Comparative Effectiveness and Safety of Non-Vitamin K Antagonist Oral Anticoagulants among Frail Nonvalvular Atrial Fibrillation Patients

Gregory Y. H. Lip[1], Allison Keshishian[2,3], Amiee Kang[4], Amol Dhamane[4], Xuemei Luo[5], Xiaoyan Li[4], Neeraja Balachander[4], Lisa Rosenblatt[4], Jack Mardekian[6], Xianying Pan[4], Manuela Di Fusco[6], Alessandra B. Garcia Reeves[4,7], Huseyin Yuce[3], Steve Deitelzweig[8]

[1]Liverpool Centre for Cardiovascular Science, University of Liverpool and Liverpool Heart & Chest Hospital, Liverpool, United Kingdom; and Aalborg Thrombosis Research Unit, Department of Clinical Medicine, Aalborg University, Aalborg, Denmark; [2]SIMR LLC, Ann Arbor, MI, USA; [3]New York City College of Technology (CUNY), New York, NY, USA; [4]Bristol-Myers Squibb Company, Lawrenceville, NJ, USA; [5]Pfizer, Inc., Groton, CT, USA; [6]Pfizer, Inc., New York, NY, USA; [7]University of North Carolina, Chapel Hill, NC, USA; [8]Ochsner Clinic Foundation, Department of Hospital Medicine, New Orleans, LA, and The University of Queensland School of Medicine, Ochsner Clinical School, New Orleans, LA, USA

INTRODUCTION

- Atrial fibrillation (AF) is the most common arrhythmia in the elderly and is a suggested risk factor of frailty in this population.[1,2]
- It is estimated that non-valvular AF (NVAF) patients have a 4-times higher odds of being classified as frail compared to non-NVAF patients.[3]
- Non-vitamin K antagonist oral anticoagulants (NOACs)—including apixaban, dabigatran, edoxaban, and rivaroxaban—have been proven to be non-inferior for stroke prevention and safety compared to warfarin in randomized controlled trials.[4]
- Frail patients are less likely to receive OACs but have a higher risk of stroke and mortality compared to non-frail patients.[5] As the elderly US population increases, it has become increasingly important to assess anticoagulant treatment among frail elderly patients.

OBJECTIVES

This subgroup analysis of the ARISTOPHANES (Anticoagulants for Reduction In STroke: Observational Pooled analysis on Health outcomes ANd Experience of patientS; ClinicalTrials.gov ID: NCT03087487) study used multiple data sources to compare stroke/systemic embolism (SE) and major bleeding (MB) risk among frail elderly patients (aged ≥65 years) with NVAF who were prescribed NOACs.

METHODS

Data Source

- This was a retrospective observational study using the US Centers for Medicare and Medicaid Services database and three US commercial databases: the commercial part of the IQVIA (formerly IMS) PharMetrics Plus™ Database ("PharMetrics"), Optum Clinformatics™ Data Mart ("Optum"), and Humana Research Database ("Humana"), covering >123 million beneficiaries annually (~38% of the US population).

Study Sample

- Among patients included in the ARISTOPHANES study, AF patients (International Classification of Diseases, 9th Revision, Clinical Modification [ICD-9-CM] code 427.31) classified as frail and aged ≥ 65 years with ≥1 pharmacy claim for apixaban, dabigatran, or rivaroxaban from 01JAN2013-30SEP2015 were included in the study. Edoxaban was not included in the study given the small sample size.
- The first NOAC prescription date was designated as the index date for patients with NOAC claim(s)
- Inclusion and exclusion criteria are shown in Figure 1.
- The follow-up period ranged from the day after the index date to 30 days after the discontinuation date, switch date, death (only inpatient death for the commercial populations and all-cause death for the Medicare population), end of continuous medical or pharmacy plan enrollment, or end of the study period (30SEP2015), whichever occurred first.
- Frailty was defined using the algorithm defined by Segal et al. In the Segal et al study, 44 conditions were considered and evaluated during a 6-month baseline period prior to their classification. Twenty-one variables were selected in the model; with a predicted probability cutoff of 0.20 to classify individuals as frail, the sensitivity was 35% and the specificity was 91%.[6] This algorithm and cutoff have been used in previous real-word studies to identify frail patients with AF who were treated with NOACs.[7]

