Effectiveness and Safety of Non-Vitamin K Antagonist Oral Anticoagulants among Nonvalvular Atrial Fibrillation Patients with Multimorbidity

Poster Su3233

Steve Deitelzweig[1], Allison Keshishian[2,3], Amiee Kang[4], Amol Dhamane[4], Xuemei Luo[5], Christian Klem[4], Lisa Rosenblatt[4], Jack Mardekian[6], Jenny Jiang[4], Huseyin Yuce[3], Gregory Y. H. Lip[7]

[1]Ochsner Clinic Foundation, Department of Hospital Medicine, New Orleans, LA, USA, and The University of Queensland School of Medicine, Ochsner Clinical School, New Orleans, LA, USA;
[2]STATinMED Research, Ann Arbor, MI, USA; [3]New York City College of Technology, City University of New York, New York, NY, USA; [4]Bristol-Myers Squibb Company, Lawrenceville, NJ, USA; [5]Pfizer, Inc., Groton, CT, USA; [6]Pfizer, Inc., New York, NY, USA;
[7]Liverpool Centre for Cardiovascular Science, University of Liverpool and Liverpool Heart & Chest Hospital, Liverpool, United Kingdom; and Aalborg Thrombosis Research Unit, Department of Clinical Medicine, Aalborg University, Aalborg, Denmark

Introduction

- Within the United States, the median number of chronic conditions is six, and atrial fibrillation (AF) is the 11th most common.[1]
- Multimorbidity is associated with an increased risk of AF, cerebrovascular events, and mortality.[2,3]
- Anticoagulation therapy in multimorbid patients is associated with an increased risk of stroke and worse outcomes post-stroke relative to patients without multiple concomitant comorbidites.[1]
- Non-vitamin K oral anticoagulants (NOACs; apixaban, dabigatran, edoxaban, and rivaroxaban) have been incorporated into treatment guidelines as a non-inferior, safe, and effective treatment choice for AF without the restrictions of conventional therapy.[4]
- There is a lack of real-world evidence of risk of stroke/systemic embolism (SE) and major bleeding (MB) directly comparing NOACs in a multimorbid nonvalvular AF (NVAF) population.

Objective

The objective of this study was to compare the risk of stroke/SE and MB of NOACs versus NOACs among NVAF patients with multimorbidity.

Methods

Data Source

This was an observational retrospective cohort analysis using de-identified patient data from CMS Medicare and four US commercial claims databases (Optum, PharMetrics, Humana, and MarketScan), covering >180 million beneficiaries annually (~56% of the US population).

Study Sample

- AF patients (International Classification of Diseases, Ninth Revision, Clinical Modification [ICD-9-CM] code 427.31) with multimorbidity—defined as the presence of ≥6 comorbid conditions among the 16 conditions listed in Table 1—who initiated treatment with apixaban, dabigatran, or rivaroxaban from 01JAN2013-30SEP2015 were included in the study. For Medicare patients, only a proportion of newly diagnosed patients was available for analysis for years 2014 and 2015. Edoxaban was not included in the study due to its small sample size.
- The first NOAC (apixaban, dabigatran, rivaroxaban) prescription date was designated as the index date.
- Inclusion and exclusion criteria are shown in Figure 1.
- The follow-up period ranged from one day post-index date until the earliest of the following: 30 days post-discontinuation, medication switch, death, end of study period, or end of continuous medical of pharmacy enrollment.

Outcome Definitions

- The outcome measures were time to first stroke/SE and MB events based on the principal or first-listed ICD-9-CM diagnosis of inpatient claims. The diagnosis codes used for stroke/SE and MB were based on validated, administrative claim-based algorithms and the *International Society on Thrombosis and Hemostasis'* definition of MB.[5,6]
- Stroke/SE included ischemic stroke, hemorrhagic stroke, and SE; MB included gastrointestinal bleeding, intracranial hemorrhage, and bleeding at other sites.

Methods (continued)

