Comparison of Effectiveness, Safety, and Healthcare Costs of Direct Oral Anticoagulants with Warfarin in Non-valvular Atrial Fibrillation Patients with Heart Failure

A. Amin[1], A. Bassalobre Garcia[2,3], X. Li[3], A. Dhamane[3], X. Luo[4], M. Di Fusco[5], A. Nadkarni[3], K. Friend[3], L. Rosenblatt[3], J. Mardekian[5], X. Pan[6], O. Baser[7], A. Keshishian[8]

[1]The University of California, Irvine, CA, USA; [2]The University of North Carolina, Chapel Hill, NC, USA; [3]Bristol-Myers Squibb Company, Lawrenceville, NJ, USA; [4]Pfizer, Inc, Groton, CT, USA; [5]Pfizer, Inc, New York, NY, USA; [6]Bristol-Myers Squibb Company, Wallingford, CT, USA; [7]Columbia University, New York, NY, USA; [8]STATinMED Research, Ann Arbor, MI, USA

BACKGROUND

- Atrial fibrillation (AF), the most common cardiac dysrhythmia in the United States, increases the risk of stroke five-fold.[1]
- Non-valvular AF (NVAF) and heart failure (HF) are some of the most common cardiac conditions.[2]
- HF prevalence among NVAF patients ranges from 27-64% in randomized control trials[3,4,5,6] and 21-48% in real-world studies.[7,8,9,10]
- Both conditions affect the elderly and share many of the same risk factors, which synergistically increase the risk of stroke/systemic embolism (SE) and the economic burden.[11,12]
- Few observational studies have evaluated outcomes of direct oral anticoagulants (DOACs) vs warfarin among NVAF patients with HF in a real-world setting.

OBJECTIVES

This study was conducted to compare the rate of stroke/SE, major bleeding (MB), and major adverse cardiac events (MACE) and healthcare costs of DOACs versus warfarin among NVAF patients with HF.

METHODS

Data Source

This was a retrospective observational analysis (ClinicalTrials.gov ID: NCT03508271) using US fee-for-service (FFS) Medicare data from the Center for Medicare and Medicaid Services. Medicare provides health insurance coverage for >38 million people aged ≥65 years as well as for those with end-stage kidney disease or a disability.

Figure 1. Patient Selection Figure

AF diagnosis any time between 01JAN2012 and 30SEP2015
N = 7,841,918

Elderly patients with ≥1 pharmacy claim for warfarin, apixaban, dabigatran, edoxaban or rivaroxaban during the identification period (01JAN2013-30SEP2015)
N = 2,341,715

12 months continuous medical and pharmacy health plan enrollment pre-index date
N = 1,435,003

≥1 medical claim for AF prior to or on the index date
N = 1,347,878

≥1 diagnosis claim(s) of HF during the 12 months prior to or on the index date
N = 565,220

Excluded 205,127 patients with rheumatic mitral valvular heart disease or valve replacement procedure prior to/on the index date

Excluded 99,979 patients with VTE, transient AF, cardiac surgery prior to/on index date

Excluded 3,140 patients with claims indicating pregnancy during the study period & hip/knee replacement surgery within 6 weeks prior to index date

Excluded 193,768 patients with pharmacy claim for OAC during the 12 month baseline period, >1 OAC on the index date, follow up time = 0, or presence of ICD-10 codes

All Patients included in the study (N= 63,206)
Apixaban (N = 10,615) Dabigatran (N = 4,297)
Rivaroxaban (N = 15,921) Warfarin (N = 32,373)

Matched Patients
Apixaban-Warfarin Dabigatran-Warfarin Rivaroxaban-Warfarin
N=10,570 N=10,570 N=4,297 N= 4,297 N=15,712 N= 15,712

AF: atrial fibrillation; HF: heart failure; OAC: oral anticoagulant; VTE: venous thromboembolism