Figure 1. Patient Selection Criteria

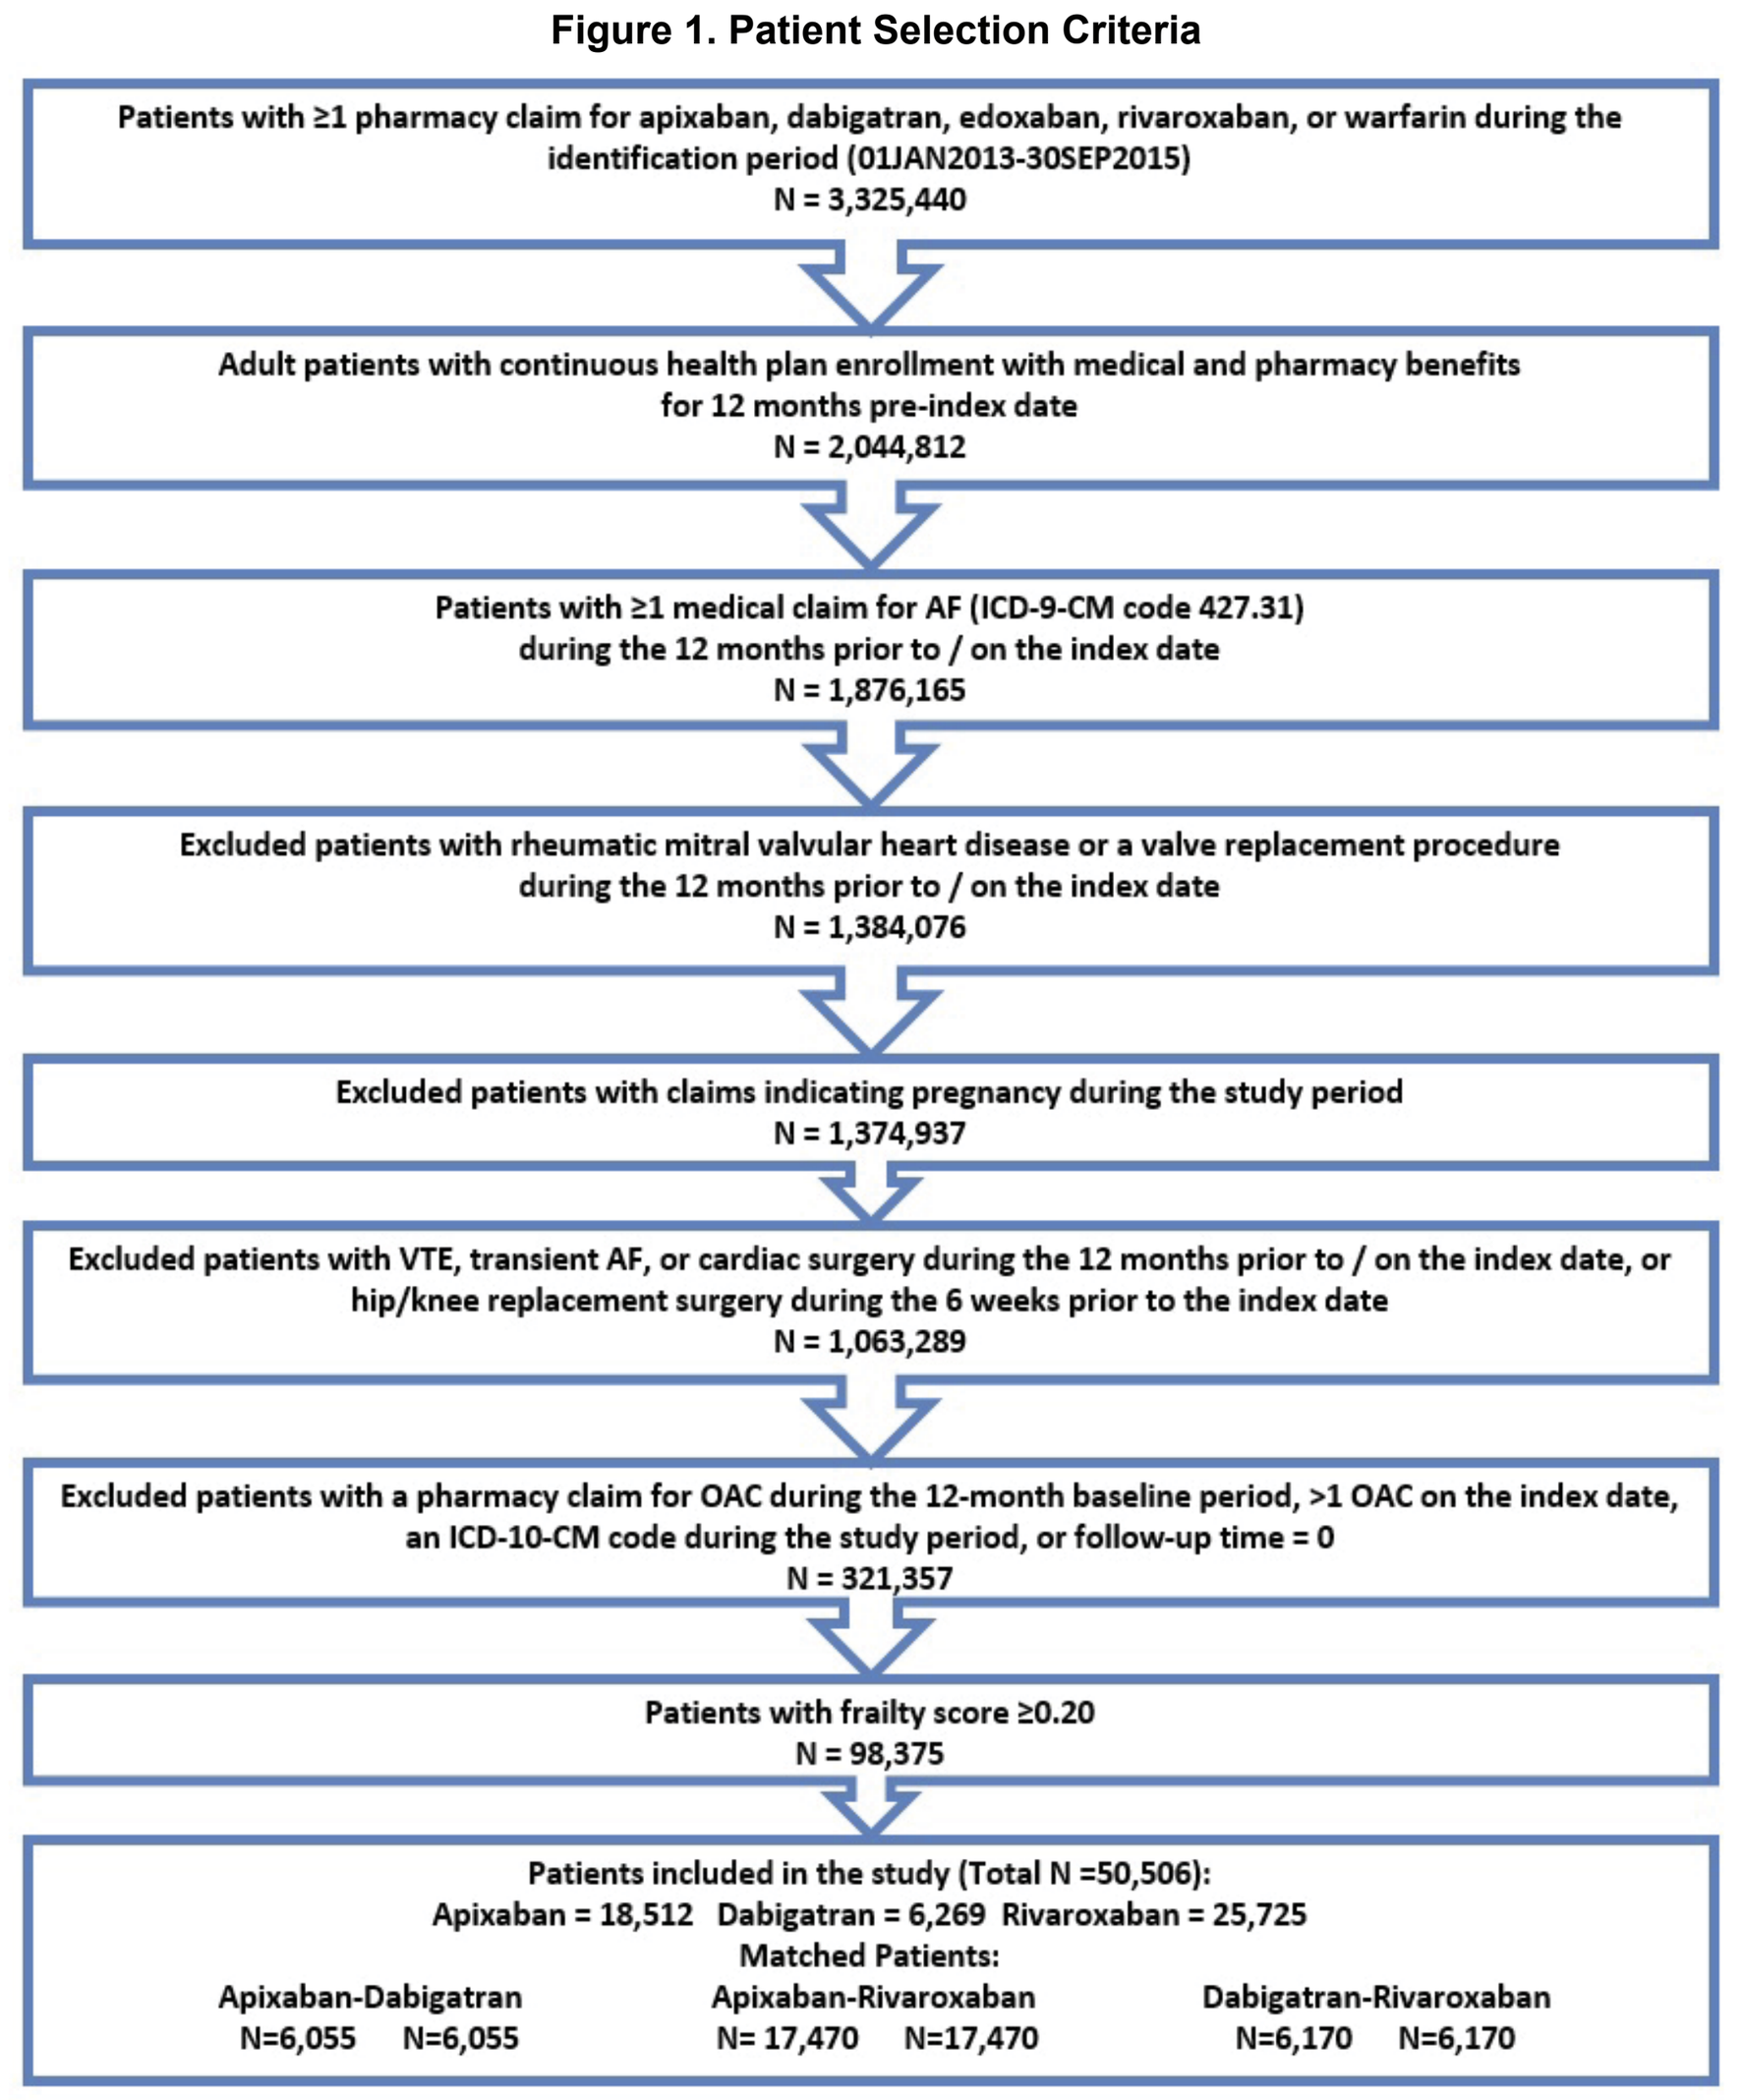

Patients with ≥1 pharmacy claim for apixaban, dabigatran, edoxaban, rivaroxaban, or warfarin during the identification period (01JAN2013-30SEP2015)
N = 3,325,440
Adult patients with continuous health plan enrollment with medical and pharmacy benefits for 12 months pre-index date
N = 2,044,812
Patients with ≥1 medical claim for AF (ICD-9-CM code 427.31) during the 12 months prior to / on the index date
N = 1,876,165
Excluded patients with rheumatic mitral valvular heart disease or a valve replacement procedure during the 12 months prior to / on the index date
N = 1,384,076
Excluded patients with claims indicating pregnancy during the study period
N = 1,374,937
Excluded patients with VTE, transient AF, or cardiac surgery during the 12 months prior to / on the index date, or hip/knee replacement surgery during the 6 weeks prior to the index date
N = 1,063,289
Excluded patients with a pharmacy claim for OAC during the 12-month baseline period, >1 OAC on the index date, an ICD-10-CM code during the study period, or follow-up time = 0
N = 321,357
Patients with frailty score ≥0.20
N = 98,375
Patients included in the study (Total N =50,506):
Apixaban = 18,512 Dabigatran = 6,269 Rivaroxaban = 25,725
Matched Patients:
