Disease Burden, Treatment Patterns, and Outcomes of Hospitalized Patients with Hepatic Encephalopathy

Xingyue Huang[1], Sara Hovland[1], Jan Stange[1], Shivani Pandya[2]
[1]Mallinckrodt Pharmaceuticals, Bedminster, NJ, USA; [2]STATinMED Research, Plano, TX, USA

BACKGROUND

- Hepatic encephalopathy (HE) describes a spectrum of potentially reversible neuropsychiatric abnormalities seen in patients with liver dysfunction and/or portosystemic shunting.[1]
- Overt hepatic encephalopathy (OHE) develops in 30 to 45 percent of patients with cirrhosis and in 10 to 50 percent of patients with transjugular intrahepatic portal-systemic shunts.[2]
- Most pharmacological treatment options are designed to lower blood ammonia concentrations (e.g., nonabsorbable disaccharides like lactulose)[3,4] or reduce proinflammatory molecules from the gut. The antibiotic rifaximin in combination with lactulose is also efficacious for the secondary prevention of HE when lactulose alone has failed.[5]
- Due to the relatively high amount of non-responders and because of the downsides of chronic antibiotic intake, primary prevention of HE represents still substantial unmet need.[3,5]
- This study descriptively evaluated the demographics, treatment patterns, hospital characteristics, and clinical and economic outcomes of patients with HE during hospitalization and after discharge.

OBJECTIVES

- Examine treatment patterns among hospitalized HE patients as well as the clinical (mortality rate, discharge disposition, and re-admission rates) and economic (health care resource utilization [HCRU] and costs) burden of hospitalized HE patients during their hospitalization and 30, 60, 90, and 180 days post discharge.

METHODS

Data Source & Study Design

- This is a retrospective study using nThrive's Health System Data (HSD) from 01NOV2013-31OCT2018.
- nThrive's HSD includes inpatient and hospital-based outpatient data from >400 hospitals across 42 states (59% South, 17% West, 13% Midwest, 12% Northeast), including urban (87%) and rural (13%) locations. Both inpatient and outpatient data are submitted by 98% of providers.
- In addition to standard patient demographic data (including age, gender, three-digit ZIP code, admit source, admit type, payer plan, and discharge status), nThrive's HSD includes:
 - chargemaster information (including detailed billing, service date, quantity, and charges) and payment information (including expected payment and allowed amounts); and
 - diagnostic information such as clinical groupings, International Classification of Diseases (ICD) diagnosis codes and identification of comorbidities, procedure codes (ICD, Current Procedural Terminology [CPT]-4, and Healthcare Common Procedure Coding System [HCPCS] codes) along with their corresponding dates.

Patient Selection

Patients were included in the study if they:

- were adults hospitalized with an HE diagnosis during the identification period (01FEB2014-30APR2018) as defined below—the first inpatient stay with an HE diagnosis was defined as the index hospitalization:
 1. HE diagnosis in primary position during the identification period; OR
 2. HE diagnosis in the secondary diagnosis position during the identification period with primary diagnosis for liver cirrhosis, renal failure, acute renal injury, infection, gastrointestinal bleeding, or ascites;
- had a liver cirrhosis diagnosis within the 90 days prior to or during the index hospitalization;
- had no evidence of incomplete data or missing information for demographic characteristics; and
- had no evidence of hospitalization for HE, in any diagnostic position, anytime within a 90-day period prior to the index hospitalization.

Study Variables

Index hospitalization:

- Demographic characteristics: age, sex, US geographical region, health plan type
- Hospital characteristics: hospital setting, teaching status, bed size
- Clinical characteristics: Quan Charlson comorbidity index (CCI) score, individual comorbidities
- Treatments medications and procedures

Study Variables (cont'd)

- HCRU and clinical and economic outcomes: during index hospital stay, including
 - inpatient length of stay (LOS);
 - total hospitalization costs and charges;
 - hospital mortality rate; and
 - discharge disposition

Follow-up post-discharge date of index hospitalization:

- Re-admission rate
- Economic outcomes:
 - Costs & Charges: inpatient, outpatient, total medical costs, and total medical charges.

Statistical Analysis

- All study variables were evaluated descriptively.
- Standard summary statistics were provided to descriptively examine all variables. Means and standard deviations (SDs) were provided for continuous variables. Numbers and percentages were provided for categorical variables.

RESULTS

Study Population

After applying the study inclusion/exclusion criteria, 24,093 hospitalized patients with HE were identified.

Table 1. Demographic and hospital characteristics of hospitalized HE patients

Baseline Patient and Hospital Characteristics	Hospitalized HE Patients N/Mean	Hospitalized HE Patients %/SD
Patient Characteristics		
Age	59.08	11.19
18-45	2428	10.08%
45-64	14644	60.78%
≥65	7021	29.14%
Sex		
Male	14868	61.71%
Female	9225	38.29%
US Geographic Region		
Midwest	1508	6.26%
Northeast	2779	11.53%
South	12949	53.75%
West	6841	28.39%
Unknown	16	0.07%
Insurance Type		
Commercial	3840	15.94%
Medicaid	5354	22.22%
Medicare	5199	21.58%
Other	1053	4.37%
Unknown	4949	20.54%
Multiple	3698	15.35%
Hospital Characteristics		
Hospital Setting		
Urban	23474	97.43%
Rural	603	2.50%
Unknown	16	0.07%
Teaching status		
Major teaching	7571	31.42%
Minor teaching	4958	20.58%
Non-teaching	11548	47.93%
Unknown	16	0.07%
Bed Size		
≤100 beds	1667	6.92%
101-199 beds	4780	19.84%
200-299 beds	3089	12.82%
300-499 beds	7670	31.83%
≥500 beds	6871	28.52%
Unknown	16	0.07%

HE: hepatic encephalopathy; SD: standard deviation

Demographic and Hospital Characteristics (Table 1)

- The majority of HE patients were males (61.71%) aged 46-64 years (60.78%; mean: 59.08 years).
- Medicaid and Medicare were the most common observed insurance types (22.22% and 21.58%, respectively).
- The majority of patients were admitted to urban medical centers (97.43%); non-teaching facilities were most common (47.93%), followed by major teaching facilities (31.42%). The facilities tended to be larger with 399-499 beds (31.83%) or ≥500 beds (28.52%).

