.9 (Table 2).

Table 1. PSM Baseline Characteristics of Patients with Treatment Duration <1 year

	Dabigatran Cohort		Apixaban Cohort		Rivaroxaban Cohort		Apixaban Cohort		Rivaroxaban Cohort		Dabigatran Cohort	
	(N = 21,228)		(N = 22,688)		(N = 48,283)		(N = 52,541)		(N = 21,173)		(N = 21,609)	
	N/Mean	%/SD	N/Mean	%/SD	N/Mean	%/SD	N/Mean	%/SD	N/Mean	%/SD	N/Mean	%/SD
Age	71.0	11.7	71.3	11.7	72.4	11.9	72.7	11.8	70.9	11.9	70.8	11.8
<65	5,766	27.2%	6,010	26.5%	11,671	24.2%	12,406	23.6%	5,900	27.9%	6,010	27.8%
65-74	6,849	32.3%	7,400	32.6%	14,374	29.8%	15,617	29.7%	6,705	31.7%	6,954	32.2%
75-79	3,550	16.7%	3,794	16.7%	7,802	16.2%	8,682	16.5%	3,515	16.6%	3,572	16.5%
≥80	5,063	23.9%	5,484	24.2%	14,436	29.9%	15,836	30.1%	5,053	23.9%	5,073	23.5%
Gender												
Male	12,787	60.2%	13,608	60.0%	27,042	56.0%	29,359	55.9%	12,942	61.1%	13,123	60.7%
Female	8,441	39.8%	9,080	40.0%	21,241	44.0%	23,182	44.1%	8,231	38.9%	8,486	39.3%
U.S. Geographic Region												
Northeast	3,752	17.7%	4,049	17.8%	6,833	14.2%	7,709	14.7%	3,920	18.5%	3,937	18.2%
North Central	5,039	23.7%	5,429	23.9%	10,525	21.8%	11,465	21.8%	5,097	24.1%	5,104	23.6%
South	8,764	41.3%	9,301	41.0%	22,955	47.5%	24,797	47.2%	8,383	39.6%	8,791	40.7%
West	3,574	16.8%	3,803	16.8%	7,744	16.0%	8,313	15.8%	3,631	17.1%	3,643	16.9%
Other	99	0.5%	106	0.5%	226	0.5%	257	0.5%	142	0.7%	134	0.6%
Baseline Comorbidity												
Deyo-Charlson Comorbidity Index	2.4	2.5	2.4	2.5	2.7	2.7	2.8	2.6	2.5	2.5	2.4	2.5
CHA_2DS_2-VASc Score	3.3	1.8	3.3	1.8	3.5	1.8	3.5	1.8	3.2	1.8	3.2	1.8
HAS-BLED Score	2.6	1.4	2.6	1.4	2.8	1.4	2.9	1.4	2.6	1.4	2.6	1.4
Bleeding History	3,450	16.3%	3,641	16.0%	9,116	18.9%	9,893	18.8%	3,603	17.0%	3,481	16.1%
Non-stroke/ SE Peripheral Vascular Disease	9,694	45.7%	10,207	45.0%	24,261	50.2%	26,541	50.5%	9,670	45.7%	9,778	45.2%
Stroke	2,022	9.5%	2,193	9.7%	5,117	10.6%	5,850	11.1%	2,042	9.6%	2,031	9.4%
Transient Ischemic Attack (TIA)	1,254	5.9%	1,375	6.1%	3,204	6.6%	3,640	6.9%	1,269	6.0%	1,259	5.8%
Anemia and Coagulation Defects	4,463	21.0%	4,653	20.5%	11,836	24.5%	12,877	24.5%	4,498	21.2%	4,497	20.8%

[a]As the INR value was not available in the data, a modified HAS-BLED score was calculated using a range of 0 to 8.