Figure 1. Selection Criteria

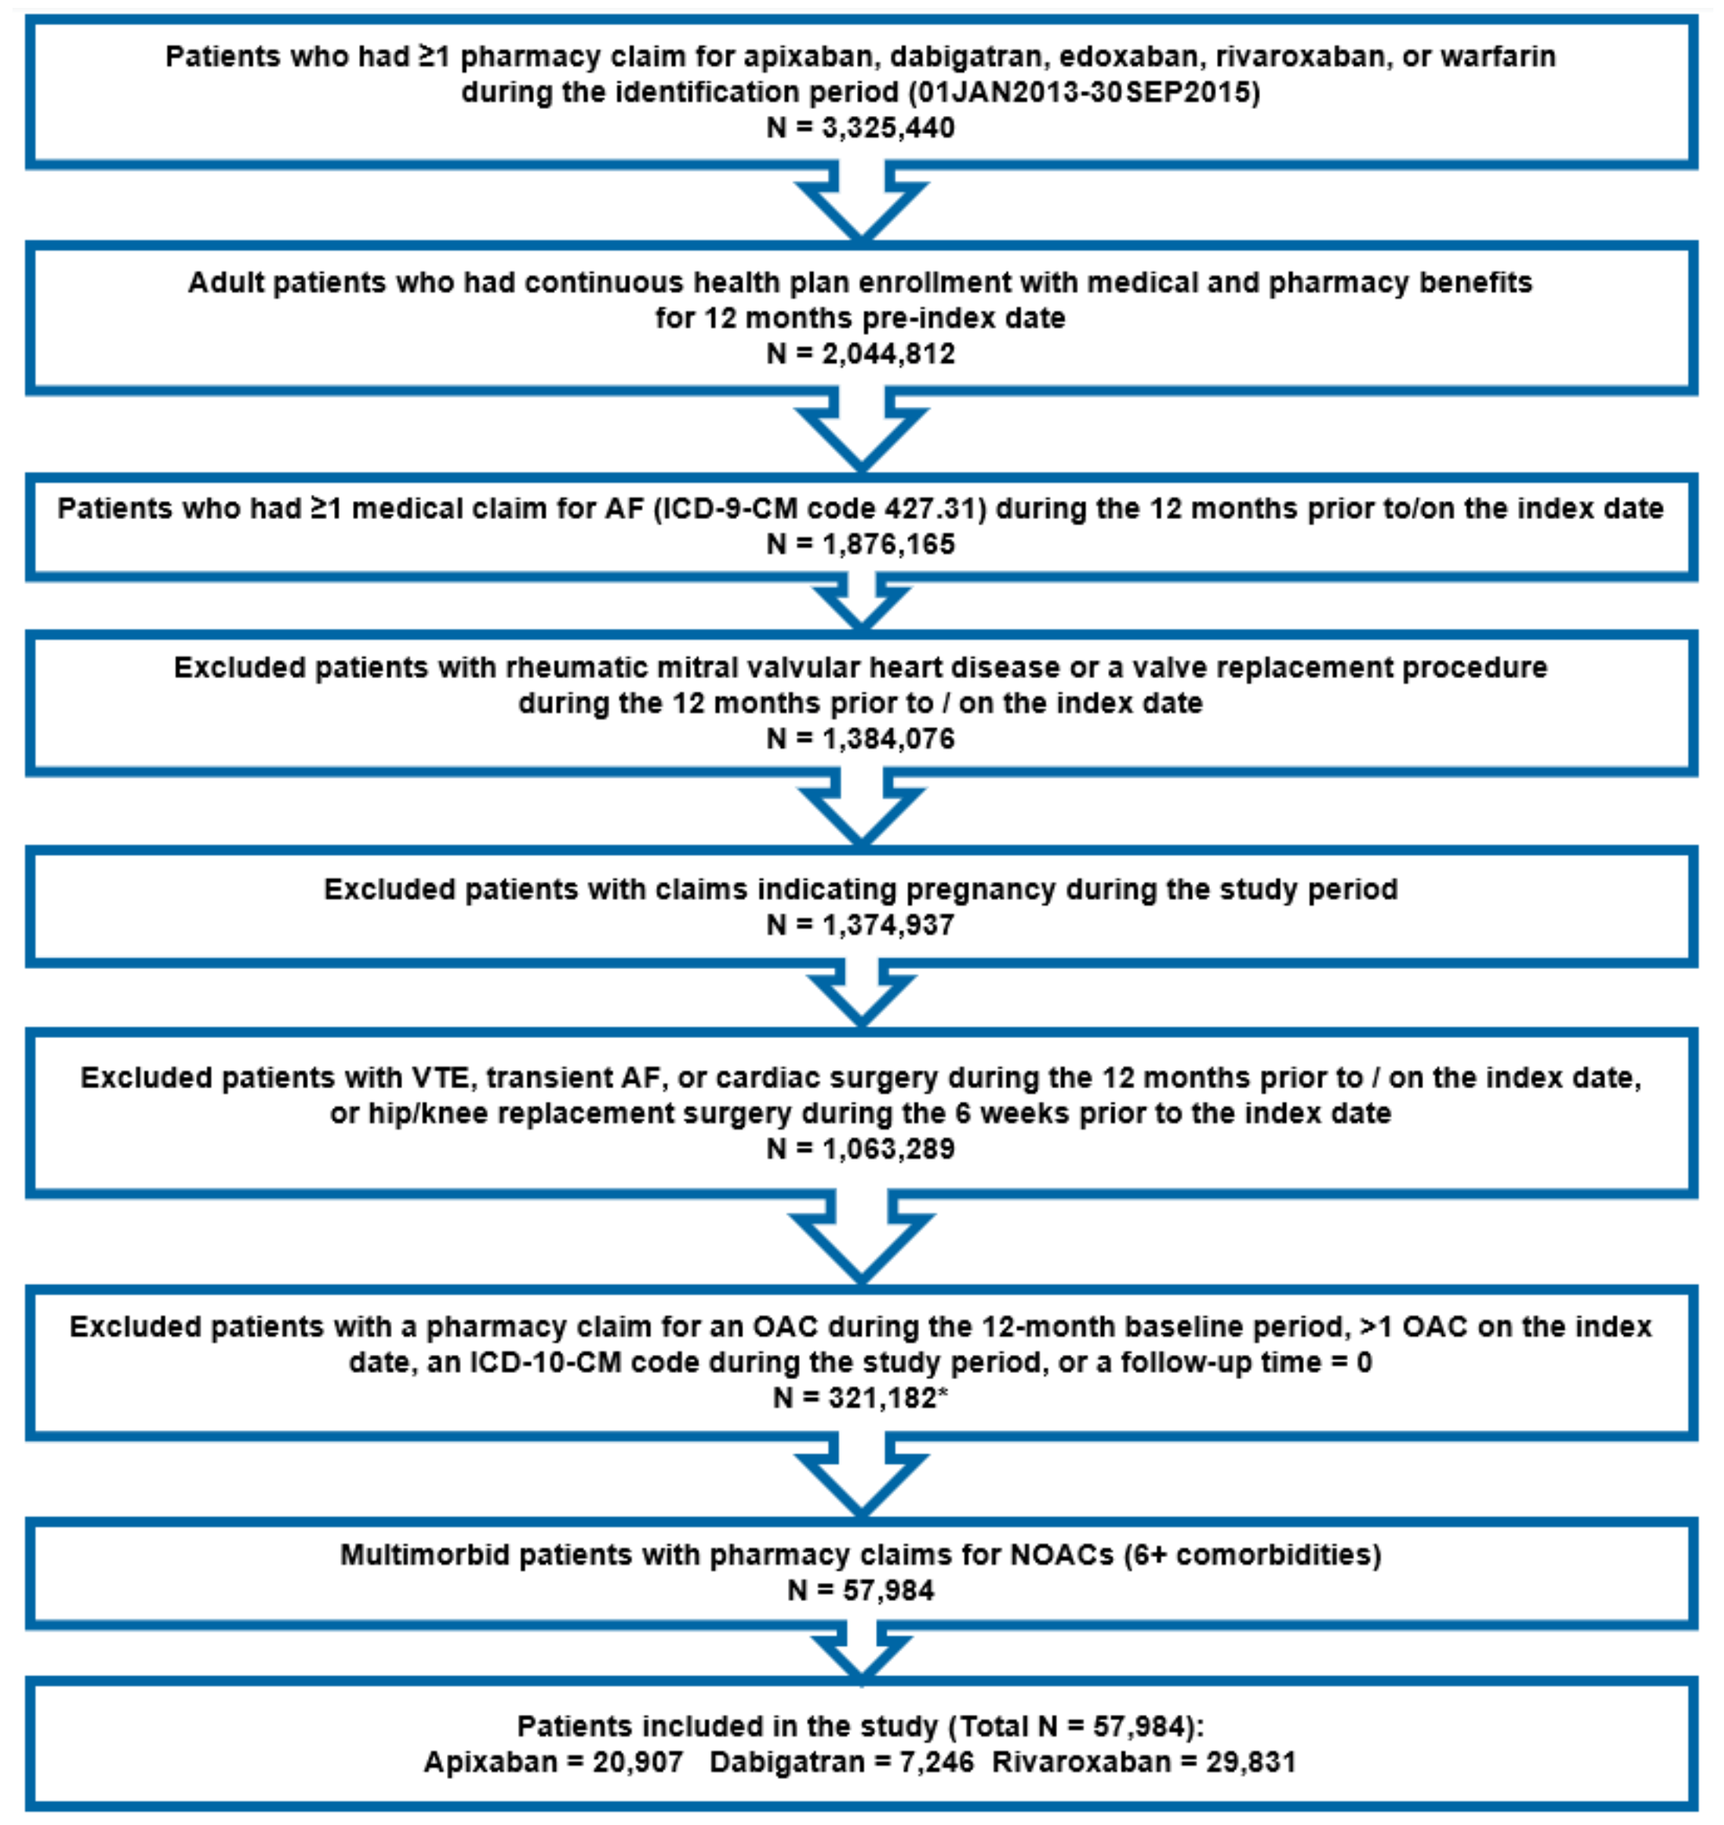
Patients who had ≥1 pharmacy claim for apixaban, dabigatran, edoxaban, rivaroxaban, or warfarin during the identification period (01JAN2013-30SEP2015)
N = 3,325,440
Adult patients who had continuous health plan enrollment with medical and pharmacy benefits for 12 months pre-index date
N = 2,044,812
Patients who had ≥1 medical claim for AF (ICD-9-CM code 427.31) during the 12 months prior to/on the index date
N = 1,876,165
Excluded patients with rheumatic mitral valvular heart disease or a valve replacement procedure during the 12 months prior to / on the index date
N = 1,384,076
Excluded patients with claims indicating pregnancy during the study period
N = 1,374,937
Excluded patients with VTE, transient AF, or cardiac surgery during the 12 months prior to / on the index date, or hip/knee replacement surgery during the 6 weeks prior to the index date
N = 1,063,289
Excluded patients with a pharmacy claim for an OAC during the 12-month baseline period, >1 OAC on the index date, an ICD-10-CM code during the study period, or a follow-up time = 0
N = 321,182*
Multimorbid patients with pharmacy claims for NOACs (6+ comorbidities)
N = 57,984
Patients included in the study (Total N = 57,984):
Apixaban = 20,907 Dabigatran = 7,246 Rivaroxaban = 29,831