Clinical Characteristics, Treatments, HCRU and Costs During Index Hospitalization

- Observed Quan-CCI scores averaged 5.43; the most common individual comorbidities were cardiovascular disease (CVD) (79.78%), electrolyte imbalance (60.98%), and acute/chronic kidney disorder ([AKD/CKD] 50.69%) (Figure 1).
- The mean LOS for the index hospitalization was 8.10 days (Figure 2), and the mortality rate was 11.41%.
- Nearly two-thirds (63.65%) patients were discharged to home/self-care, 23.58% were transferred to another facility, and the remaining patients were known to have left against medical advice, expected to return for outpatient services or transferred to inpatient setting.
- The most commonly used medications during index hospitalization included lactulose only (45.45%) followed by lactulose + rifaximin (30.03%) and metronidazole (10.13%) (Table 2).
- The most common procedures performed during index hospitalization included splenic artery embolization (2.44%) followed by liver transplantation (1.38%) and transjugular intrahepatic portosystemic shunt ([TIPS]; 1.32%) (Table 2).
- Mean index hospitalization costs were $18,297 (~$2,180 per day); mean total charges were $79,439 (~$9,538 per day).

Figure 1. Distribution of individual comorbidities observed during the index hospitalization among HE patients

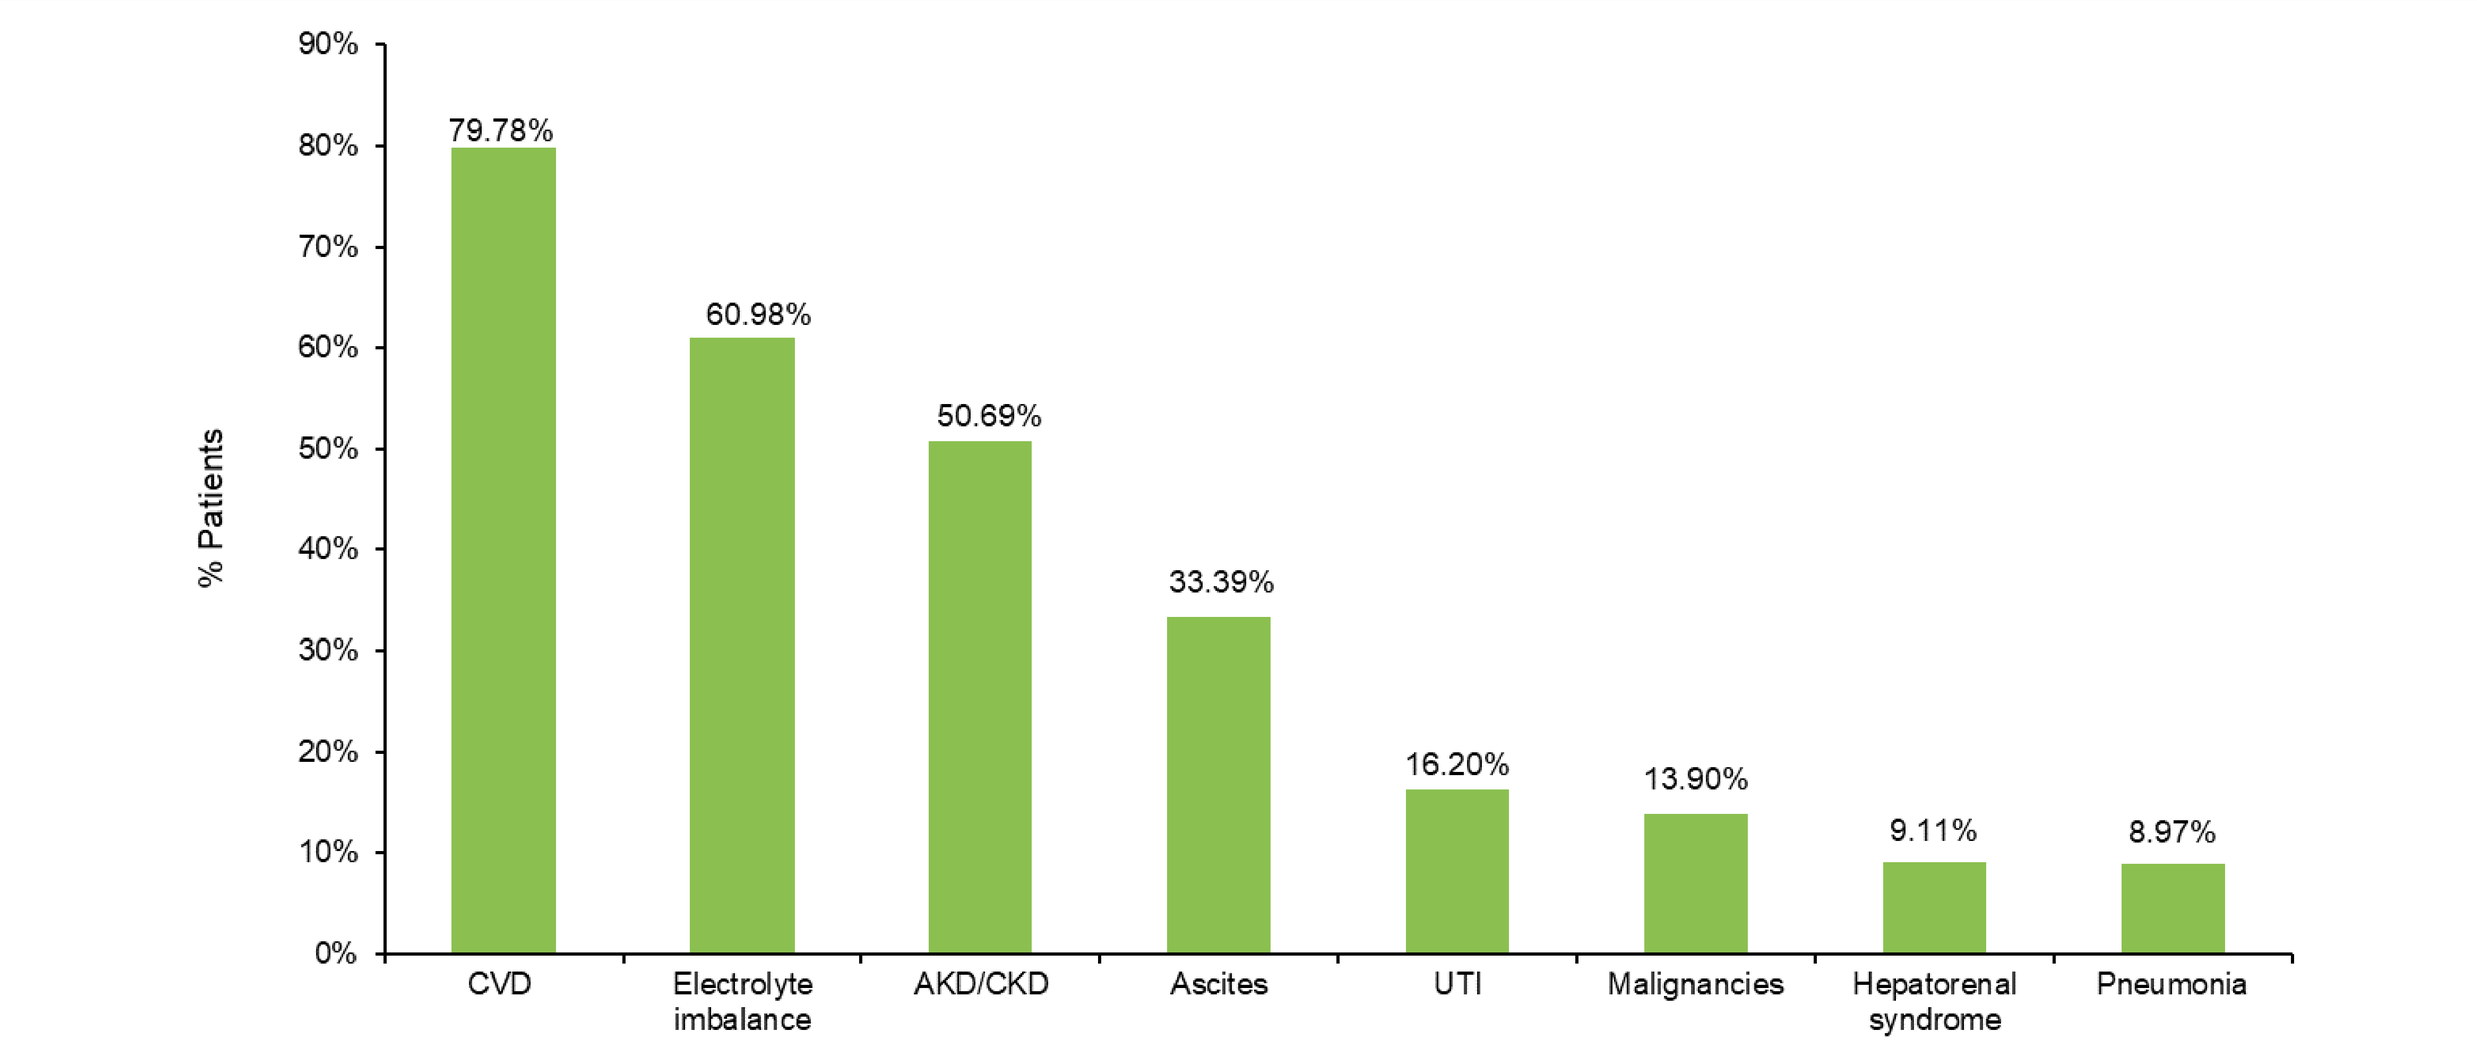
% Patients
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
79.78%
60.98%
50.69%
33.39%
16.20%
13.90%
9.11%
8.97%
CVD
Electrolyte imbalance
AKD/CKD
Ascites
UTI
Malignancies
Hepatorenal syndrome
Pneumonia