CHA_2DS_2-VASc: congestive heart failure, hypertension, age ≥75 years, diabetes mellitus, prior stroke or transient ischemic attack or thromboembolism, vascular disease, age 65-74 years, sex category; HAS-BLED: hypertension, abnormal renal and liver function, stroke, bleeding, labile INRs, elderly, drugs, and alcohol; PSM: propensity score matching; SD: standard deviation; SE: systemic embolism

Table 2. PSM Baseline Characteristics of Patients with Treatment Duration ≥1 year

	Dabigatran Cohort		Apixaban Cohort		Rivaroxaban Cohort		Apixaban Cohort		Rivaroxaban Cohort		Dabigatran Cohort	
	(N = 5,868)		(N = 4,408)		(N = 14,336)		(N = 10,078)		(N = 6,365)		(N = 5,929)	
	N/Mean	%/SD	N/Mean	%/SD	N/Mean	%/SD	N/Mean	%/SD	N/Mean	%/SD	N/Mean	%/SD
Age	73.7	9.8	73.9	10.3	74.4	10.1	74.8	10.3	73.2	9.9	73.6	9.8
<65	961	16.4%	745	16.9%	2,259	15.8%	1,584	15.7%	1,126	17.7%	996	16.8%
65-74	2,034	34.7%	1,474	33.4%	4,625	32.3%	3,065	30.4%	2,203	34.6%	2,046	34.5%
75-79	1,212	20.7%	924	21.0%	2,726	19.0%	2,013	20.0%	1,378	21.6%	1,223	20.6%
≥80	1,661	28.3%	1,265	28.7%	4,726	33.0%	3,416	33.9%	1,658	26.0%	1,664	28.1%
Gender												
Male	3,219	54.9%	2,461	55.8%	7,277	50.8%	5,194	51.5%	3,530	55.5%	3,274	55.2%
Female	2,649	45.1%	1,947	44.2%	7,059	49.2%	4,884	48.5%	2,835	44.5%	2,655	44.8%
U.S. Geographic Region												
Northeast	1,196	20.4%	844	19.1%	2,375	16.6%	1,662	16.5%	1,323	20.8%	1,229	20.7%
North Central	1,475	25.1%	1,070	24.3%	3,064	21.4%	2,182	21.7%	1,541	24.2%	1,490	25.1%
South	2,159	36.8%	1,705	38.7%	6,582	45.9%	4,546	45.1%	2,418	38.0%	2,161	36.4%
West	1,027	17.5%	784	17.8%	2,277	15.9%	1,662	16.5%	1,065	16.7%	1,033	17.4%
Other	11	0.2%	5	0.1%	38	0.3%	26	0.3%	18	0.3%	16	0.3%
Baseline Comorbidity												
Deyo-Charlson Comorbidity Index	2.4	2.3	2.2	2.2	2.5	2.4	2.5	2.4	2.4	2.3	2.4	2.3
CHA_2DS_2-VASc Score	3.6	1.6	3.5	1.6	3.7	1.6	3.7	1.6	3.5	1.6	3.6	1.6
HAS-BLED Score	2.8	1.2	2.8	1.3	2.9	1.3	2.9	1.3	2.8	1.3	2.8	1.2
Bleeding History	923	15.7%	697	15.8%	2,401	16.7%	1,829	18.1%	953	15.0%	932	15.7%
Non-stroke/ SE Peripheral Vascular Disease	2,711	46.2%	2,032	46.1%	7,114	49.6%	5,089	50.5%	2,954	46.4%	2,727	46.0%
Stroke	670	11.4%	425	9.6%	1,762	12.3%	1,160	11.5%	718	11.3%	670	11.3%
Transient Ischemic Attack (TIA)	468	8.0%	297	6.7%	1,150	8.0%	804	8.0%	467	7.3%	466	7.9%
Anemia and Coagulation Defects	1,237	21.1%	970	22.0%	3,208	22.4%	2,412	23.9%	1,277	20.1%	1,251	21.1%

[a]As the INR value was not available in the data, a modified HAS-BLED score was calculated using a range of 0 to 8.