Study Sample

- AF patients with ≥1 diagnosis claim(s) of HF during the 12 months prior to or on the index date (International Classification of Diseases, 9th Revision, Clinical Modification [ICD-9-CM] code 428.xx), aged ≥65 years, with ≥1 pharmacy claim for apixaban, dabigatran, edoxaban, rivaroxaban, or warfarin during the identification period (01JAN2013-30SEP2015) were selected. No edoxaban patients were identified after applying the selection criteria, given the recent FDA approval of this drug.
- For patients with a DOAC prescription, the first DOAC pharmacy claim date during the identification period was designated as the index date. The first warfarin prescription date was designated as the index date for patients prescribed only warfarin and without any DOAC claim.
- Inclusion and exclusion criteria are shown in Figure 1.
- Patient outcomes were assessed from the day after the index date until the earliest of the following: 30 days post-discontinuation, medication switch, death, end of study period, or end of continuous medical or pharmacy enrollment.

Outcome Definitions

- Clinical outcomes were stroke/SE, MB, and MACE. Stroke/SE included ischemic stroke, hemorrhagic stroke, and SE; MB included gastrointestinal bleeding, intracranial hemorrhage, and other sites of MB; and MACE included stroke, myocardial infarction, and all-cause mortality.
- Events were identified using the primary hospitalization discharge diagnosis. Death was obtained by validated Social Security records that include the date of death.
- Diagnosis codes used for stroke/SE and MB were based on a validated administrative claim-based algorithm and the International Society on Thrombosis and Haemostasis' definition of MB as used in the ARISTOTLE trial.[13,14]
- **Stroke/SE- and MB-related Medical Costs:** Stroke/SE- and MB-related medical costs were defined as those associated with the first stroke/SE and MB-related hospitalization plus subsequent inpatient and outpatient stroke/SE and MB costs, respectively.
- **All-cause Healthcare Costs:** Total all-cause healthcare costs were computed as the sum of inpatient, outpatient/ emergency room (ER), part D pharmacy and other (skilled nursing facility, durable medical equipment, home health agency, hospice) costs. Total medical costs were computed as the sum of inpatient, outpatient/ER, and other costs. All costs were calculated as per patient per month (PPPM).

METHODS (cont'd)

Statistical Analysis

- Patients were matched using 1:1 propensity score matching (PSM) between apixaban-warfarin, dabigatran-warfarin and rivaroxaban-warfarin patients using propensity scores generated by logistic regression based on age, gender, geographic region, socioeconomic status,[15,16] Medicaid dual eligibility, low-income Part D subsidy, Charlson comorbidity index (CCI) score, baseline bleeding and stroke/SE history, comorbidities, co-medications and inpatient stay.[17]
- The balance of patient characteristics was checked based on standardized differences with a threshold of 10%.
- Incidence of events was calculated as the number of events per 100 person-years.
- Cox proportional hazard models, with robust sandwich estimators were used to estimate the hazard ratios for stroke/SE, MB, and MACE between the matched cohorts.
- Stroke/SE- and MB-related costs were calculated using two-part generalized linear models (GLMs) that incorporated the probability of having a stroke/SE or MB.
- All-cause health care costs were compared between cohorts using GLMs with a gamma distribution and log-link function.
- P-values < 0.05 were considered significant.

RESULTS

Baseline Results

- A total of 63,206 NVAF patients with HF meeting eligibility criteria were identified, of which 10,615 (16.8%) were prescribed apixaban, 4,297 (6.8%) were prescribed dabigatran, 15,921 (25.2%) were prescribed rivaroxaban, and 32,373 (51.2%) were prescribed warfarin (Figure 1).
- Before PSM, apixaban and warfarin patients were of similar age, and dabigatran and rivaroxaban patients were younger. Warfarin and apixaban patients had similar CHA_2DS_2-VASc and HAS-BLED scores, and dabigatran and rivaroxaban patients had lower scores.
- Following PSM, there were 10,570 apixaban-warfarin, 4,297 dabigatran-warfarin, 15,712 rivaroxaban-warfarin, matched pairs.
- All baseline characteristics were balanced after PSM. Baseline characteristics are shown in Table 1.
- In this subpopulation, patients had higher CHA_2DS_2-VASc and HAS-BLED scores compared to a pooled sample of the general US NVAF population.[18]

