Effectiveness and Safety of Apixaban, Dabigatran, and Rivaroxaban versus Warfarin in Obese Patients with Nonvalvular Atrial Fibrillation: ARISTOPHANES Subgroup Analysis

Steve Deitelzweig[1], Allison Keshishian[2,3], Amiee Kang[4], Amol Dhamane[4], Xuemei Luo[5], Xiaoyan Li[4], Neeraja Balachander[4], Lisa Rosenblatt[4], Jack Mardekian[6], Xianying Pan[4], Manuela Di Fusco[6], Alessandra B. Garcia Reeves [4,7], Huseyin Yuce[3], Gregory Y. H. Lip[8]

[1]Ochsner Clinic Foundation, Department of Hospital Medicine, New Orleans, LA, USA and The University of Queensland School of Medicine, Ochsner Clinical School, New Orleans, LA ; [2]SIMR, LLC, Ann Arbor, MI; [3]New York City College of Technology, City University of New York, New York, NY; [4]Bristol-Myers Squibb Company, Lawrenceville, NJ, USA; [5]Pfizer, Inc., Groton, CT, US; [6]Pfizer, Inc., New York, NY, USA; [7]University of North Carolina, Chapel Hill, NC; [8]Liverpool Centre for Cardiovascular Science, University of Liverpool and Liverpool Heart & Chest Hospital, Liverpool, United Kingdom; and Aalborg Thrombosis Research Unit, Department of Clinical Medicine, Aalborg University, Aalborg, Denmark

INTRODUCTION

- Atrial fibrillation (AF) is the most common heart dysrhythmia diagnosed in the United States.[1] AF patients have a five-fold increase in stroke risk compared to the general population, resulting in significant morbidity and mortality.[2]
- Obesity (defined as BMI ≥30) has been linked with hypofibrinolysis, inflammation, and a prothrombotic state, bolstering the thromboembolic effects of AF.[3]
- According to the Center for Disease Control and Prevention (CDC), prevalence of obesity in the United States in 2015-16 was 39.8%.[4] Moreover, clinical trials showed that 40% patients with NVAF in the ARISTOTLE trial were obese and ~17% in the RE-LY trial had weight ≥100 kgs.[5.6] Global registries of AF patients also showed obesity prevalence of 30%.[7,8]
- Clinical trials have shown that novel oral anticoagulants (NOACs)—including apixaban, dabigatran, and rivaroxaban—are at least as effective as warfarin for preventing stroke/systemic embolism (SE) among patients with nonvalvular AF (NVAF).[6,9,10] A meta-analysis conducted among trial data in obese patients suggested a 38% lower risk for stroke/SE and 16% lower risk for major bleeding (MB), compared to normal body mass index (BMI) patients; additionally, among a subset of obese patients, NOACs showed a similar risk of stroke/SE (hazard ratio [HR]: 0.84, 95% confidence interval [CI]: 0.70-1.03) and major bleeding (HR: 1.03, 95% CI: 0.90-1.18) compared to warfarin.[11]
- Few observational studies leveraging real-world datasets have evaluated a high-risk subset of an obese NVAF population.[12]

OBJECTIVES

This study was a subgroup analysis of the ARISTOPHANES (Anticoagulants for Reduction In STroke: Observational Pooled analysis on Health outcomes ANd Experience of patientS; ClinicalTrials.gov ID: NCT03087487) study, which was conducted to compare the risk of stroke/SE and MB of NOACs versus warfarin among NVAF patients with obesity.

METHODS

Data Source

This was an observational retrospective cohort analysis using de-identified patient data from CMS Medicare data and four US commercial claims databases (Optum, PharMetrics, Humana, and MarketScan), covering >180 million beneficiaries annually (~56% of the US population).