CHA_2DS_2-VASc: congestive heart failure, hypertension, age ≥75 years, diabetes mellitus, prior stroke or transient ischemic attack or thromboembolism, vascular disease, age 65-74 years, sex category; HAS-BLED: hypertension, abnormal renal and liver function, stroke, bleeding, labile INRs, elderly, drugs, and alcohol; PSM: propensity score matching; SD: standard deviation; SE: systemic embolism

RESULTS (cont'd)

- For patients with <1 year duration of treatment, apixaban patients had a lower risk of stroke/SE and MB when compared to dabigatran and rivaroxaban patients; dabigatran patients had a lower risk of MB when compared to rivaroxaban patients, but a similar risk of stroke/SE (Figure 2).
- For patients with ≥1 year duration of treatment, apixaban patients had similar risk of stroke/SE and MB when compared to dabigatran patients; apixaban patients had a similar risk of stroke/SE when compared to rivaroxaban patients, but lower risk of MB; and dabigatran had a similar risk of stroke/SE when compared to rivaroxaban patients, but lower risk of MB (Figure 2).

Figure 2. Hazard Ratios and Incidence Rates of Stroke/SE and MB for Propensity Scored-Matched Patients

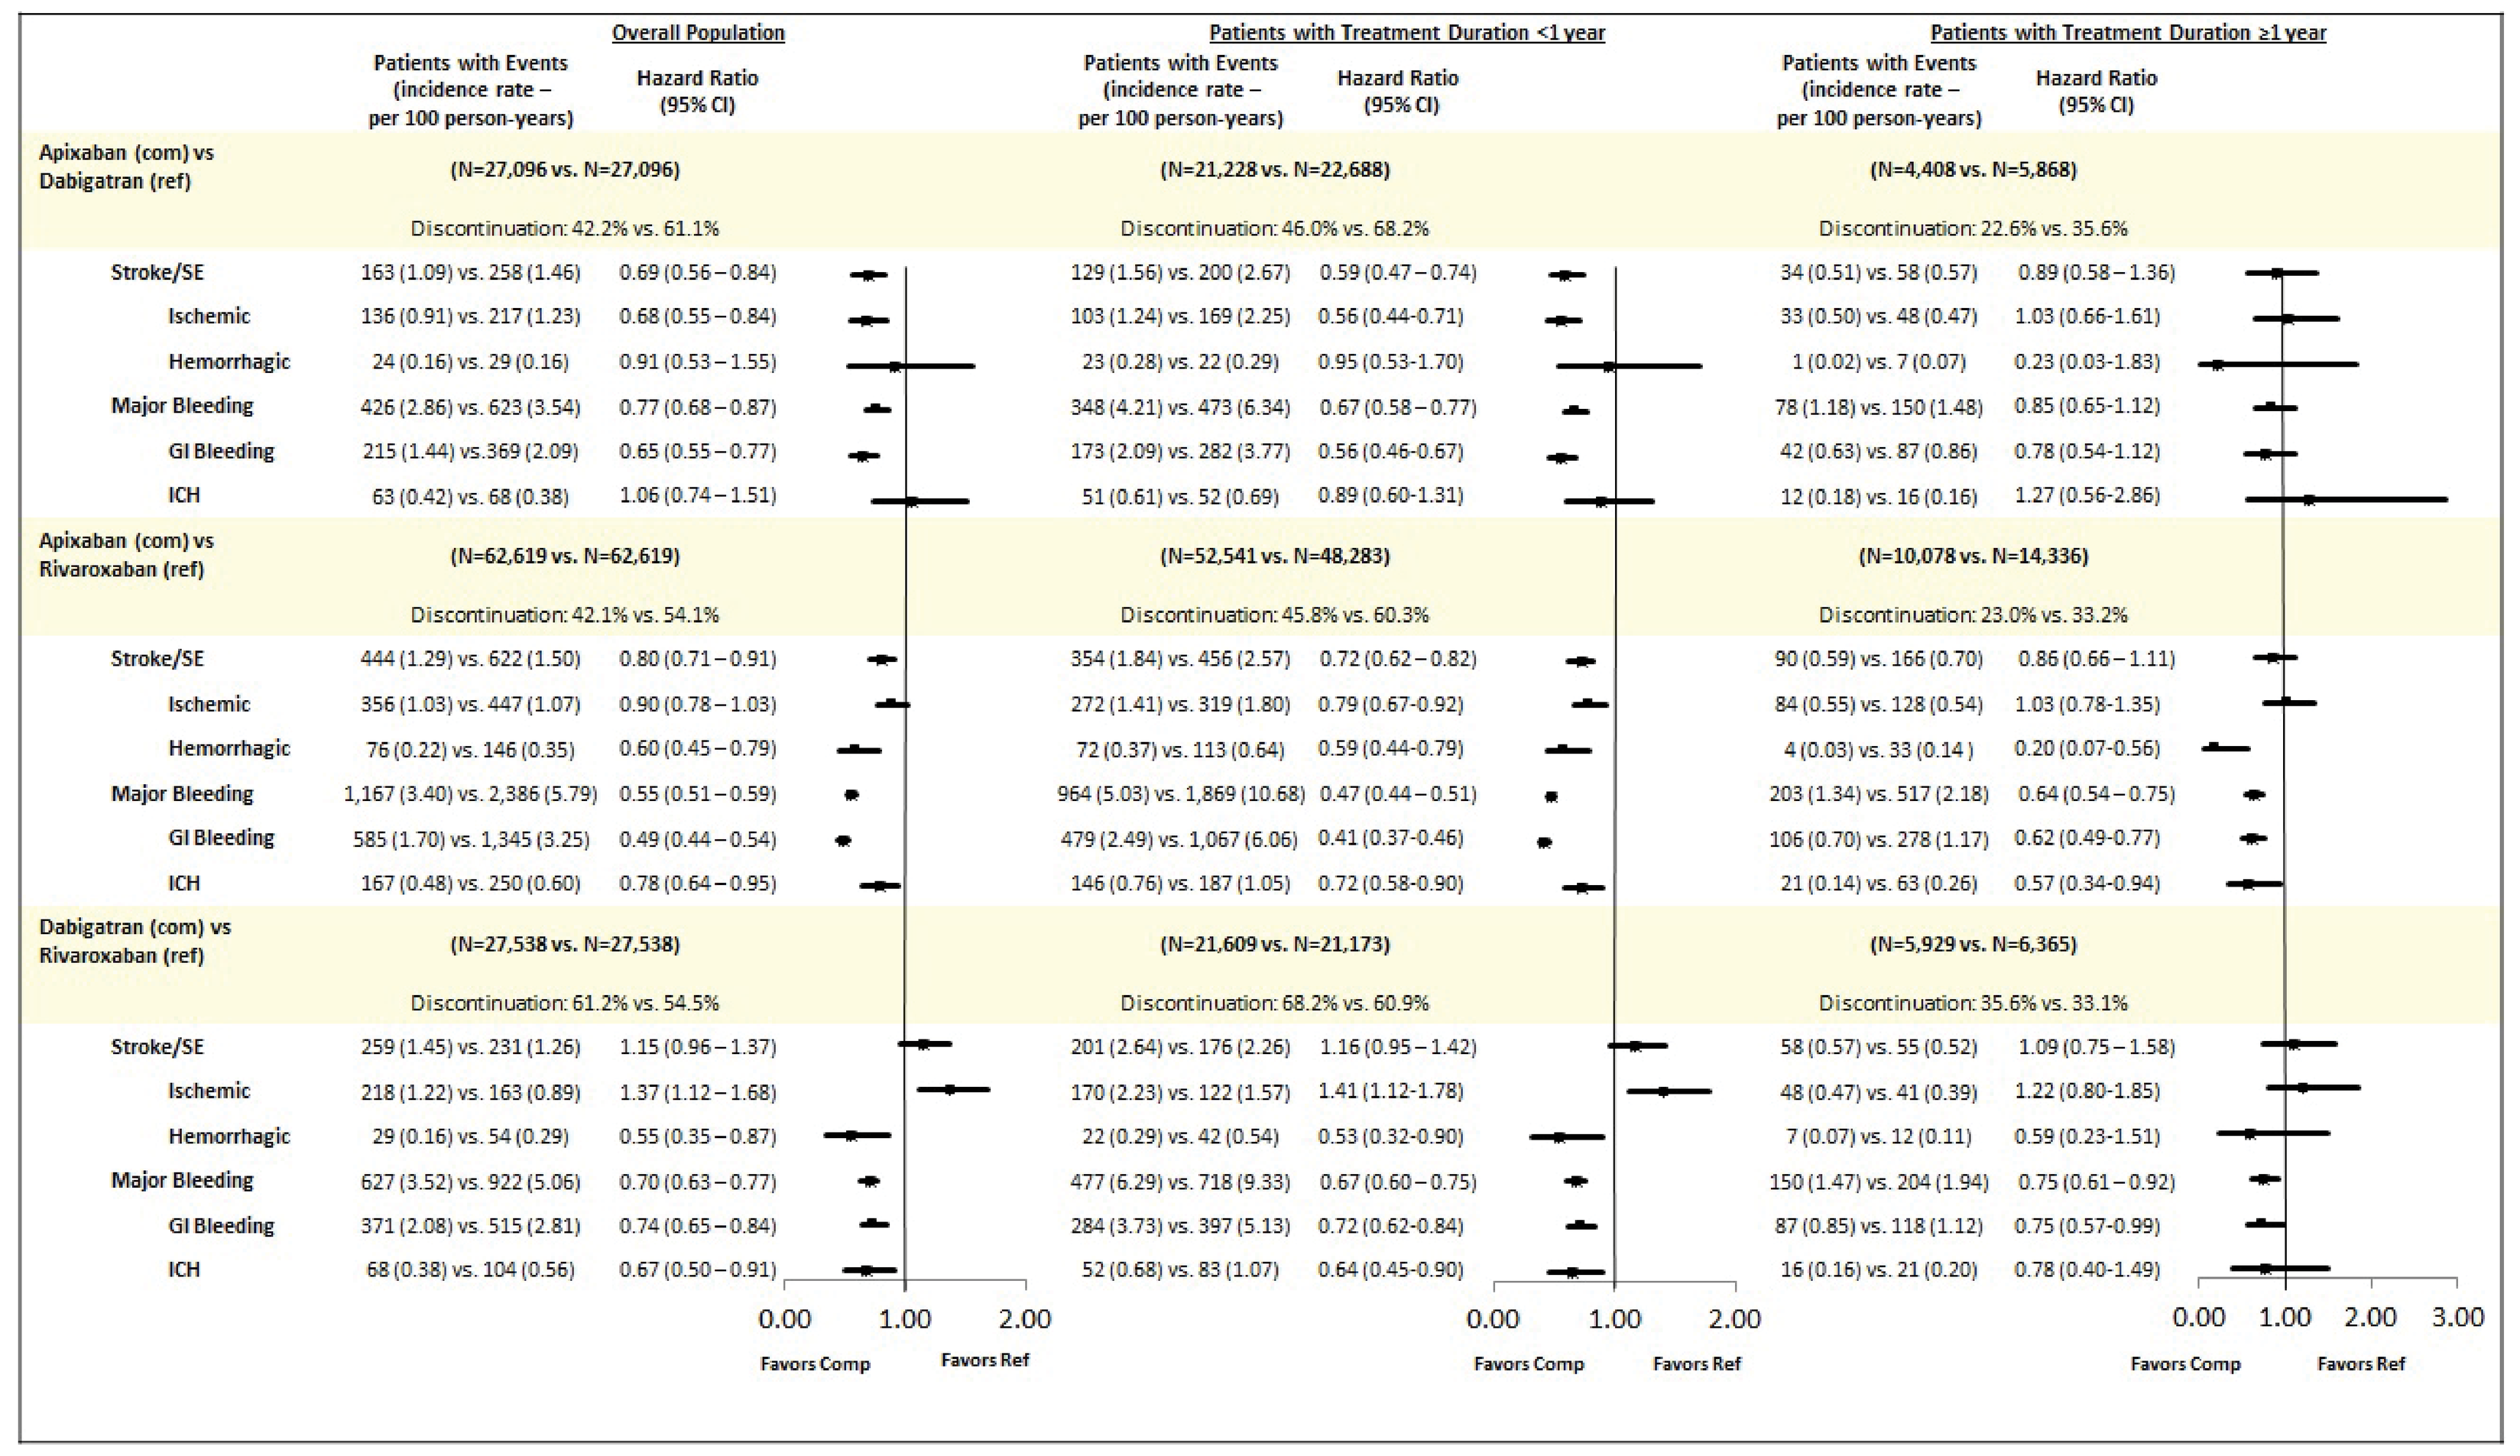
Overall Population
Patients with Treatment Duration <1 year
Patients with Treatment Duration ≥1 year
Patients with Events (incidence rate – per 100 person-years)
Hazard Ratio (95% CI)
Apixaban (com) vs Dabigatran (ref)
(N=27,096 vs. N=27,096) (N=21,228 vs. N=22,688) (N=4,408 vs. N=5,868)
Discontinuation: 42.2% vs. 61.1% Discontinuation: 46.0% vs. 68.2% Discontinuation: 22.6% vs. 35.6%
Stroke/SE 163 (1.09) vs. 258 (1.46) 0.69 (0.56 – 0.84) 129 (1.56) vs. 200 (2.67) 0.59 (0.47 – 0.74) 34 (0.51) vs. 58 (0.57) 0.89 (0.58 – 1.36)
Ischemic 136 (0.91) vs. 217 (1.23) 0.68 (0.55 – 0.84) 103 (1.24) vs. 169 (2.25) 0.56 (0.44-0.71) 33 (0.50) vs. 48 (0.47) 1.03 (0.66-1.61)