Apixaban-Dabigatran N=6,055 N=6,055
Apixaban-Rivaroxaban N= 17,470 N=17,470
Dabigatran-Rivaroxaban N=6,170 N=6,170

AF: atrial fibrillation; ICD-9 International Classification of Diseases, 9th Revision, Clinical Modification; OAC: oral anticoagulant; VTE: venous thromboembolism

Stroke and MB Definitions

- Outcome measures were time to first stroke/SE and MB events based on the principal or first-listed ICD-9-CM diagnosis on an inpatient claim. The diagnosis codes used for stroke/SE and MB were based on validated administrative claims-based algorithms and the International Society on Thrombosis and Haemostasis' definition of MB.[8,9]
- Stroke/SE included ischemic stroke, hemorrhagic stroke, and SE; MB included gastrointestinal (GI) bleeding, intracranial hemorrhage (ICH), and other MB.

Statistical Analysis

- One-to-one propensity score matching (PSM) was used to balance age, sex, US geographic region, Charlson comorbidity index (CCI) score, prior bleeding, prior stroke, baseline comorbidities, and baseline medication use between patients prescribed NOACs (apixaban vs dabigatran, apixaban vs rivaroxaban, dabigatran vs rivaroxaban). The nearest neighbor matching method without replacement with a caliper of 0.01 was used. The balance of patient characteristics was checked based on standardized differences with a threshold of 10%.[10]