AKD/CKD: acute/chronic kidney disease; CVD: cardiovascular disease; HE: hepatic encephalopathy; UTI: urinary tract infection

Figure 2. Distribution of index hospitalization length of stay among HE patients

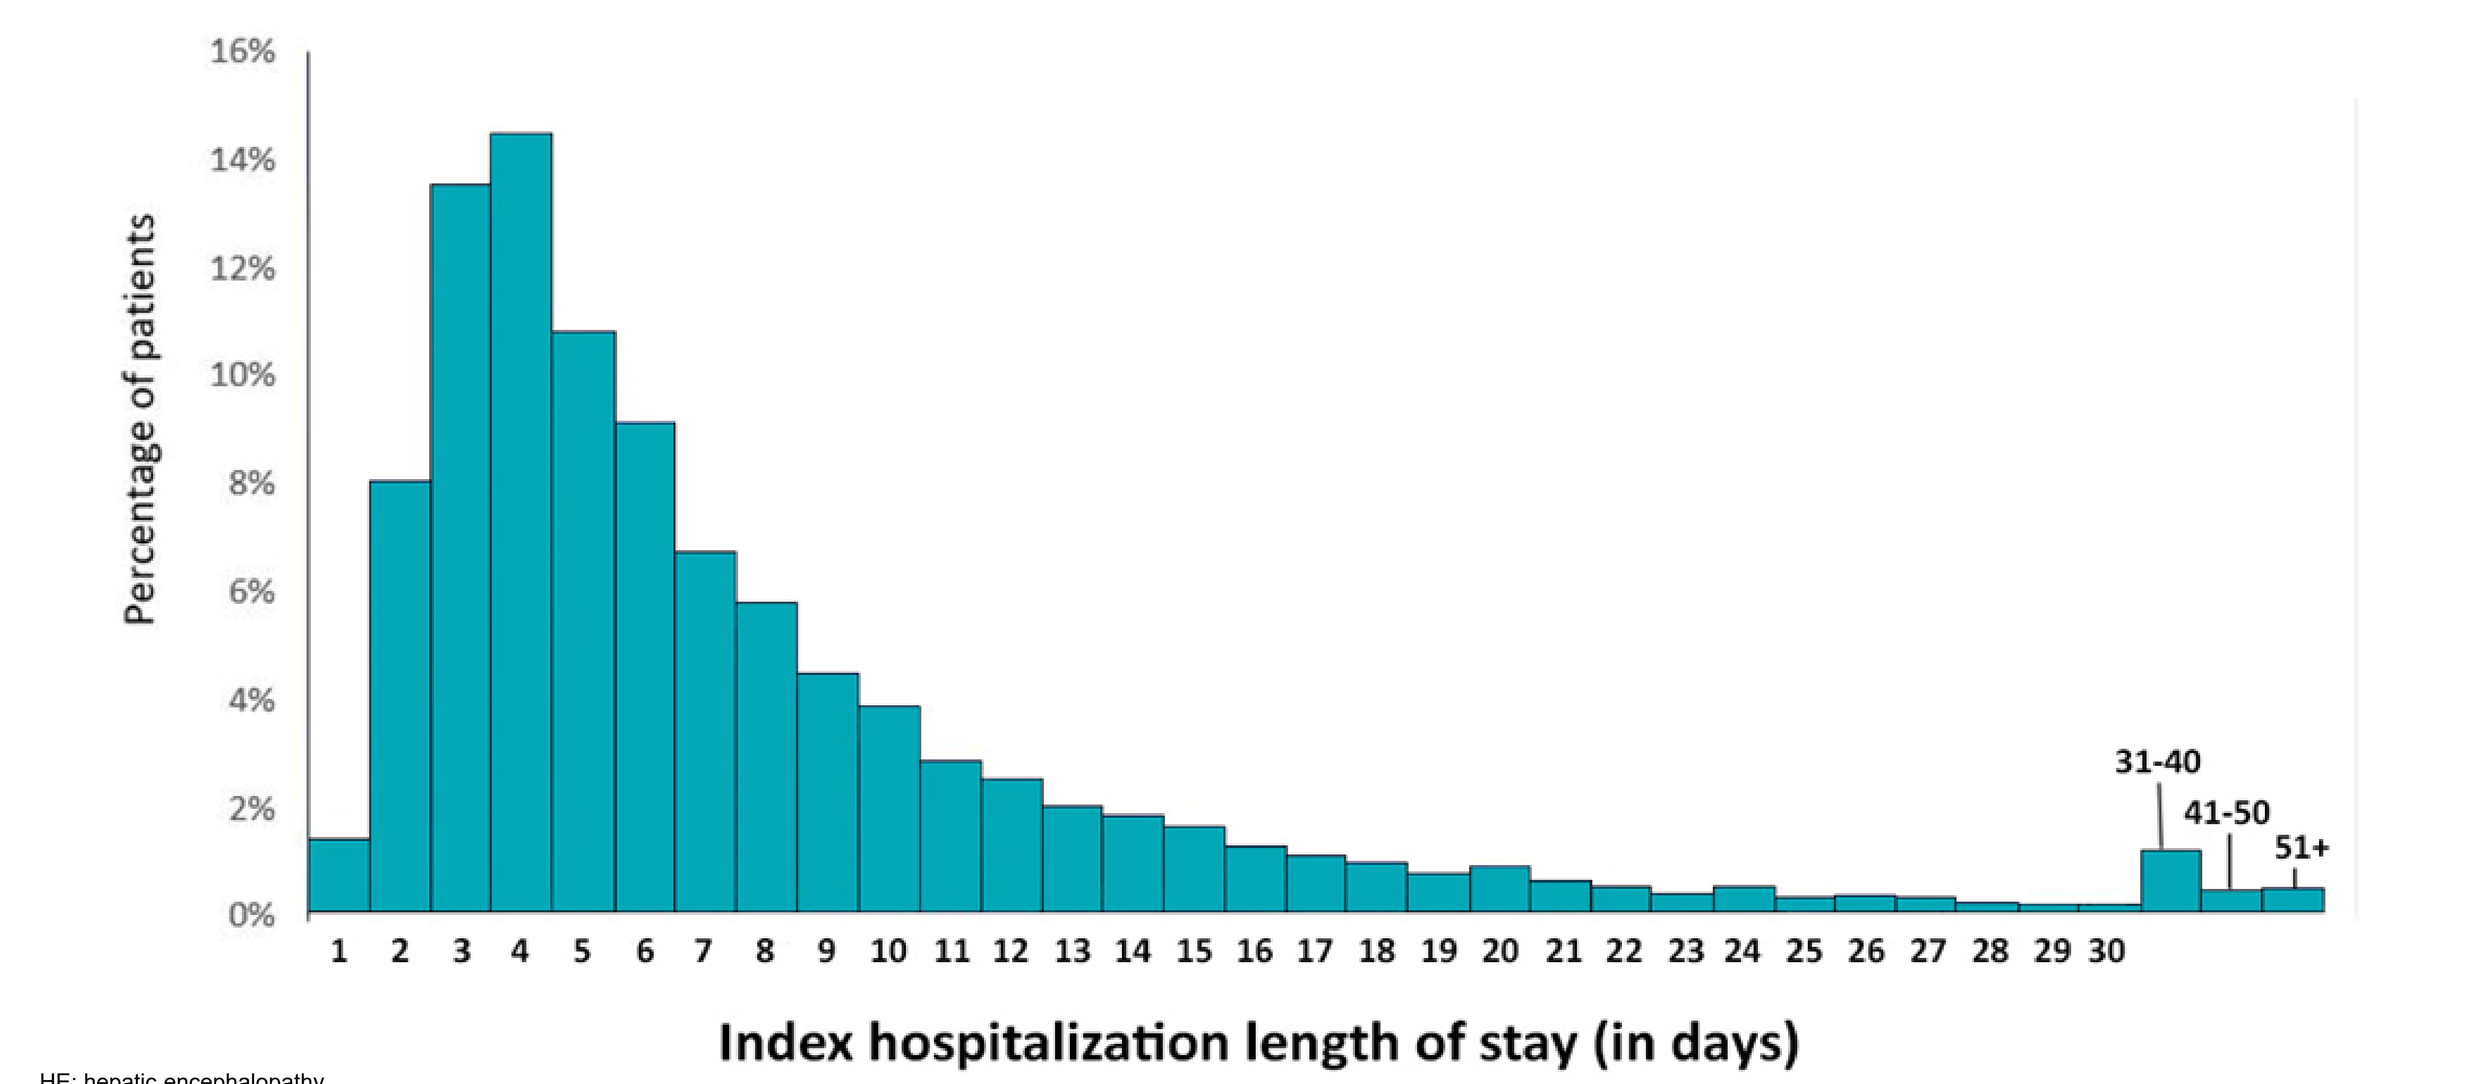
Percentage of patients
16%
14%
12%
10%
8%
6%
4%
2%
0%
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24 25 26 27 28 29 30
31-40
41-50
51+
Index hospitalization length of stay (in days)