Hemorrhagic 24 (0.16) vs. 29 (0.16) 0.91 (0.53 – 1.55) 23 (0.28) vs. 22 (0.29) 0.95 (0.53-1.70) 1 (0.02) vs. 7 (0.07) 0.23 (0.03-1.83)
Major Bleeding 426 (2.86) vs. 623 (3.54) 0.77 (0.68 – 0.87) 348 (4.21) vs. 473 (6.34) 0.67 (0.58 – 0.77) 78 (1.18) vs. 150 (1.48) 0.85 (0.65-1.12)
GI Bleeding 215 (1.44) vs. 369 (2.09) 0.65 (0.55 – 0.77) 173 (2.09) vs. 282 (3.77) 0.56 (0.46-0.67) 42 (0.63) vs. 87 (0.86) 0.78 (0.54-1.12)
ICH 63 (0.42) vs. 68 (0.38) 1.06 (0.74 – 1.51) 51 (0.61) vs. 52 (0.69) 0.89 (0.60-1.31) 12 (0.18) vs. 16 (0.16) 1.27 (0.56-2.86)
Apixaban (com) vs Rivaroxaban (ref)
(N=62,619 vs. N=62,619) (N=52,541 vs. N=48,283) (N=10,078 vs. N=14,336)
Discontinuation: 42.1% vs. 54.1% Discontinuation: 45.8% vs. 60.3% Discontinuation: 23.0% vs. 33.2%
Stroke/SE 444 (1.29) vs. 622 (1.50) 0.80 (0.71 – 0.91) 354 (1.84) vs. 456 (2.57) 0.72 (0.62 – 0.82) 90 (0.59) vs. 166 (0.70) 0.86 (0.66 – 1.11)