Table 1. Post-PSM Baseline Characteristics among DOAC vs Warfarin Users

	Apixaban		Warfarin		Dabigatran		Warfarin		Rivaroxaban		Warfarin	
	N/Mean	%/SD	N/Mean	%/SD	N/Mean	%/SD	N/Mean	%/SD	N/Mean	%/SD	N/Mean	%/SD
Sample Size	**10,570**		**10,570**		**4,297**		**4,297**		**15,712**		**15,712**	
Age*	80.4	7.8	80.4	7.8	78.7	7.6	79	7.7	79.3	7.7	79.4	7.7
65-74	2,740	25.9%	2,779	26.3%	1,443	33.6%	1,383	32.2%	4,814	30.6%	4,810	30.6%
75-79	2,064	19.5%	2,037	19.3%	916	21.3%	886	20.6%	3,368	21.4%	3,377	21.5%
≥80	5,766	54.6%	5,754	54.4%	1,938	45.1%	2,028	47.2%	7,530	47.9%	7,525	47.9%
Sex*												
Male	4,889	46.3%	4,883	46.2%	2,112	49.2%	2,081	48.4%	7,419	47.2%	7,383	47.0%
Female	5,681	53.8%	5,687	53.8%	2,185	50.9%	2,216	51.6%	8,293	52.8%	8,329	53.0%
Race*												
White	9,462	89.5%	9,472	89.6%	3,777	87.9%	3,787	88.1%	13,902	88.5%	13,892	88.4%
Black	605	5.7%	607	5.7%	278	6.5%	284	6.6%	965	6.1%	1,002	6.4%
Hispanic	152	1.4%	147	1.4%	74	1.7%	62	1.4%	316	2.0%	302	1.9%
Other/Unknown	351	3.3%	344	3.3%	168	3.9%	164	3.8%	529	3.4%	516	3.3%
US Geographic Region *												
Northeast	1,865	17.6%	1,874	17.7%	829	19.3%	826	19.2%	2,763	17.6%	2,784	17.7%
Midwest	2,319	21.9%	2,426	23.0%	962	22.4%	1,049	24.4%	3,607	23.0%	3,745	23.8%
South	4,781	45.2%	4,644	43.9%	1,717	40.0%	1,652	38.5%	6,617	42.1%	6,525	41.5%
West	1,597	15.1%	1,615	15.3%	782	18.2%	763	17.8%	2,699	17.2%	2,632	16.8%
Unknown	8	0.1%	11	0.1%	7	0.2%	7	0.2%	26	0.2%	26	0.2%
Proxy for Socioeconomic Status*												
Low	2,579	24.4%	2,590	24.5%	1,103	25.7%	1,056	24.6%	3,999	25.5%	3,978	25.3%
Mid	2,875	27.2%	2,863	27.1%	1,218	28.4%	1,245	29.0%	4,262	27.1%	4,337	27.6%
High	4,811	45.5%	4,821	45.6%	1,840	42.8%	1,857	43.2%	6,947	44.2%	6,879	43.8%
Missing	305	2.9%	296	2.8%	136	3.2%	139	3.2%	504	3.2%	518	3.3%
Medicaid dual-eligibility*	2,122	20.1%	2,168	20.5%	1,052	24.5%	1,007	23.4%	3,781	24.1%	3,742	23.8%
Part D Low-income subsidy*	3,355	31.7%	3,361	31.8%	1,601	37.3%	1,576	36.7%	5,452	34.7%	5,410	34.4%
Baseline Comorbidity												
Deyo-Charlson Comorbidity Index Score*	5.7	3	5.7	3	5.2	2.9	5.4	3	5.5	2.9	5.5	3
CHA_2DS_2-VASc Score	5.4	1.4	5.4	1.4	5.2	1.4	5.2	1.4	5.3	1.4	5.3	1.4
HAS-BLED Score[a]	3.7	1.2	3.7	1.3	3.5	1.2	3.5	1.2	3.6	1.2	3.6	1.2
Bleeding History*	3,016	28.5%	3,007	28.5%	1,108	25.8%	1,136	26.4%	4,472	28.5%	4,465	28.4%