AF: atrial fibrillation; ICD-9-CM: International Classification of Diseases, 9th Edition, Clinical Modification; VTE: venous thromboembolism

*Edoxaban was not included due to its small sample size

Results

Baseline Results

- Overall, 29.8% of the NVAF population (n=321,182) had multimorbidity.
- A total of 57,984 patients with multimorbidity were selected, including 20,907 (36.1%) apixaban, 7,246 (12.5%) dabigatran, and 29,831 (51.4%) rivaroxaban patients.
- The mean number of comorbidities ranged from 7.5 to 7.8 across the drug cohorts, with a median of 7. 23.9%- 25.8% of patients had ≥9 comorbidities.
- After propensity score matching (PSM), there were 7,208 apixaban-dabigatran, 20,610 apixaban-rivaroxaban, and 7,243 dabigatran-rivaroxaban matched pairs of multimorbid patients (Table 1).
- The mean ages were 76, 77, and 76 for the apixaban-dabigatran, apixaban-rivaroxaban, and dabigatran-rivaroxaban cohort pairs, respectively. The mean CCI scores were 4.9, 5.0, and 4.9 for the apixaban-dabigatran, apixaban-rivaroxaban, and dabigatran-rivaroxaban cohort pairs, respectively (Table 1).

Outcome Results

- Post-PSM incidence rates of stroke/SE and MB are shown in Figure 2.
- Compared to dabigatran, apixaban was associated with a lower risk of stroke/SE, and apixaban and dabigatran were associated with a similar risk of stroke/SE versus rivaroxaban.
- Apixaban use was associated with a similar risk of MB compared to dabigatran use, but apixaban and dabigatran were associated with a lower risk of MB compared to rivaroxaban.

Results (continued)

Table 1. Baseline Characterstics Among Multimorbid NVAF Patients after Propensity Score Matching Among NOACs vs NOACs

Multimorbid Patients	Apixaban Cohort		Dabigatran Cohort		Apixaban Cohort		Rivaroxaban Cohort		Dabigatran Cohort		Rivaroxaban Cohort	
	N/Mean	%/SD	N/Mean	%/SD	N/Mean	%/SD	N/Mean	%/SD	N/Mean	%/SD	N/Mean	%/SD
Sample Size	7,208	100%	7,208	100%	20,610	100%	20,610	100%	7,243	100%	7,243	100%
Age	76.64	9.47	76.35	9.14	77.57	9.66	77.44	9.52	76.33	9.13	76.47	9.32
18-54	116	1.61%	121	1.68%	343	1.66%	355	1.72%	121	1.67%	111	1.53%
55-64	505	7.01%	504	6.99%	1,402	6.80%	1,343	6.52%	504	6.96%	502	6.93%
65-74	2,389	33.14%	2,370	32.88%	5,759	27.94%	5,732	27.81%	2,397	33.09%	2,425	33.48%
75-79	1,488	20.64%	1,512	20.98%	4,060	19.70%	4,137	20.07%	1,518	20.96%	1,519	20.97%
≥75	4,198	58.24%	4,213	58.45%	13,106	63.59%	13,180	63.95%	4,221	58.28%	4,205	58.06%
≥80	2,710	37.60%	2,701	37.47%	9,046	43.89%	9,043	43.88%	2,703	37.32%	2,686	37.08%
Gender												
Male	3,658	50.75%	3,666	50.86%	9,922	48.14%	9,873	47.90%	3,692	50.97%	3,712	51.25%
Female	3,550	49.25%	3,542	49.14%	10,688	51.86%	10,737	52.10%	3,551	49.03%	3,531	48.75%
Baseline Comorbidity												
Deyo-Charlson Comorbidity Index	4.91	2.59	4.91	2.57	5.05	2.65	5.04	2.62	4.91	2.56	4.95	2.55
CHA_2DS_2-VASc Score	4.77	1.49	4.79	1.51	4.86	1.52	4.85	1.50	4.79	1.51	4.77	1.48
0-1	47	0.64%	54	0.75%	117	0.56%	128	0.62%	54	0.75%	47	0.65%
2	344	4.77%	333	4.62%	929	4.51%	887	4.30%	333	4.60%	312	4.31%
3	1,024	14.21%	1,015	14.08%	2,736	13.28%	2,753	13.36%	1,023	14.12%	1,062	14.66%
4+	5,794	80.38%	5,806	80.55%	16,828	81.65%	16,842	81.72%	5,833	80.53%	5,822	80.38%
HAS-BLED Score	3.83	1.14	3.83	1.17	3.93	1.16	3.93	1.15	3.83	1.16	3.83	1.13
0-1	98	1.36%	106	1.47%	248	1.20%	266	1.29%	106	1.46%	103	1.42%
2	764	10.60%	811	11.25%	1,953	9.48%	1,894	9.19%	819	11.31%	746	10.30%
3+	6,346	88.04%	6,291	87.28%	18,409	89.32%	18,450	89.52%	6,318	87.23%	6,394	88.28%
Bleeding history	2,184	30.30%	2,151	29.84%	6,397	31.04%	6,415	31.13%	2,165	29.89%	2,158	29.79%
Stroke/SE	1,538	21.34%	1,525	21.16%	4,495	21.81%	4,469	21.68%	1,531	21.14%	1,499	20.70%
Comorbidities for Multimorbidity Definition												
CAD (CAD, prior MI, PCI/CABG)	5,218	72.39%	5,196	72.09%	14,920	72.39%	14,795	71.79%	5,222	72.10%	5,196	71.74%
Congestive Heart Failure	3,789	52.57%	3,824	53.05%	10,968	53.22%	10,894	52.86%	3,843	53.06%	3,847	53.11%
Valvular Disease	2,071	28.73%	1,894	26.28%	5,903	28.64%	5,530	26.83%	1,907	26.33%	1,891	26.11%
Hypertension	7,063	97.99%	7,047	97.77%	20,224	98.13%	20,233	98.17%	7,080	97.75%	7,064	97.53%
PVD (PAD or aortic aneurysm)	2,978	41.32%	2,966	41.15%	8,798	42.69%	8,962	43.48%	2,984	41.20%	3,102	42.83%
CVD (carotid stenosis, TIA, stroke)	2,695	37.39%	2,669	37.03%	7,986	38.75%	7,924	38.45%	2,679	36.99%	2,683	37.04%
Depression (diagnosis or anti-depressants) or Dementia	3,814	52.91%	3,980	55.22%	11,166	54.18%	11,514	55.87%	4,002	55.25%	4,018	55.47%
Pulmonary (COPD, asthma)	2,437	33.81%	2,582	35.82%	7,075	34.33%	7,452	36.16%	2,598	35.87%	2,604	35.95%
Sleep apnea	2,050	28.44%	1,973	27.37%	5,448	26.43%	4,946	24.00%	1,988	27.45%	1,866	25.76%
GI disorder (dyspepsia, GERD, PUD)	4,349	60.34%	4,334	60.13%	12,817	62.19%	12,828	62.24%	4,346	60.00%	4,351	60.07%
Chronic liver disease	407	5.65%	384	5.33%	1,108	5.38%	1,121	5.44%	384	5.30%	409	5.65%
Hypo- or hyperthyroidism	2,849	39.53%	2,785	38.64%	8,339	40.46%	8,090	39.25%	2,797	38.62%	2,752	38.00%
Diabetes	4,266	59.18%	4,321	59.95%	11,429	55.45%	11,366	55.15%	4,353	60.10%	4,343	59.96%
Musculoskeletal (falls, osteoporosis)	1,762	24.45%	1,706	23.67%	5,268	25.56%	5,418	26.29%	1,714	23.66%	1,790	24.71%
Renal (CKD or CrCl <50 ml/min)	3,429	47.57%	3,439	47.71%	10,276	49.86%	10,339	50.16%	3,454	47.69%	3,516	48.54%
Anemia	3,571	49.54%	3,550	49.25%	10,427	50.59%	10,421	50.56%	3,564	49.21%	3,606	49.79%
Malignancy	1,736	24.08%	1,582	21.95%	4,975	24.14%	4,914	23.84%	1,588	21.92%	1,694	23.39%
Dose of the Index Prescription												
Standard Dose (5mg Apixaban, 150mg Dabigatran, 20 mg Rivaroxaban)	5,155	71.52%	5,299	73.52%	13,805	66.98%	12,224	59.31%	5,332	73.62%	4,512	62.29%
Low Dose (2.5mg Apixaban, 75 mg Dabigatran, 15mg Rivaroxaban)	2,053	28.48%	1,909	26.48%	6,805	33.02%	8,386	40.69%	1,911	26.38%	2,731	37.71%