Ischemic 356 (1.03) vs. 447 (1.07) 0.90 (0.78 – 1.03) 272 (1.41) vs. 319 (1.80) 0.79 (0.67-0.92) 84 (0.55) vs. 128 (0.54) 1.03 (0.78-1.35)
Hemorrhagic 76 (0.22) vs. 146 (0.35) 0.60 (0.45 – 0.79) 72 (0.37) vs. 113 (0.64) 0.59 (0.44-0.79) 4 (0.03) vs. 33 (0.14) 0.20 (0.07-0.56)
Major Bleeding 1,167 (3.40) vs. 2,386 (5.79) 0.55 (0.51 – 0.59) 964 (5.03) vs. 1,869 (10.68) 0.47 (0.44 – 0.51) 203 (1.34) vs. 517 (2.18) 0.64 (0.54 – 0.75)
GI Bleeding 585 (1.70) vs. 1,345 (3.25) 0.49 (0.44 – 0.54) 479 (2.49) vs. 1,067 (6.06) 0.41 (0.37-0.46) 106 (0.70) vs. 278 (1.17) 0.62 (0.49-0.77)
ICH 167 (0.48) vs. 250 (0.60) 0.78 (0.64 – 0.95) 146 (0.76) vs. 187 (1.05) 0.72 (0.58-0.90) 21 (0.14) vs. 63 (0.26) 0.57 (0.34-0.94)
Dabigatran (com) vs Rivaroxaban (ref)
(N=27,538 vs. N=27,538) (N=21,609 vs. N=21,173) (N=5,929 vs. N=6,365)
Discontinuation: 61.2% vs. 54.5% Discontinuation: 68.2% vs. 60.9% Discontinuation: 35.6% vs. 33.1%