Diabetes Mellitus*	5,059	47.9%	5,065	47.9%	2,066	48.1%	2,087	48.6%	7,577	48.2%	7,530	47.9%
Hypertension*	10,161	96.1%	10,114	95.7%	4,053	94.3%	4,057	94.4%	14,968	95.3%	14,983	95.4%
Liver Disease*	720	6.8%	722	6.8%	256	6.0%	245	5.7%	1,017	6.5%	997	6.4%
Renal Disease*	4,612	43.6%	4,633	43.8%	1,526	35.5%	1,626	37.8%	5,863	37.3%	5,944	37.8%
Myocardial Infarction*	1,667	15.8%	1,715	16.2%	612	14.2%	632	14.7%	2,525	16.1%	2,495	15.9%
Dyspepsia or Stomach Discomfort*	3,139	29.7%	3,073	29.1%	1,133	26.4%	1,137	26.5%	4,429	28.2%	4,411	28.1%
Peripheral vascular disease*	8,201	77.6%	8,219	77.8%	3,120	72.6%	3,161	73.6%	11,889	75.7%	11,954	76.1%
Prior S/SE*	1,986	18.8%	1,989	18.8%	694	16.2%	705	16.4%	2,733	17.4%	2,738	17.4%
Transient Ischemic attack*	1,091	10.3%	1,093	10.3%	361	8.4%	381	8.9%	1,469	9.4%	1,468	9.3%
Anemia and Coagulation Defects*	5,129	48.5%	5,111	48.4%	1,883	43.8%	1,934	45.0%	7,132	45.4%	7,152	45.5%
Alcoholism*	26	0.3%	30	0.3%	17	0.4%	16	0.4%	52	0.3%	55	0.4%
Baseline Medication Use												
ACEIs/ARBs*	6,940	65.7%	6,886	65.2%	2,918	67.9%	2,863	66.6%	10,564	67.2%	10,492	66.8%
Amiodarone*	2,092	19.8%	2,068	19.6%	774	18.0%	724	16.9%	2,769	17.6%	2,770	17.6%
Digoxin*	2,030	19.2%	2,043	19.3%	935	21.8%	887	20.6%	3,232	20.6%	3,190	20.3%
Diuretics*	8,176	77.4%	8,139	77.0%	3,324	77.4%	3,312	77.1%	11,905	75.8%	11,920	75.9%
Beta blockers*	6,295	59.6%	6,321	59.8%	2,454	57.1%	2,466	57.4%	9,283	59.1%	9,253	58.9%
Calcium Channel Blockers*	4,718	44.6%	4,663	44.1%	1,942	45.2%	1,910	44.5%	7,051	44.9%	7,017	44.7%
H2-receptor Blockers*	1,092	10.3%	1,078	10.2%	415	9.7%	400	9.3%	1,560	9.9%	1,572	10.0%
Proton Pump Inhibitors*	4,389	41.5%	4,309	40.8%	1,631	38.0%	1,634	38.0%	6,238	39.7%	6,203	39.5%
Statins*	6,946	65.7%	6,928	65.5%	2,598	60.5%	2,633	61.3%	9,892	63.0%	9,897	63.0%
Anti-platelets*	2,786	26.4%	2,730	25.8%	887	20.6%	846	19.7%	3,788	24.1%	3,722	23.7%
NSAIDs*	2,457	23.3%	2,445	23.1%	1,039	24.2%	962	22.4%	3,954	25.2%	3,882	24.7%
All-Cause Inpatient Admissions*	6,738	63.8%	6,782	64.2%	2,674	62.2%	2,704	62.9%	10,543	67.1%	10,567	67.3%
Index Dose [b,c]												
Lower Dose	4,393	41.6%			1,381	32.1%			7,099	45.2%		
Standard Dose	6,184	58.5%			2,919	67.9%			8,656	55.1%		
Follow Up (Mean, in days)	198	181	246	229	242	240	245	231	242	236	246	230
Median	132		155		133		157		146		155	