Figure 1. Patient Selection Flow Chart

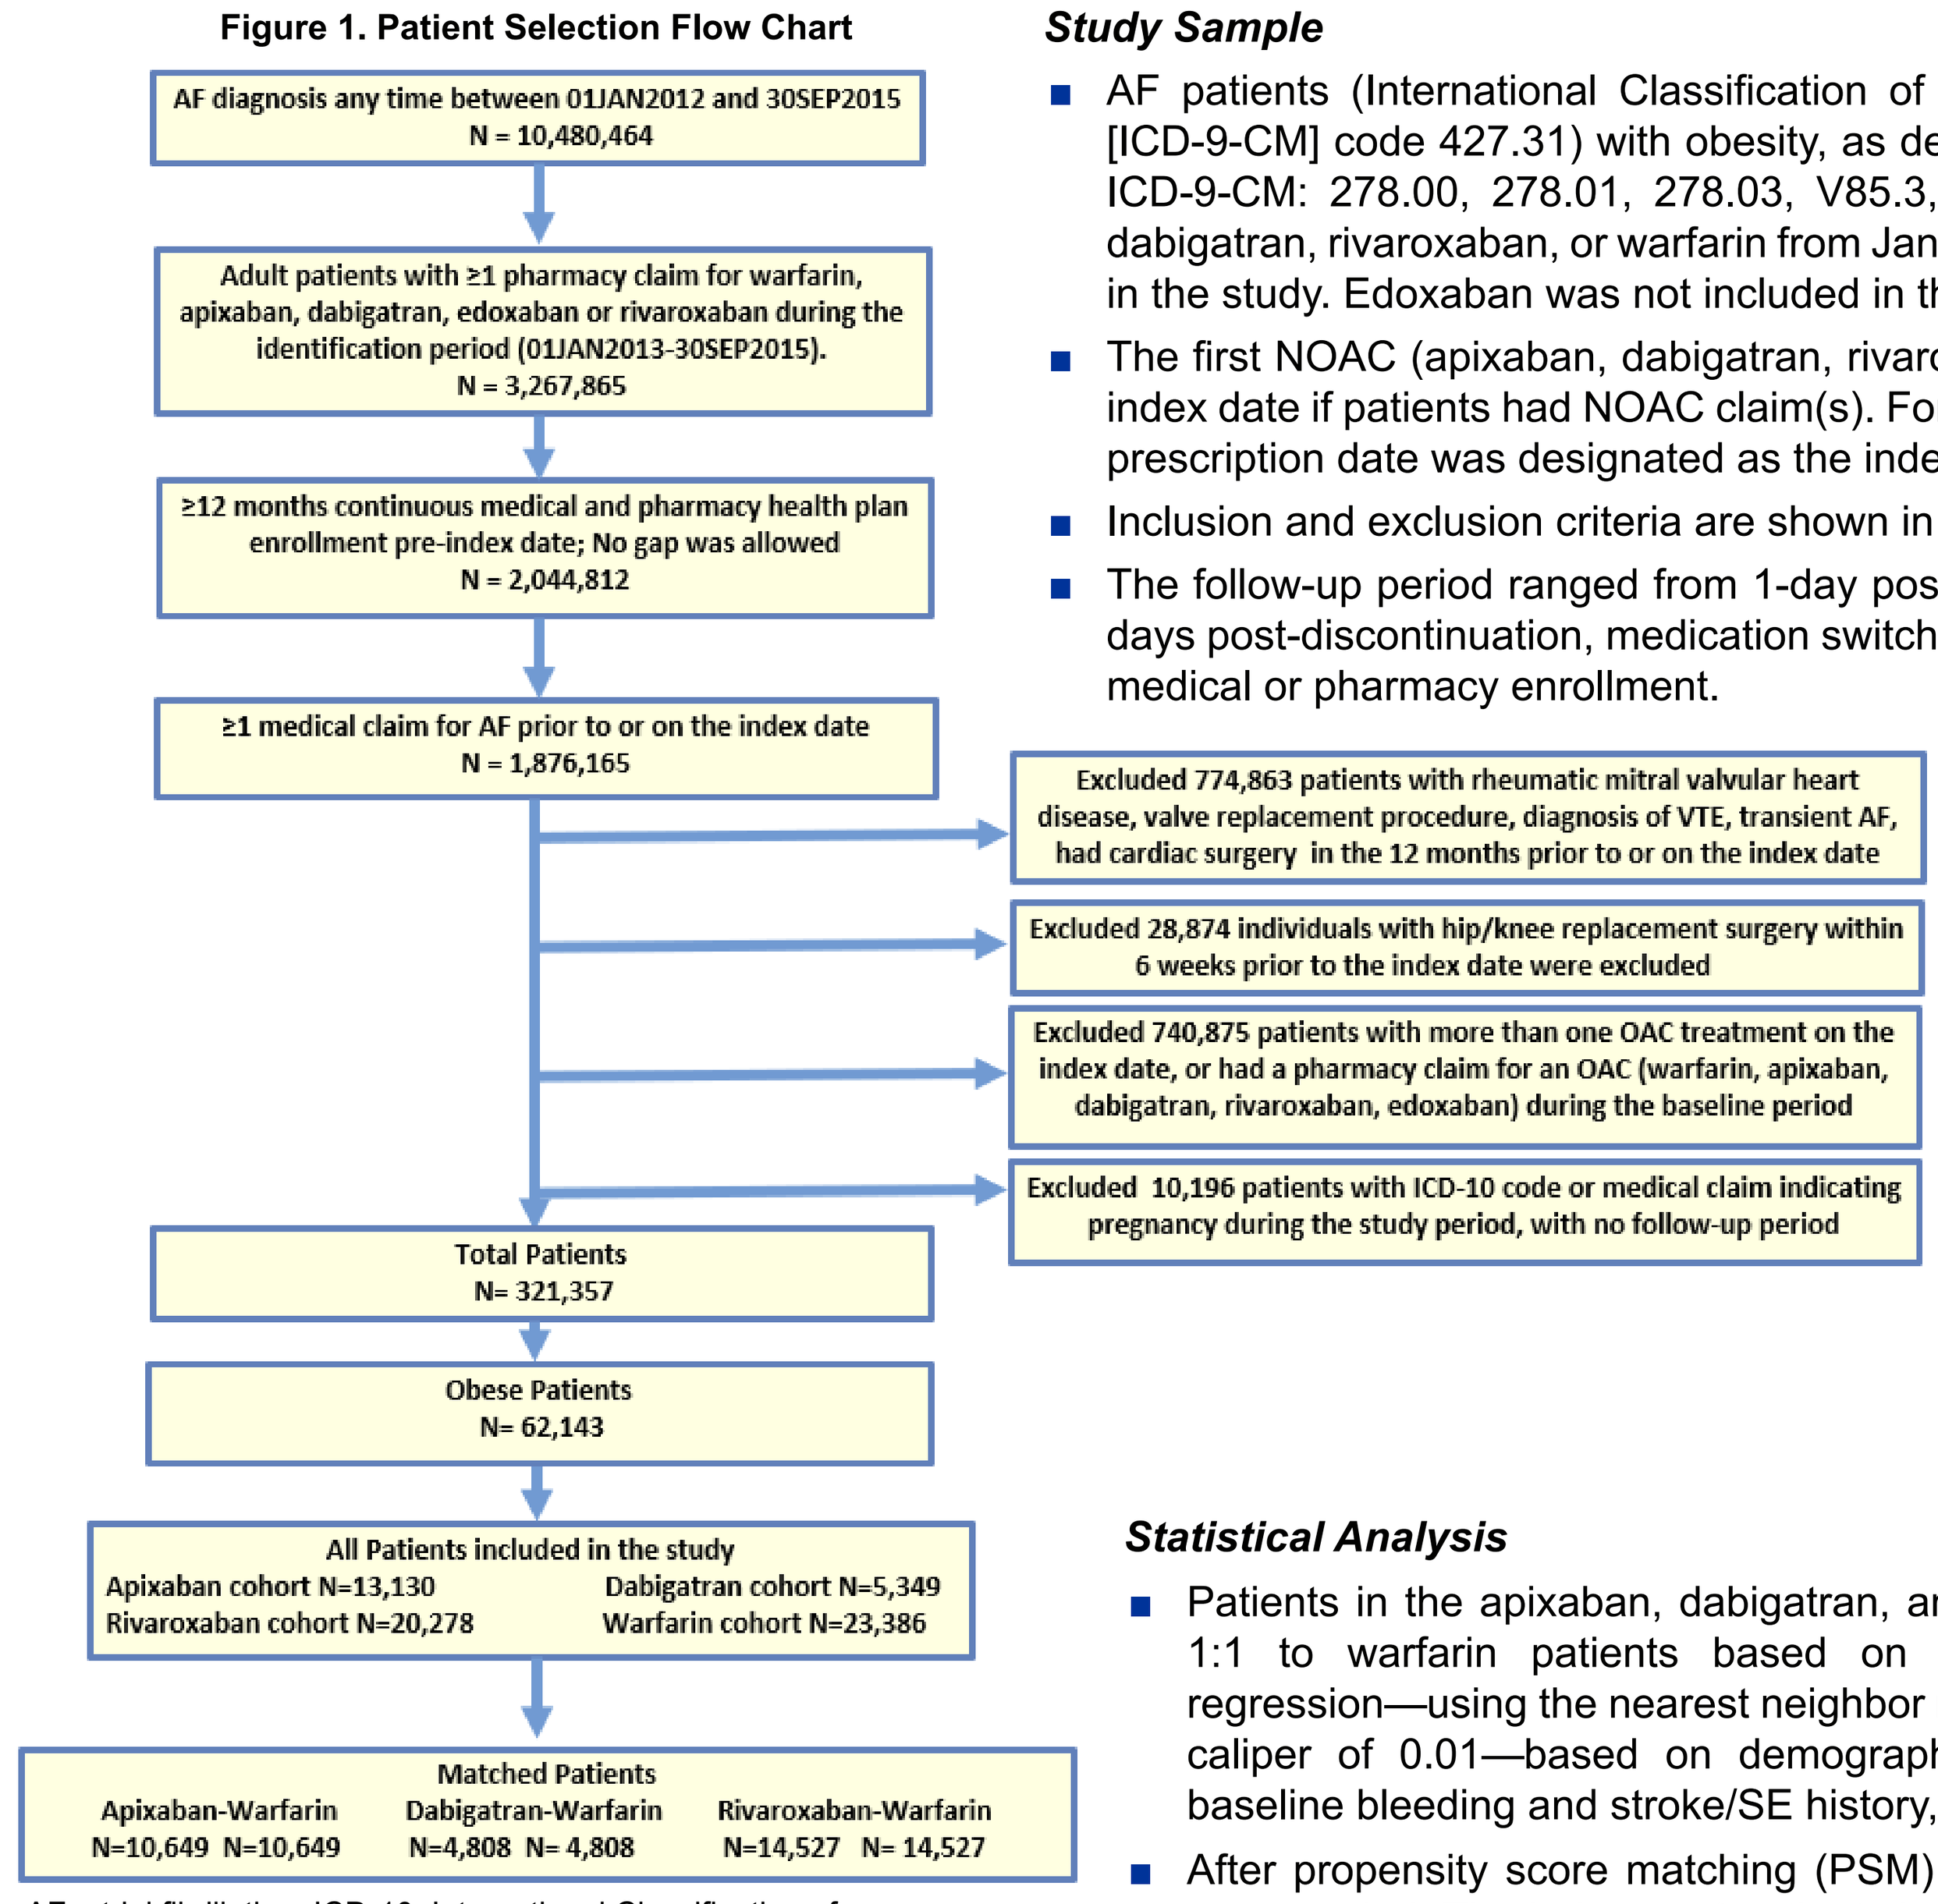

AF diagnosis any time between 01JAN2012 and 30SEP2015
N = 10,480,464
Adult patients with ≥1 pharmacy claim for warfarin, apixaban, dabigatran, edoxaban or rivaroxaban during the identification period (01JAN2013-30SEP2015).
N = 3,267,865
≥12 months continuous medical and pharmacy health plan enrollment pre-index date; No gap was allowed
N = 2,044,812
≥1 medical claim for AF prior to or on the index date
N = 1,876,165
Excluded 774,863 patients with rheumatic mitral valvular heart disease, valve replacement procedure, diagnosis of VTE, transient AF, had cardiac surgery in the 12 months prior to or on the index date
Excluded 28,874 individuals with hip/knee replacement surgery within 6 weeks prior to the index date were excluded
Excluded 740,875 patients with more than one OAC treatment on the index date, or had a pharmacy claim for an OAC (warfarin, apixaban, dabigatran, rivaroxaban, edoxaban) during the baseline period
Excluded 10,196 patients with ICD-10 code or medical claim indicating pregnancy during the study period, with no follow-up period
Total Patients
N= 321,357
Obese Patients
N= 62,143
All Patients included in the study
Apixaban cohort N=13,130
Dabigatran cohort N=5,349
Rivaroxaban cohort N=20,278
Warfarin cohort N=23,386
Matched Patients
Apixaban-Warfarin
N=10,649 N=10,649
Dabigatran-Warfarin
N=4,808 N= 4,808
Rivaroxaban-Warfarin
N=14,527 N= 14,527

AF: atrial fibrillation; ICD-10: International Classification of Diseases, Tenth Revision; OAC: oral anticoagulant; VTE: venous thromboembolism

Note: The edoxaban cohort was not included in the analysis due to the small sample size obtained among total AF patients (N=35).