HE: hepatic encephalopathy

Table 2. Medication use and procedures performed during index hospitalization among HE patients

Treatments and Procedures During Index Hospitalization	Hospitalized HE Patients N/Mean	Hospitalized HE Patients %/SD
Treatment (among patients with ≥1 charge master description; n=2211)		
Lactulose only	1005	45.45%
Rifaximin only	47	2.13%
Lactulose + Rifaximin	664	30.03%
Neomycin	6	0.27%
Metronidazole	224	10.13%
Others	8	0.36%
Procedures		
Transjugular intrahepatic portosystemic shunt	318	1.32%
Splenic artery embolization	587	2.44%
Total colectomy	4	0.02%
Liver transplantation	332	1.38%

HE: hepatic encephalopathy; SD: standard deviation
Note: Other medications and procedures will be added to this list based on the top 5 comorbidities observed during baseline

Post-discharge Clinical and Economic Outcomes

- A consistent increase in the readmission rates was noted during the 30-, 60-, 90-, and 180-day intervals after discharge from the index hospitalization (Figure 3) with associated increases in the inpatient length of stay from 10.76 days during 30-days to 22.14 days during 180-days post-discharge.
- Similar increase was also observed in costs and charges during the post-discharge period (Figure 4).

Figure 3. Re-admission rates during the post-discharge period among HE patients

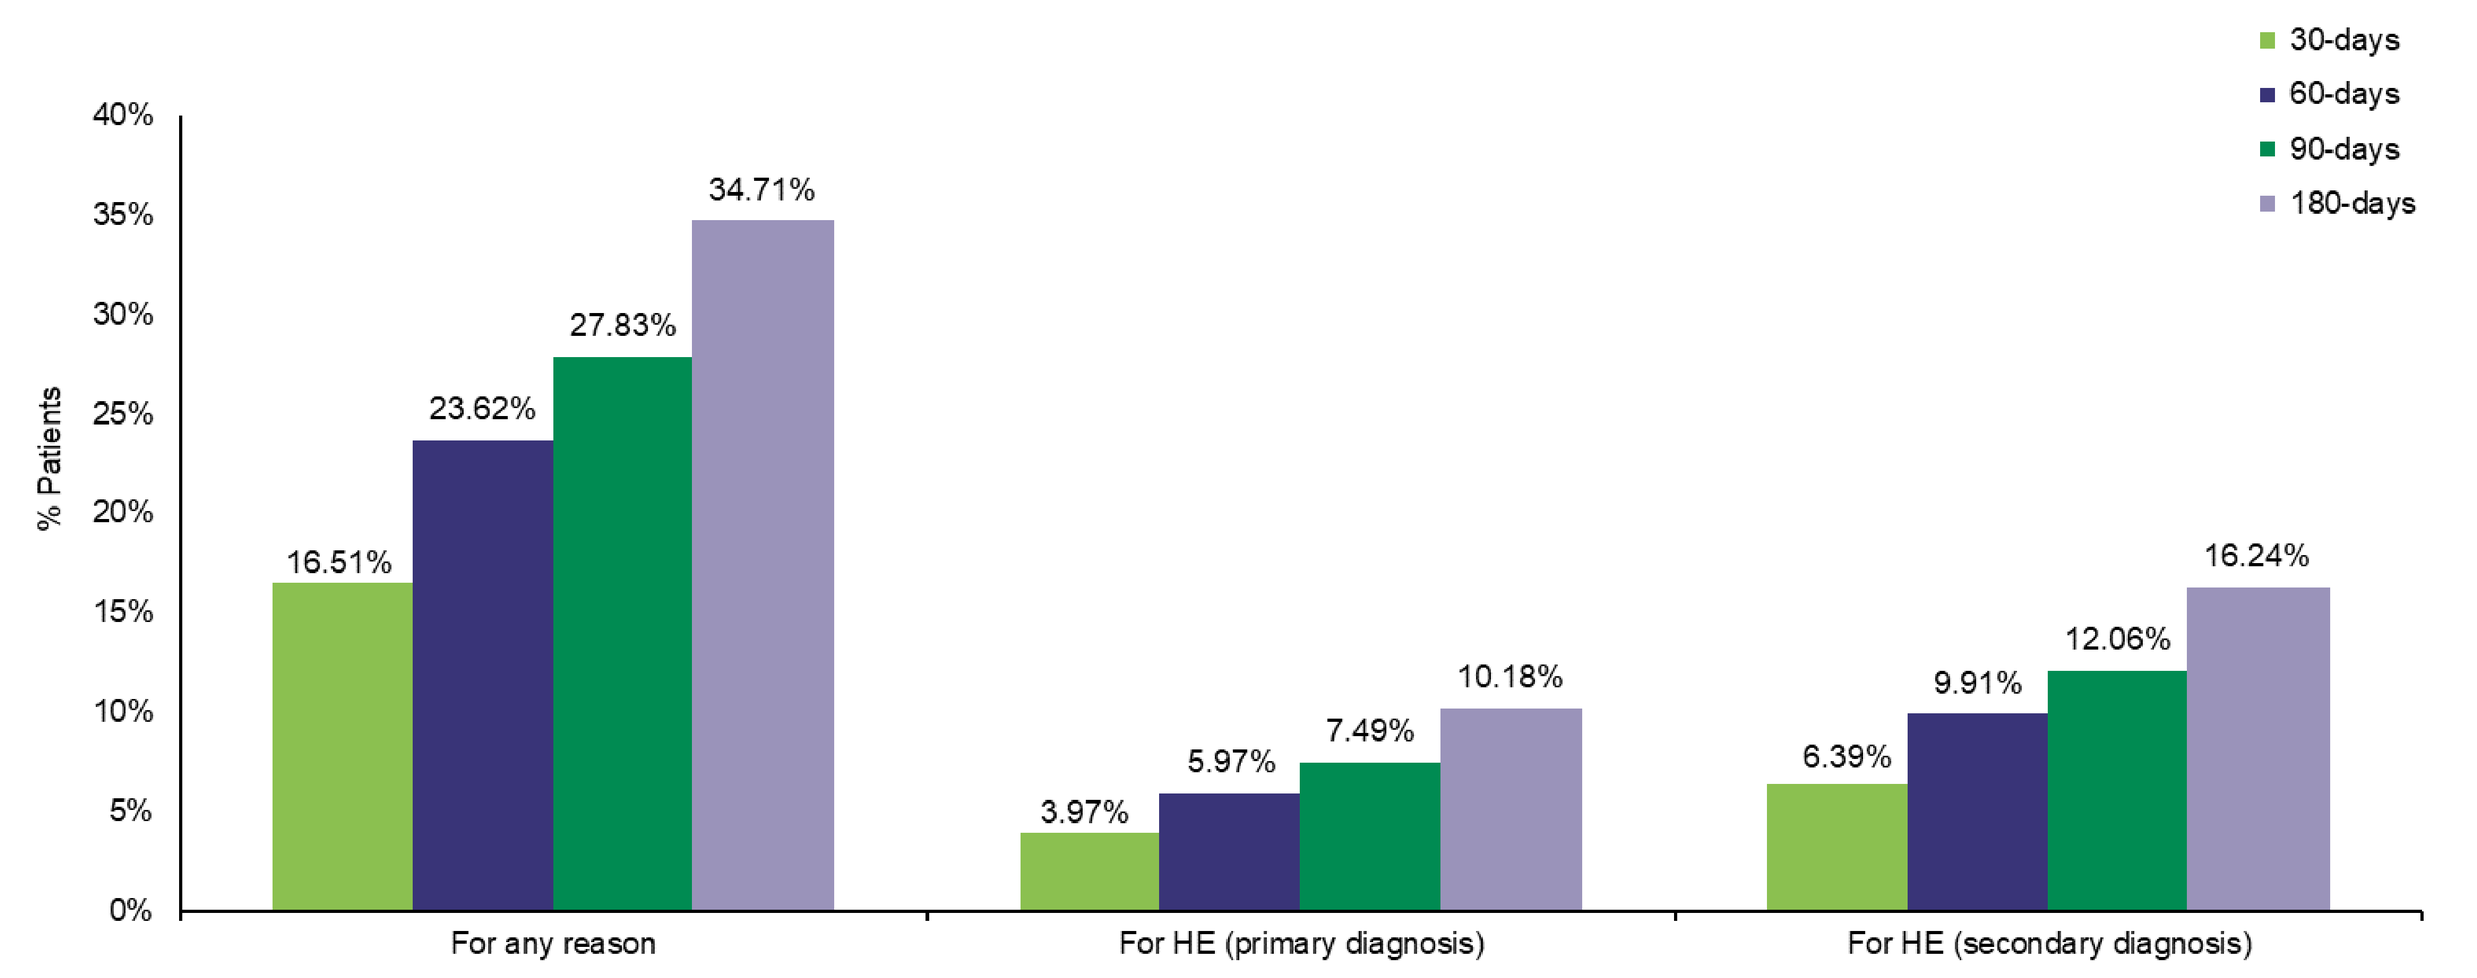
30-days
60-days
90-days
180-days
% Patients
40%
35%
30%
25%
20%
15%
10%
5%
0%
16.51%
23.62%
27.83%
34.71%
3.97%
5.97%
7.49%
10.18%
6.39%
9.91%
12.06%
16.24%
For any reason
For HE (primary diagnosis)
For HE (secondary diagnosis)

HE: hepatic encephalopathy
Note: Post-discharge readmission included only patients who survived the index hospitalization.

Figure 4. All-cause health care costs and charges during the post-discharge period among HE patients