ACEis: angiotensin-converting enzyme inhibitors; ARBs: angiotensin-receptor blockers; CHA_2DS_2-VASc: congestive heart failure, hypertension, aged ≥75 years, diabetes mellitus, prior stroke or transient ischemic attack or thromboembolism, vascular disease, aged 65-74 years, sex category; DOAC: direct oral anticoagulant; HAS-BLED: hypertension, abnormal renal and liver function, stroke, bleeding, labile international normalized ratios, elderly, drugs and alcohol; NSAIDs: non-steroidal anti-inflammatory drugs; PSM: propensity score matching; SD: standard deviation; SE: systemic embolism

[a]As the international normalized ratio value was not available in the databases, a modified HAS-BLED score was calculated with a range of 0 to 8.

[b]Lower dose: 2.5mg apixaban, 75mg dabigatran, 10mg or 15mg rivaroxaban. 1,375 (8.8%) of rivaroxaban-warfarin patients received 10 mg rivaroxaban

[c]Standard dose: 5mg apixaban, 150mg dabigatran, 20mg rivaroxaban; After PSM, baseline characteristics were balanced between cohorts.

*Covariates included in the propensity score matching

RESULTS (cont'd)

Outcome Results

- Incidence of stroke/SE, MB, and MACE are shown in Figure 2.
- After PSM, apixaban was associated with lower rates of stroke/SE, MB, and MACE, which was driven by all-cause mortality, compared to warfarin.
- Compared to warfarin, dabigatran was associated with a lower rate of MACE, which was driven by all-cause mortality; there was no significant difference for stroke/SE or MB.
- Compared to warfarin, rivaroxaban was associated with a lower rate of stroke/SE and MACE, which was driven by all-cause mortality, and an increased rate of MB, which was driven by GI bleeding (Figure 2).
- Apixaban patients incurred lower PPPM MB-related, stroke/SE-related, and all-cause healthcare costs vs warfarin.
- Dabigatran patients incurred similar PPPM stroke/SE-related and MB-related healthcare costs and lower PPPM all-cause healthcare costs vs warfarin.
- Rivaroxaban patients incurred lower PPPM stroke/SE-related and similar MB-related and all-cause healthcare costs (Table 2).

Figure 2. Incidence and Hazard Ratios of Follow-up Events for DOAC vs Warfarin Users

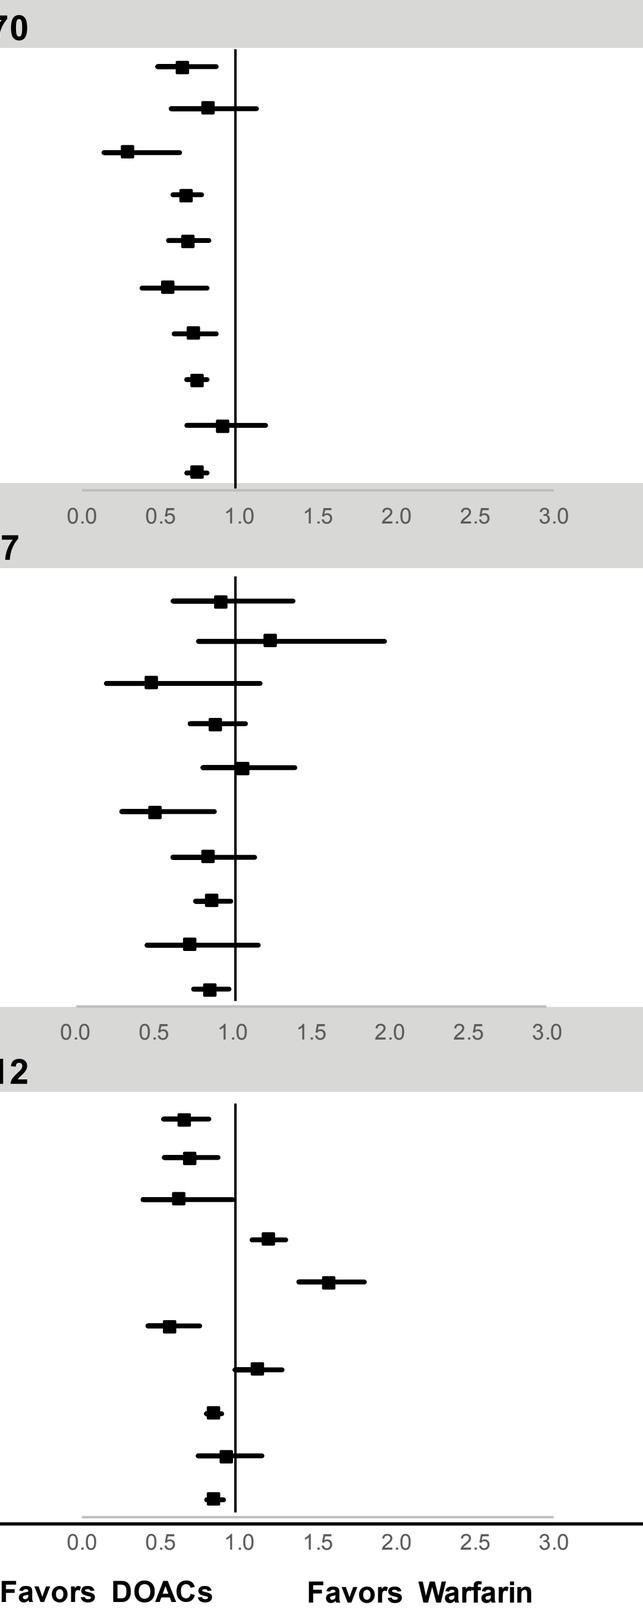
0.0 0.5 1.0 1.5 2.0 2.5 3.0
Favors DOACs
Favors Warfarin