Study Sample

- AF patients (International Classification of Diseases, Ninth Revision, Clinical Modification [ICD-9-CM] code 427.31) with obesity, as defined as BMI ≥30 (diagnosis codes for BMI ≥30: ICD-9-CM: 278.00, 278.01, 278.03, V85.3, V85.4) who initiated treatment with apixaban, dabigatran, rivaroxaban, or warfarin from January 1, 2013 - September 30, 2015 were included in the study. Edoxaban was not included in the study due to the small sample size (N=35).
- The first NOAC (apixaban, dabigatran, rivaroxaban) prescription date was designated as the index date if patients had NOAC claim(s). For patients without a NOAC claim, the first warfarin prescription date was designated as the index date.
- Inclusion and exclusion criteria are shown in Figure 1.
- The follow-up period ranged from 1-day post-index date until the earliest of the following: 30 days post-discontinuation, medication switch, death, end of study period, or end of continuous medical or pharmacy enrollment.

Outcome Definitions

- The outcome measures were time to first stroke/SE and MB events based on the principal or first-listed ICD-9-CM diagnosis of inpatient claims. The diagnosis codes used for stroke/SE and MB were based on validated administrative claim-based algorithms and the *International Society on Thrombosis and Hemostasis'* definition of MB.[13,14]
- Stroke/SE included ischemic stroke, hemorrhagic stroke, and SE; MB included gastrointestinal bleeding, intracranial hemorrhage, and bleeding at other sites.

Statistical Analysis

- Patients in the apixaban, dabigatran, and rivaroxaban treatment cohorts were matched 1:1 to warfarin patients based on the propensity scores generated by logistic regression—using the nearest neighbor matching method without replacement and with a caliper of 0.01—based on demographics, Charlson comorbidity index (CCI) score, baseline bleeding and stroke/SE history, comorbidities, and baseline co-medications.
- After propensity score matching (PSM) in each database, the resulting patient records were pooled. The balance of patient characteristics was checked based on standardized differences with a threshold of 10%.
- Incidence of events was calculated as the number of events per 100 person-years.
- Cox proportional hazards models with robust sandwich estimators were used to estimate the hazard ratios for the risk of stroke/SE and MB endpoints between the matched cohorts. P-values<0.05 were considered statistically significant.

RESULTS

Baseline Results

- After applying selection criteria, a total of 62,143 obese patients (19.3 %) with NVAF were included: 13,130 (21.1%) apixaban, 5,349 (8.6%) dabigatran, 20,278 (32.6%) rivaroxaban, and 23,386 (37.6%) warfarin patients.
- Before PSM, warfarin patients were the oldest and had the highest CCI, CHA_2DS_2-VASc, and HAS-BLED scores compared to NOAC patients.
- PSM resulted in 10,649 pairs for the apixaban-warfarin, 4,808 pairs for dabigatran-warfarin, and 14,527 pairs for rivaroxaban-warfarin cohorts (Table 1). The mean follow-up time ranged between 6 and 8 months.
- The mean ages were 71, 69, and 71 years for the apixaban-warfarin, dabigatran-warfarin, and rivaroxaban-warfarin cohort pairs, respectively. The mean CHA_2DS_2-VASc scores were 3.9, 3.7, and 3.9 the for apixaban-warfarin, dabigatran-warfarin, and rivaroxaban-warfarin cohort pairs, respectively. Baseline characteristics are shown in Table 1.

Table 1. Baseline Characteristics for Propensity Score-Matched Obese Patients prescribed NOACs vs Warfarin