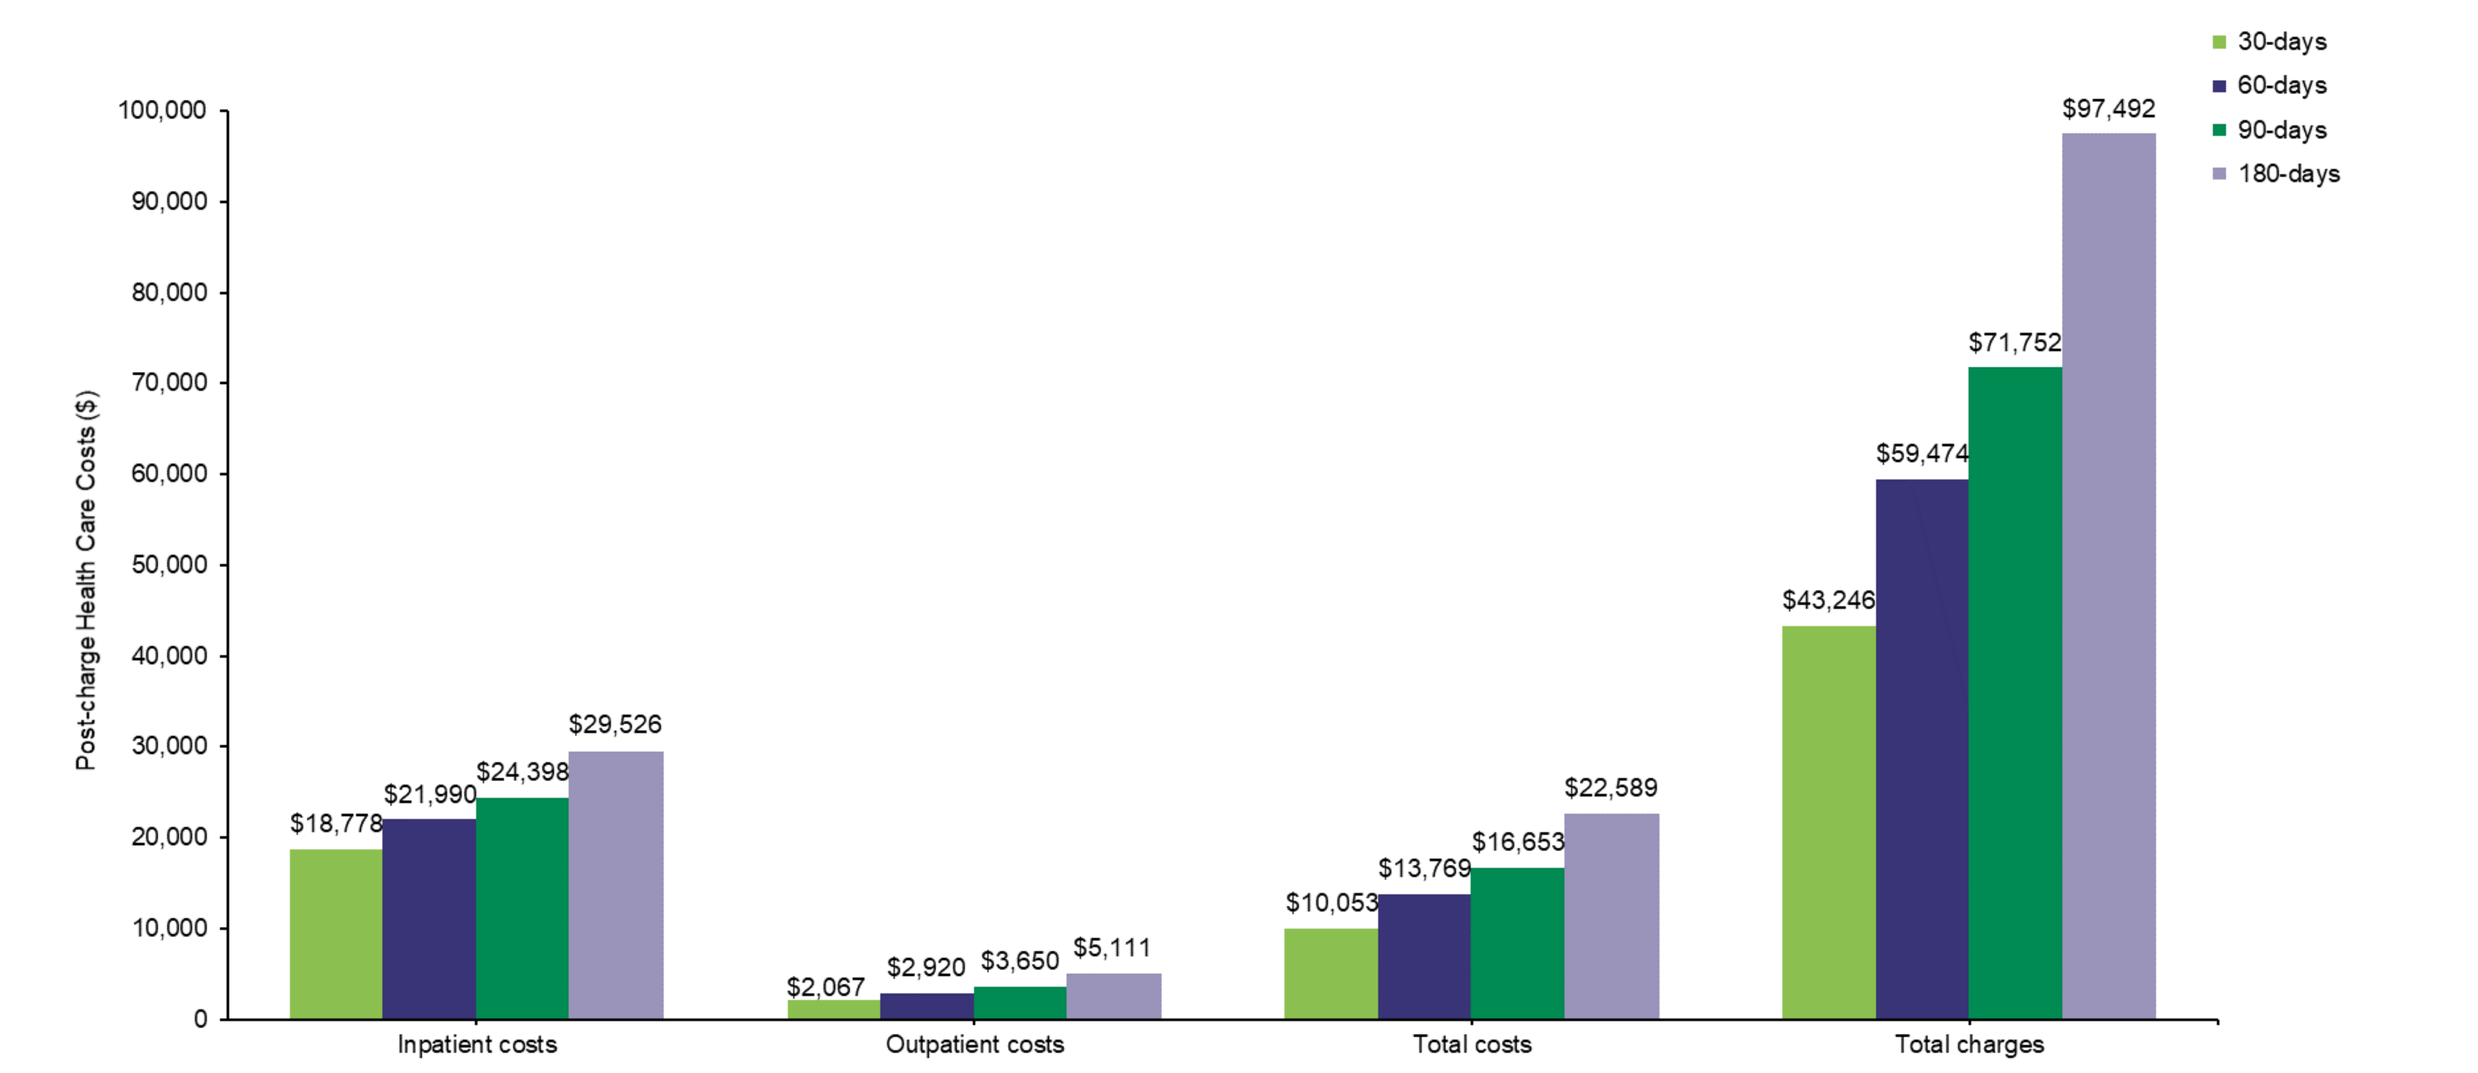
30-days
60-days
90-days
180-days
Post-charge Health Care Costs ($)
100,000
90,000
80,000
70,000
60,000
50,000
40,000
30,000
20,000
10,000
0
$18,778
$21,990
$24,398
$29,526
$2,067
$2,920
$3,650
$5,111
$10,053
$13,769
$16,653
$22,589
$43,246
$59,474
$71,752
$97,492
Inpatient costs
Outpatient costs
Total costs
Total charges