	Comparator Incidence Rate (per 100 person-years)	Reference Incidence Rate (per 100 person-years)	Hazard Ratio (95% CI)	P-value
Apixaban vs. Warfarin (ref.)	N=10,570	N=10,570		
Stroke/SE	**1.2**	**1.8**	**0.64 (0.48-0.85)**	0.002
Ischemic	1.1	1.2	0.80 (0.57-1.11)	0.172
Hemorrhagic	0.1	0.5	0.29 (0.14-0.62)	0.002
Major Bleeding	**6.1**	**8.5**	**0.66 (0.58-0.76)**	<0.001
GI Bleeding	2.8	3.8	0.67 (0.55-0.81)	<0.001
ICH	0.7	1.2	0.55 (0.38-0.80)	0.002
Other Bleeding	3.1	4.0	0.71 (0.59-0.85)	<0.001
MACE	15.7	20.1	**0.73 (0.67-0.79)**	<0.001
Myocardial Infarction	1.5	1.6	0.89 (0.67-1.17)	0.396
All-cause death	13.9	17.6	0.73 (0.67-0.80)	<0.001
Dabigatran vs. Warfarin (ref.)	N=4,297	N=4,297		
Stroke/SE	**1.6**	**1.8**	**0.93 (0.62-1.38)**	0.701
Ischemic	1.4	1.1	1.24 (0.78-1.97)	0.370
Hemorrhagic	0.2	0.5	0.48 (0.20-1.18)	0.108
Major Bleeding	**6.7**	**7.5**	**0.89 (0.73-1.08)**	0.239
GI Bleeding	3.8	3.6	1.06 (0.81-1.39)	0.681
ICH	0.7	1.3	0.51 (0.30-0.88)	0.016
Other Bleeding	2.7	3.2	0.84 (0.62-1.14)	0.269
MACE	**15.8**	**18.3**	**0.87 (0.77-0.99)**	0.032
Myocardial Infarction	1.1	1.5	0.73 (0.46-1.16)	0.182
All-cause death	13.8	16.2	0.86 (0.75-0.98)	0.021
Rivaroxaban vs. Warfarin (ref.)	N=15,712	N=15,712		
Stroke/SE	**1.3**	**1.9**	**0.65 (0.52-0.81)**	<0.001
Ischemic	0.9	1.4	0.68 (0.52-0.87)	0.003
Hemorrhagic	0.3	0.5	0.61 (0.39-0.96)	0.033
Major Bleeding	**9.4**	**7.9**	**1.18 (1.08-1.30)**	<0.001
GI Bleeding	5.5	3.5	1.57 (1.38-1.79)	<0.001
ICH	0.7	1.2	0.56 (0.42-0.75)	<0.001
Other Bleeding	4.3	3.9	1.11 (0.97-1.27)	0.136
MACE	**16.4**	**19.6**	**0.84 (0.79-0.89)**	<0.001
Myocardial Infarction	1.5	1.6	0.92 (0.74-1.14)	0.445
All-cause death	14.3	17.1	0.84 (0.79-0.90)	<0.001