	Warfarin Cohort (reference)		Apixaban Cohort		Warfarin Cohort (Reference)		Dabigatran Cohort		Warfarin Cohort (Reference)		Rivaroxaban Cohort	
	N/Mean	%/SD	N/Mean	%/SD	N/Mean	%/SD	N/Mean	%/SD	N/Mean	%/SD	N/Mean	%/SD
Sample Size	**10,649**	**100%**	**10,649**	**100%**	**4,808**	**100%**	**4,808**	**100%**	**14,527**	**100%**	**14,527**	**100%**
Age*	71.1	10.0	71	10	69.5	9.8	69.4	9.8	70.9	9.7	70.9	9.6
18-54	655	6.2%	659	6.2%	370	7.7%	391	8.1%	865	6.0%	828	5.7%
55-64	1,746	16.4%	1,709	16.0%	901	18.7%	879	18.3%	2,197	15.1%	2,210	15.2%
65-74	4,280	40.2%	4,310	40.5%	2,052	42.7%	2,082	43.3%	6,271	43.2%	6,369	43.8%
75-79	1,906	17.9%	1,949	18.3%	814	16.9%	818	17.0%	2,652	18.3%	2,626	18.1%
≥80	2,062	19.4%	2,022	19.0%	671	14.0%	638	13.3%	2,542	17.5%	2,494	17.2%
Sex*												
Male	5,738	53.9%	5,735	53.9%	2,735	56.9%	2,724	56.7%	7,760	53.4%	7,697	53.0%
Female	4,911	46.1%	4,914	46.1%	2,073	43.1%	2,084	43.3%	6,767	46.6%	6,830	47.0%
US Geographic Region*												
Northeast	1,629	15.3%	1,643	15.4%	895	18.6%	882	18.3%	2,340	16.1%	2,356	16.2%
Midwest	2,628	24.7%	2,638	24.8%	1,168	24.3%	1,202	25.0%	3,875	26.7%	3,868	26.6%
South	4,884	45.9%	4,824	45.3%	1,980	41.2%	1,968	40.9%	6,082	41.9%	6,079	41.8%
West	1,463	13.7%	1,499	14.1%	740	15.4%	726	15.1%	2,173	15.0%	2,167	14.9%
Other	45	0.4%	45	0.4%	25	0.5%	30	0.6%	57	0.4%	57	0.4%
Baseline Comorbidity												
Deyo-Charlson Comorbidity Index Score*	3.8	2.9	3.8	2.9	3.3	2.8	3.3	2.8	3.7	2.9	3.7	2.9
CHA_2DS_2-VASc Score	3.9	1.7	3.9	1.7	3.7	1.7	3.7	1.7	3.9	1.7	3.9	1.7
0	198	1.9%	165	1.5%	96	2.0%	107	2.2%	229	1.6%	222	1.5%
1	502	4.7%	534	5.0%	301	6.3%	310	6.4%	679	4.7%	719	4.9%
2	1,407	13.2%	1,515	14.2%	756	15.7%	787	16.4%	1,867	12.9%	2,002	13.8%
3	2,323	21.8%	2,293	21.5%	1,159	24.1%	1,121	23.3%	3,279	22.6%	3,125	21.5%
4+	6,219	58.4%	6,142	57.7%	2,496	51.9%	2,483	51.6%	8,473	58.3%	8,459	58.2%
HAS-BLED Score	3.2	1.4	3.2	1.4	3.0	1.4	3.0	1.3	3.2	1.4	3.2	1.4
0	184	1.7%	144	1.4%	96	2.0%	93	1.9%	224	1.5%	198	1.4%
1	894	8.4%	949	8.9%	539	11.2%	533	11.1%	1,181	8.1%	1,276	8.8%
2	2,406	22.6%	2,314	21.7%	1,263	26.3%	1,261	26.2%	3,411	23.5%	3,264	22.5%
3+	7,165	67.3%	7,242	68.0%	2,910	60.5%	2,921	60.8%	9,711	66.8%	9,789	67.4%
Bleeding History*	2,398	22.5%	2,390	22.4%	949	19.7%	934	19.4%	3,282	22.6%	3,265	22.5%
Congestive Heart Failure*	4,232	39.7%	4,196	39.4%	1,744	36.3%	1,731	36.0%	5,732	39.5%	5,771	39.7%
Diabetes Mellitus*	5,761	54.1%	5,805	54.5%	2,569	53.4%	2,561	53.3%	7,980	54.9%	8,015	55.2%
Hypertension*	10,001	93.9%	10,012	94.0%	4,479	93.2%	4,473	93.0%	13,608	93.7%	13,624	93.8%
Renal Disease*	3,327	31.2%	3,407	32.0%	1,180	24.5%	1,185	24.6%	4,298	29.6%	4,297	29.6%
Liver Disease*	751	7.1%	744	7.0%	338	7.0%	312	6.5%	1,054	7.3%	1,058	7.3%
Myocardial Infarction*	1,256	11.8%	1,279	12.0%	460	9.6%	493	10.3%	1,787	12.3%	1,769	12.2%
Dyspepsia or Stomach Discomfort*	2,546	23.9%	2,545	23.9%	1,023	21.3%	1,028	21.4%	3,481	24.0%	3,520	24.2%
Non-stroke/ SE Peripheral Vascular Disease	6,131	57.6%	6,097	57.3%	2,585	53.8%	2,570	53.5%	8,302	57.1%	8,316	57.2%
Stroke/SE*	1,265	11.9%	1,215	11.4%	479	10.0%	478	9.9%	1,718	11.8%	1,682	11.6%
Transient Ischemic Attack*	717	6.7%	697	6.5%	300	6.2%	283	5.9%	941	6.5%	948	6.5%
Anemia and Coagulation Defects*	3,405	32.0%	3,359	31.5%	1,236	25.7%	1,258	26.2%	4,465	30.7%	4,454	30.7%
Alcoholism*	144	1.4%	148	1.4%	77	1.6%	67	1.4%	193	1.3%	203	1.4%
Peripheral Artery Disease	2,523	23.7%	2,352	22.1%	1,004	20.9%	913	19.0%	3,377	23.2%	3,333	22.9%
Coronary Artery Disease	5,407	50.8%	5,481	51.5%	2,226	46.3%	2,274	47.3%	7,288	50.2%	7,368	50.7%
Dyslipidemia*	8,714	81.8%	8,720	81.9%	3,879	80.7%	3,831	79.7%	11,793	81.2%	11,795	81.2%
Morbid Obesity	4,287	40.3%	4,021	37.8%	1,957	40.7%	1,861	38.7%	5,923	40.8%	5,849	40.3%
Index Prescription Dose												
Standard Dose			9,171	86.1%			4,198	87.3%			11,139	76.7%
Low Dose			1,478	13.9%			610	12.7%			2,691	18.5%
Follow-up Time (days)	243.5	222.8	186.9	171.0	243.5	224.1	232.8	230.3	245.1	223.6	237.9	225.5

ACEis: angiotensin-converting enzyme inhibitors; ARBs: angiotensin-receptor blockers; CHA_2DS_2-VASc: congestive heart failure, hypertension, aged ≥75 years, diabetes mellitus, prior stroke or transient ischemic attack or thromboembolism, vascular disease, aged 65-74 years, sex category; HAS-BLED: hypertension, abnormal renal and liver function, stroke, bleeding, labile international normalized ratios, elderly, drugs and alcohol; NOAC: non-VKA oral anticoagulant; NSAIDs: non-steroidal anti-inflammatory drugs; PSM: propensity score matching; SD: standard deviation; SE: systemic embolism

[a]International normalized ratio values were unavailable, so a modified HAS-BLED score was calculated with a range of 0 - 8.

[b]Standard dose: 5mg apixaban, 150mg dabigatran, 20mg rivaroxaban; after PSM, baseline characteristics were balanced between cohorts.

[c]Lower dose: 2.5mg apixaban, 75mg dabigatran, 10/15mg rivaroxaban; 697 (4.8%) of rivarox-warf. patients had lower dose (10mg) rivaroxaban.

*Covariates included in the propensity score matching.