CABG: coronary artery bypass grafting; CHA_2DS_2-VASc Score: congestive heart failure, hypertension, age ≥75 years, diabetes mellitus, prior stroke or transient ischemic attack, or thromboembolism; COPD: chronic obstructive pulmonary disease; CVD: cardiovascular disease; GERD: gastroesophageal reflux disease; GI: gastrointestinal; HAS-BLED: Hypertension, Abnormal renal and liver function, Stroke, Bleeding, Labile INR, Elderly, Drugs or alcohol; MI: myocardial infarction; PCI: percutaneous coronary intervention; PUD: peptic ulcer disease; PVD: peripheral vascular disease; SD: standard deviation; TIA: transient ischemic attack

Figure 2. Incidence Rates and Hazard Ratios of Stroke/SE and Major Bleeding for Propensity Score Matched Patients

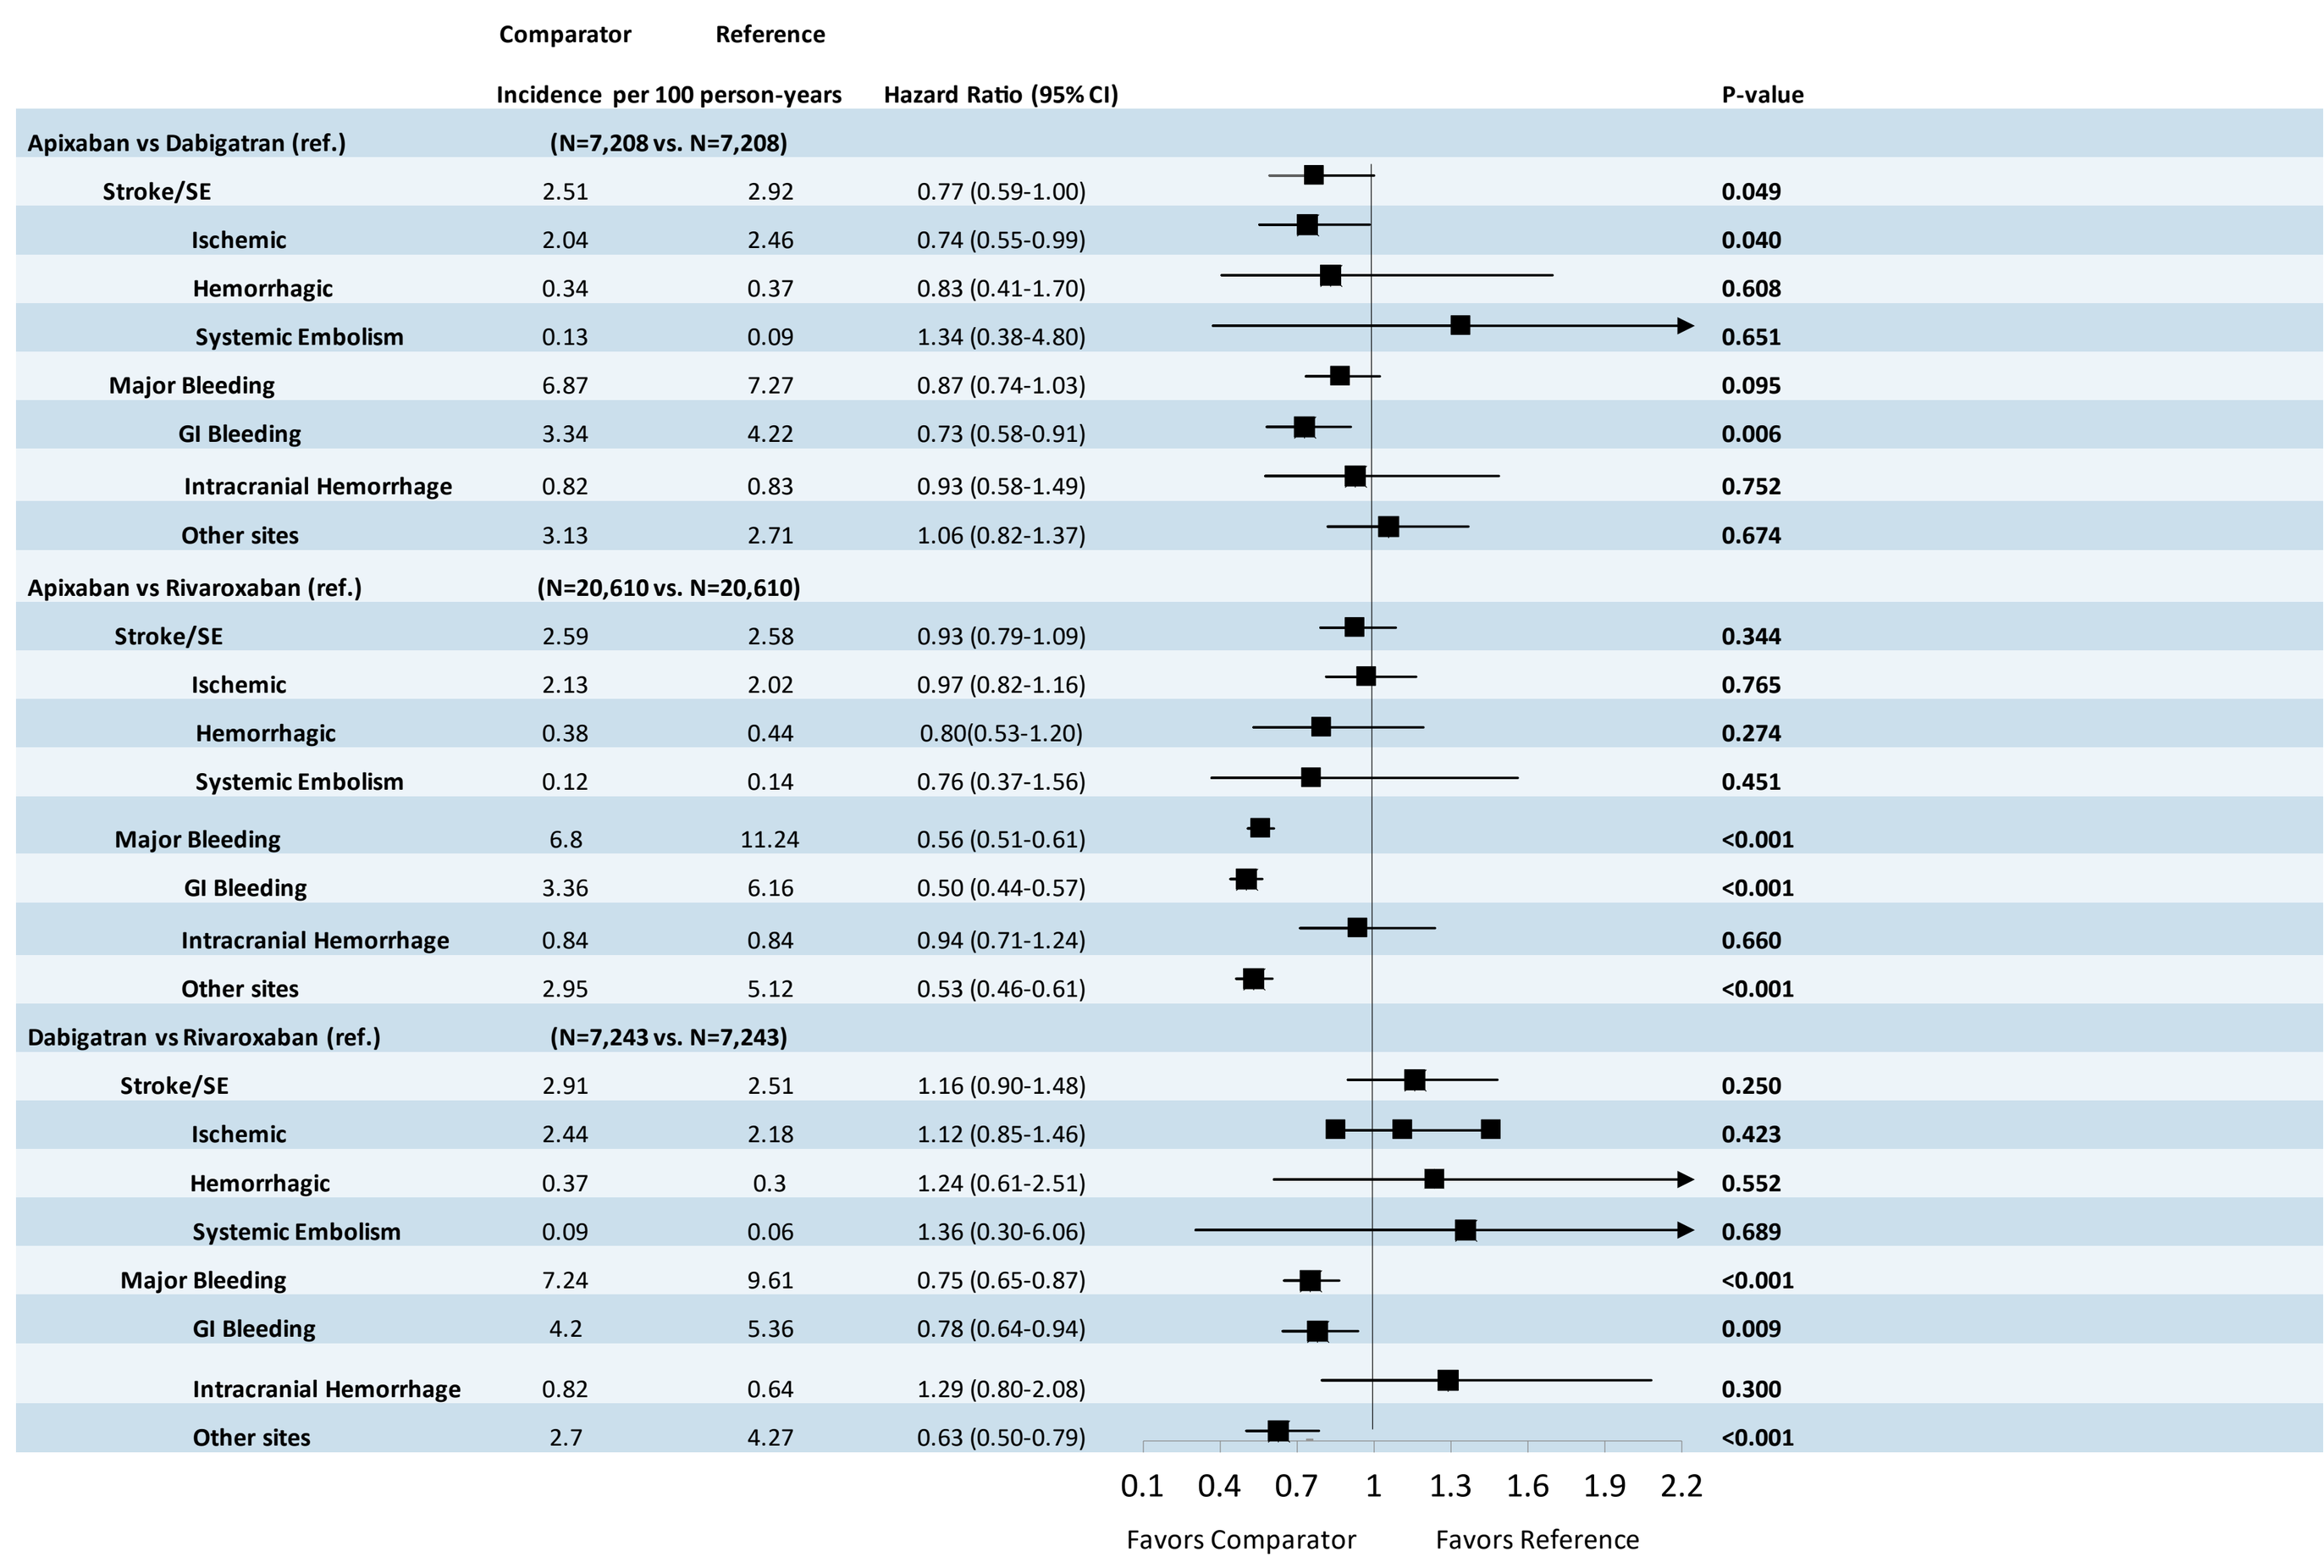
Comparator Reference
Incidence per 100 person-years
Hazard Ratio (95% CI)
P-value
Apixaban vs Dabigatran (ref.) (N=7,208 vs. N=7,208)
Stroke/SE 2.51 2.92 0.77 (0.59-1.00) 0.049
Ischemic 2.04 2.46 0.74 (0.55-0.99) 0.040
Hemorrhagic 0.34 0.37 0.83 (0.41-1.70) 0.608
Systemic Embolism 0.13 0.09 1.34 (0.38-4.80) 0.651
Major Bleeding 6.87 7.27 0.87 (0.74-1.03) 0.095
GI Bleeding 3.34 4.22 0.73 (0.58-0.91) 0.006
Intracranial Hemorrhage 0.82 0.83 0.93 (0.58-1.49) 0.752
Other sites 3.13 2.71 1.06 (0.82-1.37) 0.674
Apixaban vs Rivaroxaban (ref.) (N=20,610 vs. N=20,610)
Stroke/SE 2.59 2.58 0.93 (0.79-1.09) 0.344
Ischemic 2.13 2.02 0.97 (0.82-1.16) 0.765
Hemorrhagic 0.38 0.44 0.80(0.53-1.20) 0.274