METHODS (cont'd)

- After PSM in each database, the resulting patient records were pooled.
- Incidence of stroke/SE and MB in the PSM-adjusted cohorts were calculated as the number of stroke/SE and MB events, respectively, per 100 person-years. P-values <0.05 were considered significant.
- Cox proportional hazards models with robust sandwich estimates were used to compare the hazard ratio of stroke/SE and MB in each of the three matched cohorts.

RESULTS

- Among NVAF patients aged ≥65 years (N=146,310), 35% met frailty criteria. A total of 50,506 frail NOAC patients with NVAF were identified, including 18,512 (36.6%) apixaban, 6,269 (12.4%) dabigatran, 25,725 (50.9%) rivaroxaban.
- The mean frail indicator score was 0.4.
- Before PSM, >90% of patients had CHA_2DS_2-VASc scores ≥4, and >80% had HAS-BLED scores ≥3.
- After PSM, 6,055 apixaban-dabigatran, 17,470 apixaban-rivaroxaban, and 6,170 dabigatran-rivaroxaban pairs were included in the analysis (Table 1). The mean follow-up time was 7-8 months.
- After PSM, all baseline characteristics were balanced between the matched cohorts.
- Approximately 50% of apixaban, 39% of dabigatran, and 44% of rivaroxaban (10 or 15mg) patients were on a lower dose (2.5mg, 75mg, and 10 or 15mg, respectively).