RESULTS (cont'd)

Figure 2. Comparison of Stroke/SE and Major Bleeding Rate Among NVAF PSM Patients with Obesity Who Initiated NOACs vs Warfarin

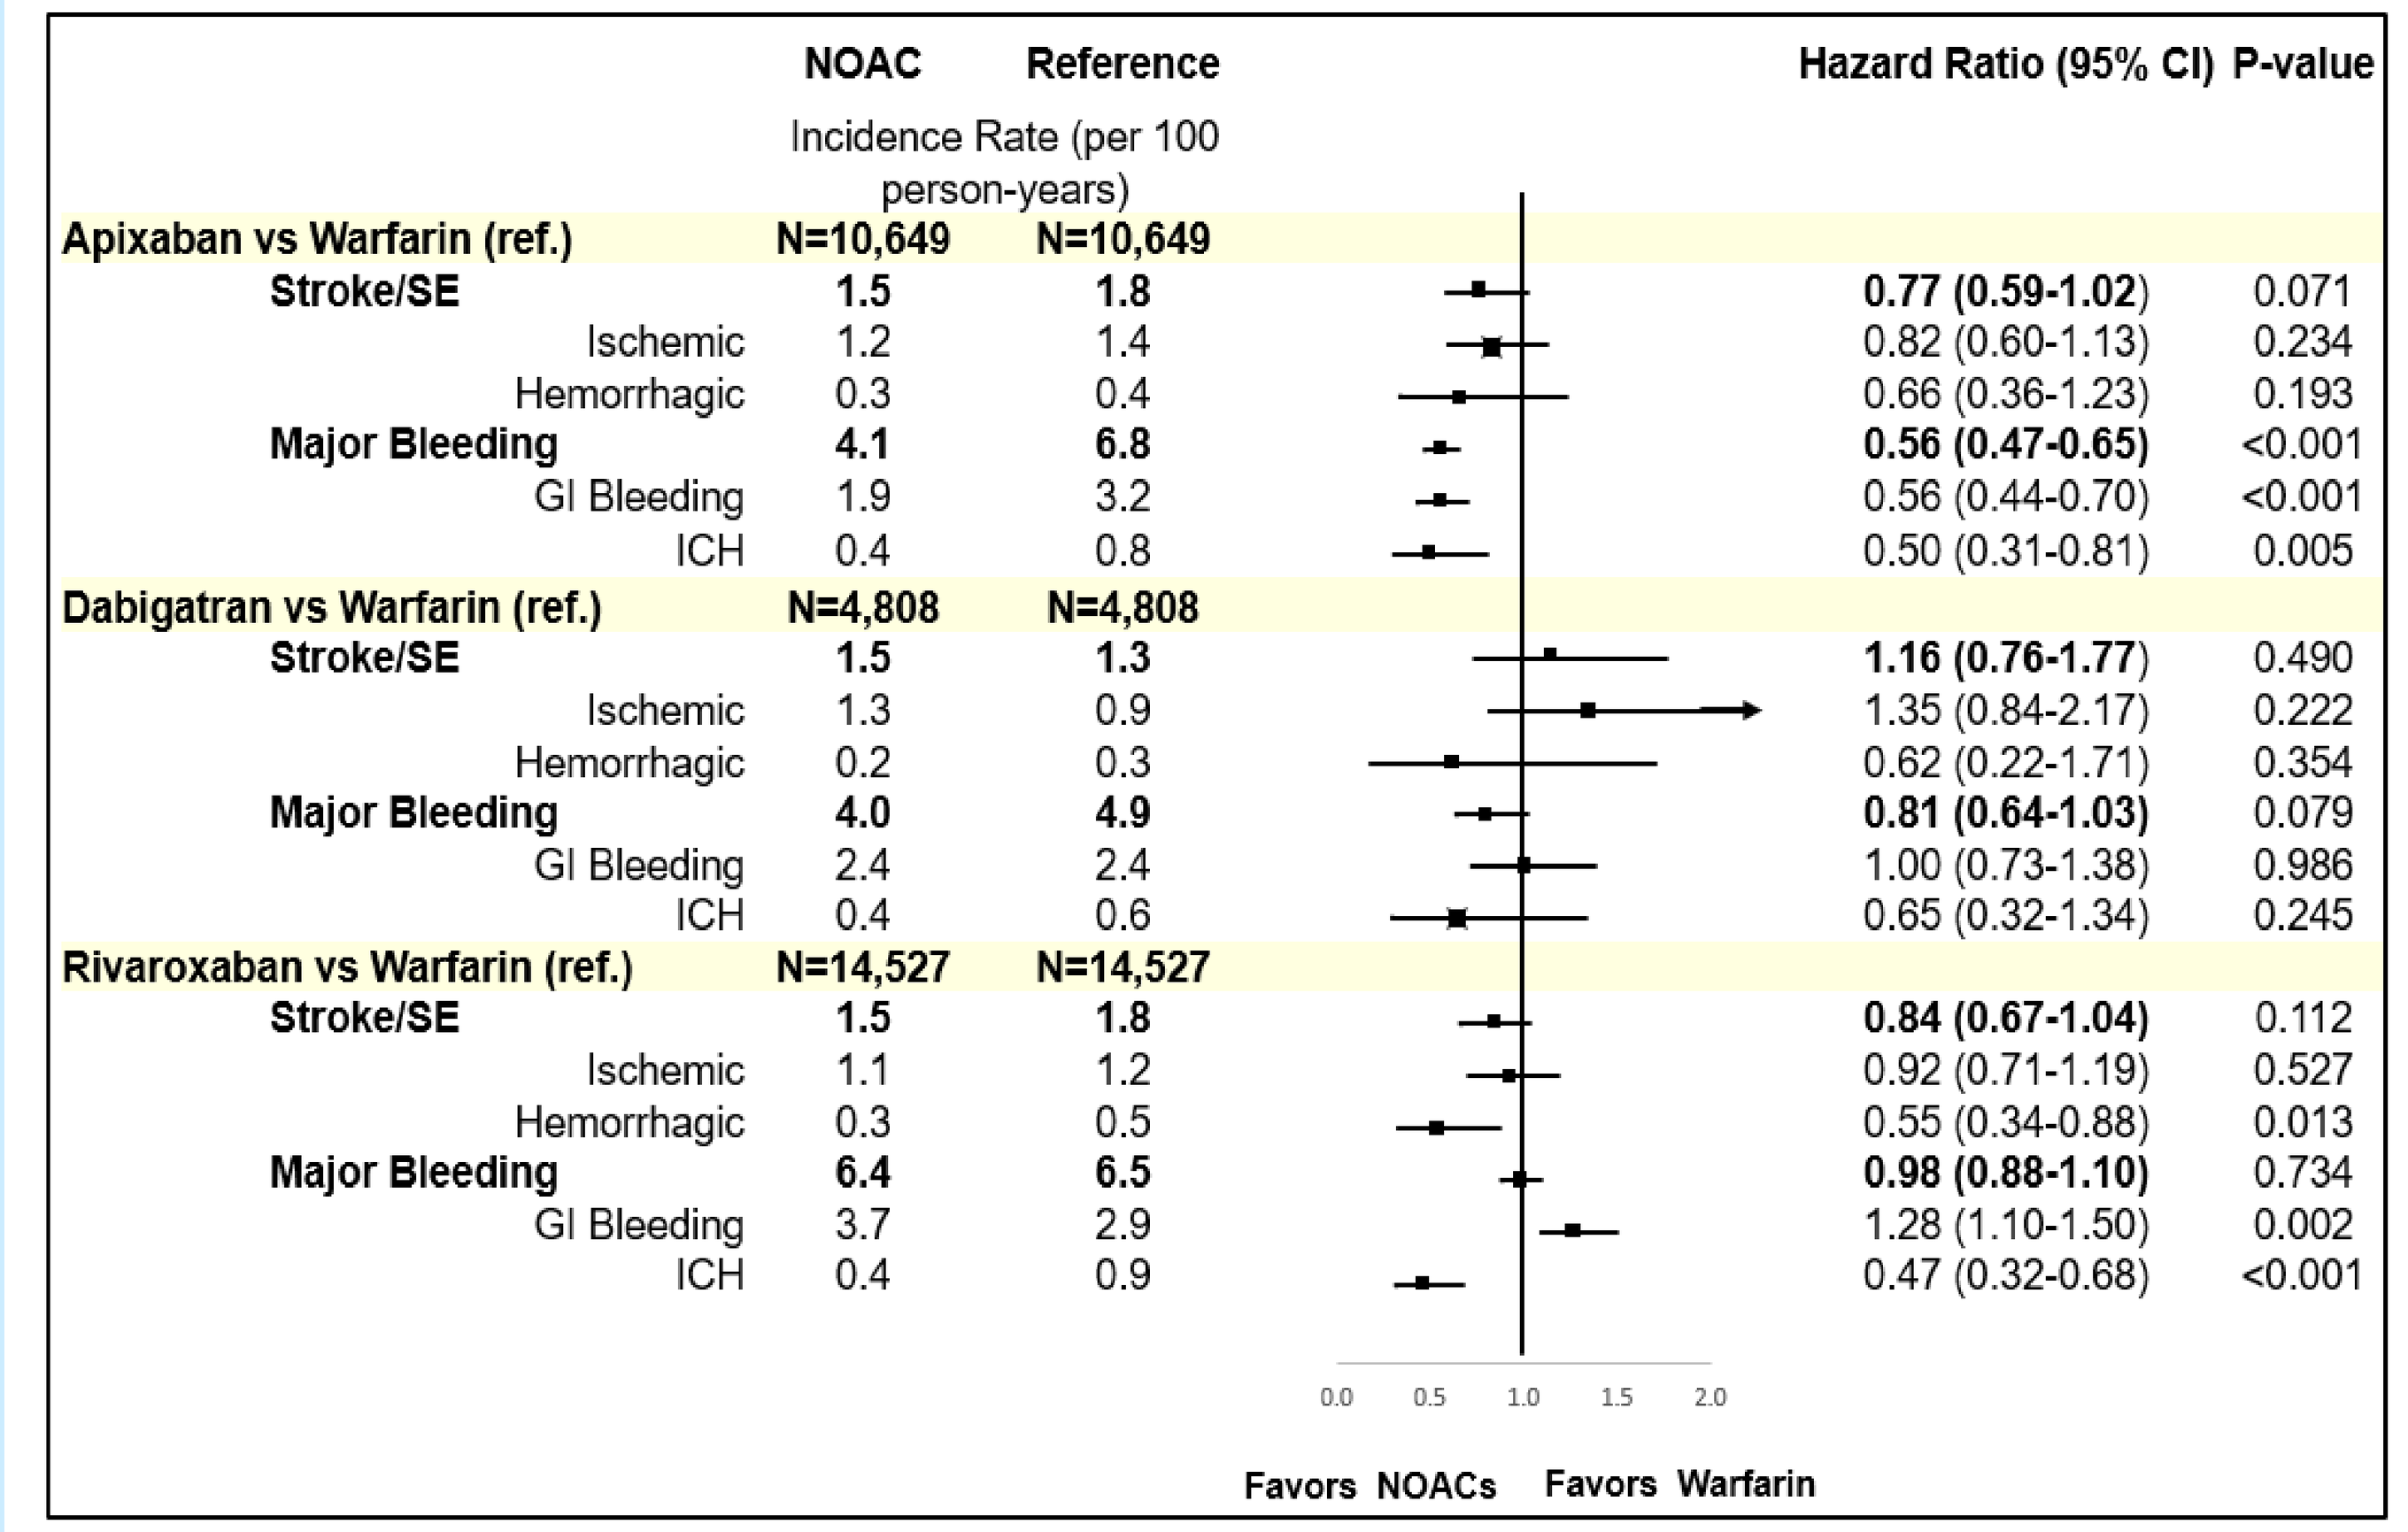

NOAC
Reference
Incidence Rate (per 100 person-years)
Hazard Ratio (95% CI)
P-value
Apixaban vs Warfarin (ref.) N=10,649 N=10,649
Stroke/SE 1.5 1.8 0.77 (0.59-1.02) 0.071
Ischemic 1.2 1.4 0.82 (0.60-1.13) 0.234
Hemorrhagic 0.3 0.4 0.66 (0.36-1.23) 0.193
Major Bleeding 4.1 6.8 0.56 (0.47-0.65) <0.001
GI Bleeding 1.9 3.2 0.56 (0.44-0.70) <0.001
ICH 0.4 0.8 0.50 (0.31-0.81) 0.005
Dabigatran vs Warfarin (ref.) N=4,808 N=4,808
Stroke/SE 1.5 1.3 1.16 (0.76-1.77) 0.490
Ischemic 1.3 0.9 1.35 (0.84-2.17) 0.222
Hemorrhagic 0.2 0.3 0.62 (0.22-1.71) 0.354
Major Bleeding 4.0 4.9 0.81 (0.64-1.03) 0.079
GI Bleeding 2.4 2.4 1.00 (0.73-1.38) 0.986
ICH 0.4 0.6 0.65 (0.32-1.34) 0.245
Rivaroxaban vs Warfarin (ref.) N=14,527 N=14,527
Stroke/SE 1.5 1.8 0.84 (0.67-1.04) 0.112
Ischemic 1.1 1.2 0.92 (0.71-1.19) 0.527
Hemorrhagic 0.3 0.5 0.55 (0.34-0.88) 0.013
Major Bleeding 6.4 6.5 0.98 (0.88-1.10) 0.734
GI Bleeding 3.7 2.9 1.28 (1.10-1.50) 0.002
ICH 0.4 0.9 0.47 (0.32-0.68) <0.001
0.0 0.5 1.0 1.5 2.0
Favors NOACs
Favors Warfarin

CI: confidence interval; GI: gastrointestinal; ICH; intracranial hemorrhage; SE: systemic embolism.

Outcome Results

- Post-PSM incidence rates of stroke/SE and MB are shown in Figure 2.
- Patients treated with apixaban had a similar risk of stroke/SE and lower risk for MB compared to warfarin (Figure 2).
- Patients treated with dabigatran and rivaroxaban had a similar risk of stroke/SE and MB compared to warfarin.

LIMITATIONS

- PSM was conducted between each cohort pair; thus, the results are not comparable across all matched populations.