CI: confidence interval; DOAC: direct oral anticoagulant; GI: gastrointestinal; ICH: intracranial hemorrhage; MACE: major adverse cardiovascular event; SE: systemic embolism

Table 2. Post-PSM Costs for DOAC vs Warfarin Users

	Apixaban vs Warfarin N = 10,570		Dabigatran vs Warfarin N = 4,297		Rivaroxaban vs Warfarin N = 15,712	
Costs	Cost PPPM	P-value	Cost PPPM	P-value	Cost PPPM	P-value
S/SE-related Medical costs	$64 vs $143	**0.003**	$117 vs $135	0.654	$83 vs $143	**0.007**
MB-related Medical costs	$249 vs $465	**<0.001**	$343 vs $392	0.365	$434 vs $441	0.825
All-cause Medical costs	$3,124 vs $3,798	**<0.001**	$3,075 vs $3,653	**0.001**	$3,464 vs $3,762	**0.001**
All-cause Pharmacy costs	$530 vs $295	**<0.001**	$450 vs $292	**<0.001**	$490 vs $298	**<0.001**
All-cause Healthcare costs	$3,654 vs $4,093	**<0.001**	$3,525 vs $3,945	**0.013**	$3,954 vs $4,060	0.242

DOAC: direct oral anticoagulant; MB: major bleeding; PPPM: per patient per month; PSM: propensity score matching; S/SE: stroke/systemic embolism

Limitations

- Only associations could be concluded from this retrospective observational study. Although cohorts were PSM-adjusted, there exist potential residual confounders such as over-the-counter use of aspirin or OAC treatment dose change.
- Current coding systems cannot effectively distinguish between systolic and diastolic HF or accurately characterize disease severity for all HF patients; hence, we did not control for type or severity of HF in the analyses.
- Given the nature of claims data, no laboratory results such as international normalized ratios were available; therefore, we could not evaluate the quality of anticoagulation therapy with warfarin. Additionally, diagnoses were identified using ICD-9-CM codes, and drug prescriptions were identified through prescription claims. Missing values, coding errors, and lack of clinical accuracy may have introduced bias into the study. In addition, information on international normalized ratio (INR) tests was not available in the claims data.
- PSM was conducted between each of the OAC cohorts; thus, the results are not comparable across matched cohorts.
- Results are applicable to the population studied only and may not be generalizable to other populations.

CONCLUSIONS

In this large retrospective observational study among elderly NVAF patients with HF, a high-risk subpopulation, DOACs were associated with lower rates of MACE, and had varying rates of stroke/SE and MB compared to warfarin. Patients using apixaban and dabigatran incurred lower all-cause healthcare costs vs warfarin. Findings from this observational analysis provide important insights regarding the use of OAC therapy among high-risk patients with comorbid NVAF and HF in a real-world setting.