Table 1. Baseline Characteristics for the PSM NOAC-NOAC Comparison Cohorts

	Apixaban (N=6,055)	Dabigatran (N=6,055)	Apixaban (N=17,470)	Rivaroxaban (N=17,470)	Dabigatran (N=6,170)	Rivaroxaban (N=6,170)
	Mean/%	Mean/%	Mean/%	Mean/%	Mean/%	Mean/%
Age	83.9	83.5	83.3	84.1	83.4	83.6
65-74	8.2%	8.3%	7.1%	7.4%	8.6%	8.7%
75-79	17.0%	16.9%	14.6%	14.1%	17.4%	16.6%
≥80	74.8%	74.8%	78.3%	78.5%	74.0%	74.7%
Sex						
Male	35.7%	35.4%	35.6%	35.2%	35.8%	35.0%
Female	64.3%	64.6%	64.4%	64.8%	64.2%	65.0%
Baseline Comorbidity						
Frailty score	0.38	0.38	0.39	0.39	0.37	0.38
CCI	4.2	4.3	4.4	4.4	4.3	4.2
CHA_2DS_2-VASc score	5.1	5.2	5.2	5.2	5.2	5.1
1	0.02%	0.02%	0.02%	-	0.02%	0.02%
2	1.1%	0.9%	0.9%	0.8%	0.9%	0.6%
3	7.4%	7.5%	7.4%	7.9%	7.5%	8.2%
4+	91.5%	91.6%	91.7%	91.4%	91.5%	91.2%
HAS-BLED score	3.6	3.6	3.7	3.7	3.6	3.6
1	2.2%	2.1%	1.8%	1.7%	2.1%	2.2%
2	15.9%	17.6%	14.5%	14.8%	17.6%	16.2%
3+	81.9%	80.3%	83.8%	83.5%	80.3%	81.6%
Bleeding history	25.1%	25.2%	26.4%	26.5%	25.2%	24.8%
Congestive heart failure (CHF)	52.7%	53.2%	53.3%	53.4%	53.3%	53.1%
Diabetes mellitus	42.5%	42.6%	40.4%	40.5%	43.0%	42.4%
Hypertension	94.2%	94.0%	94.6%	94.7%	94.0%	94.0%
Renal disease	32.6%	33.1%	36.8%	36.7%	32.7%	32.4%
Liver disease	4.9%	4.9%	5.4%	5.2%	4.9%	4.9%
Myocardial infarction	11.0%	10.9%	12.1%	12.1%	10.8%	10.9%
Dyspepsia or stomach discomfort	26.6%	26.3%	27.1%	27.3%	26.3%	26.0%
Non-stroke/ SE peripheral vascular disease	66.3%	66.6%	69.1%	69.0%	66.3%	66.5%
Stroke/SE	22.7%	22.6%	23.2%	23.3%	22.5%	22.9%
Transient ischemic attack (TIA)	12.4%	12.8%	13.3%	13.2%	12.8%	13.4%
Anemia and coagulation defects	40.6%	40.6%	42.5%	42.3%	40.7%	40.9%
Alcoholism	0.4%	0.4%	0.4%	0.4%	0.6%	0.4%
Peripheral artery disease	31.9%	32.0%	33.0%	33.9%	31.8%	32.0%
Coronary artery disease	56.4%	56.6%	59.2%	58.0%	56.3%	55.4%
Baseline Components of Frailty						
Impaired mobility	1.2%	1.0%	1.1%	1.1%	1.0%	1.2%
Depression	30.4%	31.4%	29.7%	31.2%	31.7%	31.6%
Congestive heart failure	52.6%	53.2%	53.2%	53.3%	53.3%	53.0%
Parkinson disease	3.5%	3.3%	3.4%	3.7%	3.3%	3.8%
Arthritis	55.1%	53.7%	54.1%	55.2%	54.2%	56.6%
Cognitive impairment	20.9%	24.3%	21.7%	25.2%	24.2%	25.2%
Stroke	25.5%	26.1%	25.8%	25.9%	26.0%	25.4%
Paranoia	9.0%	9.0%	8.4%	9.0%	9.1%	8.8%
Chronic skin ulcer	11.9%	11.1%	11.5%	11.8%	11.0%	11.5%
Pneumonia	21.2%	21.8%	21.4%	22.4%	21.9%	21.3%
Skin and soft tissue infection	18.6%	17.4%	17.6%	18.0%	17.5%	17.6%
Mycoses	32.3%	31.1%	31.2%	32.0%	30.9%	32.0%
Hospital admission in past 6 months	51.2%	51.7%	53.4%	58.5%	51.8%	56.7%
Gout	10.6%	9.3%	10.5%	9.5%	9.4%	8.9%
Falls	11.3%	9.9%	11.4%	11.8%	10.0%	11.6%
Muscoskeletal problems	77.0%	74.6%	76.7%	76.3%	74.9%	76.4%
Urinary tract infection	34.2%	34.9%	35.0%	37.2%	34.9%	34.8%
CCI (>0.0)	96.2%	96.4%	96.7%	96.7%	96.5%	96.7%
Baseline Medication Use						
ACE/ARB	62.51%	62.49%	61.53%	61.84%	62.64%	62.51%
Amiodarone	13.29%	13.56%	15.37%	15.53%	13.40%	13.57%
Beta blockers	58.89%	59.16%	61.61%	61.84%	58.93%	59.08%
H2-receptor antagonist	9.12%	9.28%	9.34%	9.44%	9.29%	9.34%
Proton pump inhibitor	37.11%	37.32%	39.02%	38.86%	37.15%	36.69%
Statins	58.76%	58.43%	62.37%	62.40%	58.30%	59.30%
Anti-platelets	22.92%	22.79%	27.05%	27.07%	22.59%	22.25%
NSAIDS	25.15%	25.63%	24.45%	24.15%	26.09%	25.62%
Dose of the Index Prescription						
Standard dose*	51.6%	61.3%	49.9%	46.2%	61.5%	48.1%
Lower dose**	48.4%	38.7%	50.1%	44.7%	38.5%	43.0%
Mean Follow-up Time (in days)	204.2	240.0	201.1	236.9	240.1	239.2