HE: Hepatic Encephalopathy
Note: Inpatient and outpatient costs were calculated only among patients with ≥1 encounter. Total medical costs are not a summation of inpatient and outpatient costs: means were calculated with different denominators, as a patient may have had inpatient and outpatient encounters on the same day.

DISCUSSION

- This study builds on the evidence reported in the previous US studies using nationally representative data.
- The observed mean length of stay and in-hospital mortality rate were in line with findings in the literature of hospitalized patients with HE which reported LOS ranging from ~8.1 to 8.5 days and a gradual reduction in in-hospital mortality rate since 2009 (14.4% in 2009, 13.4% in 2010, and 12.3% in 2014). However, hospital characteristics differed between the current study sample and previous studies, which may have played a role in the discrepancy in mortality due to overarching systemic variables that were not evaluated in any of the mentioned studies.
- The substantial economic burden noted in the present study accords with previous studies that have noted an upward trajectory over time of charges associated with hospitalization for HE, despite decreasing in-hospital mortality.
- Overall, the precedents observed in those studies are consistent with our observations. Although trending in the right direction, these data affirm there remains considerable mortality and lengthy hospitalization associated with HE.

LIMITATIONS

Analysis of hospital encounter database is subject to a number of data limitations including:

- Certain information that could influence study outcomes, such as clinical and disease-specific parameters, are not necessarily available in chargemaster data.
- Chargemaster billing descriptions are based on charges prior to negotiation with health insurance providers, actual cost-to-charge ratios vary by institution and are not available in the dataset.
- Treatments were identified based solely on the chargemaster description due to the lack of a separate pharmacy file with National Drug Codes; therefore, treatments have increased susceptibility to input errors and redundant coding.
- The dataset did not capture enough patients with ≥1 encounter in the pre-index period (<50% of the study population) to form a representative sample, which precluded baseline comorbidity analysis. Future analysis of comorbidities and associated outcomes among patients with and without pre-index encounters would be instructive.
- Potentially, post-discharge encounter visits at hospitals not affiliated with the nThrive's Health System won't be included in the HSD.

CONCLUSIONS

- This retrospective hospital data-based study reveals important insights into the current real-world state of OHE inpatient treatment, and related outcomes and utilization, which demonstrate unmet need in the management of this condition.
- Despite mono or dual therapy, OHE is associated with substantial mortality, high rates of recurrent/readmission, and associated costs from US hospital perspective.
- These results underscore a major unmet need and the importance research for more effective HE treatment options.

REFERENCES

1. Romero-Gómez M, Boza F, García-Valdecasas MS, et al. Subclinical hepatic encephalopathy predicts the development of overt hepatic encephalopathy. *Am J Gastroenterol*. 2001;96:2718.
2. Boyer TD, Haskal ZJ, American Association for the Study of Liver Diseases. The role of transjugular intrahepatic portosystemic shunt in the management of portal hypertension. *Hepatology*. 2005;41:386.
3. Stepanova M, Mishra A, Venkatesan C, Younossi ZM. In-hospital mortality and economic burden associated with hepatic encephalopathy in the United States from 2005 to 2009. *Clin Gastroenterol Hepatol*. 2012;10(9):1034-41.e1.
4. Phongsamran PV, Kim JW, Cupo Abbott J, Rosenblatt A. Pharmacotherapy for hepatic encephalopathy. *Drugs*. 2010;70(9):1131-48.
5. Hadjihambi A, Jalan R. Hepatic encephalopathy: New treatments. *Clin Liver Dis (Hoboken)*. 2015;5(5):109-11.

The International Liver Congress 2020, August 25-28, 2020: London, UK

This study was funded by Mallinckrodt Pharmaceuticals.
X Huang is a full time employee of Mallinckrodt Pharmaceuticals. At the time of the study, S Hovland and Jan Stange were employees of Mallinckrodt Pharmaceuticals and S Pandya was an employee of STATinMED Research, a paid consultant to Mallinckrodt Pharmaceuticals.

Mallinckrodt, the "M" brand mark, the Mallinckrodt Pharmaceuticals logo and other brands are trademarks of a Mallinckrodt company. © 2017 Mallinckrodt.