REFERENCES

1 Wolf PA, Abbott RD, Kannel WB. Atrial fibrillation: a major contributor to stroke in the elderly. The Framingham Study. *Arch Intern Med.* 1987;147(9):1561-1564.
2 Santhanakrishnan R, Wang N, Larson MG, et al. Atrial fibrillation begets heart failure and vice versa: temporal associations and differences in preserved versus reduced ejection fraction. *Circulation.* 2016;133(5):484-492.
3 van Diepen S, Hellkamp AS, Patel MR, et al. Efficacy and safety of rivaroxaban in patients with heart failure and nonvalvular atrial fibrillation: insights from ROCKET AF. *Circ Heart Fail.* 2013;6(4):740-747.
4 Granger CB, Alexander JH, McMurray JJ, et al. Apixaban versus warfarin in patients with atrial fibrillation. *N Engl J Med.* 2011;365(11):981-992.
5 McMurray JJ, Ezekowitz JA, Lewis BS, et al. Left ventricular systolic dysfunction, heart failure, and the risk of stroke and systemic embolism in patients with atrial fibrillation: insights from the ARISTOTLE trial. *Circ Heart Fail.* 2013;6(3):451-460.
6 Patel MR, Mahaffey KW, Garg J, et al. Rivaroxaban versus warfarin in nonvalvular atrial fibrillation. *N Engl J Med.* 2011;365(10):883-891.
7 Adeboyeje G, Sylwestrzak G, Barron JJ, et al. Major bleeding risk during anticoagulation with warfarin, dabigatran, apixaban, or rivaroxaban in patients with nonvalvular atrial fibrillation. *J Manag Care Pharm.* 2017;23(9):968-978.
8 Bassand JP, Accetta G, Camm AJ, et al. Two-year outcomes of patients with newly diagnosed atrial fibrillation: results from GARFIELD-AF. *Euro Heart J.* 2016;37(38):2882-2889.
9 Lip GYH, Laroche C, Dan GA, et al. A prospective survey in European Society of Cardiology member countries of atrial fibrillation management: baseline results of EURObservational Research Programme Atrial Fibrillation (EORP-AF) Pilot General Registry. *Europace.* 2013;16(3):308-319.
10 Lip GY, Laroche C, Popescu MI, et al. Heart failure in patients with atrial fibrillation in Europe: a report from the EURObservational Research Programme Pilot survey on Atrial Fibrillation. *Euro J Heart Fail.* 2015;17(6):570-582.
11 Kotecha D, Piccini JP. Atrial fibrillation in heart failure: what should we do? *Eur Heart J.* 2015;36(46):3250-3257.
12 Zhao L, Wang WYS, Yang X. Anticoagulation in atrial fibrillation with heart failure. *Heart Fail Rev.* 2018;22:1-9.
13 Thigpen JL, Dillon C, Forster KB, et al. Validity of international classification of disease codes to identify ischaemic stroke and intracranial hemorrhage among individuals with associated diagnosis of atrial fibrillation. *Circ Cardiovasc Qual Outcomes.* 2015;8(1):8-14.
14 Cunningham A, Stein CM, Chung CP, et al. An automated database case definition for serious bleeding related to oral anticoagulant use. *Pharmacoepidemiol Drug Saf.* 2011;20(6):560-566.
15 Birkmeyer NJ, Gu N, Baser O, et al. Socioeconomic status and surgical mortality in the elderly. *Med Care.* 2008;46(9):893-899.
16 Diez Roux AV, Merkin SS, Arnett D, et al. Neighborhood of residence and incidence of coronary heart disease. *N Engl J Med.* 2001;345(2):99-106.
17 Rosenbaum PR, Rubin DB. The central role of the propensity score in observational studies for causal effects. *Biometrika.* 1983;70(1):41-55.
18 Li X, Deitelzweig S, Keshishian A, et al. Effectiveness and safety of apixaban versus warfarin in non-valvular atrial fibrillation patients in "real-world" clinical practice. *Thrombosis Haemostasis.* 2017;117(6):1072-82.

DISCLOSURES

Funding for this research was provided by Bristol-Myers Squibb and Pfizer Inc.
A. Amin is a paid consultant for Bristol-Myers Squibb.
X. Li, A. Dhamane, A. Nadkarni, K. Friend, L. Rosenblatt, A. Bassalobre Garcia, and X. Pan are employees of Bristol-Myers Squibb.
X. Luo, M. Di Fusco and J. Mardekian are employees of Pfizer Inc.
O. Baser has nothing to disclose.
A. Keshishian is an employee of STATinMED Research, which received funding from Bristol-Myers Squibb and Pfizer Inc. for conducting this study and for the development of this poster.

Presented at the *European Society of Cardiology ESC Congress 2018*, Munich, Germany, 25-29 August 2018