CCI: Charlson comorbidity index; CHA_2DS_2-VASc: congestive heart failure, hypertension, aged ≥75 years, diabetes mellitus, prior stroke or transient ischemic attack or thromboembolism, vascular disease, aged 65-74 years, sex category; HAS-BLED: hypertension, abnormal renal and liver function, stroke, bleeding, labile international normalized ratios, elderly, drugs and alcohol; NOAC: Non-vitamin K oral anticoagulant; PSM: propensity score matching

* Standard dose: 5 mg BID apixaban, 150 mg BID dabigatran, 20 mg QD rivaroxaban; Lower dose: 2.5 mg BID apixaban, 75 mg BID dabigatran, 10 or 15 mg QD rivaroxaban. 2,238 (8.2%) rivaroxaban patients received 10 mg QD

** The different types of stroke/SE and major bleeding were evaluated separately.

RESULTS (cont'd)

- Patients with apixaban use were associated with a similar risk of stroke/SE compared to dabigatran (Figure 2).
- Apixaban use was associated with a lower risk of stroke/SE compared to rivaroxaban.
- Patients with apixaban use were associated with a lower risk of major bleeding compared to dabigatran and rivaroxaban. Dabigatran patients had a lower risk of major bleeding compared to rivaroxaban patients.

Figure 2. Post-PSM Incidence & Hazard Ratios of Stroke/SE and Major Bleeding