Stroke/SE 259 (1.45) vs. 231 (1.26) 1.15 (0.96 – 1.37) 201 (2.64) vs. 176 (2.26) 1.16 (0.95 – 1.42) 58 (0.57) vs. 55 (0.52) 1.09 (0.75 – 1.58)
Ischemic 218 (1.22) vs. 163 (0.89) 1.37 (1.12 – 1.68) 170 (2.23) vs. 122 (1.57) 1.41 (1.12-1.78) 48 (0.47) vs. 41 (0.39) 1.22 (0.80-1.85)
Hemorrhagic 29 (0.16) vs. 54 (0.29) 0.55 (0.35 – 0.87) 22 (0.29) vs. 42 (0.54) 0.53 (0.32-0.90) 7 (0.07) vs. 12 (0.11) 0.59 (0.23-1.51)
Major Bleeding 627 (3.52) vs. 922 (5.06) 0.70 (0.63 – 0.77) 477 (6.29) vs. 718 (9.33) 0.67 (0.60 – 0.75) 150 (1.47) vs. 204 (1.94) 0.75 (0.61 – 0.92)
GI Bleeding 371 (2.08) vs. 515 (2.81) 0.74 (0.65 – 0.84) 284 (3.73) vs. 397 (5.13) 0.72 (0.62-0.84) 87 (0.85) vs. 118 (1.12) 0.75 (0.57-0.99)
ICH 68 (0.38) vs. 104 (0.56) 0.67 (0.50 – 0.91) 52 (0.68) vs. 83 (1.07) 0.64 (0.45-0.90) 16 (0.16) vs. 21 (0.20) 0.78 (0.40-1.49)
0.00 1.00 2.00
0.00 1.00 2.00
0.00 1.00 2.00 3.00
Favors Comp Favors Ref