- Only associations could be concluded from this retrospective observational study. Although cohorts were PSM-adjusted, there are potential residual confounders such as over-the-counter use of aspirin or warfarin treatment dose change.
- As with all claims data, laboratory results such as international normalized ratios were unavailable, diagnoses were identified using ICD-9-CM codes, and drug prescriptions were identified through prescription claims. Missing values, coding errors, and lack of clinical accuracy may have introduced bias into the study. Body measurements such as weight or lean body mass were unavailable in the claims data to validate obesity diagnosis codes. Our current obesity cohort classification was based purely on diagnosis codes related to BMI ≥30.
- Although some of the datasets contain data from different insurance plans without plan-level overlap, others are employer-based claims datasets that may have duplicate patient records when pooled, but based on a published estimate of 0.5%, there are likely too few duplicates to significantly affect results.[15]

CONCLUSIONS

In this large real-world obese subset of NVAF patients in the United States, all NOACs had a similar risk of stroke/SE compared to warfarin, while apixaban had a lower risk of major bleeding compared to warfarin. Findings from this observational analysis provide important insights regarding the use of oral anticoagulant therapy among high-risk patients with NVAF and obesity in a real-world setting.

REFERENCES

1 Shea JB, Sears SF. *Circulation.* 2008;117(20):e340-3.
2 Wolf PA, Abbott RD, Kannel WB. *Arch Intern Med.* 1987;147(9):1561-4.
3 Darvall KA, Sam RC, Silverman SH, et al. *Eur J Vasc Endovasc Surg.* 2007;33(2):223-33.