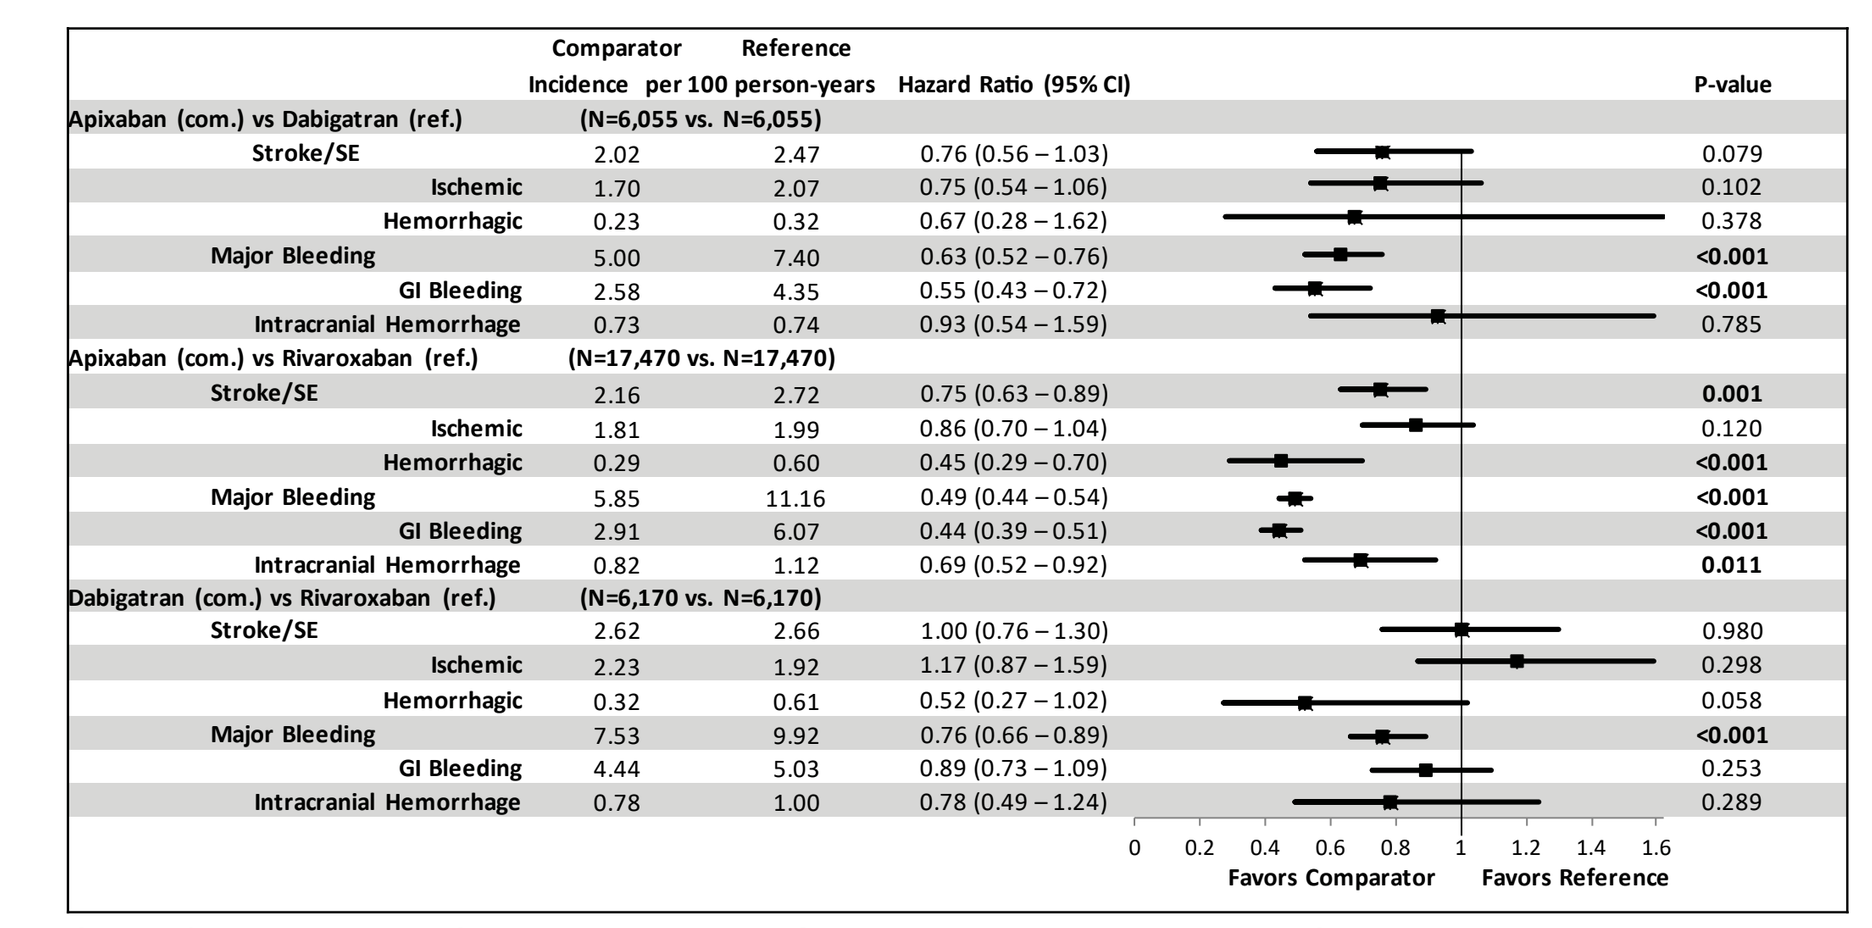

Comparator Reference
Incidence per 100 person-years Hazard Ratio (95% CI) P-value
Apixaban (com.) vs Dabigatran (ref.) (N=6,055 vs. N=6,055)
Stroke/SE 2.02 2.47 0.76 (0.56 – 1.03) 0.079
Ischemic 1.70 2.07 0.75 (0.54 – 1.06) 0.102
Hemorrhagic 0.23 0.32 0.67 (0.28 – 1.62) 0.378
Major Bleeding 5.00 7.40 0.63 (0.52 – 0.76) <0.001
GI Bleeding 2.58 4.35 0.55 (0.43 – 0.72) <0.001
Intracranial Hemorrhage 0.73 0.74 0.93 (0.54 – 1.59) 0.785
Apixaban (com.) vs Rivaroxaban (ref.) (N=17,470 vs. N=17,470)
Stroke/SE 2.16 2.72 0.75 (0.63 – 0.89) 0.001
Ischemic 1.81 1.99 0.86 (0.70 – 1.04) 0.120
Hemorrhagic 0.29 0.60 0.45 (0.29 – 0.70) <0.001
Major Bleeding 5.85 11.16 0.49 (0.44 – 0.54) <0.001
GI Bleeding 2.91 6.07 0.44 (0.39 – 0.51) <0.001
Intracranial Hemorrhage 0.82 1.12 0.69 (0.52 – 0.92) 0.011
Dabigatran (com.) vs Rivaroxaban (ref.) (N=6,170 vs. N=6,170)
Stroke/SE 2.62 2.66 1.00 (0.76 – 1.30) 0.980
Ischemic 2.23 1.92 1.17 (0.87 – 1.59) 0.298
Hemorrhagic 0.32 0.61 0.52 (0.27 – 1.02) 0.058
Major Bleeding 7.53 9.92 0.76 (0.66 – 0.89) <0.001
GI Bleeding 4.44 5.03 0.89 (0.73 – 1.09) 0.253
Intracranial Hemorrhage 0.78 1.00 0.78 (0.49 – 1.24) 0.289
0 0.2 0.4 0.6 0.8 1 1.2 1.4 1.6
Favors Comparator Favors Reference