CI: confidence interval; GI: gastrointestinal; ICH: intracranial hemorrhage; MB: major bleeding; SE: systemic embolism

LIMITATIONS

- PSM was conducted between each cohort pair; thus, the results are not comparable across the matched populations.
- Only associations could be concluded from this retrospective observational study. Although cohorts were matched through PSM, potential residual confounders exist such as over-the-counter aspirin use and dose changes in OAC treatment.
- Given the nature of claims data, laboratory test results and biomarkers were not available. Diagnoses and drug prescriptions were identified using claims. Missing values, coding errors, and lack of clinical accuracy may have introduced bias into the study. Although some of the datasets contain information from different insurance plans that do not overlap at the plan level, others are employer-based claims datasets which may contain duplicate patient records when pooled together; however, the number of such duplicates is likely to be low—based on a published estimate of 0.5%—and therefore unlikely to have a significant impact on the results.[8]

CONCLUSIONS

- **Across NVAF patients with a treatment duration of <1 and ≥1 year in the ARISTOPHANES study, both apixaban and dabigatran were associated with a lower risk of MB compared to rivaroxaban. These findings indicate varying long-term safety outcomes among different NOACs.**
- **Apixaban in patients with treatment duration of <1 year was associated with lower risk of stroke/SE versus dabigatran and rivaroxaban, although stroke/SE risk with treatment duration of ≥1 year was similar across NOACs.**

REFERENCES

[1]Ruff CT, Giugliano RP, Braunwald E, et al. Comparison of the efficacy and safety of new oral anticoagulants with warfarin in patients with atrial fibrillation: a meta-analysis of randomized trials. *Lancet.* 2014;383(9921):955-62.

[2]Lip GYH, Keshishian A, Li X, et al. Effectiveness and safety of oral anticoagulants among nonvalvular atrial fibrillation patients.*Stroke.* 2018;49(12):2933-44.

[3]Dentali F, Riva N, Crowther M, et al. Efficacy and safety of the novel oral anticoagulants in atrial fibrillation: a systematic review and meta-analysis of the literature. *Circulation.* 2012;126(20), 2381-2391.

[4]Seeger JD, Bykov K, Bartels DB, et al. Safety and effectiveness of dabigatran and warfarin in routine care of patients with atrial fibrillation. *Thromb Haemost.* 2015;114(12), 1277-1289.

[5]Thigpen JL, Dillon C, Forster KB, et al. Validity of international classification of disease codes to identify ischemic stroke and intracranial hemorrhage among individuals with associated diagnosis of atrial fibrillation. *Circ Cardiovasc Qual Outcomes.* 2015;8(1):8-14.

[6]Cunningham A, Stein CM, Chung CP, Daugherty JR, Smalley WE, Ray WA. An automated database case definition for serious bleeding related to oral anticoagulant use. *Pharmacoepidemiol Drug Saf.* 2011;20(6):560-6.

[7]Austin PC. The use of propensity score methods with survival or time-to-event outcomes: reporting measures of effect similar to those used in randomized experiments. *Stat Med.* 2014;33(7):1242-58.

[8]Broder MS, Neary MP, Chang E, Cherepanov D, Katznelson L. Treatments, complications, and healthcare utilization associated with acromegaly: a study in two large United States databases. *Pituitary.* 2014;17(4):333-41.

DISCLOSURES

Funding for this research was provided by Bristol-Myers Squibb Company and Pfizer Inc.

S Deitelzweig is a consultant for Bayer/Janssen, Bristol Myers Squibb Company/Pfizer Inc., Daiichi-Sankyo, Portola, and Boehringer Ingelheim, and has been on the speakers' bureau for Janssen, Bristol Myers Squibb Company/Pfizer Inc., and Boehringer Ingelheim.

A Keshishian is a paid employee of STATinMED Research, which is a paid consultant to Pfizer and Bristol-Myers Squibb in connection with this study and the development of this poster.

X Li, A Kang, A Dhamane, N Balachander, L Rosenblatt, and J Jiang are paid employees of Bristol-Myers Squibb with ownership of stocks in Bristol-Myers Squibb Company.

X Luo, J Mardekian, and M Di Fusco are paid employees of Pfizer Inc., with ownership of stocks in Pfizer Inc.

H Yuce has nothing to disclose.

GYH Lip is a consultant for Bayer/Janssen, BMS/Pfizer, Medtronic, Boehringer Ingelheim, Novartis, Verseon and Daiichi-Sankyo. Speaker for Bayer, BMS/Pfizer, Medtronic, Boehringer Ingelheim, and Daiichi-Sankyo. No fees are directly received personally.

Presented at the *European Society of Cardiology ESC Congress 2019*, Paris, France, 31 August - 04 September 2019