Systemic Embolism 0.12 0.14 0.76 (0.37-1.56) 0.451
Major Bleeding 6.8 11.24 0.56 (0.51-0.61) <0.001
GI Bleeding 3.36 6.16 0.50 (0.44-0.57) <0.001
Intracranial Hemorrhage 0.84 0.84 0.94 (0.71-1.24) 0.660
Other sites 2.95 5.12 0.53 (0.46-0.61) <0.001
Dabigatran vs Rivaroxaban (ref.) (N=7,243 vs. N=7,243)
Stroke/SE 2.91 2.51 1.16 (0.90-1.48) 0.250
Ischemic 2.44 2.18 1.12 (0.85-1.46) 0.423
Hemorrhagic 0.37 0.3 1.24 (0.61-2.51) 0.552
Systemic Embolism 0.09 0.06 1.36 (0.30-6.06) 0.689
Major Bleeding 7.24 9.61 0.75 (0.65-0.87) <0.001
GI Bleeding 4.2 5.36 0.78 (0.64-0.94) 0.009
Intracranial Hemorrhage 0.82 0.64 1.29 (0.80-2.08) 0.300
Other sites 2.7 4.27 0.63 (0.50-0.79) <0.001
0.1 0.4 0.7 1 1.3 1.6 1.9 2.2
Favors Comparator
Favors Reference