CI: confidence interval; GI: gastrointestinal; PSM: propensity score matching; SE: systemic embolism

LIMITATIONS

- Only associations could be concluded from this retrospective observational study. Although cohorts were matched through PSM, potential residual confounders may exist.
- PSM was conducted between each NOAC cohort; thus, the results are not comparable across the matched populations.
- Laboratory results (eg, creatinine clearance, international normalized ratio) and biomarkers (eg, body weight) are not comprehensively captured in the data sources. Dose selection criteria for each NOAC cannot be ascertained.
- Some of the datasets do not include specific age data beyond a maximum age (eg, 84 or 89 years) due to privacy policies.
- To avoid potential duplicates of beneficiaries, patients with Medicare supplemental plans in PharMetrics data were not included in the study because those patients may also have Medicare Part A and Part B coverage. Based on prior publications, we expect minimal overlap (<0.5%) across the databases.
- The definition and "cutoff" for frailty used in our study was reported to provide a sensitivity of 35% and a specificity of 91%, which indicate the possibility of misclassification of frail patients.[6] It is likely that our percentage of frail patients included is low due to low sensitivity.
- Although the analysis added evidence to the frail elderly NVAF patient population, limited generalizability of the results to a broader population may be expected.

CONCLUSIONS

- In this large observational study regarding NOAC comparisons among frail elderly NVAF patients, compared to rivaroxaban, apixaban use was associated with a lower risk of stroke/SE and major bleeding and dabigatran use was associated with a lower risk of major bleeding. Apixaban use was also associated with a lower risk of major bleeding compared to dabigatran.
- These results may be helpful for clinicians in evaluating the risk-benefit ratio of NOACs for frail NVAF patients.
- Randomized controlled trials are needed to confirm the findings of this study.

REFERENCES

1. Fumagalli S, Tarantini F, Guarducci L, et al. Atrial fibrillation is a possible marker of frailty in hospitalized patients: results of the GIFA Study. *Aging Clin Exp Res.* 2010;22(2):129-33.
2. Udompanich S, Lip GY, Apostolakis S, Lane DA. Atrial fibrillation as a risk factor for cognitive impairment: a semi-systematic review. *QJM.* 2013;106(9):795-802.
3. Polidoro A, Stefanelli F, Ciacciarelli M, Pacelli A, Di Sanzo D, Alessandri C. Frailty in patients affected by atrial fibrillation. *Arch Gerontol Geriatr.* 2013;57(3):325-7.
4. Ruff CT, Giugliano RP, Braunwald E, et al. Comparison of the efficacy and safety of new oral anticoagulants with warfarin in patients with atrial fibrillation: a meta-analysis of randomised trials. *Lancet.* 2014;383(9921):955-62.
5. Perera V, Bajorek BV, Matthews S, Hilmer SN. The impact of frailty on the utilisation of antithrombotic therapy in older patients with atrial fibrillation. *Age Ageing.* 2009;38(2):156-62.
6. Segal JB, Chang HY, Du Y, Walston JD, Carlson MC, Vardhan R. Development of a claims-based frailty indicator anchored to a well-established frailty phenotype. *Med Care.* 2017;55(7):716-22.
7. Fried LP, Tangen CM, Walston J, et al. Frailty in older adults: evidence for a phenotype. *J Gerontol A Biol Sci Med Sci.* 2001;56:M146–M156.
8. Thigpen JL, Dillon C, Forster KB, et al. Validity of international classification of disease codes to identify ischemic stroke and intracranial hemorrhage among individuals with associated diagnosis of atrial fibrillation. *Circ Cardiovasc Qual Outcomes.* 2015;8(1):8-14.
9. Cunningham A, Stein CM, Chung CP, Daugherty JR, Smalley WE, Ray WA. An automated database case definition for serious bleeding related to oral anticoagulant use. *Pharmacoepidemiol Drug Saf.* 2011;20(6):560-6.
10. Austin PC. The use of propensity score methods with survival or time-to-event outcomes: reporting measures of effect similar to those used in randomized experiments. *Stat Med.* 2014;33(7):1242-58.

DISCLOSURES

Funding for this research was provided by Bristol-Myers Squibb Company and Pfizer Inc.
G.Y.H. Lip and S. Deitelzweig are paid consultants to Bristol-Myers Squibb Company and Pfizer Inc.
A. Kang, A. Dhamane, X. Li, N. Balachander, L. Rosenblatt, X. Pan, and A.B. Garcia Reeves are employees of Bristol-Myers Squibb Company.
X. Luo, J. Mardekian, and Manuela Di Fusco are employees of Pfizer Inc.
A. Keshishian is an employee of SIMR LLC, which is a paid consultant to Bristol-Myers Squibb Company and Pfizer Inc.

Presented at the *American College of Cardiology (ACC) 2019 Scientific Sessions*, New Orleans, LA, March 16-18, 2019