4 Cdc.gov. (2019). Adult Obesity Facts Overweight & Obesity CDC. https://www.cdc.gov/obesity/data/adult.html. Accessed January 15, 2019.
5 Sandhu RK, Ezekowitz J, Andersson U, et al. *Eur Heart J.* 2016;37(38):2869-78.
6 Connolly SJ, Ezekowitz MD, Yusuf S, et al. *N Engl J Med.* 2009;361(12):1139-51.
7 Boriani G, Laroche C, Diemberger et al. *J Cardiovasc Electrophysiol.* 2018;29(4):566-72.
8 Goldhaber SZ, Bassand JP, Accetta G et al. I *European Society of Cardiology Congress.* Aug 26, 2017.
9 Patel MR, Mahaffey KW, Garg J, et al. *N Engl J Med.* 2011;365(10):883-91.
10 Granger CB, Alexander JH, McMurray JJ, et al. *N Engl J Med.* 2011;365(11):981-92.
11 Proietti M, Guiducci E, Cheli P, Lip GY. *Stroke.* 2017;48(4):857-66.
12 Peterson ED, Ashton V, Chen YW, et al. *Circulation.* 2018;138(Suppl 1):A16245.
13 Thigpen JL, Dillon C, Forster KB, et al. *Circ Cardiovasc Qual Outcomes.* 2015;8(1):8-14.
14 Cunningham A, Stein CM, Chung CP, et al. *Pharmacoepidemiol Drug Saf.* 2011;20(6):560-6.
15 Broder MS, Neary MP, Chang E, et al. *Pituitary.* 2014;17(4):333-41.

Disclosure Statement

Funding for this research was provided by Bristol-Myers Squibb Company and Pfizer Inc.

S. Deitelzweig and G.Y.H. Lip are paid consultants to Bristol-Myers Squibb Company and Pfizer Inc.

A. Kang, A. Dhamane, X. Li, N. Balachander, L. Rosenblatt, X. Pan, and A.B. Garcia Reeves are employees of Bristol-Myers Squibb Company.

X. Luo, J. Mardekian, and Manuela Di Fusco are employees of Pfizer Inc.

A. Keshishian is an employee of SIMR, LLC, which is a paid consultant to Bristol-Myers Squibb Company and Pfizer Inc.

Presented at the *American College of Cardiology (ACC) 2019 Scientific Sessions*, New Orleans, LA, March 16-18, 2019