CI: confidence interval; GI: gastrointestinal; SE: systemic embolism

Limitations

- PSM was conducted between each cohort pair; thus, the results are not comparable across all matched populations.
- Only associations could be concluded from this retrospective observational study. Although cohorts were PSM-adjusted, there are potential residual confounders such as over-the-counter aspirin use.
- While all patients were required to have at least 6 comorbidities, the severity of these conditions may not be equal among patients.
- As with all claims data, laboratory results such as international normalized ratios were unavailable, diagnoses were identified using ICD-9-CM codes, and drug prescriptions were identified through prescription claims. Missing values, coding errors, and lack of clinical accuracy may have introduced bias into the study.

Conclusions

- In this real-world subset of NVAF patients with multimorbidity in the United States, NOACs were associated with varying risks of stroke/SE and MB when compared to each other.
- Findings from this observational study provide insight into the safety and effectiveness of NOAC therapy among high-risk NVAF patients with multiple comorbidities.

References

[1]Parks AL, Fang MC. Anticoagulation in older adults with multimorbidity. *Clin Geriatr Med.* 2016;32(2):331-46.
[2]Vanbeselaere V, Truyers C, Elli S, et al. Association between atrial fibrillation, anticoagulation, risk of cerebrovascular events and multimorbidity in general practice: A registry-based study. *BMC Cardiovasc Disord.* 2016;16:61.
[3]Jani BD, Nicholl BI, McQueenie R, et al. Multimorbidity and co-morbidity in atrial fibrillation and effects on survival: Findings from UK Biobank cohort. *Europace.* 2018;20(FI_3):f329-36.
[4]Ruff CT, Giugliano RP, Braunwald E, et al. Comparison of the efficacy and safety of new oral anticoagulants with warfarin in patients with atrial fibrillation: A meta-analysis of randomised trials. *Lancet.* 2014;383(9921):955-62.
[5]Thigpen JL, Dillon C, Forster KB, et al. Validity of international classification of disease codes to identify ischemic stroke and intracranial hemorrhage among individuals with associated diagnosis of atrial fibrillation. *Circ Cardiovasc Qual Outcomes.* 2015;8(1):8-14.
[6]Cunningham A, Stein CM, Chung CP, Daugherty JR, Smalley WE, Ray W. An automated database case definition for serious bleeding related to oral anticoagulant use. *Pharmacoepidemiol Drug Saf.* 2011;20(6):560-6.

Disclosures

Funding for this research was provided by Bristol-Myers Squibb Company and Pfizer Inc.

S. Deitelzweig and G.Y.H. Lip are paid consultants to Bristol-Myers Squibb Company and Pfizer Inc.

A. Kang, A. Dhamane, C. Klem, L. Rosenblatt, and J. Jiang are employees of Bristol-Myers Squibb Company.

X. Luo and J. Mardekian are employees of Pfizer Inc.

A. Keshishian is an employee of STATinMED Research, which is a paid consultant to Bristol-Myers Squibb Company and Pfizer Inc., the study sponsors.

Download an e-copy here

Link

QR Code

Poster presented at the *American Heart Association (AHA) Scientific Sessions*, Philadelphia, PA: November 16-18, 2019